Concurrent degradation of BTK and IMiD neosubstrates by NX-2127 enhances multiple mechanisms of tumor killing

Mark Noviski*, Jun Ma*, Ernestine Lee*, Nivetha Brathaban*, May Tan*, Luz Perez*, Daniel W Robbins*, Austin Tenn-McClellan*, Janine Powers*, Arthur Sands*, Gwenn Hansen*, Cristiana Guiducci*, Ryan Rountree*

*Nurix Therapeutics, San Francisco, CA, USA

Permanent Abstract Number 1126

Abstract

B cell receptor (BCR) signaling is integral for the development, adhesion, growth, and survival of human B cells. Chronic activation of BTK-mediated BCR signaling is a hallmark of many B cell malignancies, making BTK an attractive therapeutic target. While covalent inhibitors of BTK have proven effective, they have been associated with acquired resistance mutations. IMiDs (immunomodulatory imide drugs) exert direct anti-cancer effects in B cell malignancies as well as enhance T cell activity. Based on clinical data of BTK covalent inhibitors and IMiDs in B cell malignancies, a strategy combining BTK and IMiD mechanisms in a single oral, small molecule may improve anti-tumor activity.

Here we describe NX-2127, a bifunctional molecule that catalyzes the degradation of BTK and IMiD neosubstrates Aiolos and Ikaros. NX-2127 degrades BTK in a range of B cell lymphoma cell lines (DC_{50}=4-13 nM), including a DLBCL line harboring the most common ibrutinib-resistant mutation found in the clinic (C481S). In primary human T cells, NX-2127 degrades Aiolos (DC_{50}=25 nM) and Ikaros (DC_{50}=54 nM) and enhances IL-2 secretion. NX-2127 promotes superior in vitro killing of a DLBCL cell line, TMD8, and a MCL cell line, REC-1, as compared to BTK inhibitors ibrutinib, acalabrutinib, and pirtobrutinib, or IMiDs pomalidomide and lenalidomide. BTK inhibitors have limited effects on viability of TMD8 cells (E_{max}=17-33% viable) and REC-1 cells (E_{max}=46-52% viable), whereas the dual BTK and IMiD activity of NX-2127 promotes complete killing of TMD8 and REC-1 cells and does so more potently than IMiDs alone. In REC-1 cells, suppression of Ikaros protein levels by IMiDs, BTK inhibitors, and NX-2127 correlates closely with downstream viability reduction, independent of drug mechanism. In vivo, oral administration of NX-2127 demonstrates dose proportional degradation of BTK and Aiolos in a TMD8 xenograft model, closely correlating with the degree of tumor control.

RNAseq analysis performed on REC-1 cells demonstrates that NX-2127 modulates not only pathways associated with each mechanism, but also shows distinct regulation of gene expression when compared to ibrutinib or pomalidomide dosed independently. Gene set enrichment analysis (GSEA) reveals that NX-2127 downregulates gene sets involved in DNA replication, DNA repair, cell cycle, and survival signaling pathways. Furthermore, a key mediator of T cell recognition, CD1c, is more strongly upregulated by NX-2127 than pomalidomide and ibrutinib at both the transcript and cell surface expression levels in REC-1 cells, implying a potential immune-mediated anti-neoplastic mechanism. The gene expression signature of NX-2127 exposure supports the hypothesis that its anti-tumor effects include both tumor cell-intrinsic and tumor cell-extrinsic mechanisms and suggests that the combination of BTK degradation and IMiD activity may achieve enhanced efficacy in certain B cell malignancies. A phase 1a/b trial of NX-2127 for patients with relapsed or refractory B-cell malignancies is ongoing.

Results

Figure 1. NX-2127 promotes degradation of BTK and BTK-C481S in B cell lymphoma lines

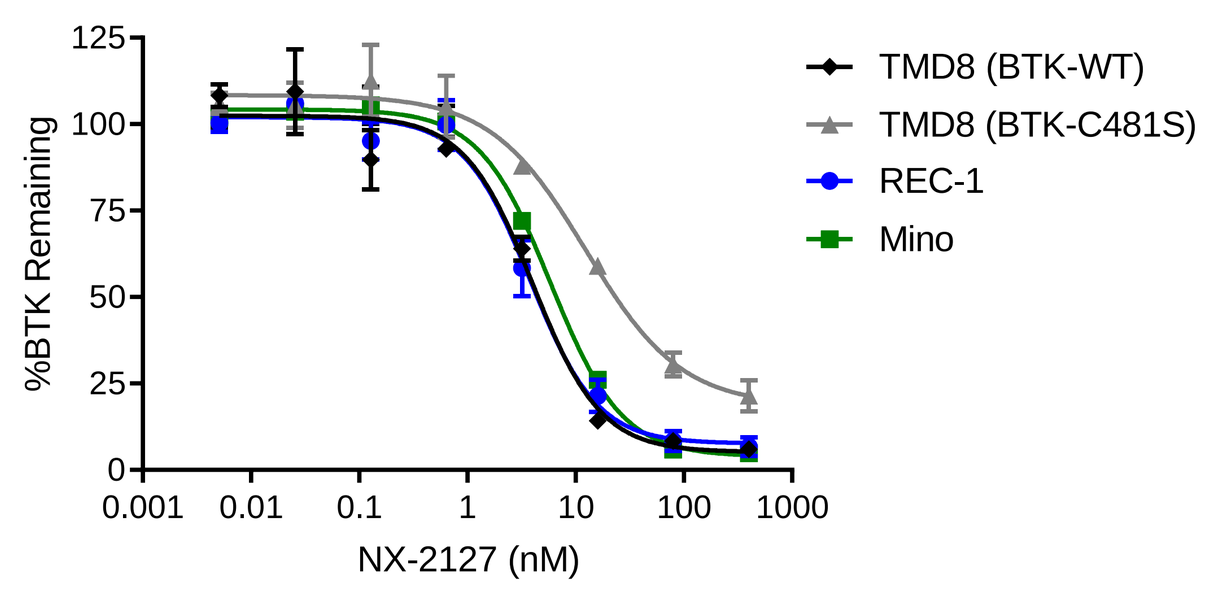
%BTK Remaining
NX-2127 (nM)
TMD8 (BTK-WT)
TMD8 (BTK-C481S)
REC-1
Mino

Cell Line	Cell Type	BTK DC_{50}
TMD8 (BTK-WT)	ABC-DLBCL	4 nM
TMD8 (BTK-C481S)	ABC-DLBCL	13 nM
REC-1 (BTK-WT)	MCL	4 nM
Mino (BTK-WT)	MCL	6 nM

TMD8, REC-1, and Mino cells were treated with NX-2127 for 4 hours *in vitro*, and BTK levels were quantified by homogenous time-resolved FRET kit (Cisbio). BTK levels were plotted as a function of NX-2127 concentration, and a four-parameter non-linear curve fit was calculated. DC_{50} values represent the concentration of NX-2127 that promotes half-maximal BTK degradation. ABC-DLBCL: activated B cell-like diffuse large B cell lymphoma. MCL: mantle cell lymphoma.

References

[1] Patro et al. 2017. Nature Methods **14:** 417-419.
[2] https://www.reneshbedre.com/blog/deseq2.html
[3] https://www.gsea-msigdb.org/gsea/msigdb/

Results (Continued)

Figure 2. NX-2127 promotes Aiolos and Ikaros degradation in primary human T cells and enhances IL-2 secretion

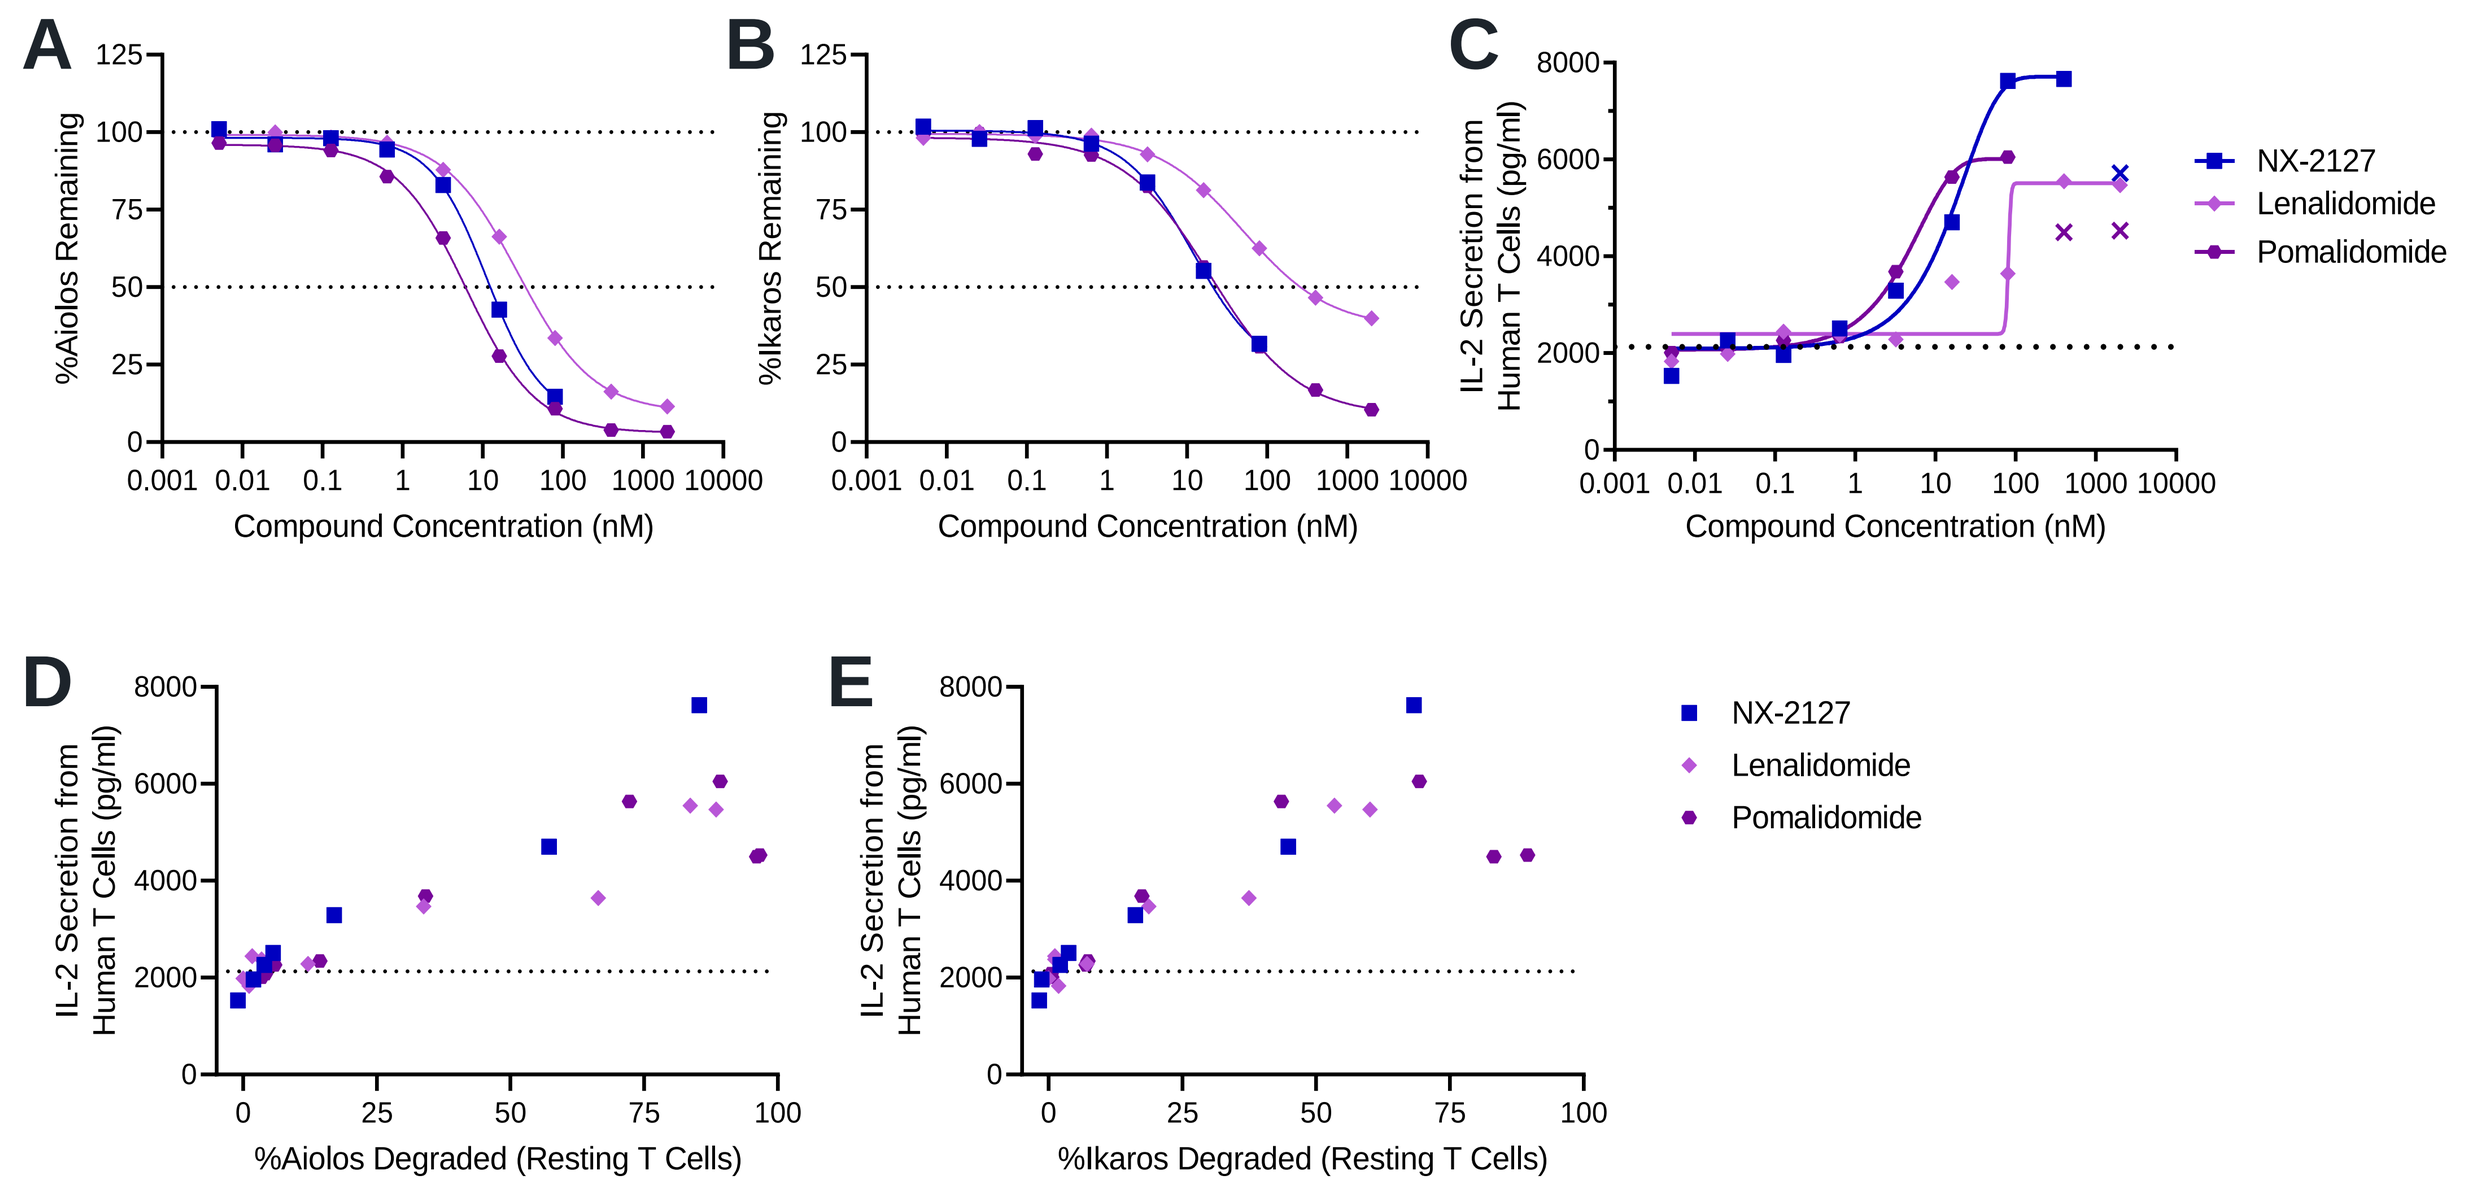
A
%Aiolos Remaining
Compound Concentration (nM)
B
%Ikaros Remaining
Compound Concentration (nM)
C
IL-2 Secretion from Human T Cells (pg/ml)
Compound Concentration (nM)
NX-2127
Lenalidomide
Pomalidomide
D
IL-2 Secretion from Human T Cells (pg/ml)
%Aiolos Degraded (Resting T Cells)
E
IL-2 Secretion from Human T Cells (pg/ml)
%Ikaros Degraded (Resting T Cells)
NX-2127
Lenalidomide
Pomalidomide

(A-B) Human peripheral blood mononuclear cells (PBMCs) were treated with compound for 24 hours, and Aiolos (A) and Ikaros (B) degradation in CD3+ T cells were quantified by intracellular flow cytometry. Background staining was quantified using isotype control antibodies, and percent Aiolos and Ikaros remaining were calculated by normalizing background-subtracted mean fluorescence intensities (MFIs) to DMSO-treated cells. The top two concentrations of NX-2127 are excluded due to compound autofluorescence. (C) T cells were purified by negative selection, treated with compound for 1 hour, and then stimulated with anti-CD3 and anti-CD28 for 24 hours in the continued presence of compound. IL-2 secretion was quantified by ELISA. (D-E) IL-2 secretion from stimulated T cells in (C) is plotted as a function of Aiolos and Ikaros degradation in resting T cells from (A-B). %Aiolos or Ikaros degraded is defined as 100 - %Aiolos or Ikaros remaining. Full degradation of Aiolos and Ikaros is not required to achieve enhanced IL-2 secretion.

Figure 3. NX-2127 promotes superior killing of TMD8 and REC-1 cells compared to BTK inhibitors and IMiD drugs

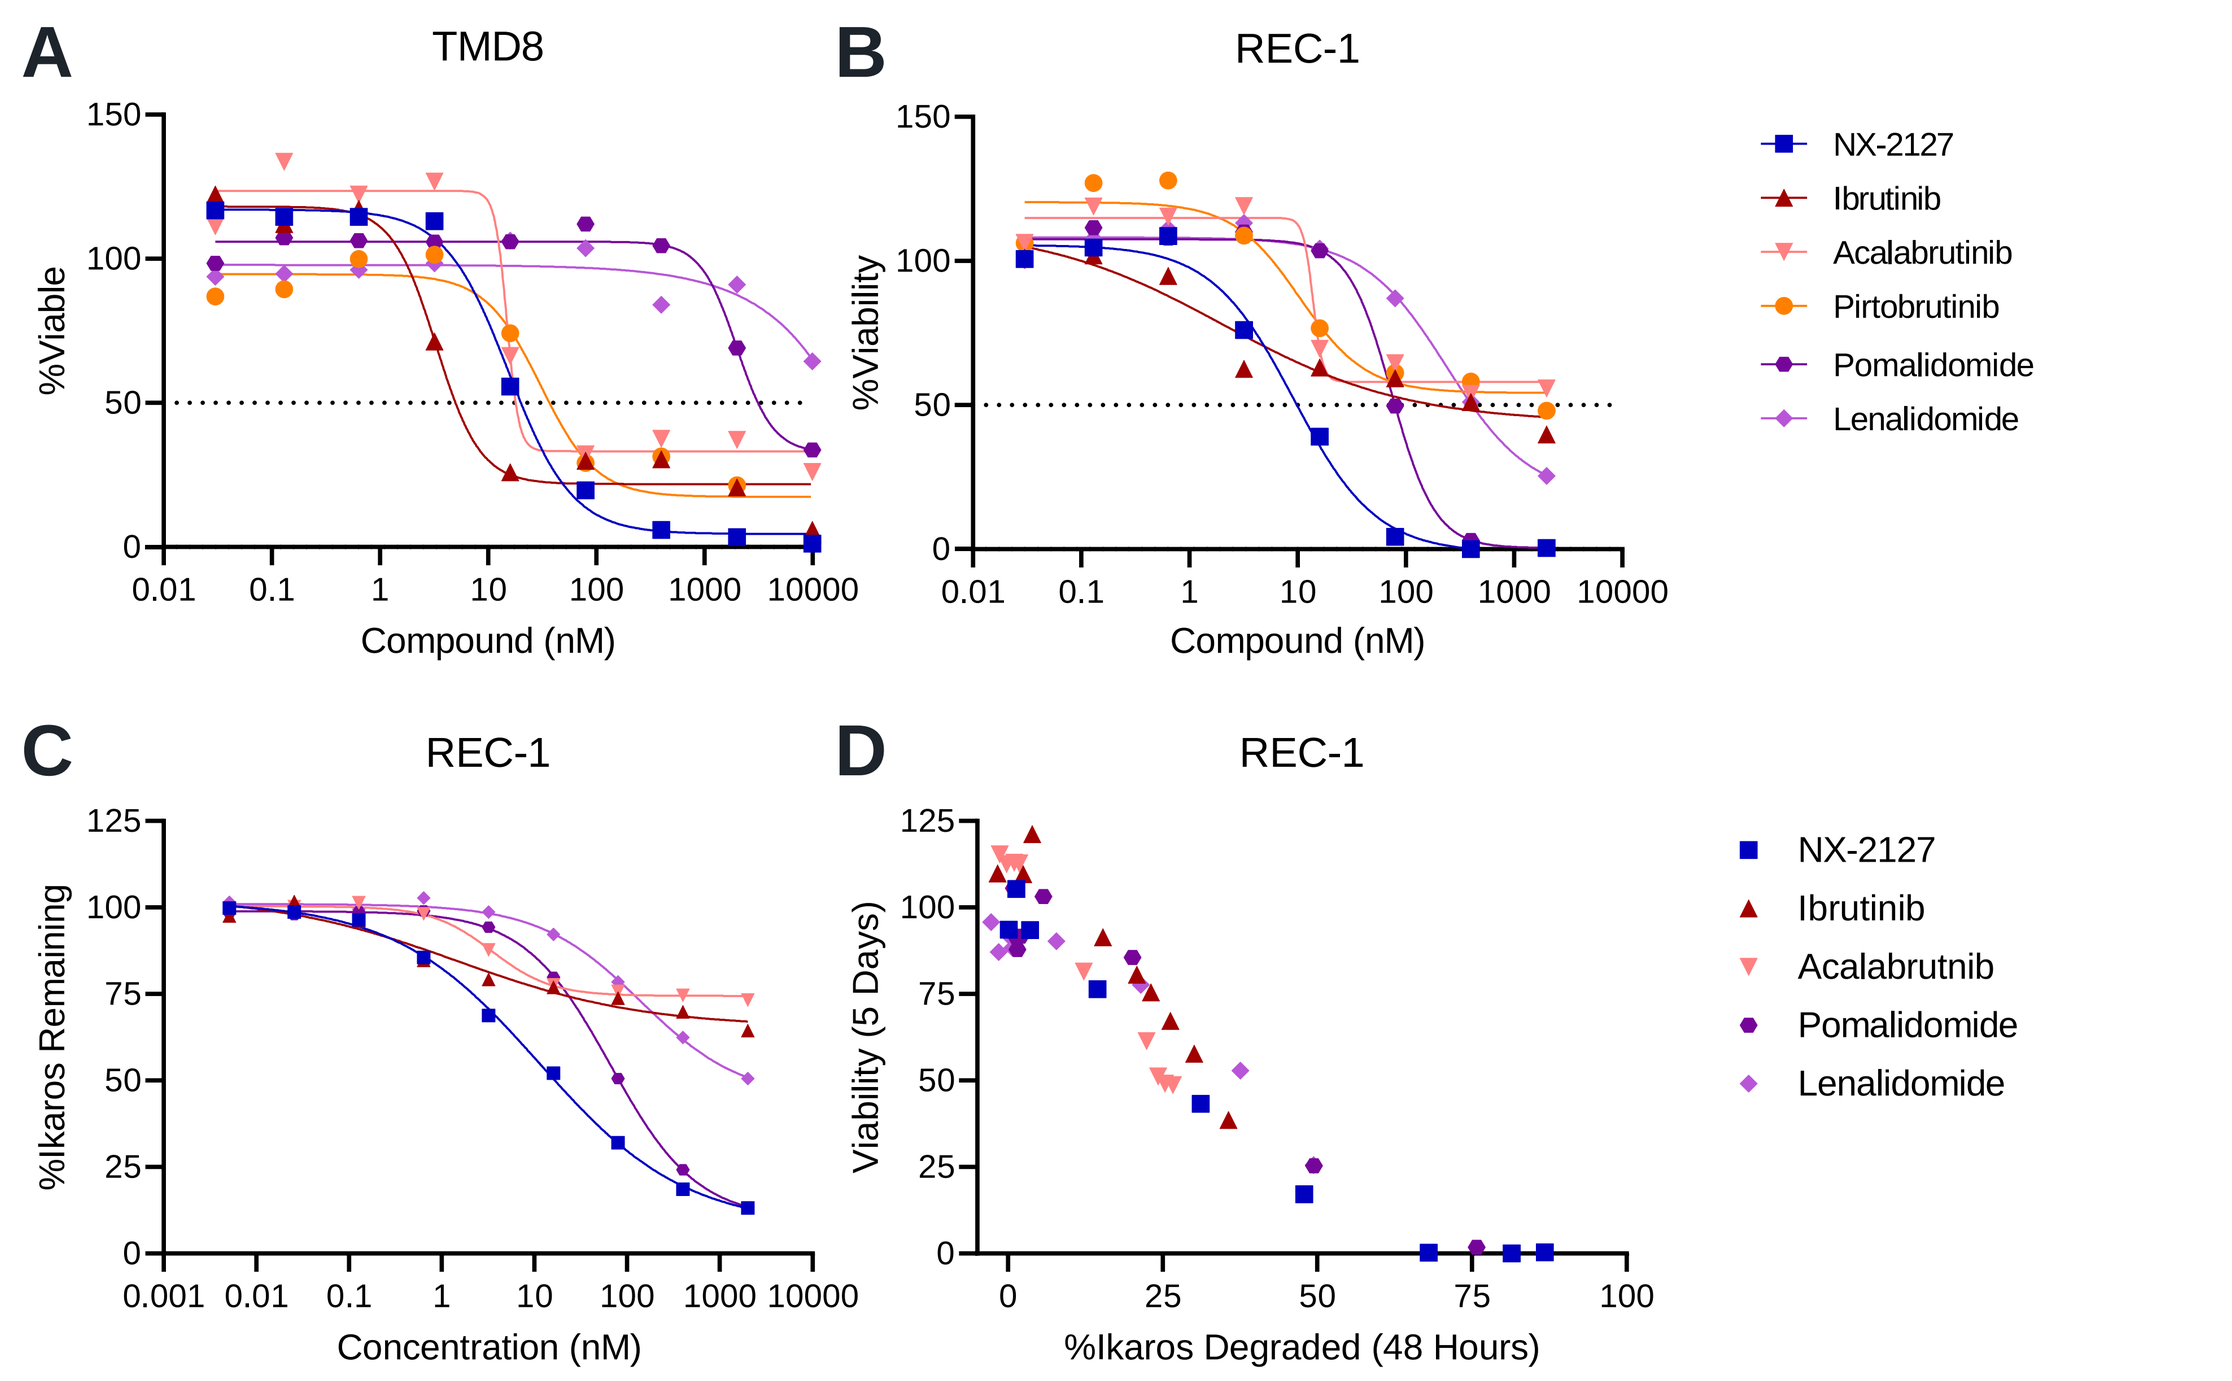
A
TMD8
%Viable
Compound (nM)
B
REC-1
%Viability
Compound (nM)
NX-2127
Ibrutinib
Acalabrutinib
Pirtobrutinib
Pomalidomide
Lenalidomide
C
REC-1
%Ikaros Remaining
Concentration (nM)
D
REC-1
Viability (5 Days)
%Ikaros Degraded (48 Hours)
NX-2127
Ibrutinib
Acalabrutnib
Pomalidomide
Lenalidomide

(A-B) TMD8 cells (A) and REC-1 cells (B) were incubated with compound for 3 days and 5 days, respectively. Viability was quantified using CellTiter-Glo® 2.0 (Promega) and normalized to DMSO-treated cells. (C) REC-1 cells were treated with compound for 48 hours, and intracellular Ikaros protein was quantified by flow cytometry and normalized to DMSO-treated cells. (D) Viability of REC-1 cells after 5 days of compound treatment plotted as a function of Ikaros degradation after 48 hours of compound treatment, where %Ikaros degraded = 100 - %Ikaros remaining. Viability was also plotted as a function of Aiolos and IRF4 levels, but the amount of Aiolos or IRF4 suppression required to promote cell death differed by drug mechanism of action (data not shown).

Figure 4. NX-2127 induces differential gene expression in REC-1 cells compared to ibrutinib and pomalidomide

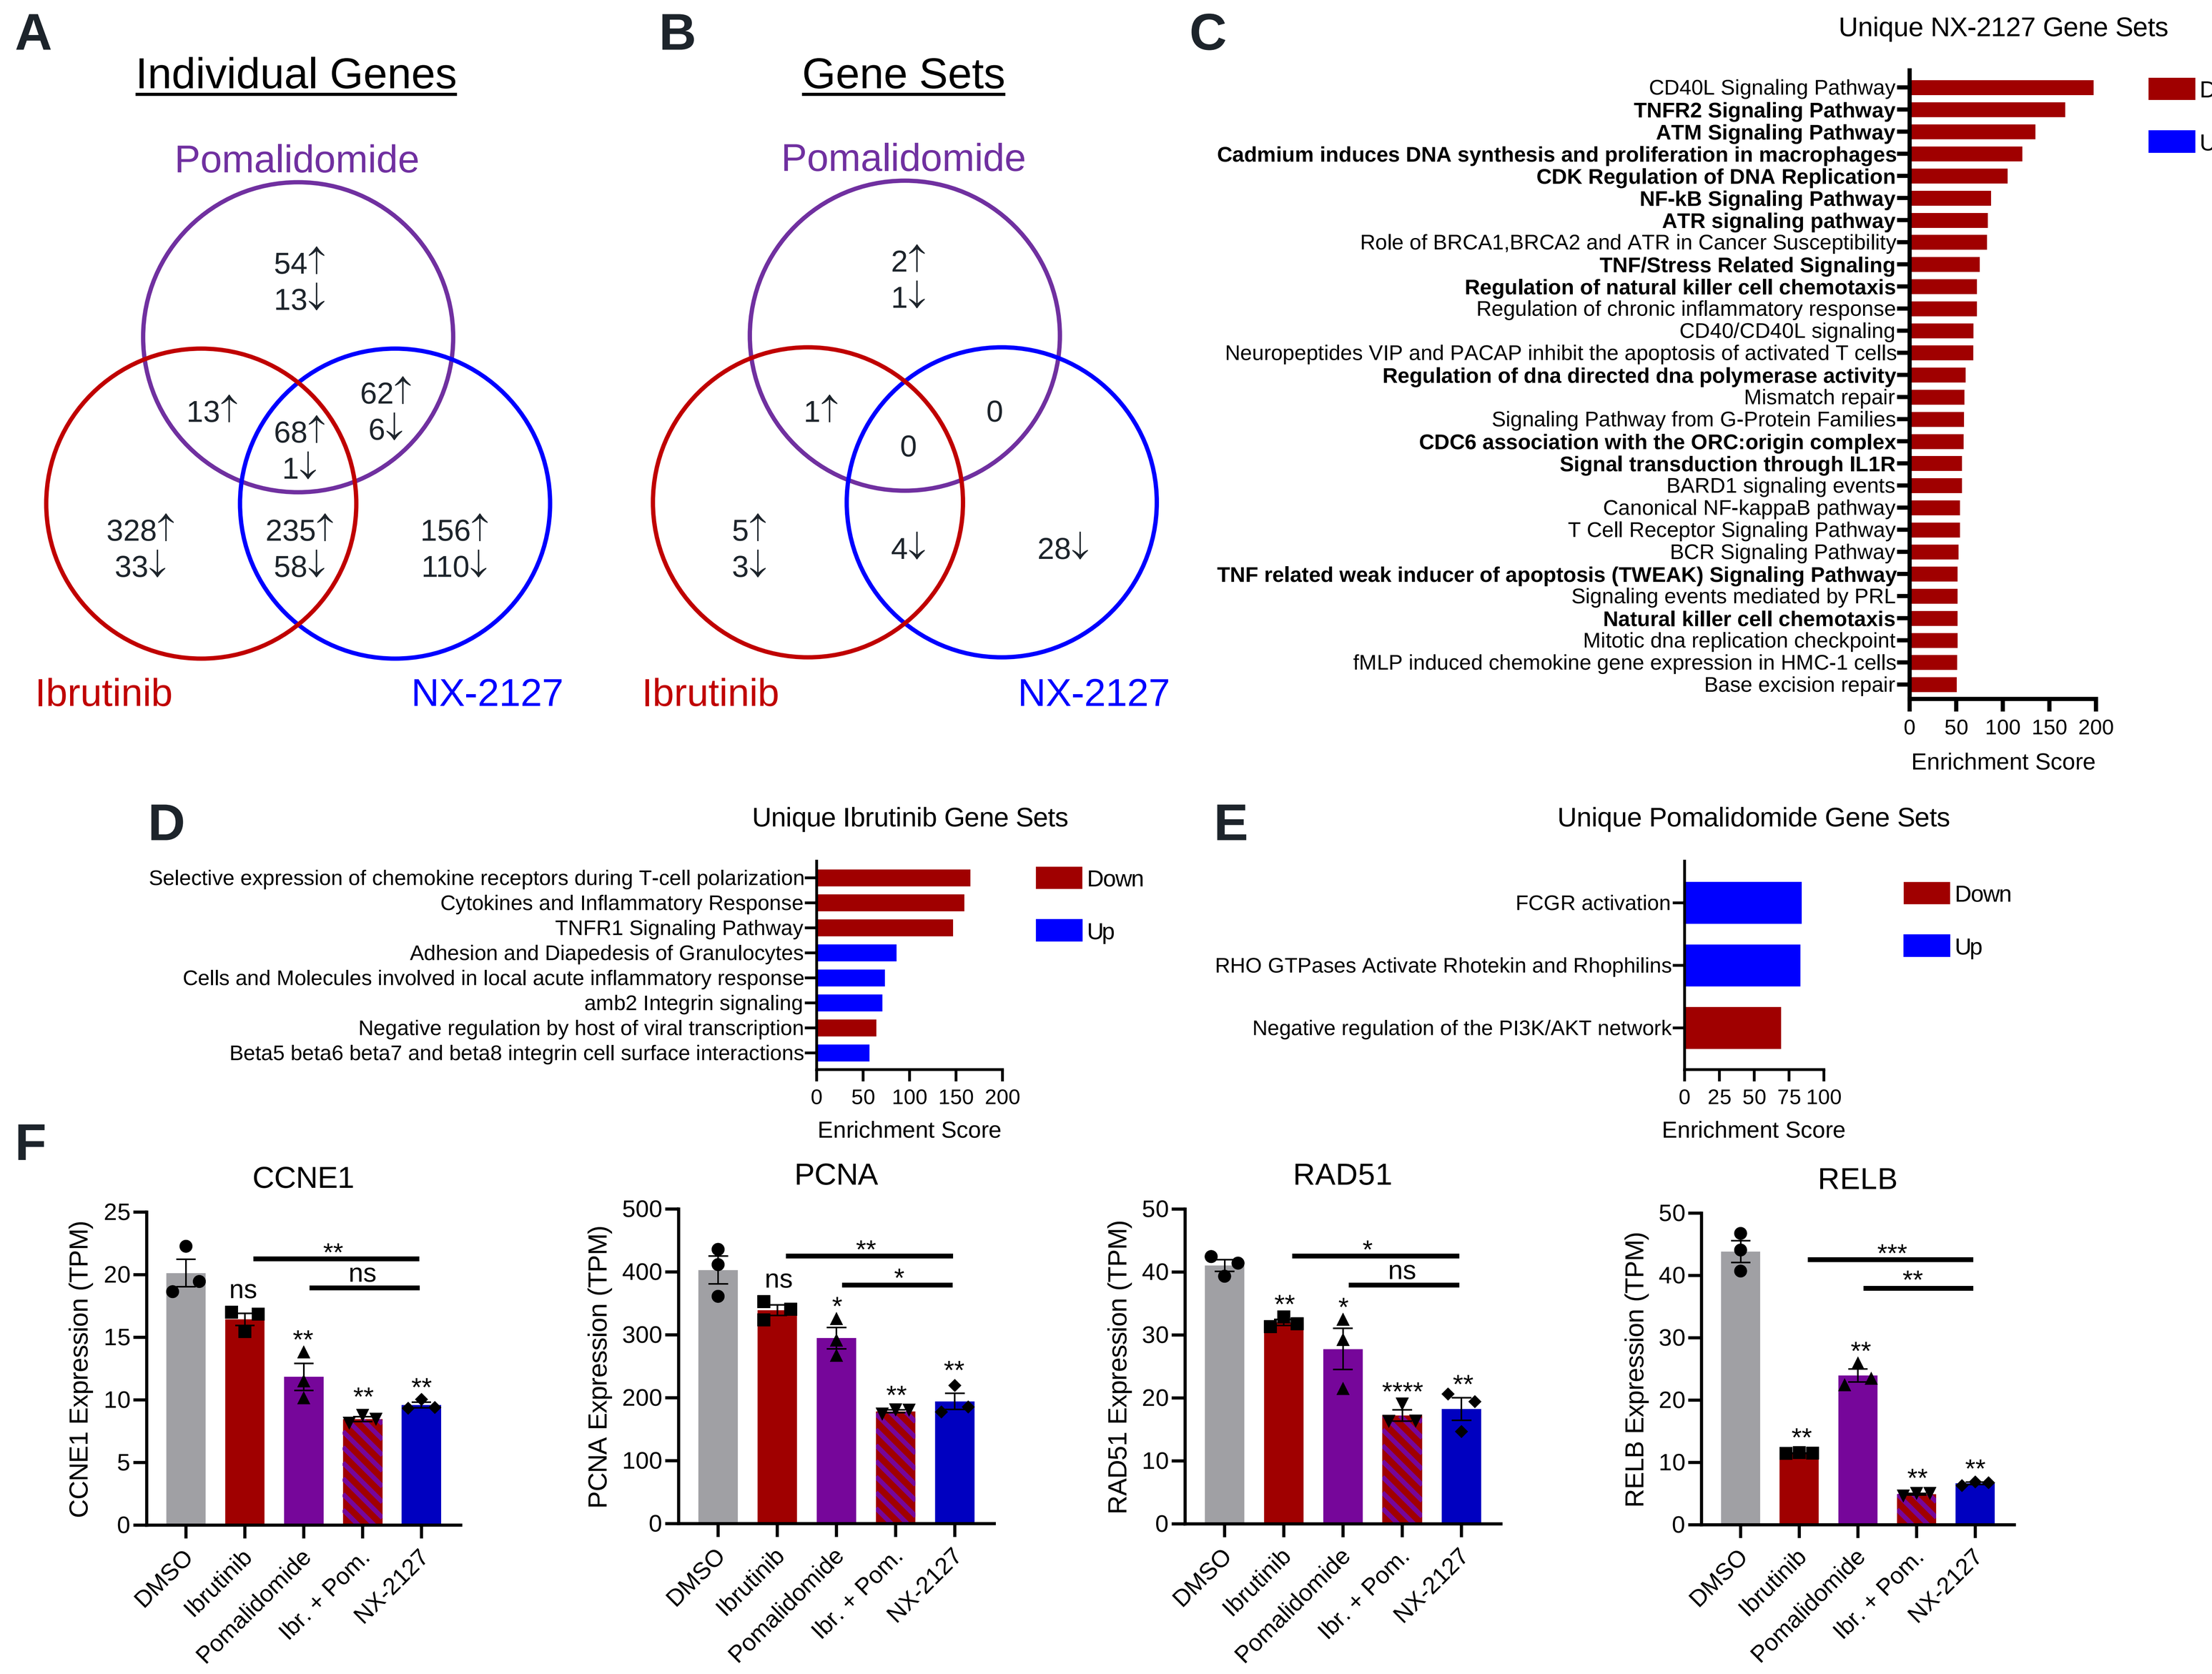
A
Individual Genes
Pomalidomide
54↑
13↓
13↑
62↑
6↓
68↑
1↓
328↑
33↓
235↑
58↓
156↑
110↓
Ibrutinib
NX-2127
B
Gene Sets
Pomalidomide
2↑
1↓
1↑
0
0
5↑
3↓
4↓
28↓
Ibrutinib
NX-2127
C
Unique NX-2127 Gene Sets
CD40L Signaling Pathway
TNFR2 Signaling Pathway
ATM Signaling Pathway
Cadmium induces DNA synthesis and proliferation in macrophages
CDK Regulation of DNA Replication
NF-kB Signaling Pathway
ATR signaling pathway
Role of BRCA1,BRCA2 and ATR in Cancer Susceptibility
TNF/Stress Related Signaling
Regulation of natural killer cell chemotaxis
Regulation of chronic inflammatory response
CD40/CD40L signaling
Neuropeptides VIP and PACAP inhibit the apoptosis of activated T cells
Regulation of dna directed dna polymerase activity
Mismatch repair
Signaling Pathway from G-Protein Families
CDC6 association with the ORC:origin complex
Signal transduction through IL1R
BARD1 signaling events
Canonical NF-kappaB pathway
T Cell Receptor Signaling Pathway
BCR Signaling Pathway
TNF related weak inducer of apoptosis (TWEAK) Signaling Pathway
Signaling events mediated by PRL
Natural killer cell chemotaxis
Mitotic dna replication checkpoint
fMLP induced chemokine gene expression in HMC-1 cells
Base excision repair
Enrichment Score
Down
Up
D
Unique Ibrutinib Gene Sets
Selective expression of chemokine receptors during T-cell polarization
Cytokines and Inflammatory Response
TNFR1 Signaling Pathway
Adhesion and Diapedesis of Granulocytes
Cells and Molecules involved in local acute inflammatory response
amb2 Integrin signaling
Negative regulation by host of viral transcription
Beta5 beta6 beta7 and beta8 integrin cell surface interactions
Enrichment Score
Down
Up
E
Unique Pomalidomide Gene Sets
FCGR activation
RHO GTPases Activate Rhotekin and Rhophilins
Negative regulation of the PI3K/AKT network
Enrichment Score
Down
Up
F
CCNE1
CCNE1 Expression (TPM)
PCNA
PCNA Expression (TPM)
RAD51
RAD51 Expression (TPM)
RELB
RELB Expression (TPM)
DMSO
Ibrutinib
Pomalidomide
Ibr. + Pom.
NX-2127

(A) REC-1 cells were treated with 100 nM compound or DMSO for 24 hours, and RNA sequencing was performed. RNAseq transcripts were quantified and normalized using Salmon and DESeq2, respectively [1-2]. Normalized gene expression in compound-treated samples was compared DMSO-treated samples, and differentially expressed genes were defined as having a twofold or greater change in expression and p<0.05 by both the paired and unpaired Welch's t-test. Counts of common and unique differentially-expressed genes are displayed. (B) Gene set enrichment analysis (GSEA) was performed on differentially-expressed genes for each compound in (A) using the following gene sets from The Molecular Signatures Database [3]: PID, KEGG, Biocarta, Reactome, WikiPathways, and GO: Biological process. Significantly enriched gene sets were defined as having an enrichment score $\geq$ 50 and FDR $\leq$ 0.05. Significantly enriched gene sets were compared between treatments, and common and unique gene sets are displayed. (C) Gene sets that are modulated by NX-2127 treatment, but not ibrutinib or pomalidomide single treatment. Gene sets that are modulated by both NX-2127 and a combination of ibrutinib and pomalidomide are displayed in bold. (D) Gene sets that are modulated by ibrutinib treatment, but not NX-2127 or pomalidomide treatment. (E) Gene sets that are modulated by pomalidomide treatment, but not NX-2127 or ibrutinib treatment. (F) Selected genes uniquely modulated by NX-2127 and/or ibrutinib and pomalidomide combination. P-values for individual comparisons were calculated using Welch's unpaired t-test. P-values above each bar correspond to the DMSO comparison, and p-values above lines correspond to the NX-2127 comparison. Ibr.+Pom.: 100 nM ibrutinib + 100 nM pomalidomide combination.

Figure 5. NX-2127 more strongly upregulates CD1c expression in REC-1 cells than ibrutinib or pomalidomide

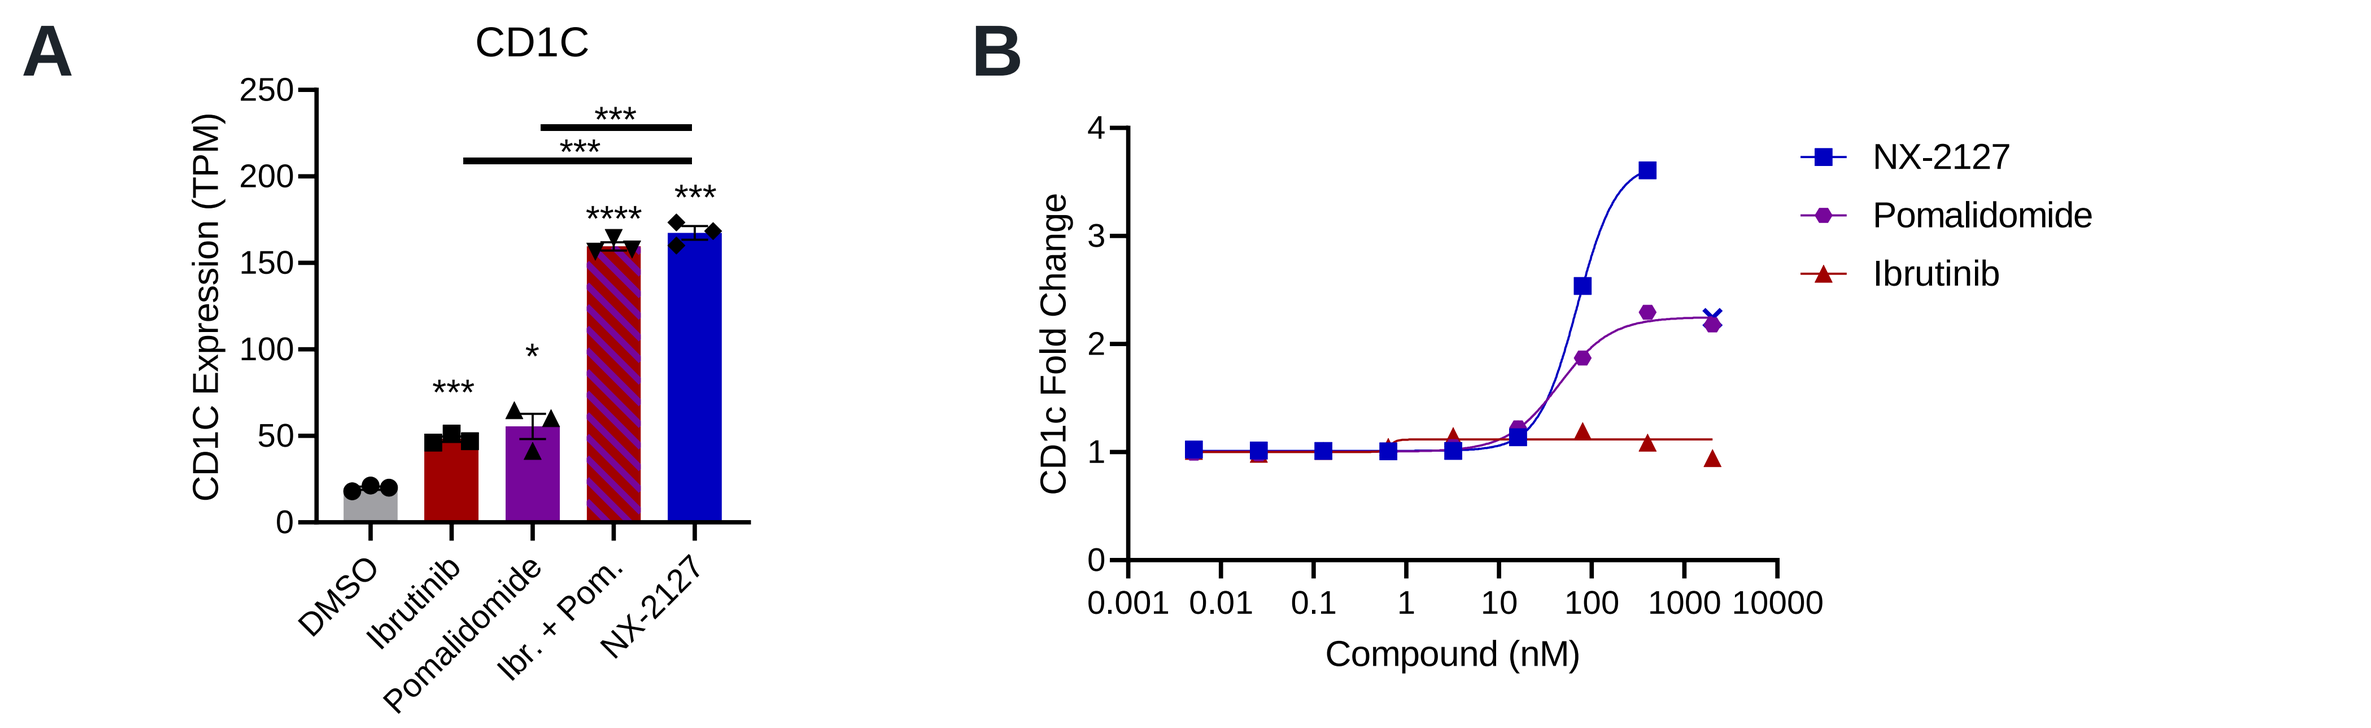
A
CD1C
CD1C Expression (TPM)
DMSO
Ibrutinib
Pomalidomide
Ibr. + Pom.
NX-2127
B
CD1c Fold Change
Compound (nM)
NX-2127
Pomalidomide
Ibrutinib

(A) CD1c transcript was upregulated by NX-2127, ibrutinib, and pomalidomide treatment, but upregulation was strongest with NX-2127. Statistics were calculated as described in (4F). (B) REC-1 cells were treated with NX-2127, ibrutinib, or pomalidomide for 48 hours, and surface CD1c expression was quantified by flow cytometry. Fold change was calculated relative to DMSO-treated cells.

Figure 6. NX-2127 promotes BTK and Aiolos degradation in TMD8 tumors and drives complete tumor regression

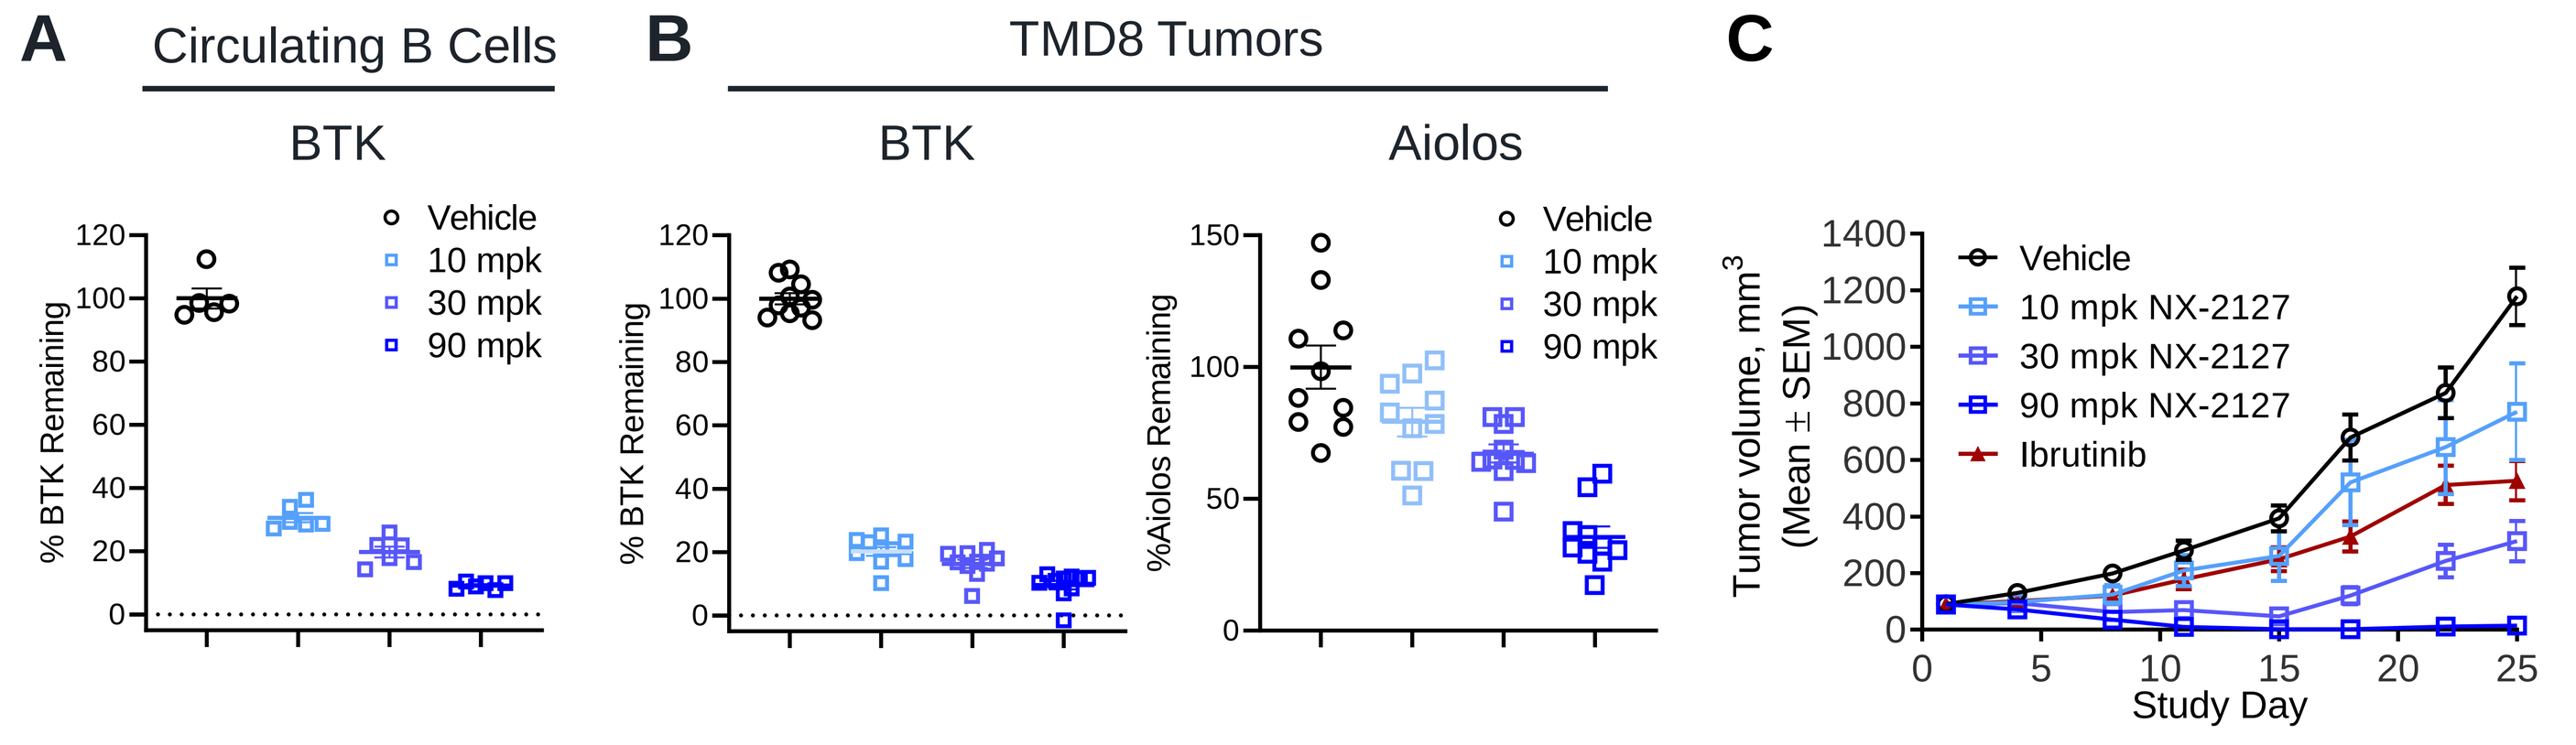
A
Circulating B Cells
BTK
% BTK Remaining
Vehicle
10 mpk
30 mpk
90 mpk
B
TMD8 Tumors
BTK
% BTK Remaining
Aiolos
%Aiolos Remaining
Vehicle
10 mpk
30 mpk
90 mpk
C
Tumor volume, mm³ (Mean ± SEM)
Study Day
Vehicle
10 mpk NX-2127
30 mpk NX-2127
90 mpk NX-2127
Ibrutinib

D

Treatment	Oral gavage dose (mg/kg)	% BTK degradation in circulating B cells	% BTK degradation in TMD8 tumor tissue	% TGI vs Vehicle (Day 24)	*P* value vs Vehicle
Vehicle	0	0.0±3.2	0.0±1.8	N/A	0
NX-2127	10	69.3±1.5	79.8±1.4	58%	0.0492
	30	80.2±1.8	83.7±1.3	74%	<0.0001
	90	90.8±0.4	90.4±1.4	100%	<0.0001
Ibrutinib	30	N/A	N/A	62%	0.0004

(A) NX-2127 was administered to CD-1 mice by daily oral gavage, and BTK levels in circulating B cells were quantified by flow cytometry 24 hours after the fifth dose and normalized to the vehicle group. (B) 1x10^7 TMD8 tumor cells were implanted subcutaneously in female CB.17 SCID mice, and mice were randomized into treatment groups when tumors reached 200 mm^3. Five daily doses of compound were administered by oral gavage. 24 hours after the final dose, tumors were dissociated, and BTK and Aiolos levels were quantified by Simple Western™ (ProteinSimple) or western blot. BTK and Aiolos protein levels were normalized to the vehicle group. (C) 1 x 10^7 TMD8 tumor cells were implanted into female CB.17 SCID mice. 17 days after implantation (Day 1), mice were sorted by tumor volume and evenly distributed into five groups of ten animals per group. NX-2127 at 10, 30, or 90 mg/kg (mpk) or ibrutinib at 30 mg/kg was administered orally once a day from Day 2-24. Tumor volumes were monitored twice a week, and mean tumor volumes and tumor growth inhibition (TGI) were calculated (D).

Conclusions

- NX-2127:
 - Degrades BTK in ABC-DLBCL (DC_{50} = 4 nM) and MCL (DC_{50} = 4-6 nM) lines and can degrade the C481S mutant form of BTK (DC_{50} = 13 nM)
 - Promotes Aiolos and Ikaros degradation and enhances IL-2 secretion by primary human T cells
 - Promotes complete killing of REC-1 cells, and cell death correlates with the ability of NX-2127 to suppress neosubstrate levels, as exemplified by Ikaros suppression. BTK inhibitors partially suppress Ikaros levels in REC-1 cells and only promote partial cell death.
 - Suppresses a variety of gene sets involved in DNA replication, DNA repair, and cell cycle progression that are not suppressed by ibrutinib or pomalidomide treatment alone.
 - Promotes upregulation of CD1c in REC-1 cells at both the transcript and surface expression level. This may lead to enhanced recognition of certain tumor cells by T cells.
 - Degrades BTK and Aiolos in tumor cells *in vivo* and drives complete regression of TMD8 tumors
- BTK degradation may represent a potentially informative biomarker for predicting clinical responses to NX-2127
 - >80% BTK degradation in circulating B cells is associated with 74% TMD8 tumor growth inhibition, and >90% BTK degradation in circulating B cells is associated with 100% tumor growth inhibition
- A phase 1a/b trial of NX-2127 for patients with relapsed or refractory B cell malignancies is ongoing (NCT04830137)

Disclosures

All authors are past or current employees of Nurix Therapeutics and hold company stock or stock options.

Presented at the American Association for Cancer Research (AACR) Annual Meeting. April 8-13, 2022. Do not post.