NX-0255, a Small-Molecule CBL-B Inhibitor, Expands and Enhances Tumor-Infiltrating Lymphocytes for Use in Adoptive Cancer Immunotherapy

Poster 98

Sarah Whelan[1], Jennifa Gosling[1], Monisha Mani[1], Thomas Cummins[1], Frederick Cohen[1], Austin Tenn-McClellan[1], Janine Powers[1], Gwenn Hansen[1], Michael T. Lotze[1], and Arthur T. Sands[1]

[1]*Nurix Therapeutics, San Francisco, CA, USA*

BACKGROUND

- Adoptive cell transfer (ACT) of tumor-infiltrating lymphocytes (TIL) effects durable responses in patients with melanoma and some epithelial tumors[1-4]
- It is thought that poor in vitro cell expansion and inefficient T-cell migration to the tumor limits the broader application of this approach[5,6]
- The E3 ubiquitin ligase, Casitas B-lineage lymphoma b (CBL-B) is expressed in T cells in which it functions as a regulator of immune cell activation, in part by requiring CD28 costimulation in addition to T-cell receptor activation[7,8]
- We have developed NX-0255, a highly potent, small-molecule inhibitor of CBL-B, demonstrating its ability to increase T-cell–derived cytokine secretion and proliferation in the presence or absence of costimulation
- Here, we investigated the effects of NX-0255 on the ex vivo growth and characteristics of human TIL to create drug-enhanced TIL (DeTIL-0255) as an ACT product for treating patients with cancer
- We hypothesize that CBL inhibition during TIL ex vivo expansion will promote the generation of a product with superior characteristics (**Figure 1**)

Figure 1. Schema for Expanding DeTIL-0255

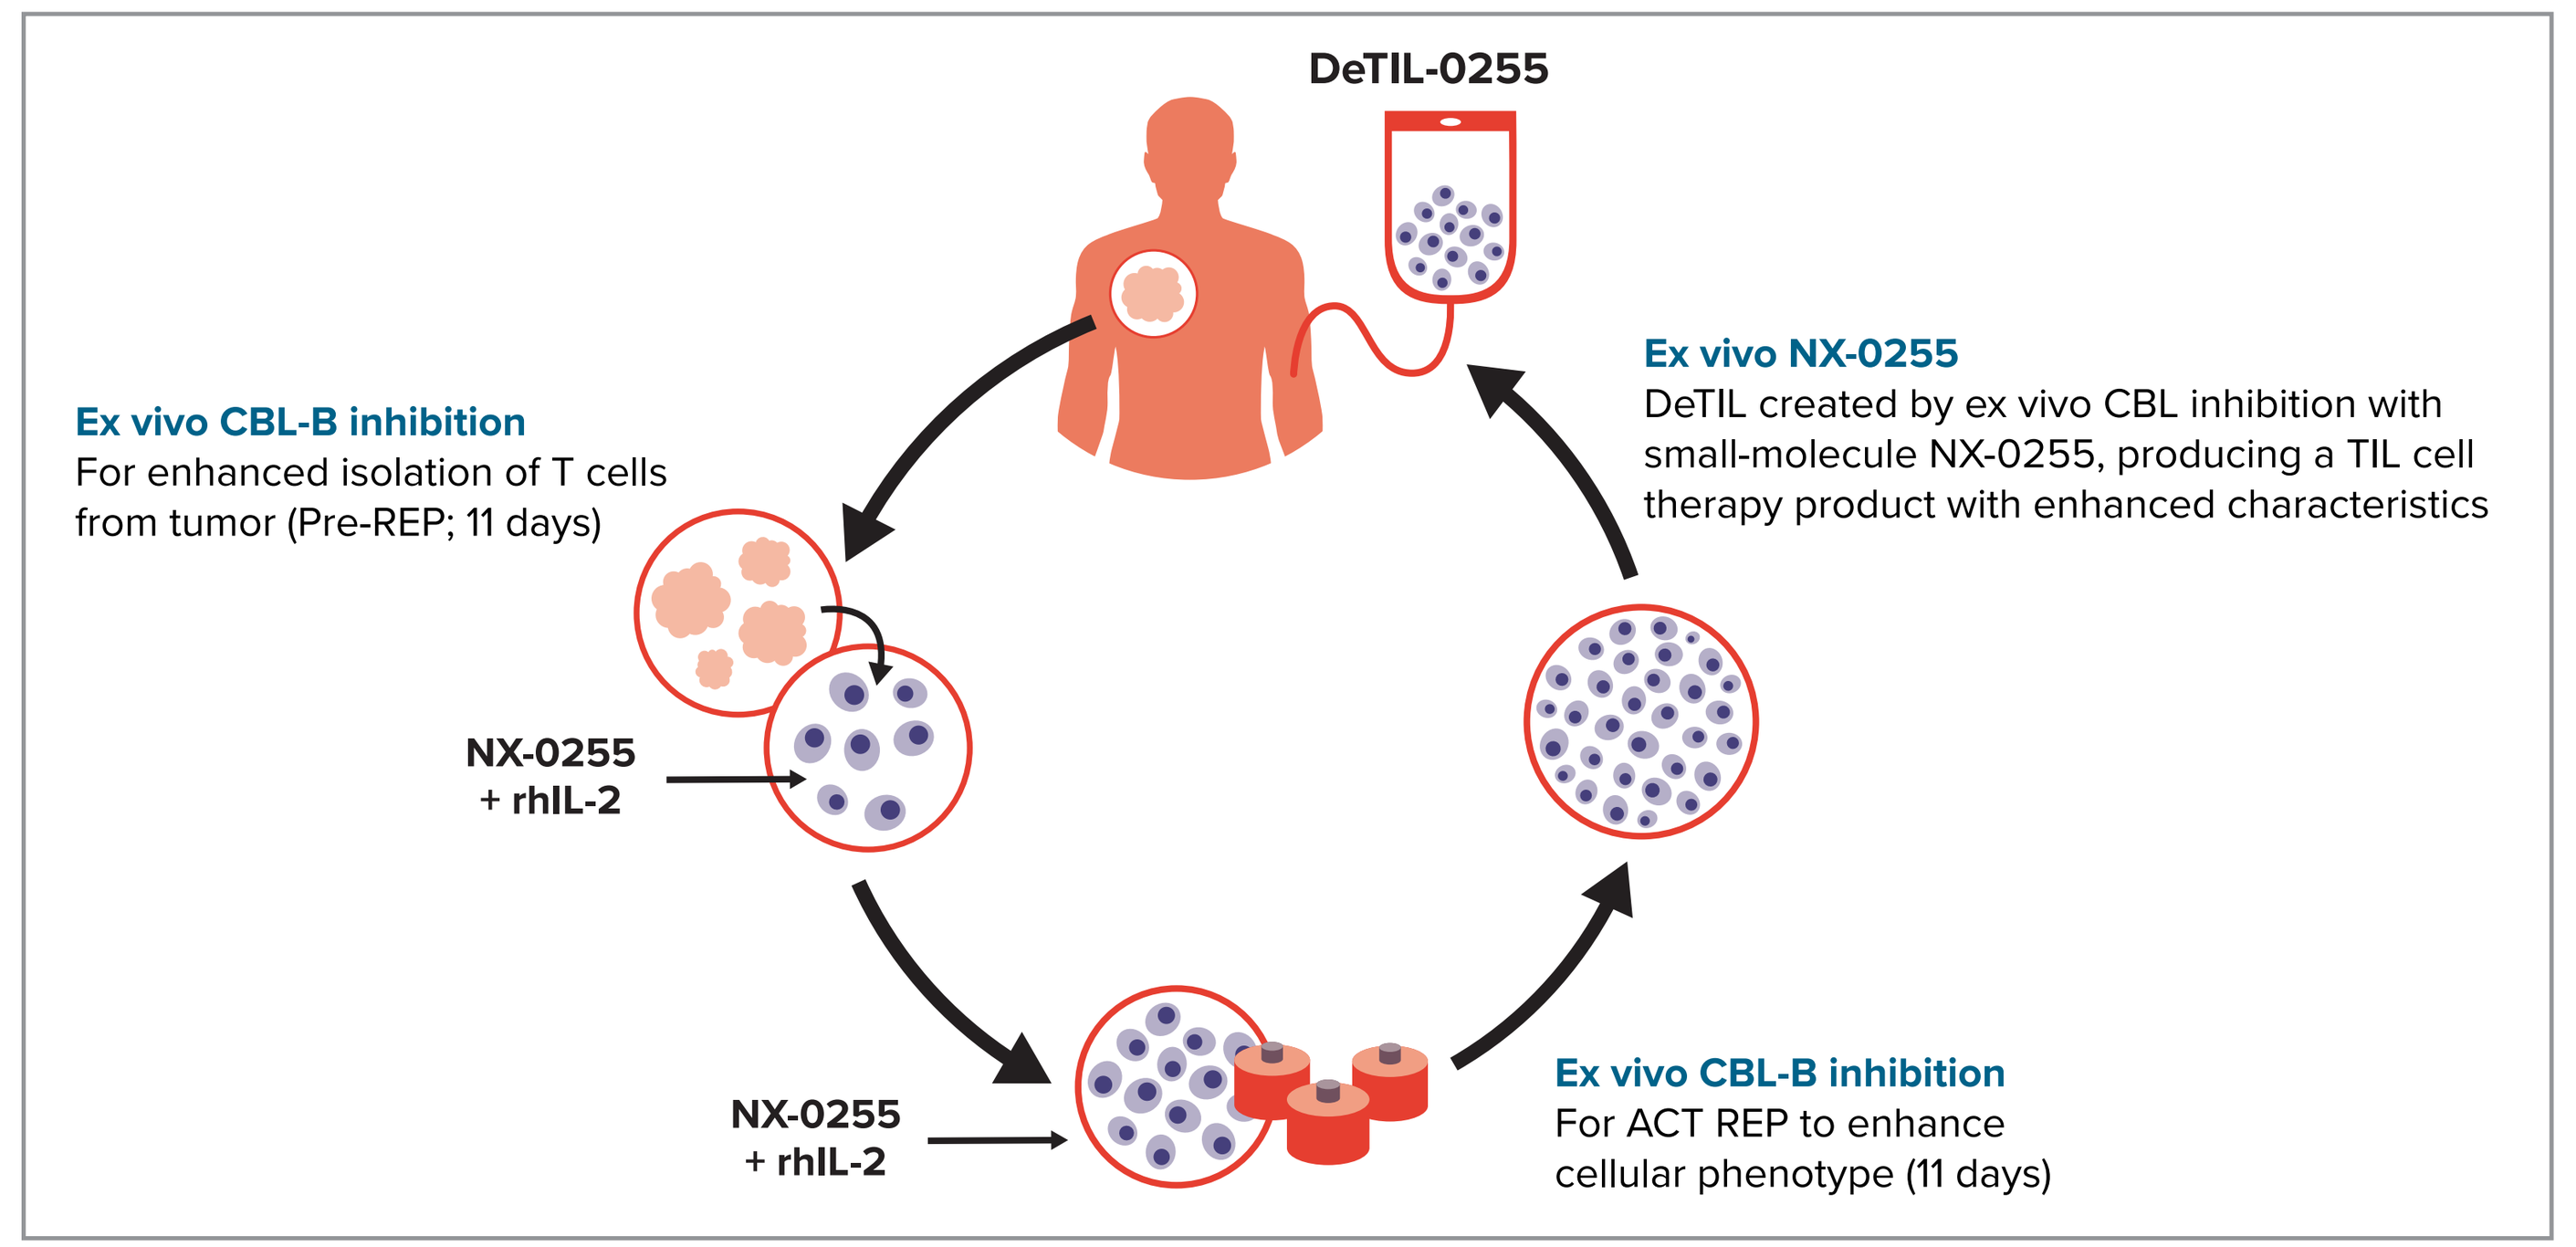
DeTIL-0255
Ex vivo CBL-B inhibition
For enhanced isolation of T cells from tumor (Pre-REP; 11 days)
Ex vivo NX-0255
DeTIL created by ex vivo CBL inhibition with small-molecule NX-0255, producing a TIL cell therapy product with enhanced characteristics
NX-0255 + rhIL-2
NX-0255 + rhIL-2
Ex vivo CBL-B inhibition
For ACT REP to enhance cellular phenotype (11 days)

ACT, adoptive cell transfer; DeTIL, drug-enhanced tumor infiltrating lymphocytes; CBL-B, Casitas B-lineage lymphoma b; REP, rapid expansion phase; rhIL-2, recombinant human interleukin-2.

METHODS

- To understand the role of NX-0255 during the expansion of TIL, tumor fragments from ovary and colon tumors were cultured with recombinant human interleukin-2 (rhIL-2) and compared in 2 experimental groups: NX-0255 without rhIL-2, or NX-0255 in combination with rhIL-2
 - After 28 days of culture, cell yield and viability were assessed
- After process development, cells were cultured from tumor fragments from ovary, colon, lung, head and neck, breast, and vulva carcinomas in G-REX flasks (Wilson Wolf, St Paul, MN) with either rhIL-2 alone (TIL) or rhIL-2 in combination with NX-0255 (DeTIL-0255) for a period of 22 days
 - After expansion, TIL and DeTIL-0255 were evaluated for yield (cell count), phenotype (flow cytometry), cytokine/chemokine secretion (cytometric bead assays), functionality (anti-CD3/CD28 stimulation), T-cell receptor (TCR) Vβ repertoire (RNA sequencing), and single-cell transcriptomics (10× Genomics)

RESULTS

- Culturing of TIL in the presence of NX-0255 alone resulted in the expansion of TIL to a similar extent as standard rhIL-2. The combination of NX-0255 with rhIL-2 dramatically increased TIL expansion in 3 of 4 samples (**Figure 2**)
- DeTIL-0255 significantly increased the number of cells expanded from tumor tissue (ovary, lung, head and neck, melanoma, breast, and vulva carcinomas) compared with conventionally cultured TIL (n=16, *P*=0.004) (**Figure 3**)
- Flow cytometric analysis demonstrated that DeTIL-0255 CD8+ T cells were significantly less exhausted than TIL, as shown by the significant reduced expression of programmed cell death protein 1 (PD-1; *P*=0.02), and coexpressing PD-1+ TIM-3 (T cell immunoglobulin and mucin domain-containing protein 3; *P*=0.03) and PD-1+ LAG-3 (lymphocyte activation gene 3; *P*=0.03) (**Figure 4**)
- The functional capacity of DeTIL-0255 was differentially enhanced, with significant increases in the absolute numbers of CD8+ T-cells expressing intracellular perforin (*P*=0.001), granzyme B (*P*=0.005), and CD107a (*P*=0.01) when comparing DeTIL-0255 with TIL. An increase of CD8+ T cells expressing CD137/4-1BB, a biomarker of CD8+ T-cell tumor reactivity was also observed (*P*=0.03) (**Figure 5**)
- TCR repertoire analysis showed that DeTIL-0255 had a significant increase in diversity index (Di) and unique CDR3 β chains in comparison with TIL, suggesting an increase in CD8+ T-cell effector function and TCR diversity is associated with antitumor effects (**Figure 6**)
- Single-cell sequencing analysis demonstrated that DeTIL-0255 expanded in the pre-rapid expansion phase had increased expression of genes associated with stemness (CCR7+,CD62L+[SELL], CD127+[IL-7R]) and cytotoxicity (GNLY+,GZMB+,NKG7+) (**Figure 7**)

RESULTS

Figure 2. NX-0255 in Combination With rhIL-2 Yields Superior T-Cell Expansion

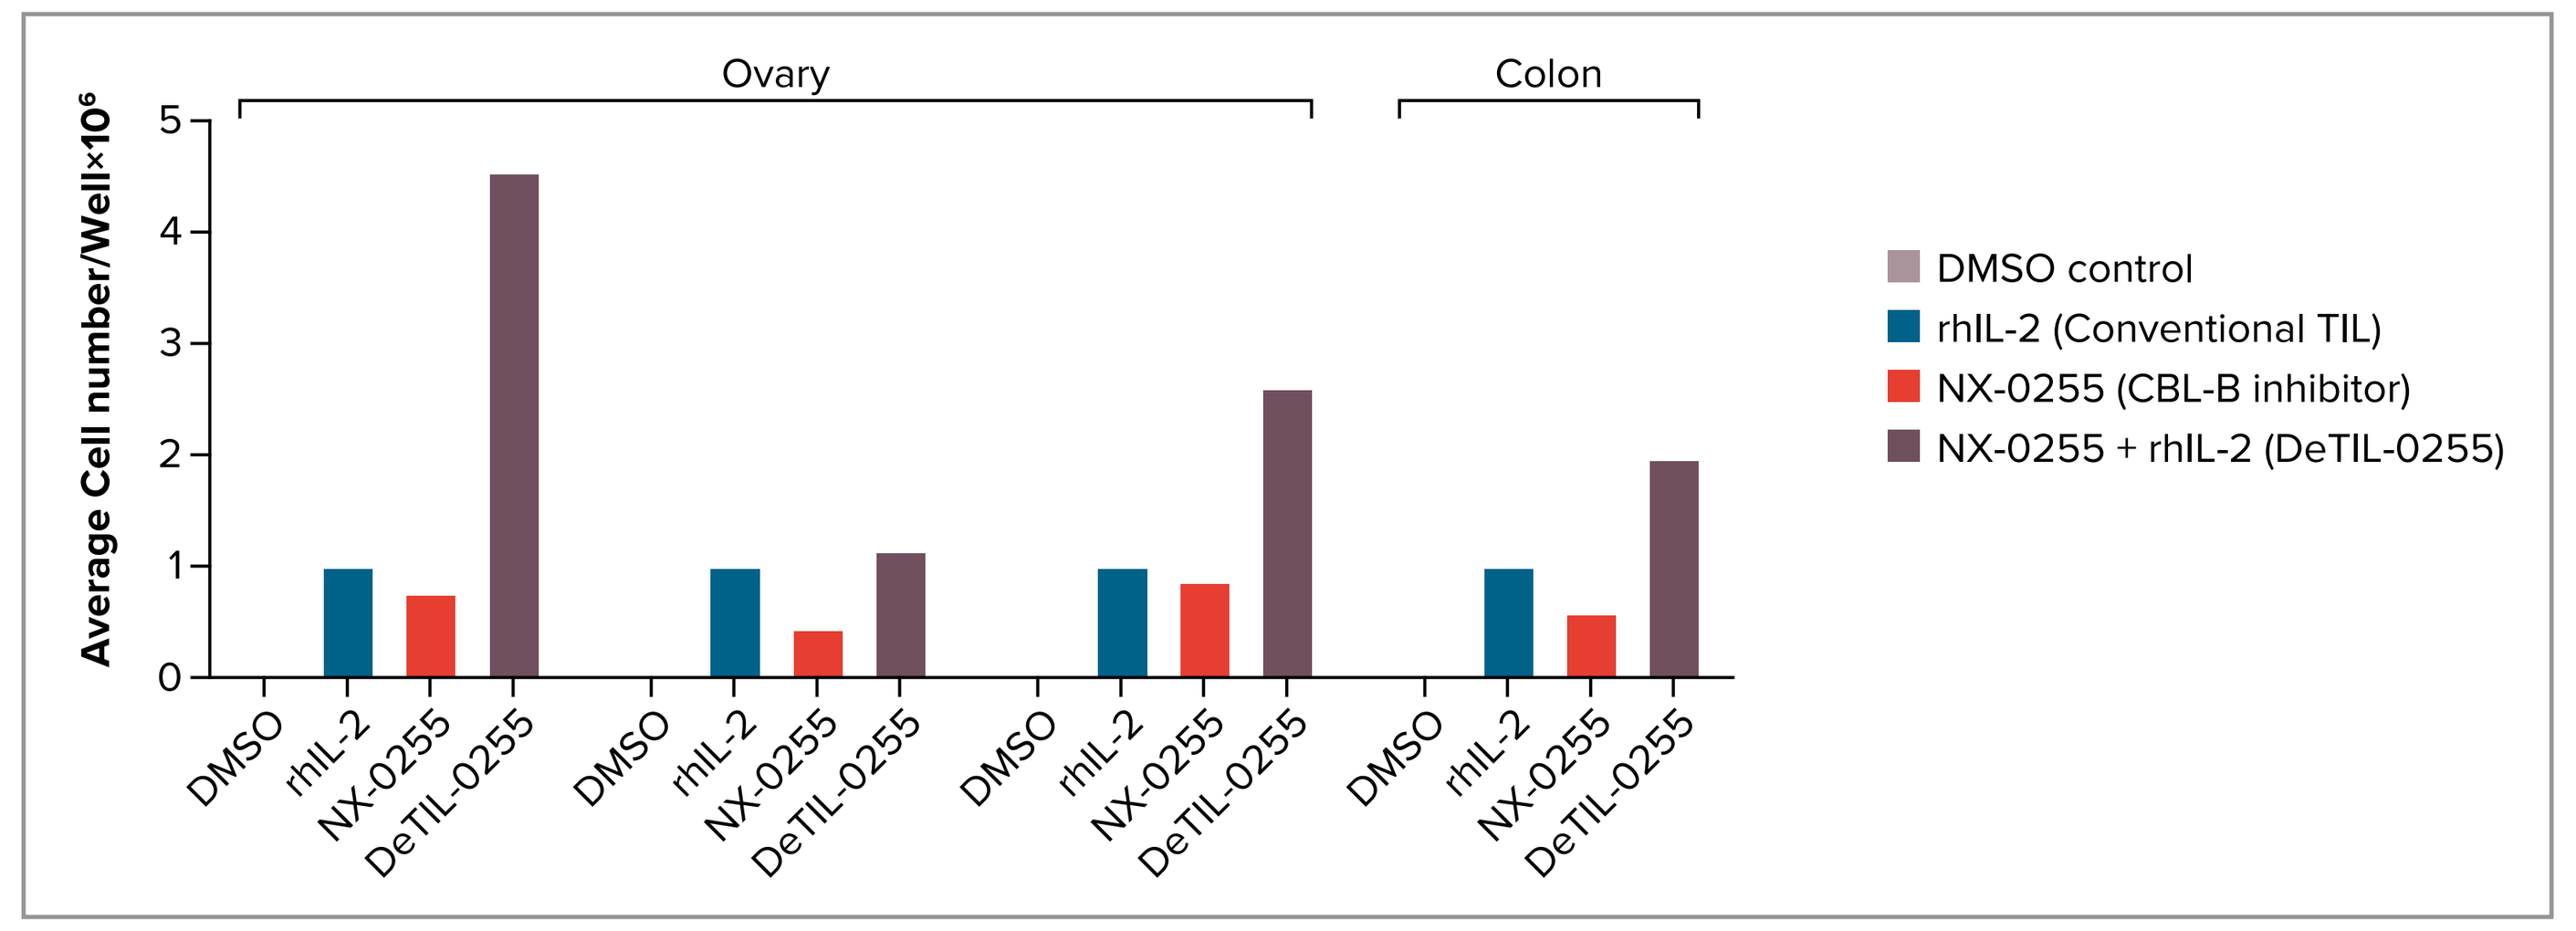
Ovary
Colon
Average Cell number/Well×10⁶
DMSO
rhIL-2
NX-0255
DeTIL-0255
DMSO control
rhIL-2 (Conventional TIL)
NX-0255 (CBL-B inhibitor)
NX-0255 + rhIL-2 (DeTIL-0255)

CBL-B, Casitas B-lineage lymphoma b; DeTIL-0255, drug-enhanced tumor-infiltrating lymphocyte; DMSO, dimethyl sulfoxide; rhIL-2, recombinant human interleukin-2; TIL, tumor-infiltrating lymphocyte.

- Culturing of TIL in the presence of NX-0255 alone resulted in the expansion of TIL to a similar extent as standard rhIL-2. The combination of NX-0255 with rhIL-2 dramatically increased TIL expansion in 3 of 4 samples

Figure 3. NX-0255 in Combination With rhIL-2 (DeTIL-0255) Significantly Increased the Number of T Cells Expanded From Tumor Tissue Compared With rhIL-2 Alone (TIL)

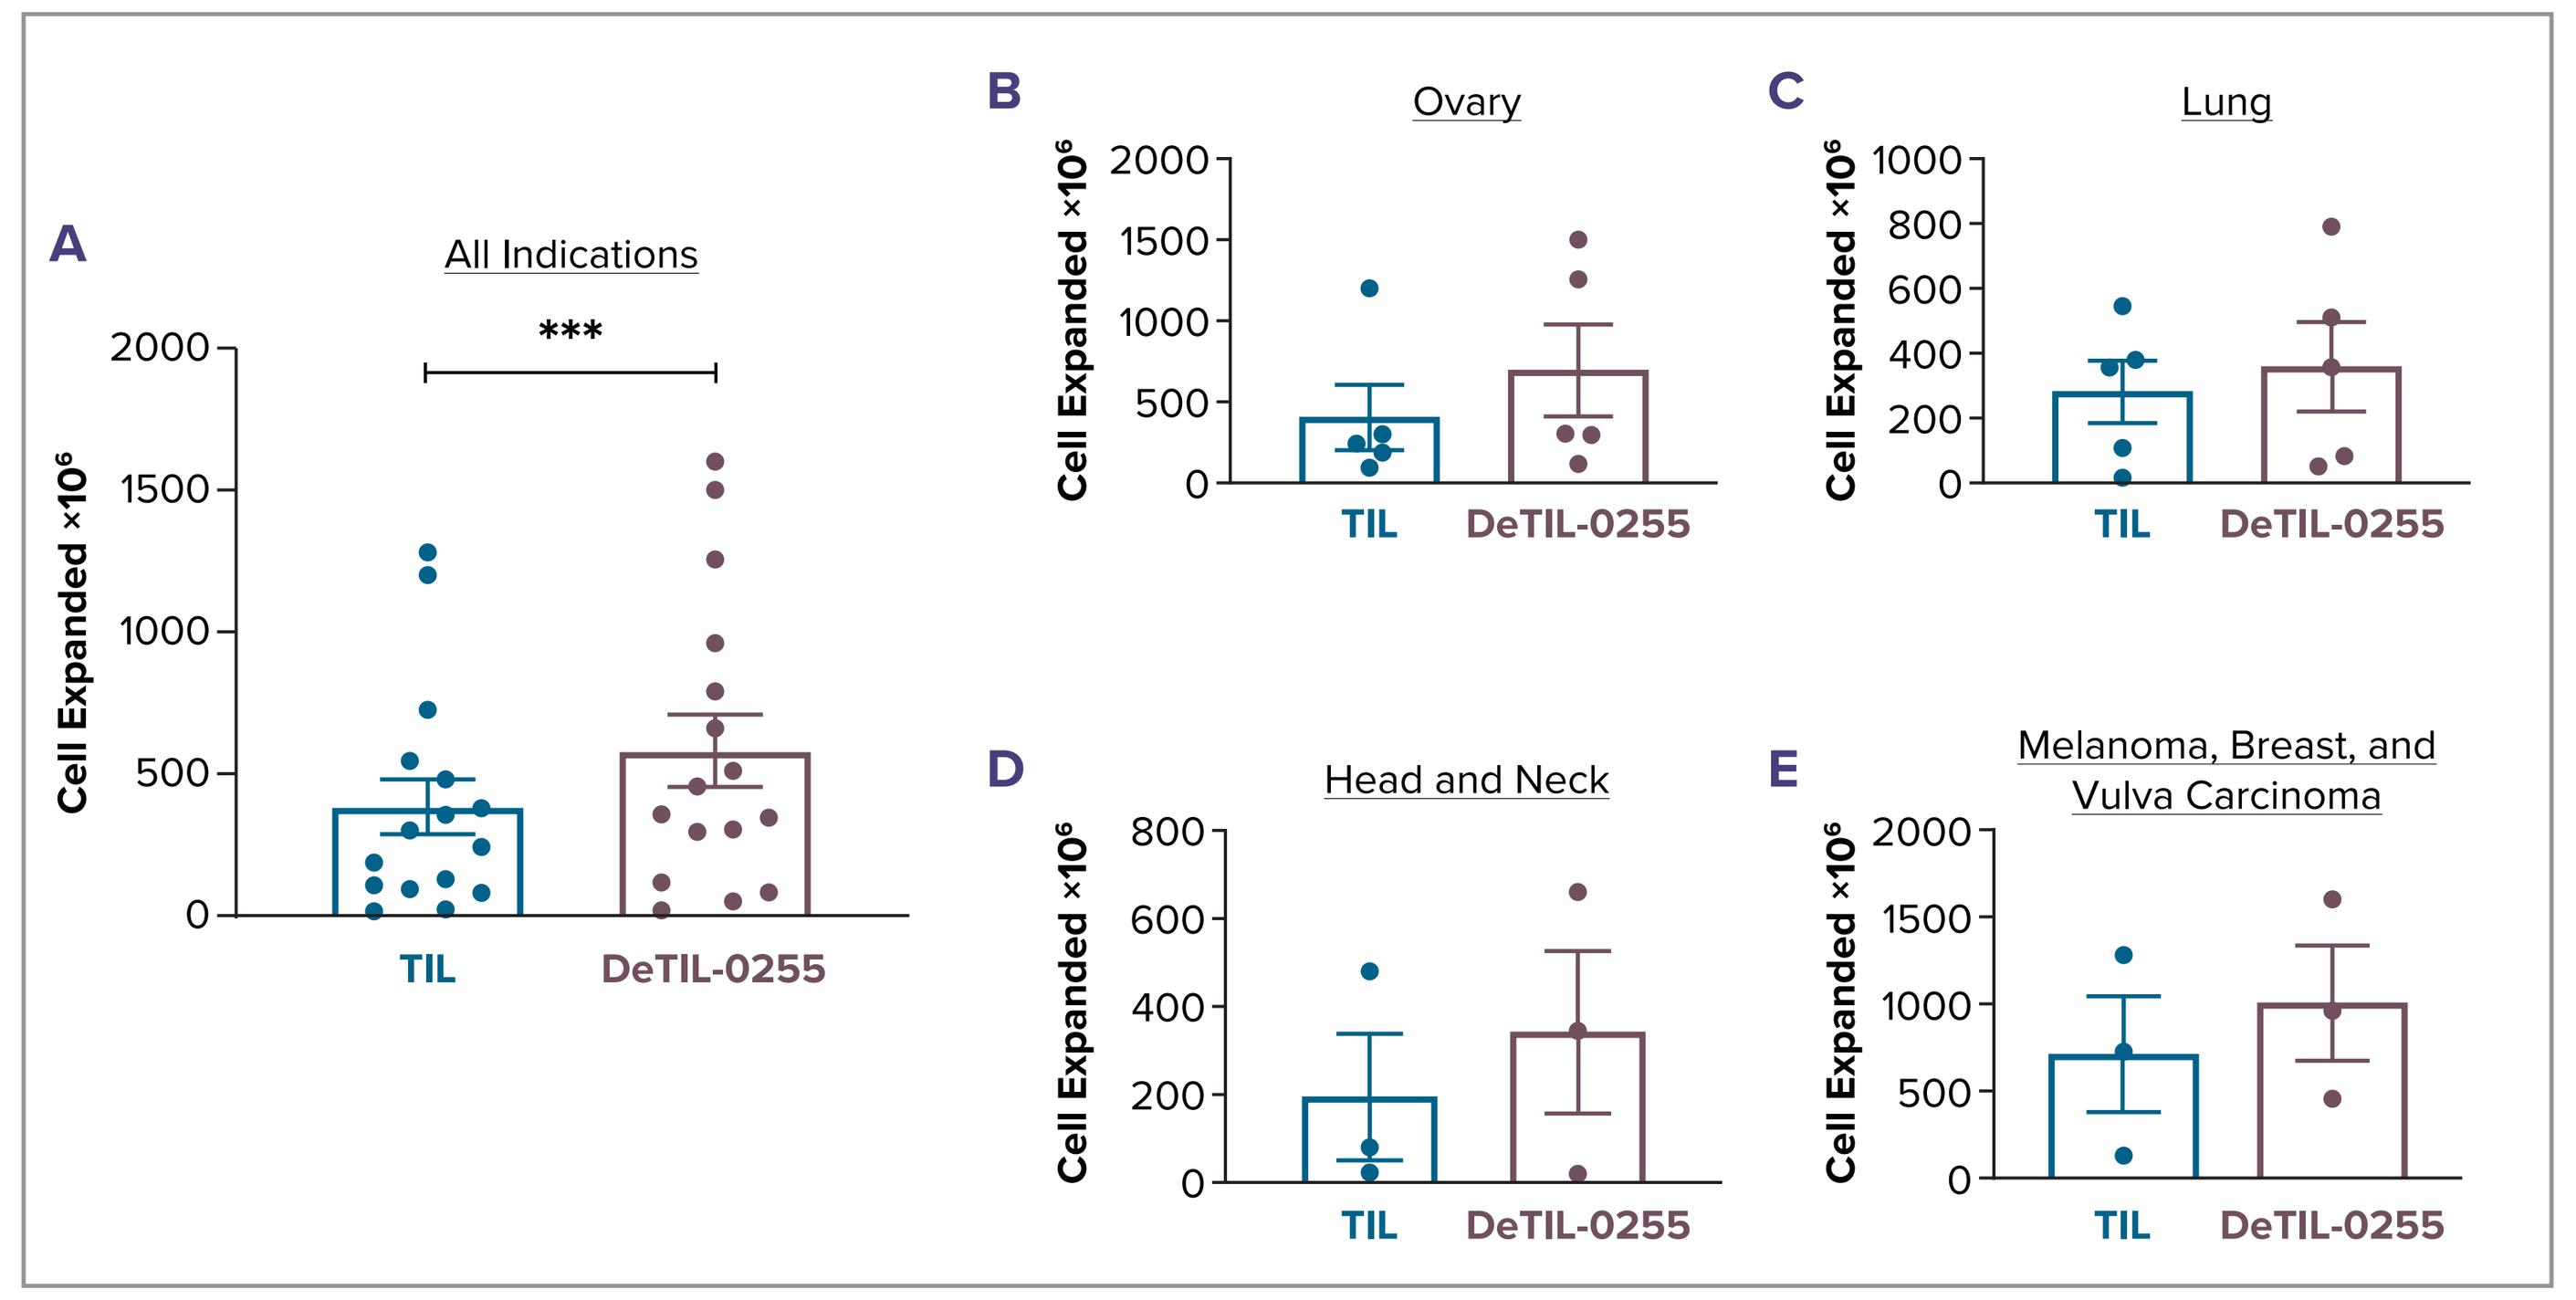
A
All Indications

Cell Expanded ×10⁶
B
Ovary
C
Lung
D
Head and Neck
E
Melanoma, Breast, and Vulva Carcinoma
TIL
DeTIL-0255

The number of T cells expanded from all indications (A); ovary tumor tissue (B); lung tumor tissue (C); head and neck tumor tissue (D); melanoma, breast and vulva carcinoma tissue (E). Statistical significance was assessed using 2-tailed Wilcoxon signed-rank test; *$P<0.05$, **$P<0.005$, and ***$P<0.001$. Error bars depict standard error of the mean (SEM).
DeTIL-0255, drug-enhanced tumor-infiltrating lymphocyte; rhIL-2, recombinant human interleukin-2; TIL, tumor-infiltrating lymphocyte.

- DeTIL-0255 significantly increased the number of cells expanded from tumor tissue (ovary, lung, head and neck, melanoma, breast, and vulva carcinomas) compared with conventionally cultured TIL (n=16, *P*=0.004)

Figure 4. CD8+ T DeTIL-0255 Are Less Exhausted Than CD8+ TIL

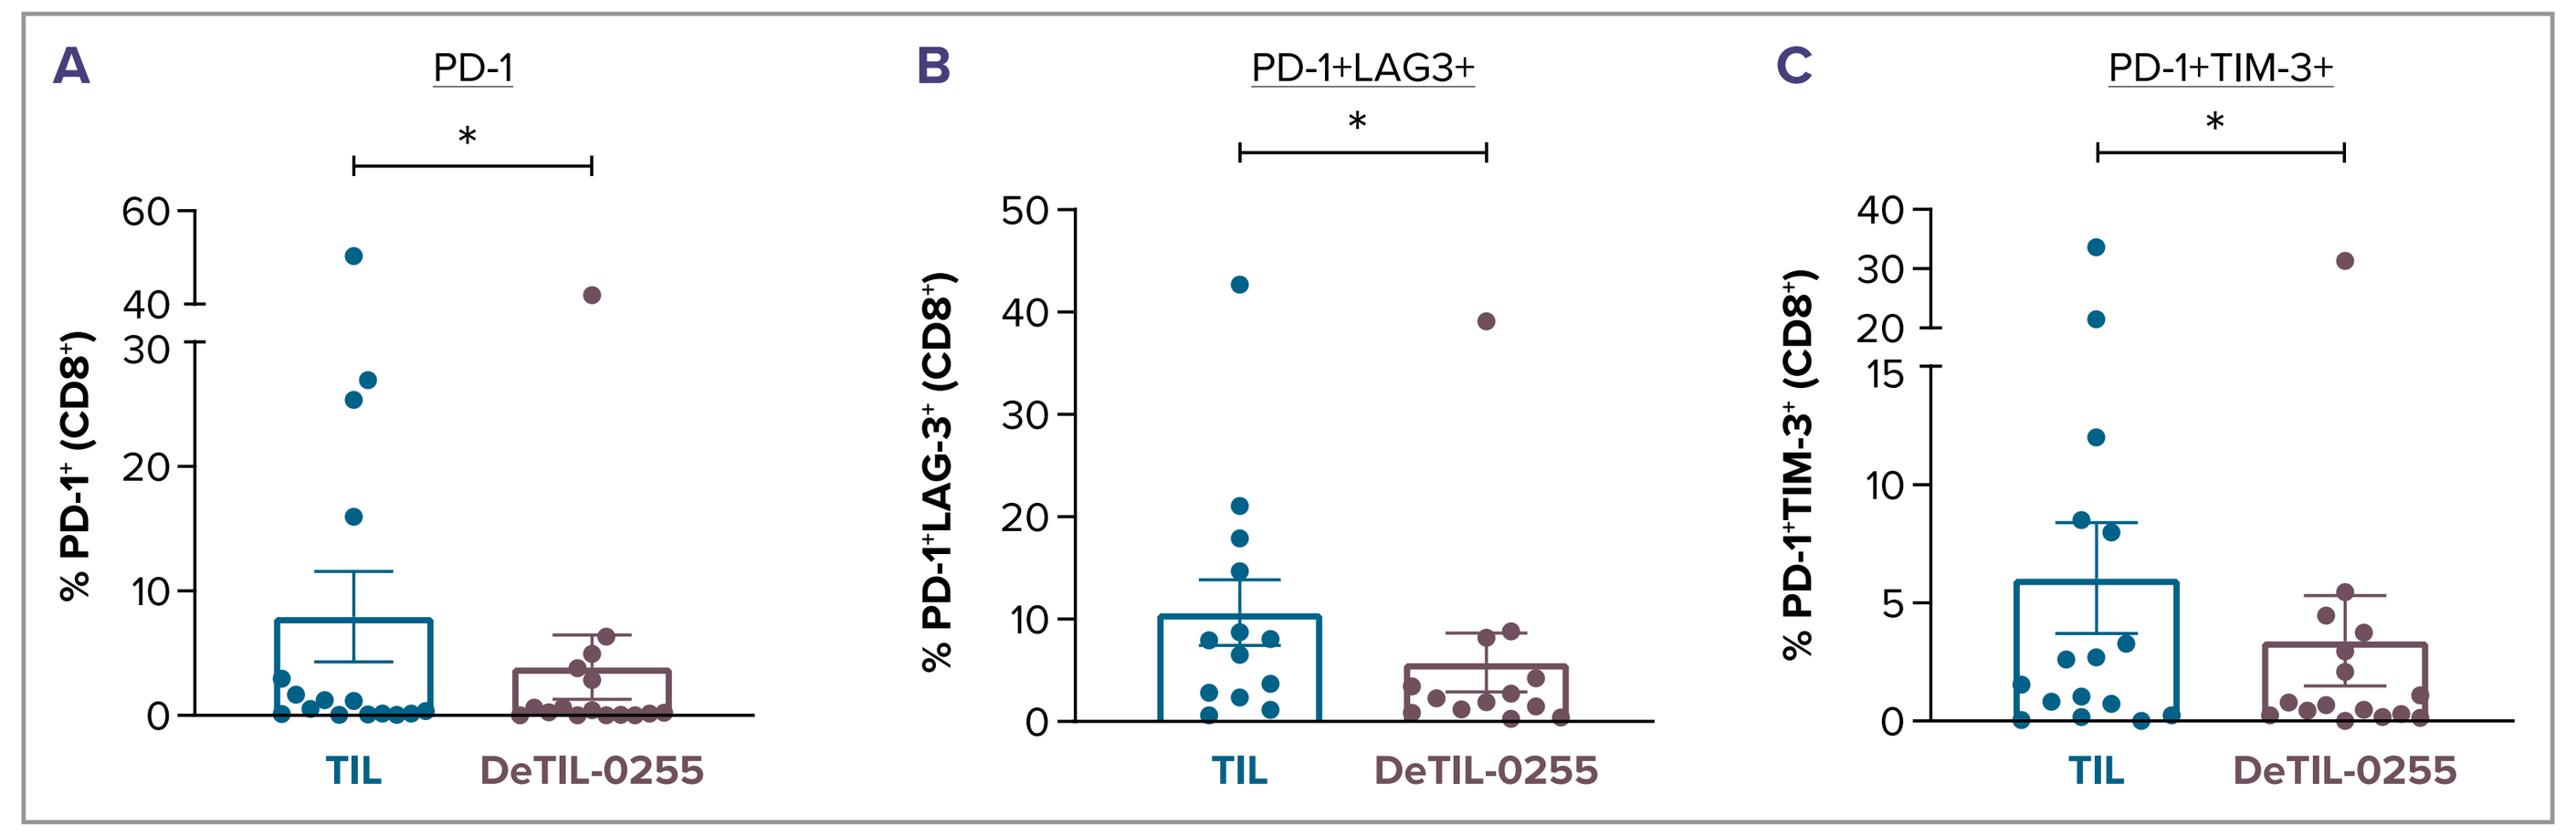
A
PD-1
% PD-1⁺ (CD8⁺)
B
PD-1+LAG3+
% PD-1⁺LAG-3⁺ (CD8⁺)
C
PD-1+TIM-3+
% PD-1⁺TIM-3⁺ (CD8⁺)
*
TIL
DeTIL-0255

Flow cytometry analysis of CD8+ T cells for PD-1 (A), PD-1 and LAG-3 (B), PD-1 and TIM-3 (C) expression. Statistical significance was assessed using 2-tailed Wilcoxon signed-rank test; *$P<0.05$, **$P<0.005$, and ***$P<0.001$. Error bars depict standard error of the mean (SEM).
DeTIL-0255, drug-enhanced tumor infiltrating lymphocyte; LAG-3, lymphocyte-activation gene 3; PD-1, programmed cell death protein 1; TIL, tumor-infiltrating lymphocyte; TIM-3, T-cell immunoglobulin and mucin domain-containing protein 3.

- (A) Flow cytometric analysis demonstrated that DeTIL-0255 CD8+ T cells were significantly less exhausted than TIL CD8+ T cells, as shown by the significant reduced expression of PD-1 (*P*=0.02) (B) and coexpressing PD-1+TIM-3+ (*P*=0.03) and (C) PD-1+LAG-3+ (*P*=0.03)

Figure 5. DeTIL-0255 Are Significantly More Cytotoxic Than TIL

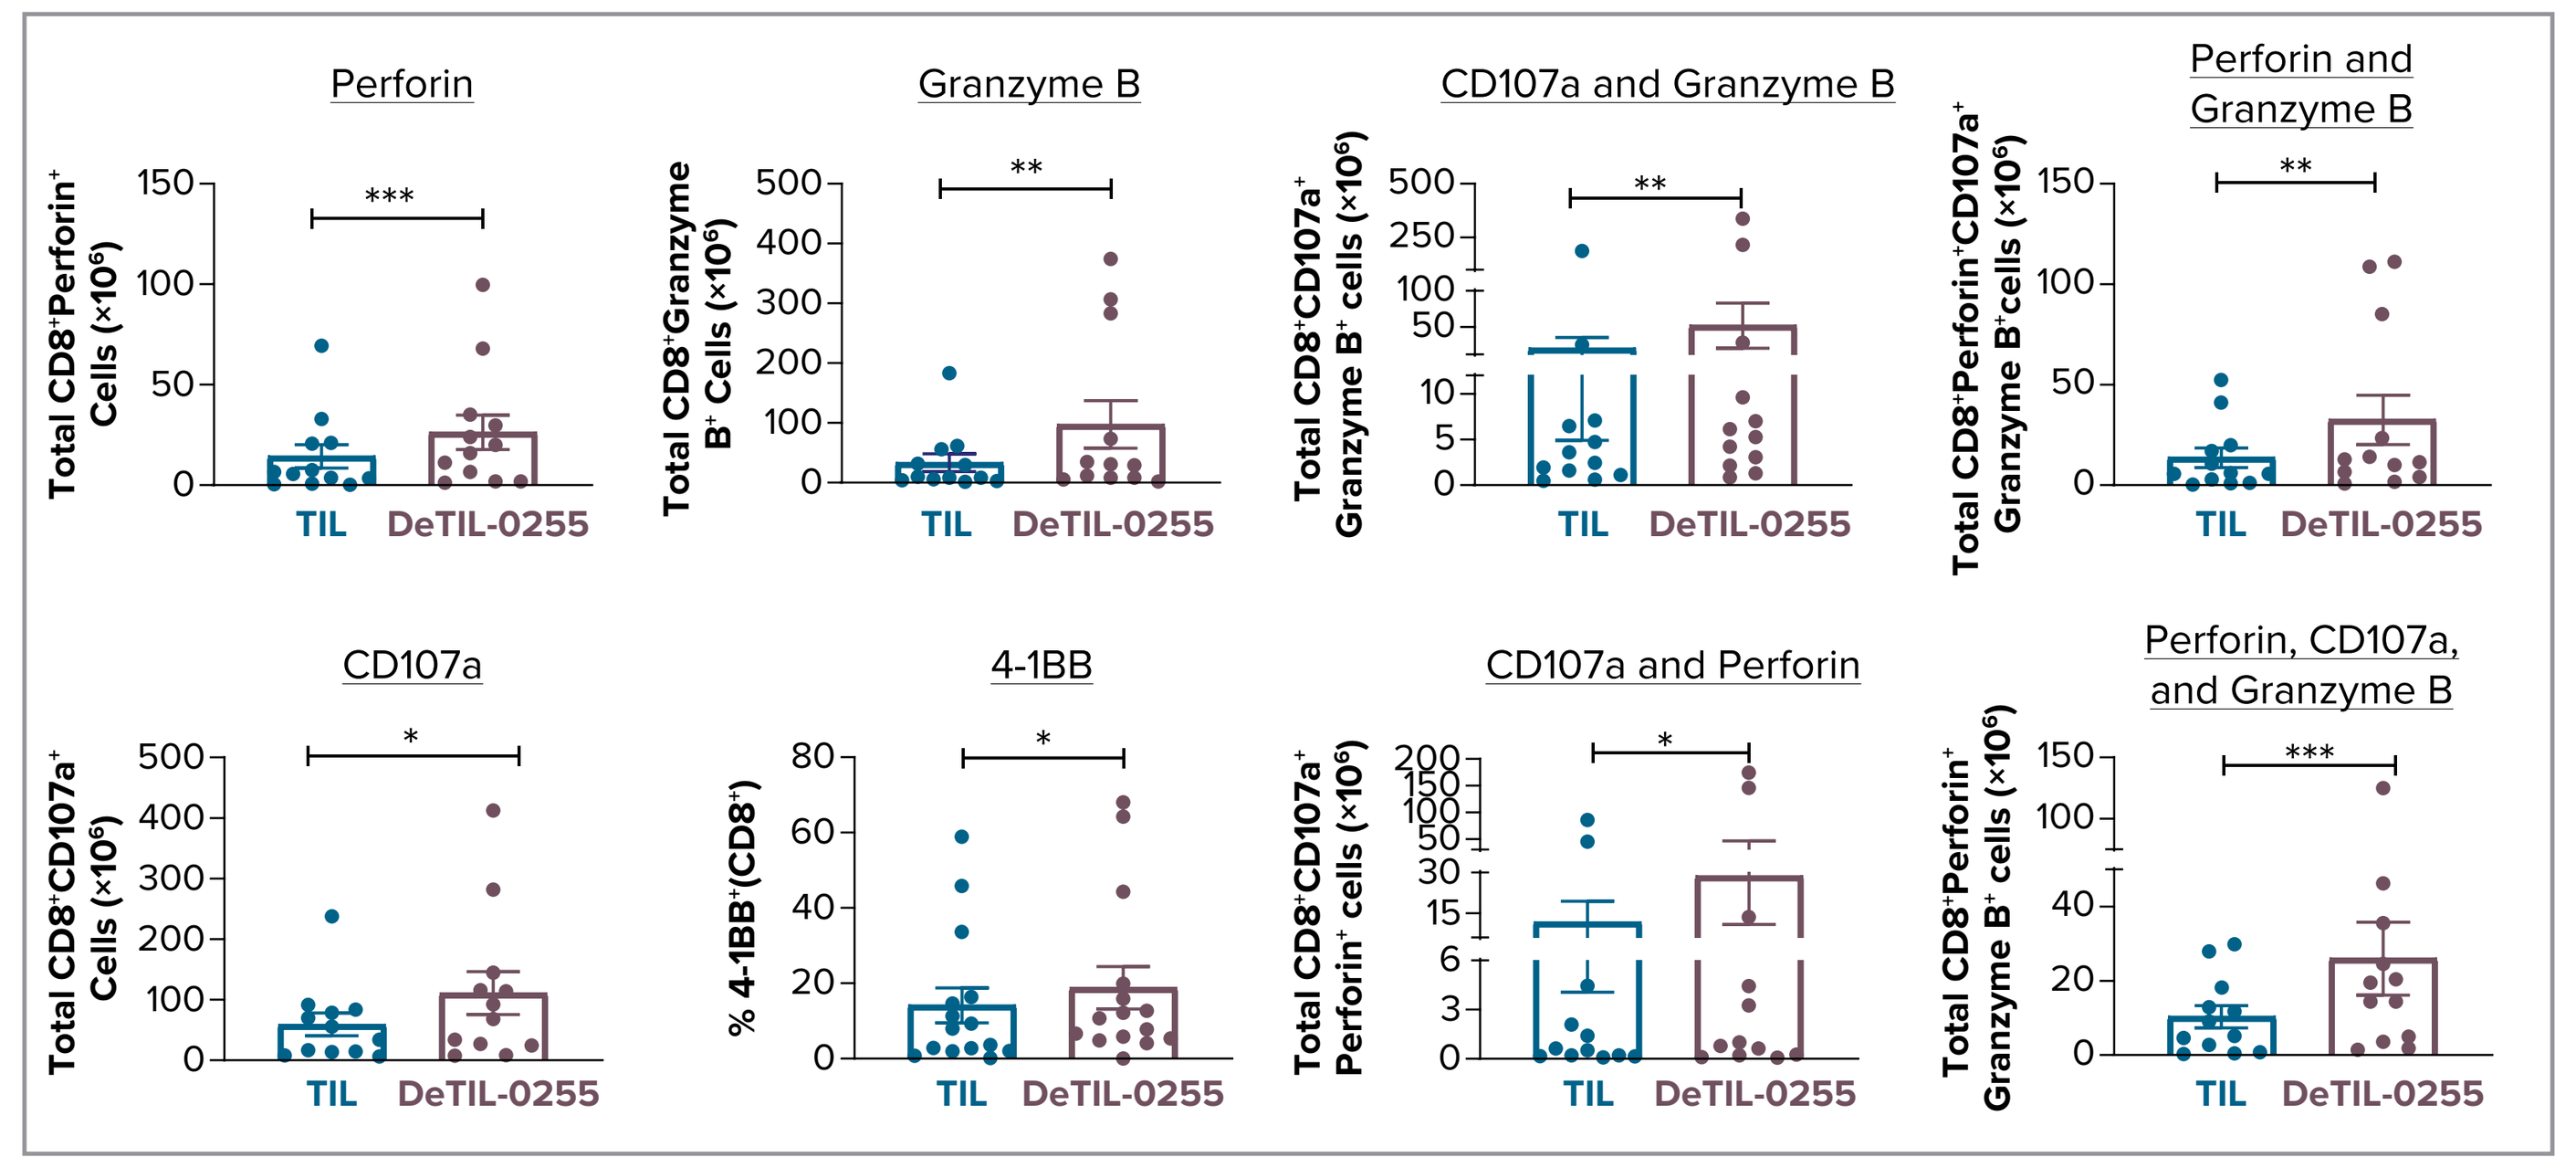
Perforin
Granzyme B
CD107a and Granzyme B
Perforin and Granzyme B
CD107a
4-1BB
CD107a and Perforin
Perforin, CD107a, and Granzyme B
Total CD8⁺Perforin⁺ Cells (×10⁶)
Total CD8⁺Granzyme B⁺ Cells (×10⁶)
Total CD8⁺CD107a⁺ Granzyme B⁺ cells (×10⁶)
Total CD8⁺Perforin⁺CD107a⁺ Granzyme B⁺cells (×10⁶)
Total CD8⁺CD107a⁺ Cells (×10⁶)
% 4-1BB⁺(CD8⁺)
Total CD8⁺CD107a⁺ Perforin⁺ cells (×10⁶)
Total CD8⁺Perforin⁺ Granzyme B⁺ cells (×10⁶)
TIL
DeTIL-0255

Statistical significance was assessed using 2-tailed Wilcoxon signed-rank test; *$P<0.05$, **$P<0.005$, and ***$P<0.001$. Error bars depict standard error of the mean (SEM).
DeTIL-0255, drug-enhanced tumor-infiltrating lymphocyte; TIL, tumor-infiltrating lymphocyte.

- The functional capacity of DeTIL-0255 was differentially enhanced, with significant increases in the absolute numbers of CD8+ T cells expressing intracellular perforin (*P*=0.001), granzyme B (*P*=0.005) and CD107a (*P*=0.01) when comparing DeTIL-0255 with TIL. An increase of CD8+ T cells expressing CD137/4-1BB, a biomarker of CD8+ T-cell tumor reactivity, was also observed (*P*=0.03)

Figure 6. DeTIL-0255 Exhibit Significantly Increased TCR Diversity Compared With TIL

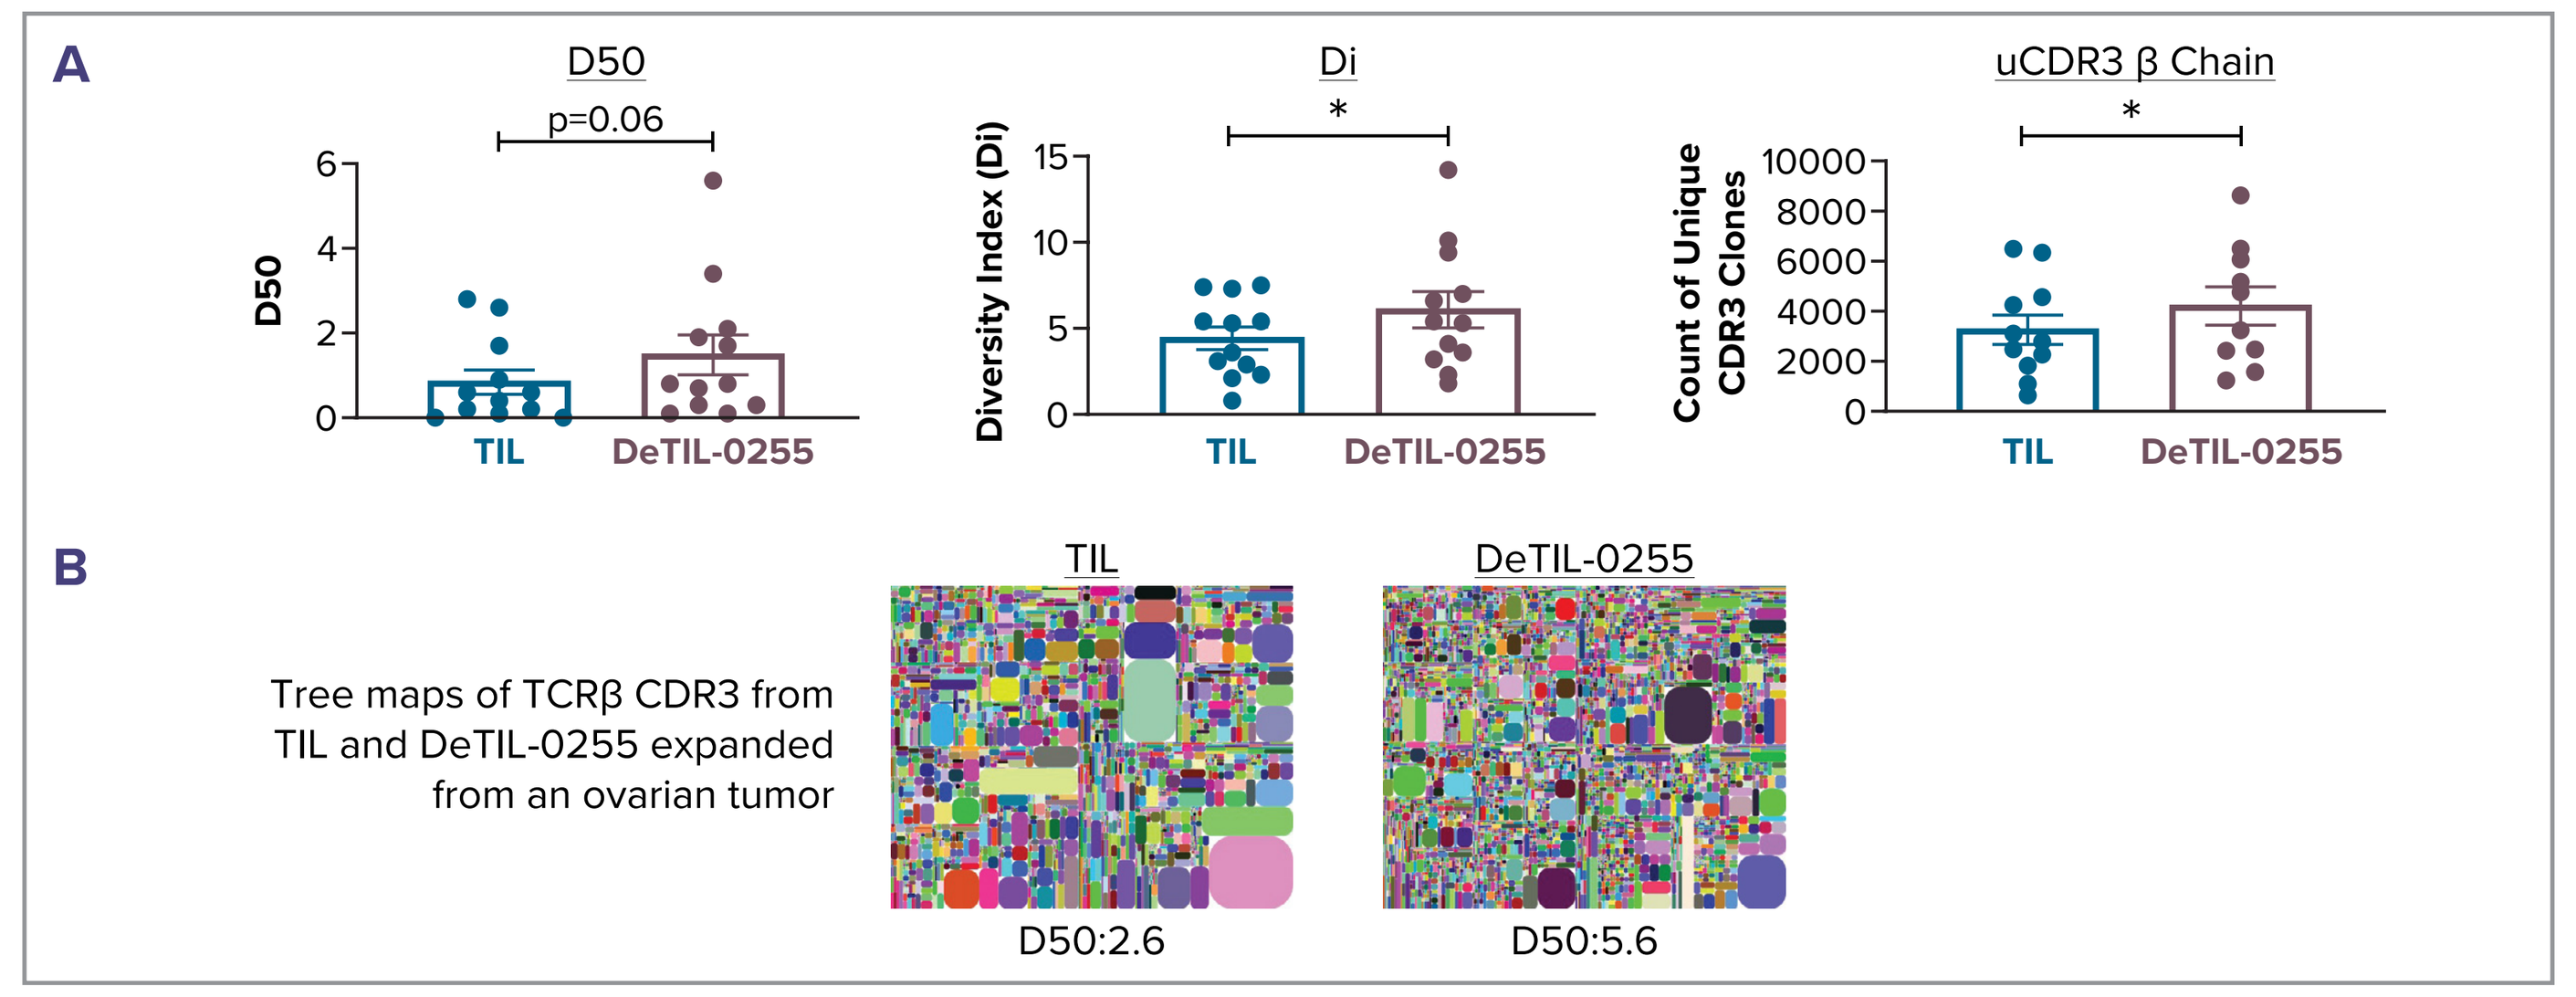
A
D50
p=0.06
Di
*
Diversity Index (Di)
uCDR3 β Chain
*
Count of Unique CDR3 Clones
TIL
DeTIL-0255
B
Tree maps of TCRβ CDR3 from TIL and DeTIL-0255 expanded from an ovarian tumor
TIL
DeTIL-0255
D50:2.6
D50:5.6

TCR repertoire diversity is quantified (A) and illustrated in tree maps where each rounded rectangle represents a unique entry: V–J–uCDR3, and the size of the spot denotes the relative frequency (B). Error bars depict standard error of the mean (SEM).
D50, diversity 50 value; DeTIL-0255, drug-enhanced tumor-infiltrating lymphocyte; Di, diversity index; TCR, T-cell receptor; TIL, tumor-infiltrating lymphocyte.

- TCR repertoire analysis showed that DeTIL-0255 had a significant increase in diversity index (Di) and unique CDR3 β chains in comparison with TIL, suggesting that an increase in CD8+ T-cell effector function and TCR diversity is associated with antitumor effects

Figure 7. DeTIL-0255 Are Less Differentiated and More Cytotoxic Than TIL Cells

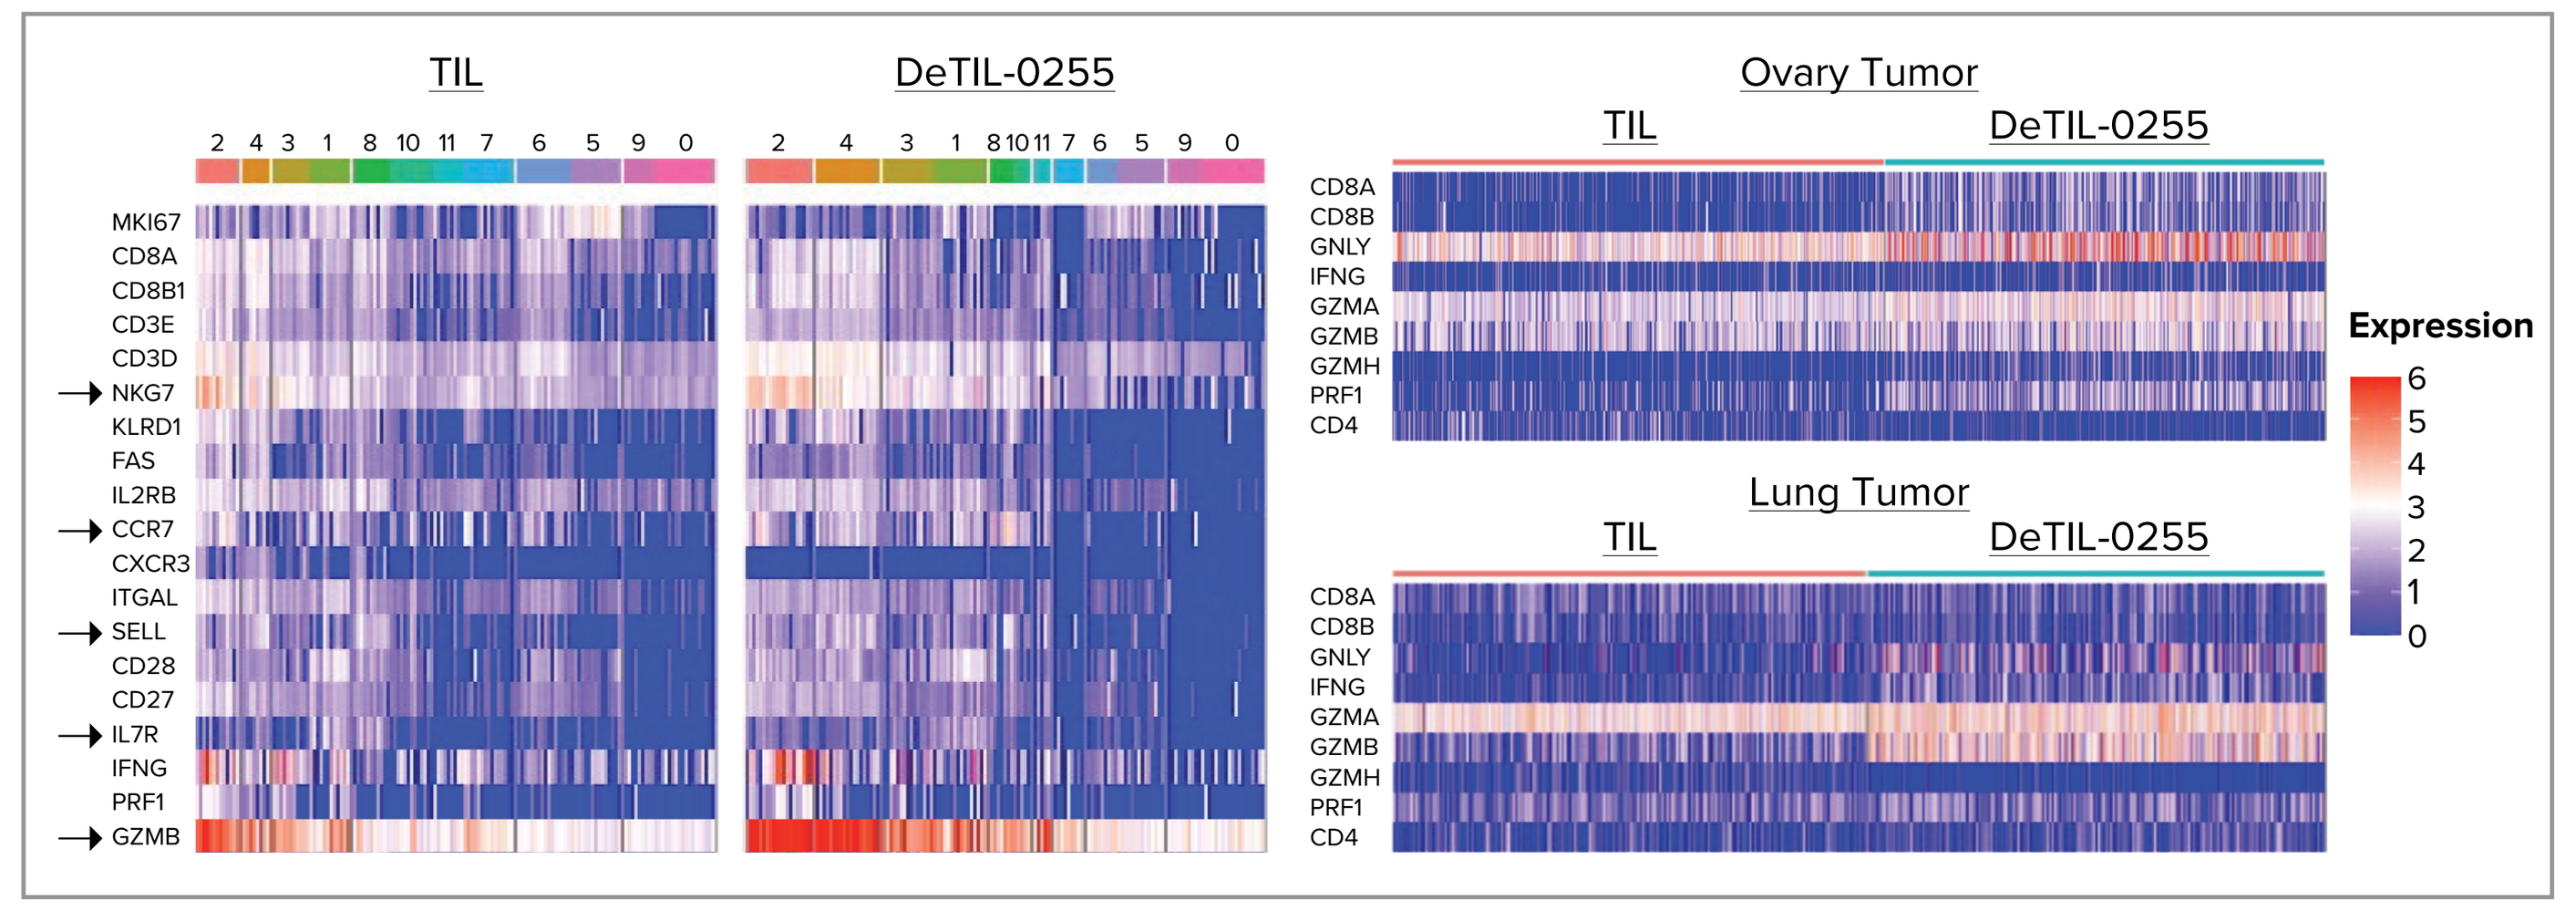
TIL
DeTIL-0255
MKI67
CD8A
CD8B1
CD3E
CD3D
NKG7
KLRD1
FAS
IL2RB
CCR7
CXCR3
ITGAL
SELL
CD28
CD27
IL7R
IFNG
PRF1
GZMB
Ovary Tumor
Lung Tumor
CD8A
CD8B
GNLY
IFNG
GZMA
GZMB
GZMH
PRF1
CD4
Expression

CCR, C-C Motif Chemokine Receptor; CXCR, C-X-C chemokine receptor; CD, cluster of differentiation; DeTIL-0255, drug-enhanced tumor-infiltrating lymphocyte; FAS, Fas cell surface death receptor; GNLY, Granulysin; GZM, granzyme; IFN, interferon; IL, interleukin; KLR, Killer Cell Lectin Like Receptor; MKI, marker of proliferation; NKG, Natural Killer Cell Granule Protein; PRF1, Perforin 1; SELL, selectin L. TIL, tumor-infiltrating lymphocyte.

- Single-cell sequencing analysis demonstrated that DeTIL-0255 expanded in the pre-REP had increased expression of genes associated with stemness (CCR7+,CD62L+[SELL],CD127+[IL-7R]) and cytotoxicity (GNLY+,GZMB+,NKG7+)

Table 1: Summary of DeTIL-0255 Expansion Studies Compared With TIL

Exhaustion	
Marker	**% of CD8+**
Total PD-1+	↓
Total PD-1+ TIM-3+	↓
Total PD-1+ LAG-3+	↓

Cytotoxic Function	
Marker	**Absolute No. of CD8**
CD107a+	↑
GrB+	↑
Perforin+	↑
CD107a+ GrB+	↑
CD107a+ Perforin	↑
GrB+ Perforin	↑
GrB+ Perforin CD107A+	↑

Chemokine Secretion	
Secretion	**pg/mL**
RANTES	↑
MCP-1	↑
IL-8	↑

Cytokine Secretion	
Secretion	**pg/mL**
7 CRS-associated cytokines (IL-2, IL-4, IL-6, IL-9, IL-10, IFN-γ, TNF-α)	–

Tumor Reactivity	
CD8	**% of CD8+**
Total 41BB+	↑

Arrows indicate a statistically significant (*P*<0.05) change in DeTIL-0255 compared with TIL.
CRS, cytokine release syndrome; DeTIL-0255, drug-enhanced tumor-infiltrating lymphocytes; GrB, granzyme B; IFN-γ, interferon gamma; IL, interleukin; LAG-3, lymphocyte-activation gene 3; MCP-1, monocyte chemoattractant protein 1; PD-1, programmed cell death protein 1; TIL, tumor-infiltrating lymphocytes; TNF-α, tumor necrosis factor alpha.

CONCLUSIONS

- Culturing of TIL in the presence of NX-0255 without rhIL-2 resulted in the expansion of cells, with numbers comparable with that of conventionally cultured TIL with rhIL-2
- Furthermore, the addition of NX-0255 in combination with rhIL-2 significantly increased the number of cells expanded compared with conventionally cultured TIL
- DeTIL-0255 expansion resulted in T cells that were significantly less exhausted and more cytotoxic than conventional TIL.
 - TCR repertoire and single-cell sequencing analysis showed that DeTIL-0255 had increased TCR diversity and validated that DeTIL-0255 expressed genes associated with stemness and cytotoxicity
- DeTIL-0255 significantly increased chemokine and cytokine secretion upon TCR and CD28 costimulation but did not elicit significantly more cytokines than TIL, alleviating additional safety concerns with the addition of NX-0255
- Increased expansion, reduced exhaustion, increased cytotoxicity, increased TCR diversity, and gene expression consistent with stemness all predict broader functional activity of DeTIL-0255 compared with conventional TIL, potentially conferring improved antitumor activity.
- Taken together, these data support the clinical development of DeTIL-0255 for the treatment of patients with cancer

REFERENCES

1. Krishna S, et al. *Science*. 2020;370:1328-1334.
2. Stevanović S, et al. *Clin Cancer Res*. 2019;25:1486-1493.
3. Chandran SS, et al. *Lancet* Oncol. 2017;18:792-802.
4. Goff SL, et al. *N Engl J Med*. 1988;319:1676-1680.
5. Met Ö, et al. *Semin Immunopathol*. 2019;41:49-58.
6. Bachmaier K, et al. *Nature*. 2000;403:211-216.
7. Chiang YJ, et al. *Nature*. 2000;403:216-220.

ACKNOWLEDGMENTS

Medical writing support was provided by Sarah Whelan of Nurix Therapeutics and Phylicia Aaron, PhD, of Second City Science, with funding from Nurix Therapeutics

DISCLOSURES

SW: Employment with and stock or other ownership in Nurix Therapeutics; patents, royalties, or other intellectual property with Nurix Therapeutics and Compugen Ltd. **JG:** Stock or other ownership in Chemocentryx. **MM:** Employment with and stock or other ownership in Nurix Therapeutics and Spotlight Therapeutics. **TC:** Employment with Nurix Therapeutics, Amgen, and Bristol Meyer Squibb; stock or other ownership with and patents, royalties, or other intellectual property with Nurix Therapeutics. **FC:** Employment with and stock or other ownership with Nurix Therapeutics. **AT-M:** Employment with and stock or other ownership in Nurix Therapeutics and Gilead. **JP:** Employment with Nurix Therapeutics; stock or other ownership in Nurix Therapeutics; patents or other intellectual property with Five Prime Therapeutics. **GH:** Employment and leadership with Nurix Therapeutics; stock or other ownership with Nurix Therapeutics and Lexicon Pharmaceuticals. **MTL:** Employment, leadership, stock or other ownership, honoraria, consulting or advisory role, research funding, patents, royalties, other intellectual property, and travel, accommodations, expenses, and other relationships with Nurix Therapeutics; honoraria with Nurix Therapeutic and Adicet. **ATS:** Employment, leadership, and stock or other ownership with Nurix Therapeutics.

Copies of this presentation obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

Presented at the Society for Immunotherapy of Cancer (SITC) Annual Meeting, November 10-14, 2021.