Modeling Limit of Detection for Dominant T Cell Receptor Clones Reveals Comparable Measures Across RNA and DNA Based Sequencing

Sagar N. Sharma BS[1,2], Samuel C. Butler BS[1], Jennifer M. Bone PhD[1], Pranav Murthy MS[1], Michael T. Lotze MD[1,2]

[1]Department of Surgery, University of Pittsburgh, Pittsburgh, PA, USA; [2]Nurix Therapeutics, San Francisco, CA

Dominant Tumor-Infiltrating Lymphocytes (TIL) Clones are Tumor Reactive

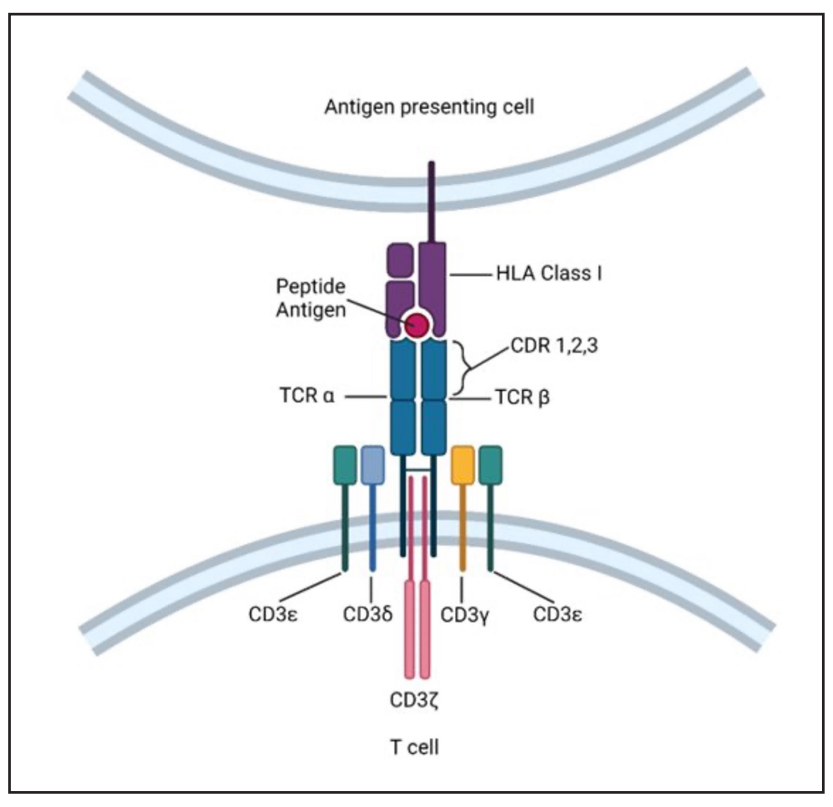
Antigen presenting cell
Peptide Antigen
HLA Class I
CDR 1,2,3
TCR α
TCR β
CD3ε
CD3δ
CD3γ
CD3ε
CD3ζ
T cell

Figure 1. The T Cell Receptor (TCR) complex is a heterodimer composed of an α and β chain along with CD3-associated proteins. Complimentary-determining regions (CDRs) are related to binding with peptide antigens loaded on the human leukocyte antigen (HLA) complex. Sequencing of the CDR3 region of the β chain allows for identification of unique clones and thus quantification of the T cell repertoire.

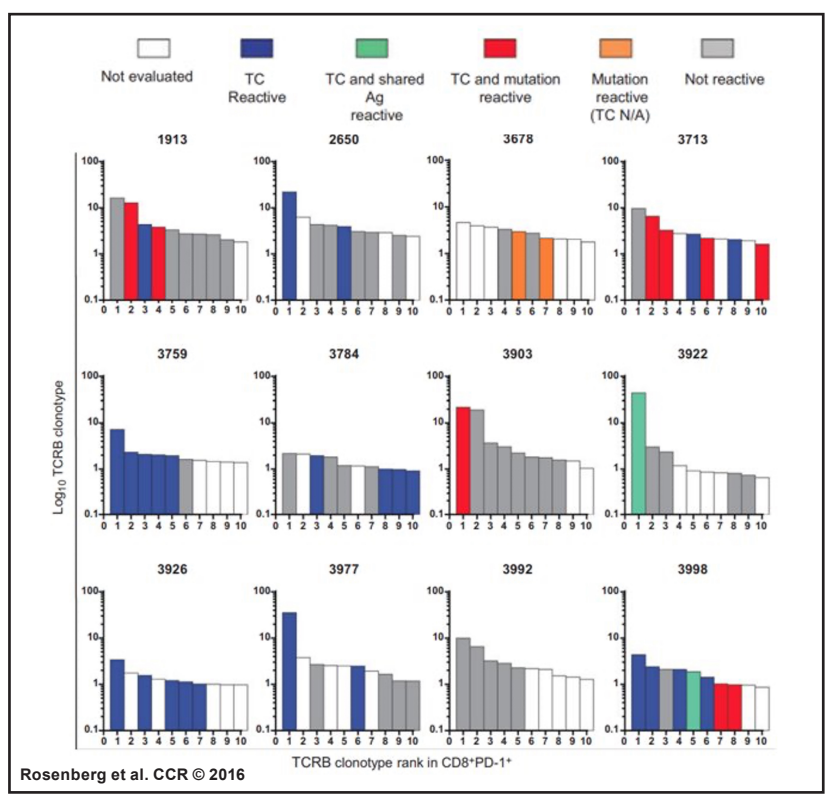
Not evaluated
TC Reactive
TC and shared Ag reactive
TC and mutation reactive
Mutation reactive (TC N/A)
Not reactive
1913
2650
3678
3713
3759
3784
3903
3922
3926
3977
3992
3998
Log10 TCRB clonotype
TCRB clonotype rank in CD8+PD-1+
Rosenberg et al. CCR © 2016

Figure 2. CD8+ PD-1+ T cells were isolated from tumor digest obtained from 12 patients with metastatic melanoma. DNA based β chain sequencing was combined with pairSEQ to identify the cells' αβ pairing. Transduction of top 10 most abundant TCRs revealed that 11 out of 12 patients possess at least 2 or more of these TCRs that show *in vitro* reactivity.

TCRβ Sequencing: RNA vs DNA

Method	RNA[1]	DNA[2]
Minimum input quantity for PBMC	300-500ng	3380-4700ng
CDR3 sequencing	Functional sequences	Includes introns, exons, and nonproductive sequences
CDR3 expression	Multiple transcripts per cell	Single copy per cell
Error control	Unique Molecular Identifiers (UMI)	Synthetic TCRG template
Cost per sample	$1375	$2200
TCR chains	β α δ γ	β, α/δ, γ
Time to service	2 weeks	2 weeks
Data sharing	By request	IMMUNE-ACCESS Public Database

[1]iRepertoire, Inc. [2]Adaptive Biotechnologies

Table 1. A comparison of some of the key similarities and differences in RNA vs DNA based sequencing methods. Calculation of pricing for Adaptive includes computational analysis and expedited data fee. Adaptive offers services for either β, α and δ, or γ chain sequencing, whereas iRepertoire offers any custom TCR chain combination

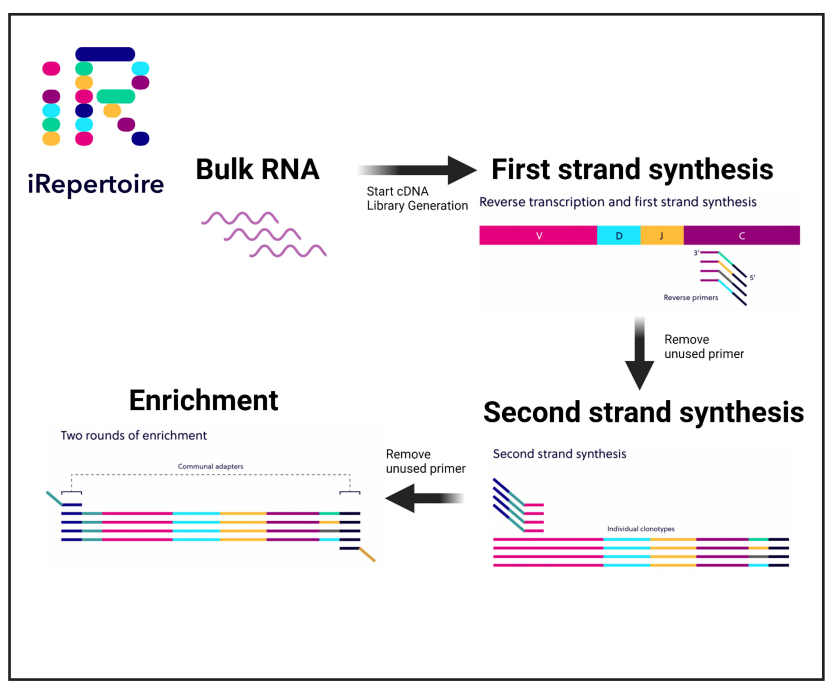
iRepertoire
Bulk RNA
Start cDNA Library Generation
First strand synthesis
Reverse transcription and first strand synthesis
Remove unused primer
Second strand synthesis
Second strand synthesis
Enrichment
Two rounds of enrichment
Remove unused primer

Figure 3. Sample bulk RNA is first reverse transcribed into cDNA for the synthesis of the first strand using a gene-specific primer mix. Physical and chemical means remove residual primers to reduce the number of primer-dimers. A second PCR round completes the cDNA synthesis followed by another wash step. Two rounds of enrichment are completed using the communal adapter sequences introduced earlier before pooling for sequencing. Unique molecule identifiers (UMI) are included in the initial PCR steps to reduce amplification bias.

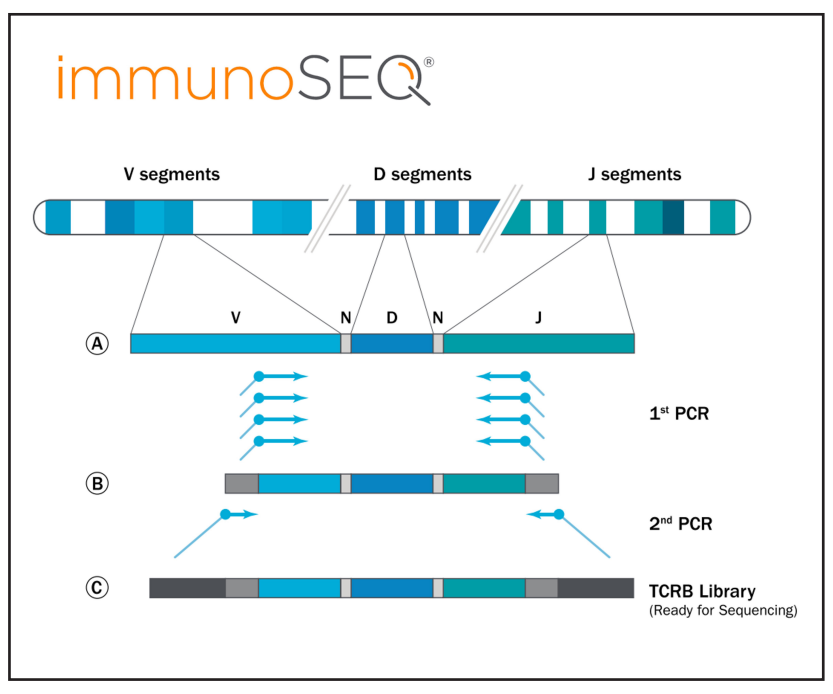
immunoSEQ
V segments
D segments
J segments
V N D N J
1st PCR
2nd PCR
TCRB Library (Ready for Sequencing)

Figure 4. A synthetic TCRG repertoire was created to represent all possible VJ-gene combinations to titrate primers before the first round of PCR to reduce amplification bias. Sample bulk DNA is mixed with multiplex V- and J- primers before going through two rounds of PCR and the addition of barcodes and adaptor sequences for sequencing. Post amplification, the synthetic template can be further used to computationally correct any residual bias.

Experimental Overview: Modeling Limit of Detection

Sample	MNCs	PBMCs	Dilution Factor
1	5 million	0	-
2	0	5 million	-
3	5	5 million	10^6
4	50	5 million	10^5
5	500	5 million	10^4
6	5000	5 million	10^3

Table 2. Mononuclear cells (MNCs) from a single donor were serially diluted in a diverse pool of peripheral blood mononuclear cells (PBMCs). Sample 1 and 2 were used as reference repertoires for MNCs and PBMCs, respectively.

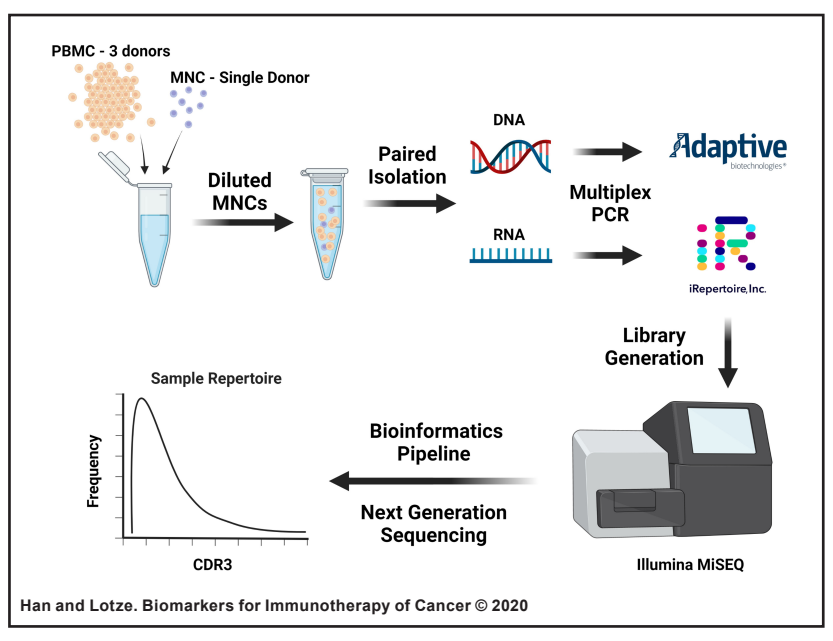
PBMC - 3 donors
MNC - Single Donor
Diluted MNCs
Paired Isolation
DNA
RNA
Multiplex PCR
Adaptive
iRepertoire,Inc.
Library Generation
Illumina MiSEQ
Next Generation Sequencing
Bioinformatics Pipeline
Sample Repertoire
Frequency
CDR3
Han and Lotze. Biomarkers for Immunotherapy of Cancer © 2020

Figure 5. RNA and DNA was isolated and sent for cDNA library generation and Next-Generation Sequencing (NGS) by iRepertoire and Adaptive, respectively. The proprietary bioinformatic protocols were followed to generate CDR3 maps that contained the amino acid sequence and its respective frequency within the sample.

Top 10 Most Abundant Clones Capture Head of CDR3 Distribution

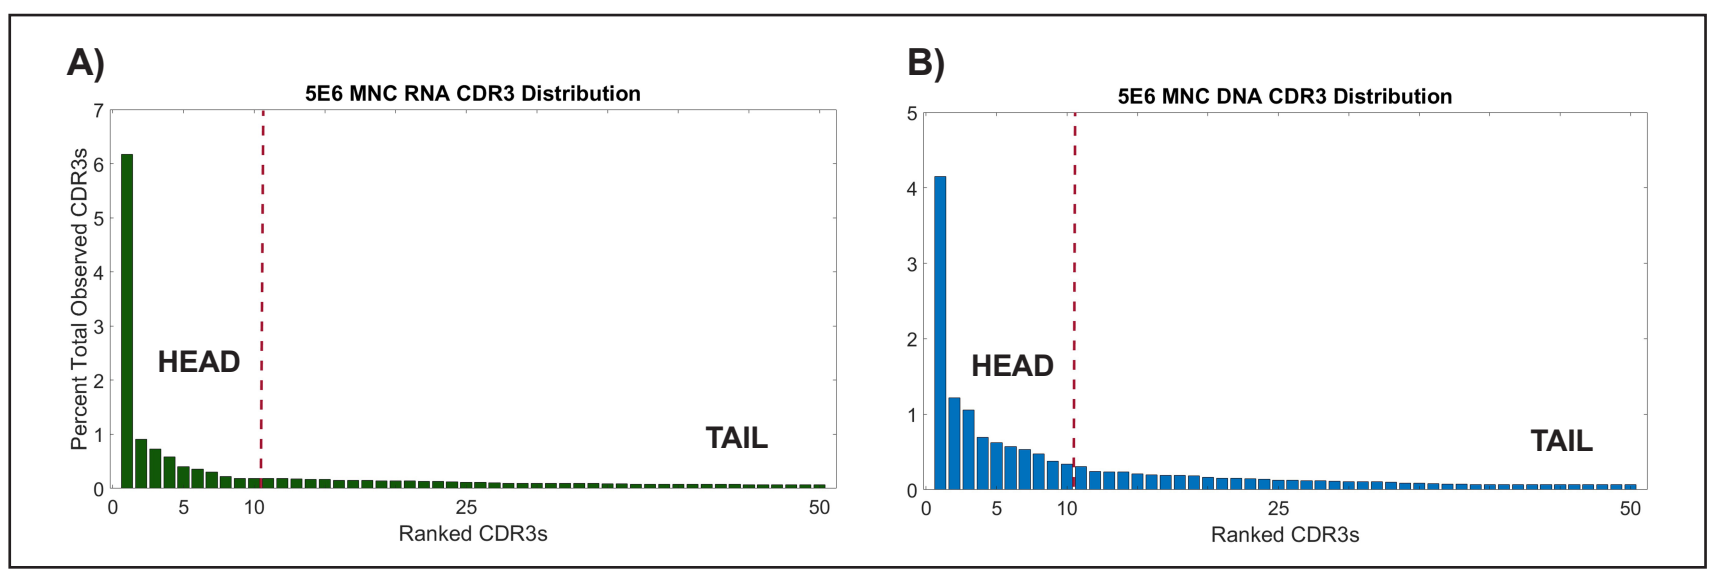
A)
5E6 MNC RNA CDR3 Distribution
Percent Total Observed CDR3s
HEAD
TAIL
Ranked CDR3s
B)
5E6 MNC DNA CDR3 Distribution
HEAD
TAIL
Ranked CDR3s

Figure 6. Shown are the top 50 most abundant CDR3 sequences for the undiluted RNA (**A**) and DNA (**B**) MNC samples. The x-axis denotes the rank or relative abundance of the CDR3, and the y-axis shows the percent of the sample's repertoire that CDR3 sequence occupies. For example, the most abundant MNC clone in the RNA CDR3 distribution makes up 6.177% of the total reads from that sample. The dashed red line denotes the head (left) or top 10 most abundant CDR3s and the tail (right) illustrates the decreasingly abundant CDR3s. Clones to the left of the cutoff are deemed dominant for the purposes of all following analyses.

Dominant CDR3s are Reproducibly Detected

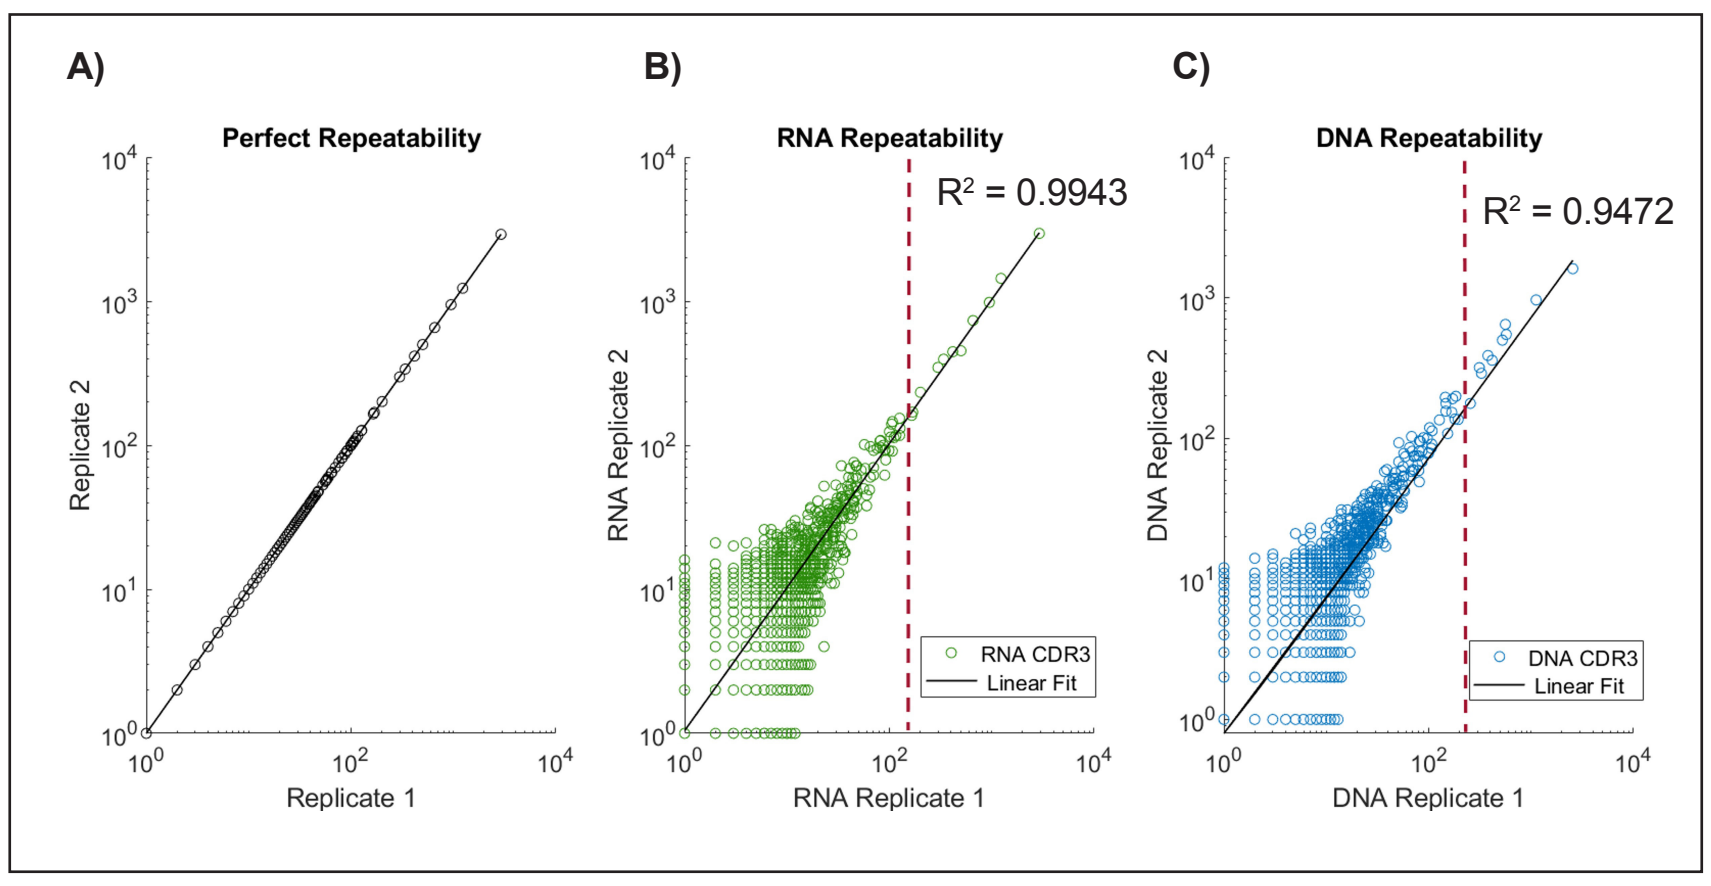
A)
Perfect Repeatability
Replicate 2
Replicate 1
B)
RNA Repeatability
R² = 0.9943
RNA Replicate 2
RNA CDR3
Linear Fit
RNA Replicate 1
C)
DNA Repeatability
R² = 0.9472
DNA Replicate 2
DNA CDR3
Linear Fit
DNA Replicate 1

Figure 7. Open circles on plots (**A-C**) represent a unique CDR3 sequence where the x- or y-value would be its abundance on a log scale in replicate 1 and 2, respectively. The right most CDR3s are the most abundant. Perfect reproducibility (**A**) is a straight line through the origin where the same CDR3s have the exact same abundance in each replicate. Plots for RNA (**B**) and DNA (**C**) show significant deviation from linearity with CDR3s that have a low abundance. The dashed red line divides each plot into the top 10 clones (right) and the rest (left). In both RNA and DNA, the top 10 clones were highly linear (R^2 = 0.9943 and 0.9472, respectively). The rest of the clones for RNA and DNA were poorly correlated between the replicates (R^2 = 0.5001 and 0.5991, respectively).

RNA and DNA Detect Dominant MNC CDR3s and Maintain Sample Hierarchy

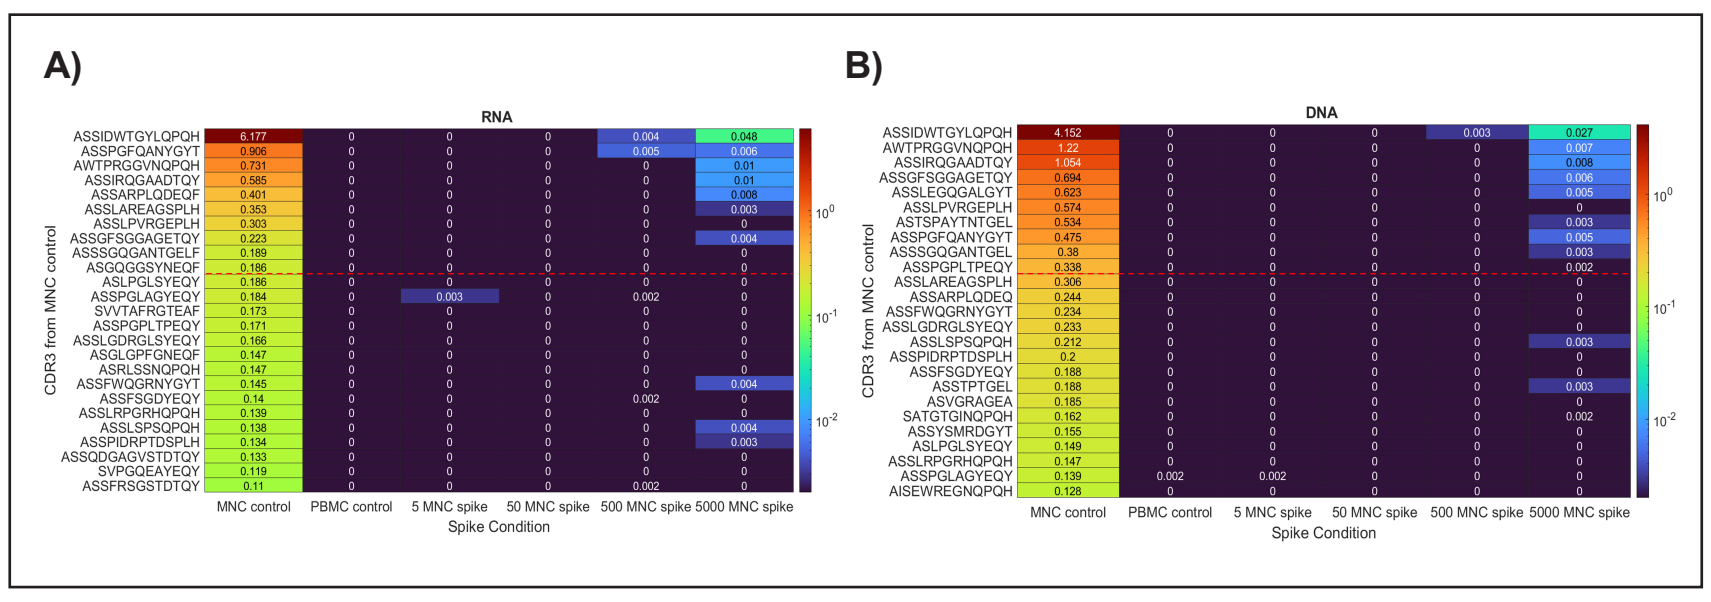
A)
RNA
CDR3 from MNC control
ASSIDWTGYLQPQH
ASSPGFQANYGYT
AWTPRGGVNQPQH
ASSIRQGAADTQY
ASSARPLQDEQF
ASSLAREAGSPLH
ASSLPVRGEPLH
ASSGFSGGAGETQY
ASSSGQGANTGELF
ASGQGGSYNEQF
ASLPGLSYEQY
ASSPGLAGYEQY
SVVTAFRGTEAF
ASSPGPLTPEQY
ASSLGDRGLSYEQY
ASGLGPFGNEQF
ASRLSSNQPQH
ASSFWQGRNYGYT
ASSFSGDYEQY
ASSLRPGRHQPQH
ASSLSPSQPQH
ASSPIDRPTDSPLH
ASSQDGAGVSTDTQY
SVPGQEAYEQY
ASSFRSGSTDTQY
MNC control
PBMC control
5 MNC spike
50 MNC spike
500 MNC spike
5000 MNC spike
Spike Condition
B)
DNA
CDR3 from MNC control
ASSIDWTGYLQPQH
AWTPRGGVNQPQH
ASSIRQGAADTQY
ASSGFSGGAGETQY
ASSLEGQGALGYT
ASSLPVRGEPLH
ASTSPAYTNTGEL
ASSPGFQANYGYT
ASSSGQGANTGEL
ASSPGPLTPEQY
ASSLAREAGSPLH
ASSARPLQDEQ
ASSFWQGRNYGYT
ASSLGDRGLSYEQY
ASSLSPSQPQH
ASSPIDRPTDSPLH
ASSFSGDYEQY
ASSTPTGEL
ASVGRAGEA
SATGTGINQPQH
ASSYSMRDGYT
ASLPGLSYEQY
ASSLRPGRHQPQH
ASSPGLAGYEQY
AISEWREGNQPQH
MNC control
PBMC control
5 MNC spike
50 MNC spike
500 MNC spike
5000 MNC spike
Spike Condition

Figure 8. Sharing of the 25 most abundant clones from the undiluted reference MNC sample is shown. Each value shown is the percent of that sample's repertoire that is occupied by the specific CDR3 amino acid sequence that borders the left side of the MNC control column. The dashed red line delineates the top 10 (top) from the rest of the clones (bottom). All values ≤ 0.001% were considered equal to zero for visual clarity. There is no significant sharing between the MNC and PBMC reference columns for both RNA (**A**) and DNA (**B**). Samples with MNCs diluted at factors of 10^6 and 10^5 did not harbor detectable dominant clones. The most dominant clonotype from the MNC control (ASSIDWTGYLQPQH) was the most dominant of shared clones found in the 5000(1:10^3) and 500(1:10^4) MNC spike in samples for both RNA and DNA. The hierarchy of the original MNC sample is maintained in both the 10^3 and 10^4 diluted MNC sample for RNA and DNA. Clones outside the top 10 had little to no detectable signal.

Summary

Conclusions

- The top 10 CDR3s are reproducible and capture a substantial portion of the head of the long tail CDR3 distribution.
- The LOD for diluted populations is 1:10^3 for the most abundant clones and 1:10^4 for the top clone.
- Detection of dominant clones is consistent across DNA and RNA based methods.

Limitations

- Dilution factors 10^2 and 10^1 were not tested and may have provided a clearer illustration of sample hierarchy maintenance.
- Cutoff for dominant clone classification should be validated on another test cohort.

Future Directions

- Due to codon degeneracy, use of nucleotide sequences may be a more accurate method of detecting shared clones.
- Screening shared TCRs through a public database may help ensure shared clones are truly private.

Presented at the 24th Annual TRCCC Meeting, June 6-8, 2022, Seven Springs PA, USA