Universal Expansion of CBL-B–inhibited Tumor-Infiltrating Lymphocytes, DeTIL-0255, from Women with Ovarian Cancer: Process Validation

Pranav Murthy,[1] Nell Narasappa,[1] Xiaoyan Liang,[1] Xianzhu Wu,[1] Irene Luu,[1] Jeevitha Jeevan,[1] Sagar Sharma,[1] Alison Ross,[1] Caleb Lampenfeld,[1] Sam Zahn,[1] Thomas Musial,[1] Jennifer Bone,[1] John Nakayama,[2] David L. Bartlett,[2] Jocelyn S. Chapman,[3] Greta Garrido,[1] Nicholas Shinners,[1] Arthur T. Sands,[1] Michael Blackton,[1] Ena Wang,[1] and Michael T. Lotze[1]

[1]Nurix Therapeutics, San Francisco, CA, USA; [2]Allegheny Health Network Cancer Institute, Pittsburgh, PA, USA; [3]UCSF Medical Center, San Francisco, CA, USA

361

Background

Ovarian cancer immunotherapy clinical response dependent on TIL CD28 signaling and CD226

- Tumor-infiltrating lymphocytes (TIL), especially stem-like tissue resident memory T cells, are a positive prognostic factor in patients with ovarian cancer.[1-3]
- Immune checkpoint inhibition has yielded limited durable clinical responses in patients with recurrent ovarian cancer:
 - KEYNOTE-100 phase II study: pembrolizumab:[4]
 - 1–3 prior therapies (n=285): objective response rate (ORR) 7.4%; median progression-free survival (PFS) 2.1 months.
 - 4–6 prior therapies (n=91): ORR 9.9%; median PFS 2.1 months.
 - NRG Oncology randomized phase II study:[5]
 - Nivolumab (n=49): ORR 12.2%; median PFS 3.9 months.
 - Nivolumab + ipilimumab (n=51): ORR 31.4%; median PFS 2 months.
 - Response to programmed cell death protein-1 (PD-1) inhibition is dependent on antigen-presenting cells (APC) – TIL CD28 signaling[6] and TIL CD226.[7]

Limited response to conventional TIL therapy in patients with ovarian cancer in published clinical trials

- Conventional TIL therapy has demonstrated durable complete responses in several solid tumors, including metastatic melanoma,[8] cervical,[9] lung,[10] and breast cancer.[11]
- Conversely, in two clinical trials of conventional TIL therapy in patients with advanced ovarian cancer, limited clinical responses have been observed (**Table 1**).[12,13]
- We hypothesize that expanding TIL in a shorter manufacturing time, yielding enhanced memory phenotype and reduced exhaustion, will improve clinical responses.

Table 1. Conventional TIL therapy in patients with ovarian cancer: published clinical trials

		NCT02482090	NCT03287674
TIL therapy regimen		Resection + NMA + TIL + IL-2	αCTLA-4 + Resection + NMA + TIL + IL-2 + αPD-1
Median lines of prior therapy (range)		3 (2–6)	4 (3–7)
Median total TIL manufacturing time, days (range)		38.5 (24–47)	40 (32–56)
Median composition (range)	CD3% (of singlets)	98.3 (95.5–99.4)	99.3 (93.6–99.7)
	CD4% (of CD3%)	58.3 (23.4–82.1)	41.0 (7.8–96.2)
	CD8% (of CD3%)	37.4 (16.0–75.5)	50.3 (3.1–88.5)
Median T cell memory (range)	T_{CM}% (of CD4)	0.4 (0.1–0.7)	NR (<5%)
	T_{EM}% (of CD4)	99.6 (99.3–99.9)	NR (>70%)
	T_{CM}% (of CD8)	8.2 (3.1–13.0)	NR (<5%)
	T_{EM}% (of CD8)	91.1 (87.0–96.9)	NR (>70%)
	PD-1% (of CD4)	8.7 (1.5–18.7)	13.5 (NR)
	LAG-3% (of CD4)	85.2 (65.9–93.6)	43.6 (NR)
	PD-1% (of CD8)	16.4 (6.0–51.5)	34.8 (NR)
	LAG-3% (of CD8)	98.2 (95.6–99.8)	94.1 (NR)
Best clinical response		SD (n=6)	PR (n=1); SD (n=5)
Reference		12	13

Abbreviations: αCTLA-4, anti-CTLA-4; αPD-1, anti-PD-1; IL-2, interleukin-2; LAG-3, lymphocyte activation gene 3; NMA, non-myeloablative chemotherapy; NR, not reported; PD-1, programmed cell death protein-1; PR, partial response; SD, stable disease; T_{CM}, T central memory; T_{EM}, T effector memory; TIL, tumor-infiltrating lymphocytes.

CBL-B: a modulator of T cell activation and novel immunotherapy target

- T cell receptor (TCR) engagement (Signal 1) in the absence of CD28 (Signal 2) results in T cell anergy.
- Casitas B lineage lymphoma-B (CBL-B) is an E3 ubiquitin ligase that regulates the CD28 dependence of T cell activation.[14]
- Mice deficient in CBL-B demonstrate enhanced signal dependent T cell activation, reduced Treg, myeloid-derived suppressor cells (MDSC) and TGF-β dependent immunosuppression, and robust T and NK cell dependent anti-tumor activity.
- Inhibition of CBL-B with the small molecule inhibitor NX-0255 enhances signal dependent T cell interleukin-2 and interferon-γ production and proliferation (**Figure 1**).
- *Ex vivo* culture of human TIL with NX-0255 increases TIL expansion, stemness, TCR diversity, and cytolytic potential (**Figure 2**).[15]
- *Ex vivo* inhibition of CBL-B with NX-0255 enhances the anti-tumor activity and persistence of adoptively transferred CD8 cells in pre-clinical models.[16]

Figure 1. CBL-B: A modulator of T-cell activation and novel target for immuno-oncology

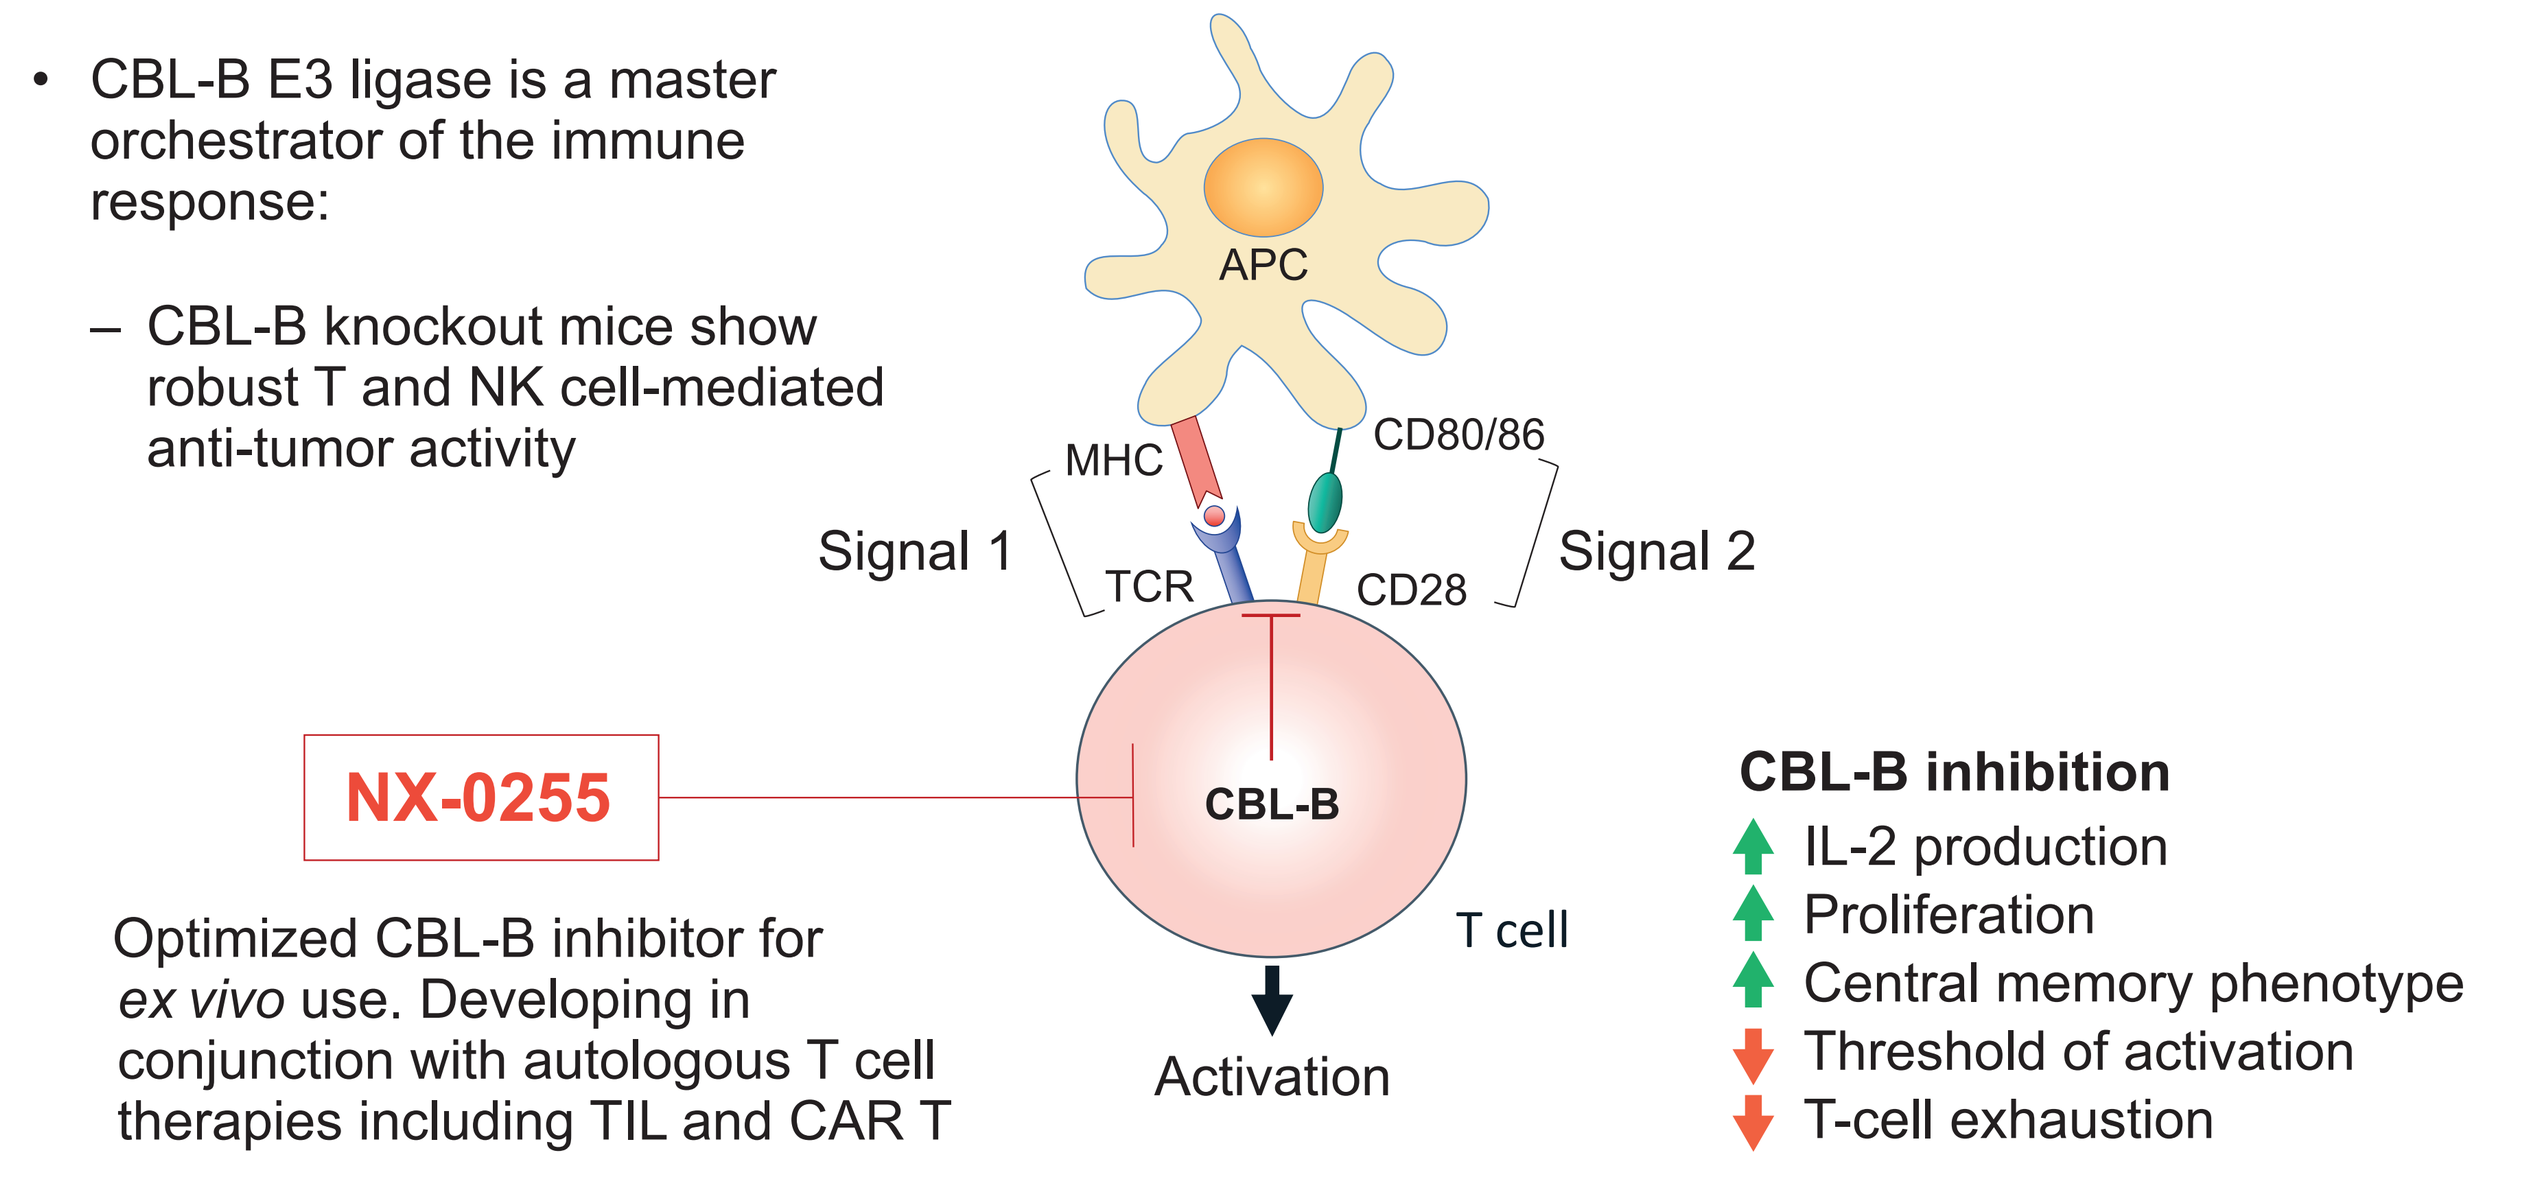

• CBL-B E3 ligase is a master orchestrator of the immune response:
– CBL-B knockout mice show robust T and NK cell-mediated anti-tumor activity
APC
MHC
CD80/86
Signal 1
TCR
CD28
Signal 2
NX-0255
CBL-B
T cell
Optimized CBL-B inhibitor for ex vivo use. Developing in conjunction with autologous T cell therapies including TIL and CAR T
Activation
CBL-B inhibition
IL-2 production
Proliferation
Central memory phenotype
Threshold of activation
T-cell exhaustion

Abbreviations: CBL-B, Casitas B-lineage lymphoma B; IL-2, interleukin-2; TCR, T-cell receptor; TIL, tumor-infiltrating lymphocytes

Figure 2. Manufacturing of drug-enhanced tumor-infiltrating lymphocytes (DeTIL-0255)

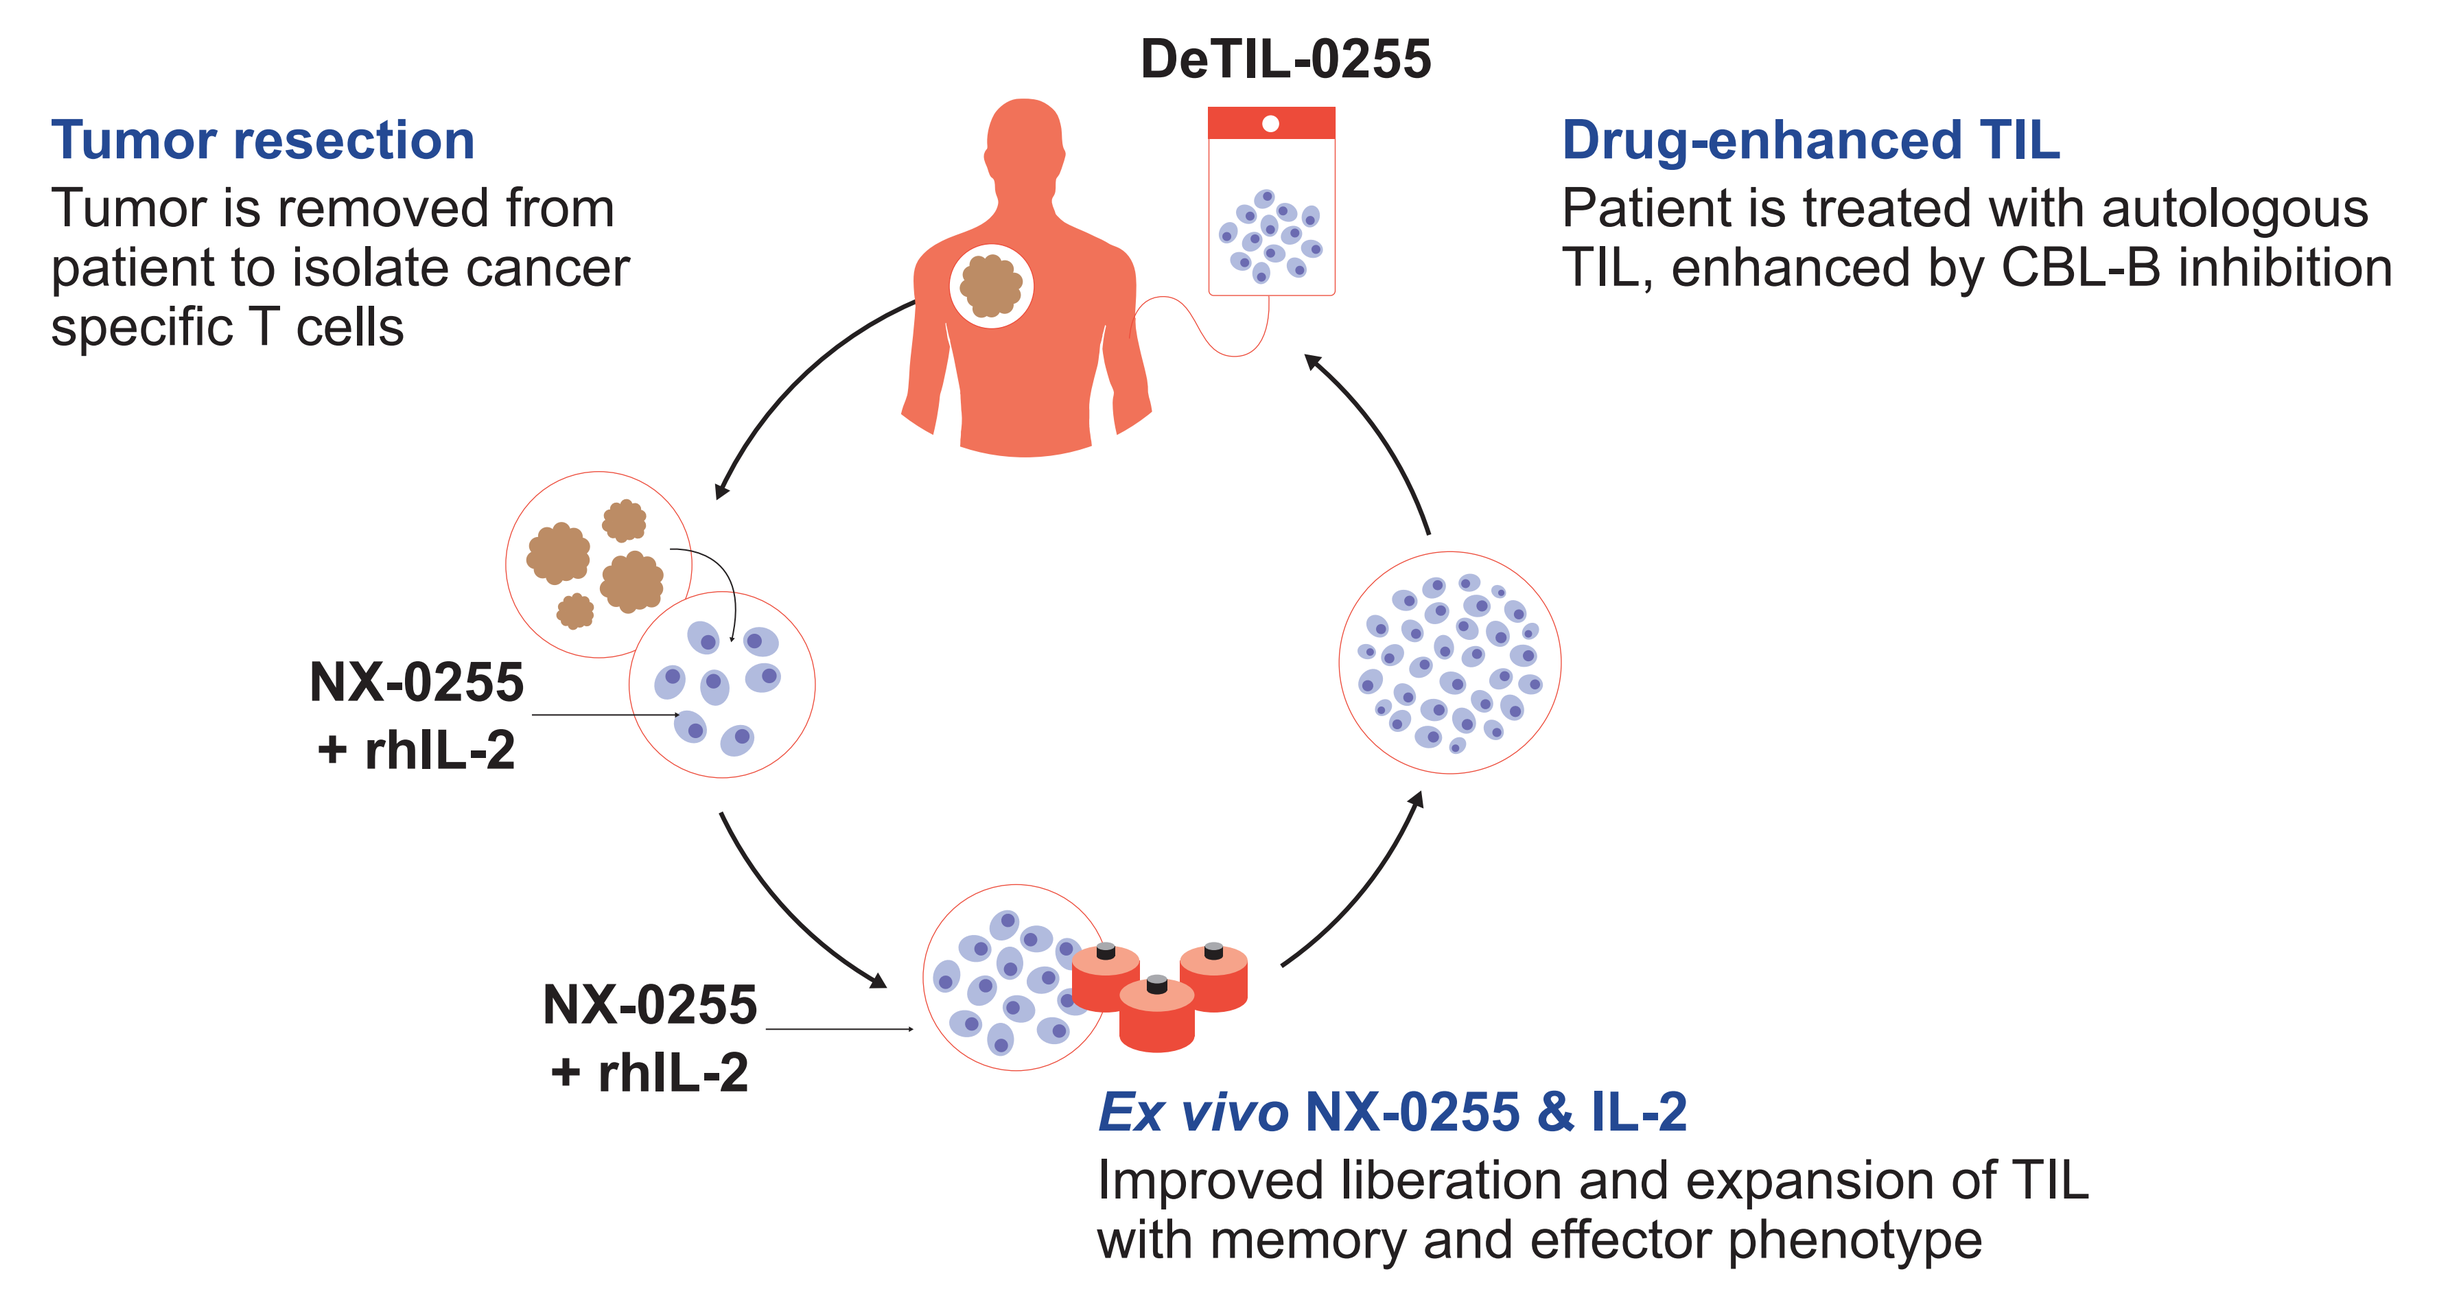

Tumor resection
Tumor is removed from patient to isolate cancer specific T cells
DeTIL-0255
Drug-enhanced TIL
Patient is treated with autologous TIL, enhanced by CBL-B inhibition
NX-0255 + rhIL-2
NX-0255 + rhIL-2
Ex vivo NX-0255 & IL-2
Improved liberation and expansion of TIL with memory and effector phenotype

Abbreviations: CBL-B, Casitas B lineage lymphoma-B; (rh)IL-2, (recombinant human) interleukin-2; TIL, tumor-infiltrating lymphocytes.

Objectives

- Characterize pre-clinical and early clinical manufacturing experience of DeTIL-0255 from consenting donors (n=18) with pathologically proven ovarian cancer.

Results

- Robust expansion of ovarian DeTIL-0255 in 22 days across all subsets (**Figure 3**).

Figure 3. DeTIL-0255 22-day expansion summary

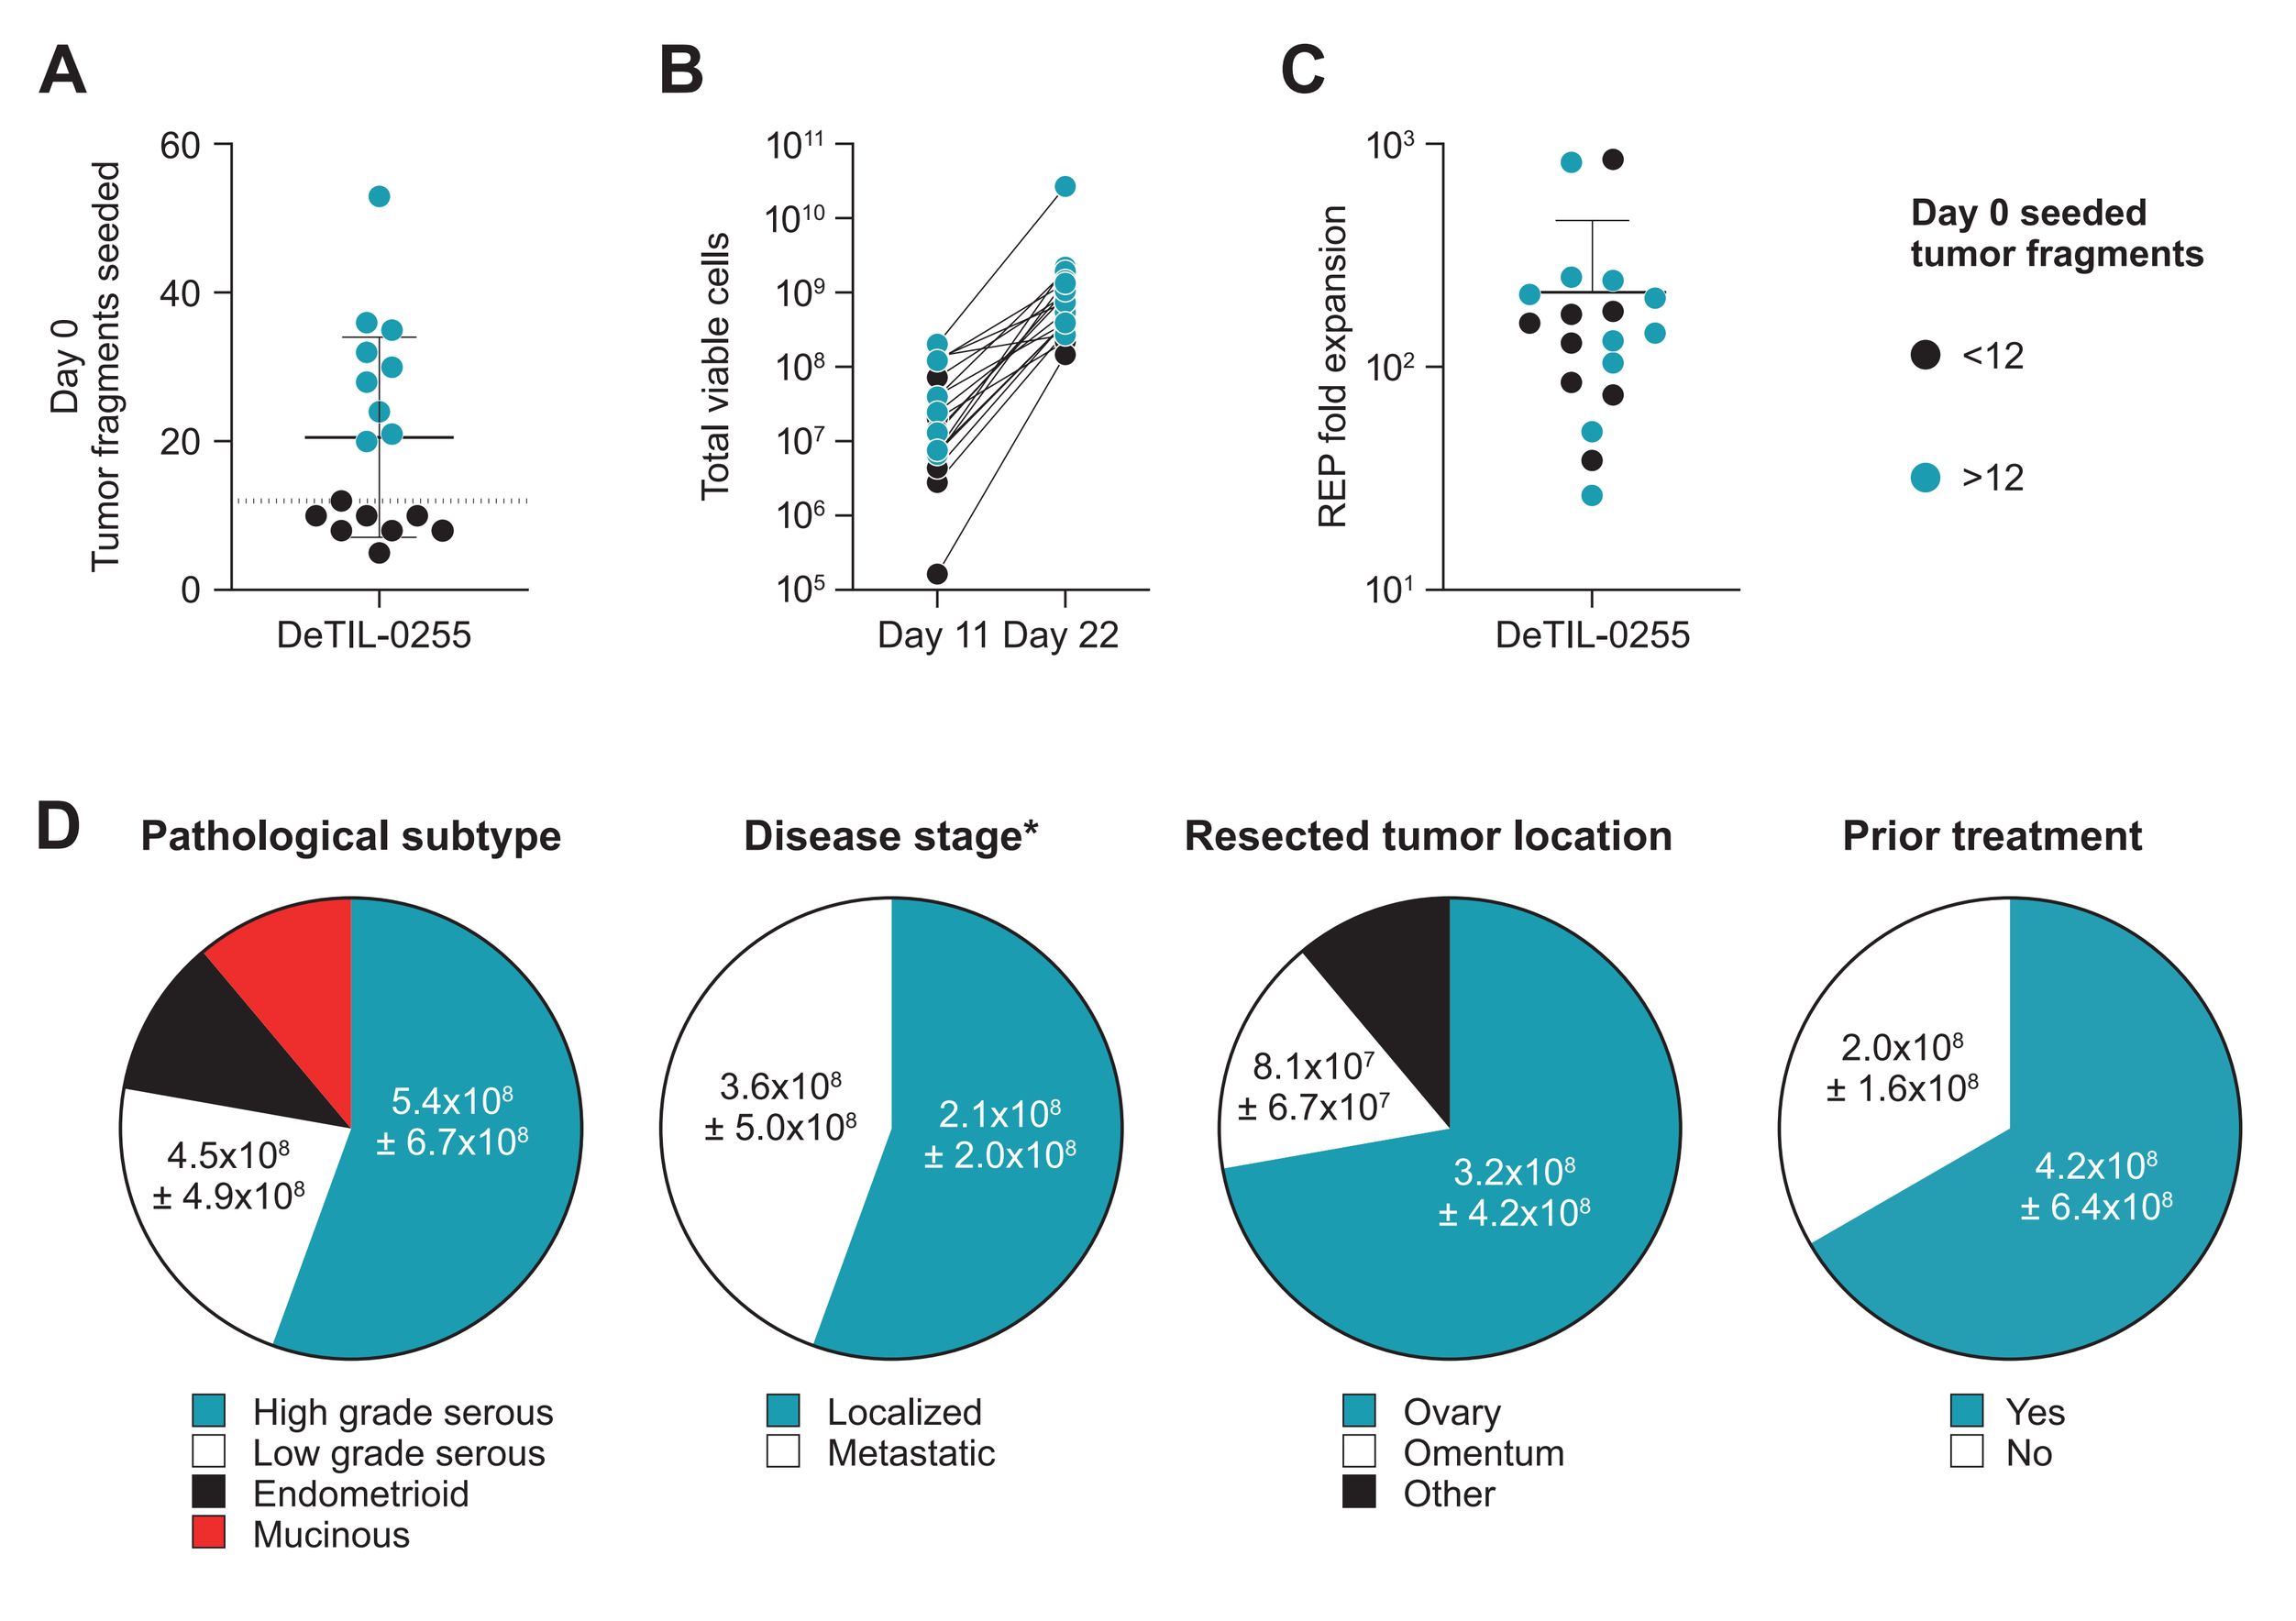

A
Day 0 Tumor fragments seeded
DeTIL-0255
B
Total viable cells
Day 11 Day 22
C
REP fold expansion
DeTIL-0255
Day 0 seeded tumor fragments
<12
>12
D
Pathological subtype
5.4x10^8 ± 6.7x10^8
4.5x10^8 ± 4.9x10^8
High grade serous
Low grade serous
Endometrioid
Mucinous
Disease stage*
3.6x10^8 ± 5.0x10^8
2.1x10^8 ± 2.0x10^8
Localized
Metastatic
Resected tumor location
8.1x10^7 ± 6.7x10^7
3.2x10^8 ± 4.2x10^8
Ovary
Omentum
Other
Prior treatment
2.0x10^8 ± 1.6x10^8
4.2x10^8 ± 6.4x10^8
Yes
No

(A) Number of 1-2mm² tumor fragments seeded for DeTIL-0255 expansion on day 0 of manufacturing. **(B)** Day 11 and 22 total viable cells (TVC) as assessed on NC-200. **(C)** Fold expansion (day 22 TVC / day 11 TVC seeded) during rapid expansion protocol (REP). No significant differences between less than 12 vs greater than 12 day 0 fragments seeded across all expansion variables. **(D)** Clinical characteristics of patients whose tumors were used for DeTIL expansion, including resected tumor pathology, disease stage at the time of resection (localized to ovary or metastatic spread beyond ovary), anatomic site of resected tumor used for DeTIL-0255 expansion, and indication of any systemic oncology therapy prior to resection. No significant differences observed with DeTIL-0255 expansion or phenotype across all clinical subsets. Normalized day 22 TVC (Day 22 TVC / Day 0 tumor fragments seeded) mean ± standard deviation indicated for all major clinical subsets. *FIGO or AJCC staging unavailable for all patients evaluated.

- Ovarian DeTIL-0255 demonstrate a balanced CD4 and CD8 central memory and effector memory phenotype (**Figure 4**).

Figure 4. Flow spectral cytometry phenotype of day 22 DeTIL-0255

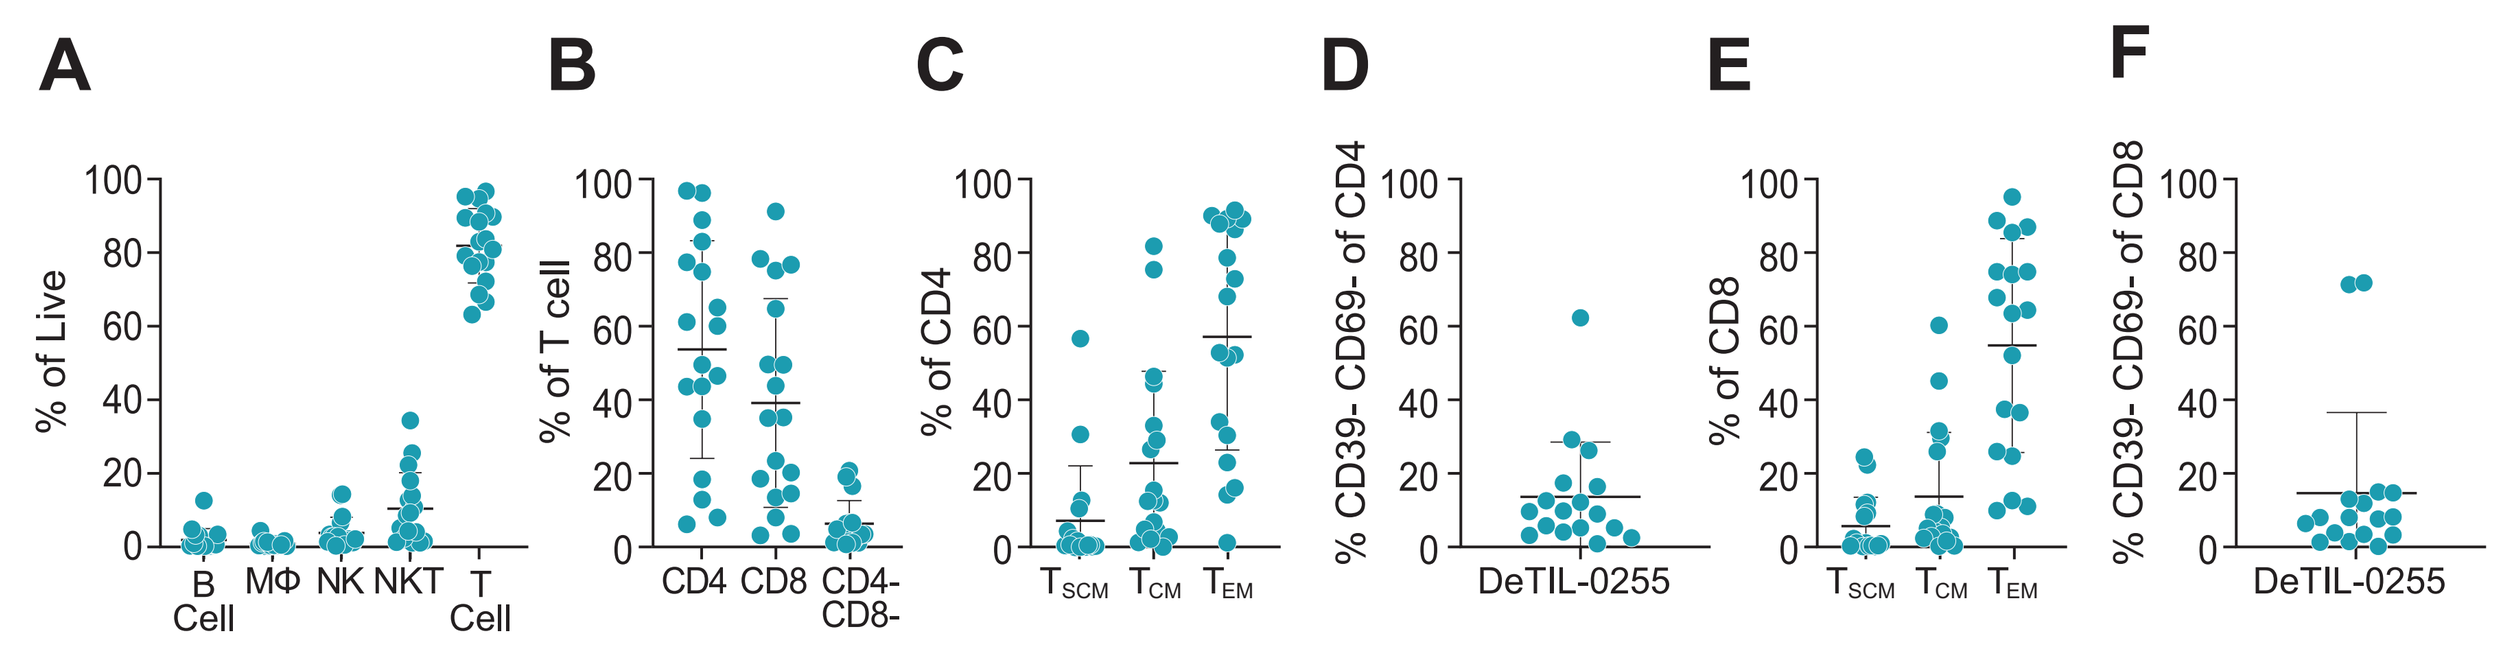

A
% of Live
B Cell MΦ NK NKT T Cell
B
% of T cell
CD4 CD8 CD4- CD8-
C
% of CD4
T_{SCM} T_{CM} T_{EM}
D
% CD39- CD69- of CD4
DeTIL-0255
E
% of CD8
T_{SCM} T_{CM} T_{EM}
F
% CD39- CD69- of CD8
DeTIL-0255

(A) Immune cell composition (CD19+ B cells, CD14+ monocytes/macrophages, CD3- CD56+ NK cells, CD3+ CD56+ NKT cells, and CD3+ CD56- T cells of live cells), **(B)** T cell composition (CD4+, CD8+, CD4-CD8- of T cells), and **(C-F)** memory subpopulations of CD4 **(C-D)** or CD8 **(E-F)** cells [T stem cell memory (T_{SCM}, CCR7+ CD45RO-), T central memory (T_{CM}, CCR7+ CD45RO+), T effector memory (T_{EM}, CCR7- CD45RO+), and stem-like memory cells (CD39- CD69-)].

- Ovarian DeTIL-0255 possess a CD226 high phenotype with limited PD-1, LAG-3, and CD57 expression (**Figure 5**).

Figure 5. Extended flow spectral cytometry phenotype of day 22 DeTIL-0255

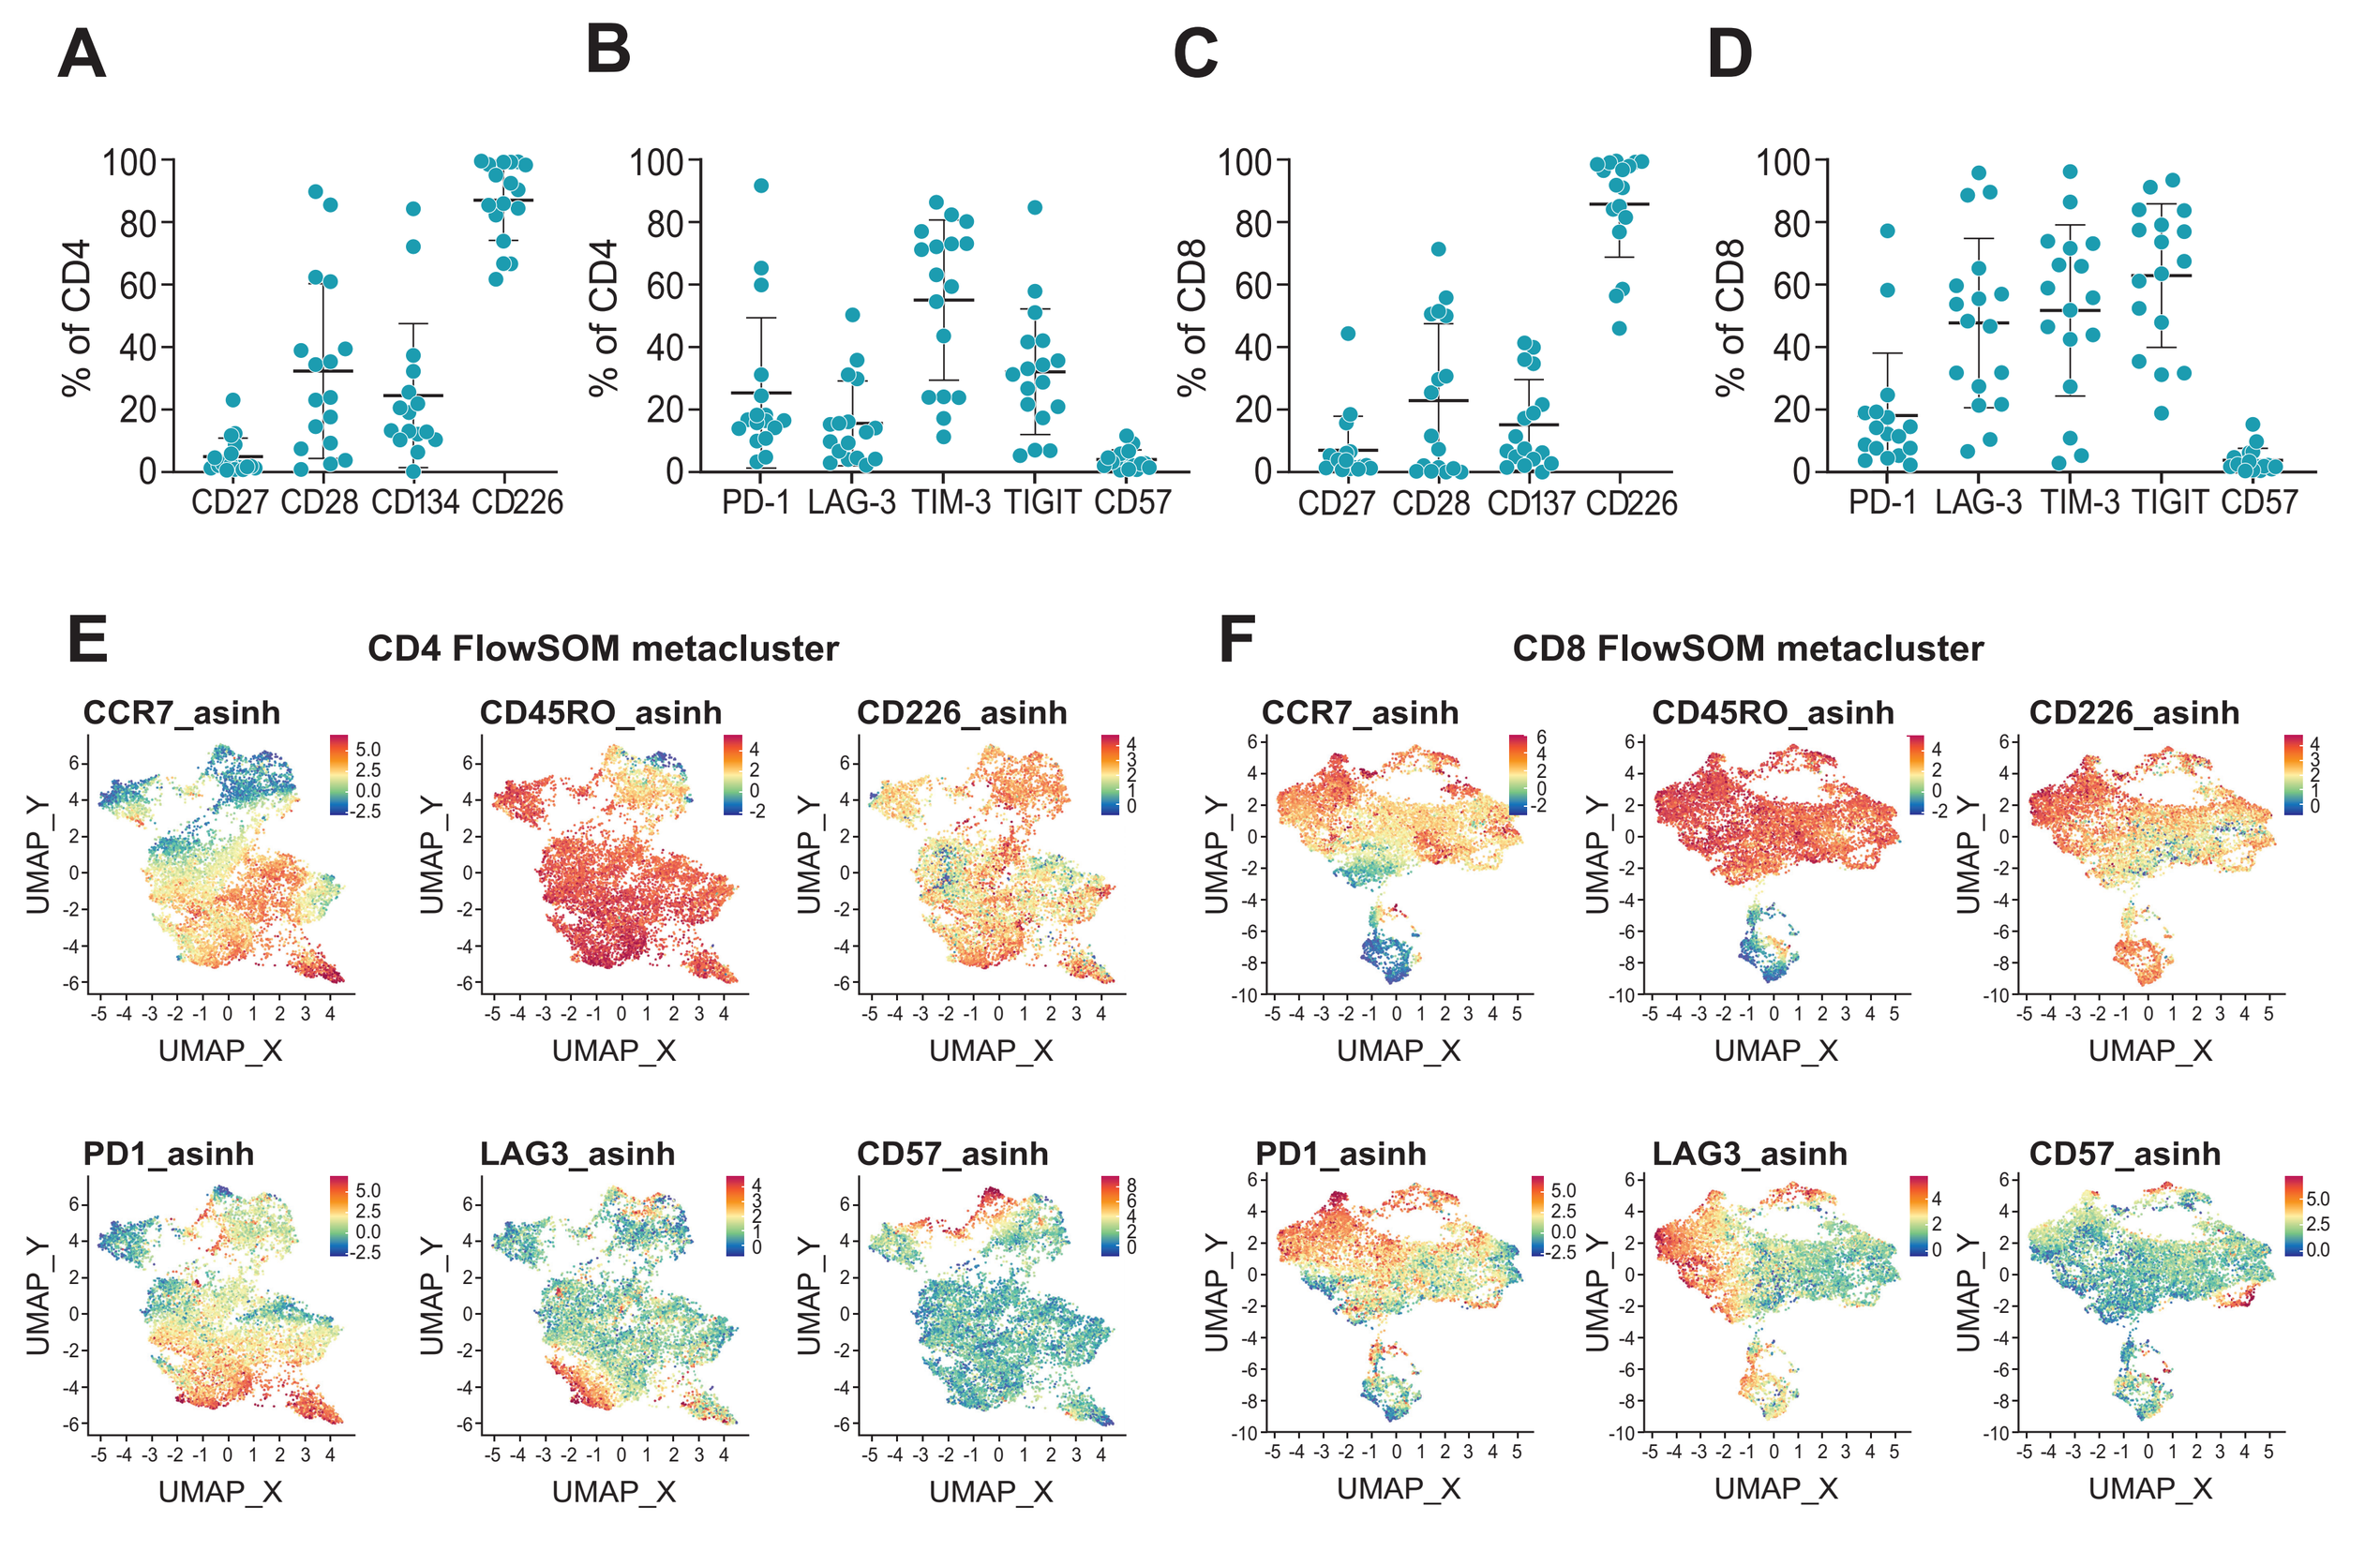

A
% of CD4
CD27 CD28 CD134 CD226
B
% of CD4
PD-1 LAG-3 TIM-3 TIGIT CD57
C
% of CD8
CD27 CD28 CD137 CD226
D
% of CD8
PD-1 LAG-3 TIM-3 TIGIT CD57
E
CD4 FlowSOM metacluster
CCR7_asinh
CD45RO_asinh
CD226_asinh
PD1_asinh
LAG3_asinh
CD57_asinh
F
CD8 FlowSOM metacluster
CCR7_asinh
CD45RO_asinh
CD226_asinh
PD1_asinh
LAG3_asinh
CD57_asinh
UMAP_X
UMAP_Y

(A, B) CD4 and CD8 **(C, D)** percent positive for co-stimulatory domains (**A, C**, CD27, CD28, CD134/OX-40, CD137/4-1BB, CD226/DNAM-1), or exhaustion markers (**B, D,** programmed cell death protein 1 (PD-1), lymphocyte activation gene 3 (LAG-3), T cell immunoglobulin and mucin-domain containing-3 (TIM-3), T cell immunoreceptor with Ig and ITIM domains (TIGIT), CD57). Arcsinh fluorescence intensity of CCR7, CD45RO, CD226, PD-1, LAG-3, and CD57 of CD4 **(E)** and CD8 **(F)** FlowSOM[17] metaclusters visualized by the Uniform Manifold Approximation and Project (UMAP) dimensionality reduction algorithm validate CD226 high DeTIL-0255 phenotype with balance of memory and effector populations and limited PD-1, LAG-3, and CD57 expression.

- Robust Th1 response of stimulated DeTIL-0255 (**Figure 6**).

Figure 6. Functional potential of DeTIL-0255

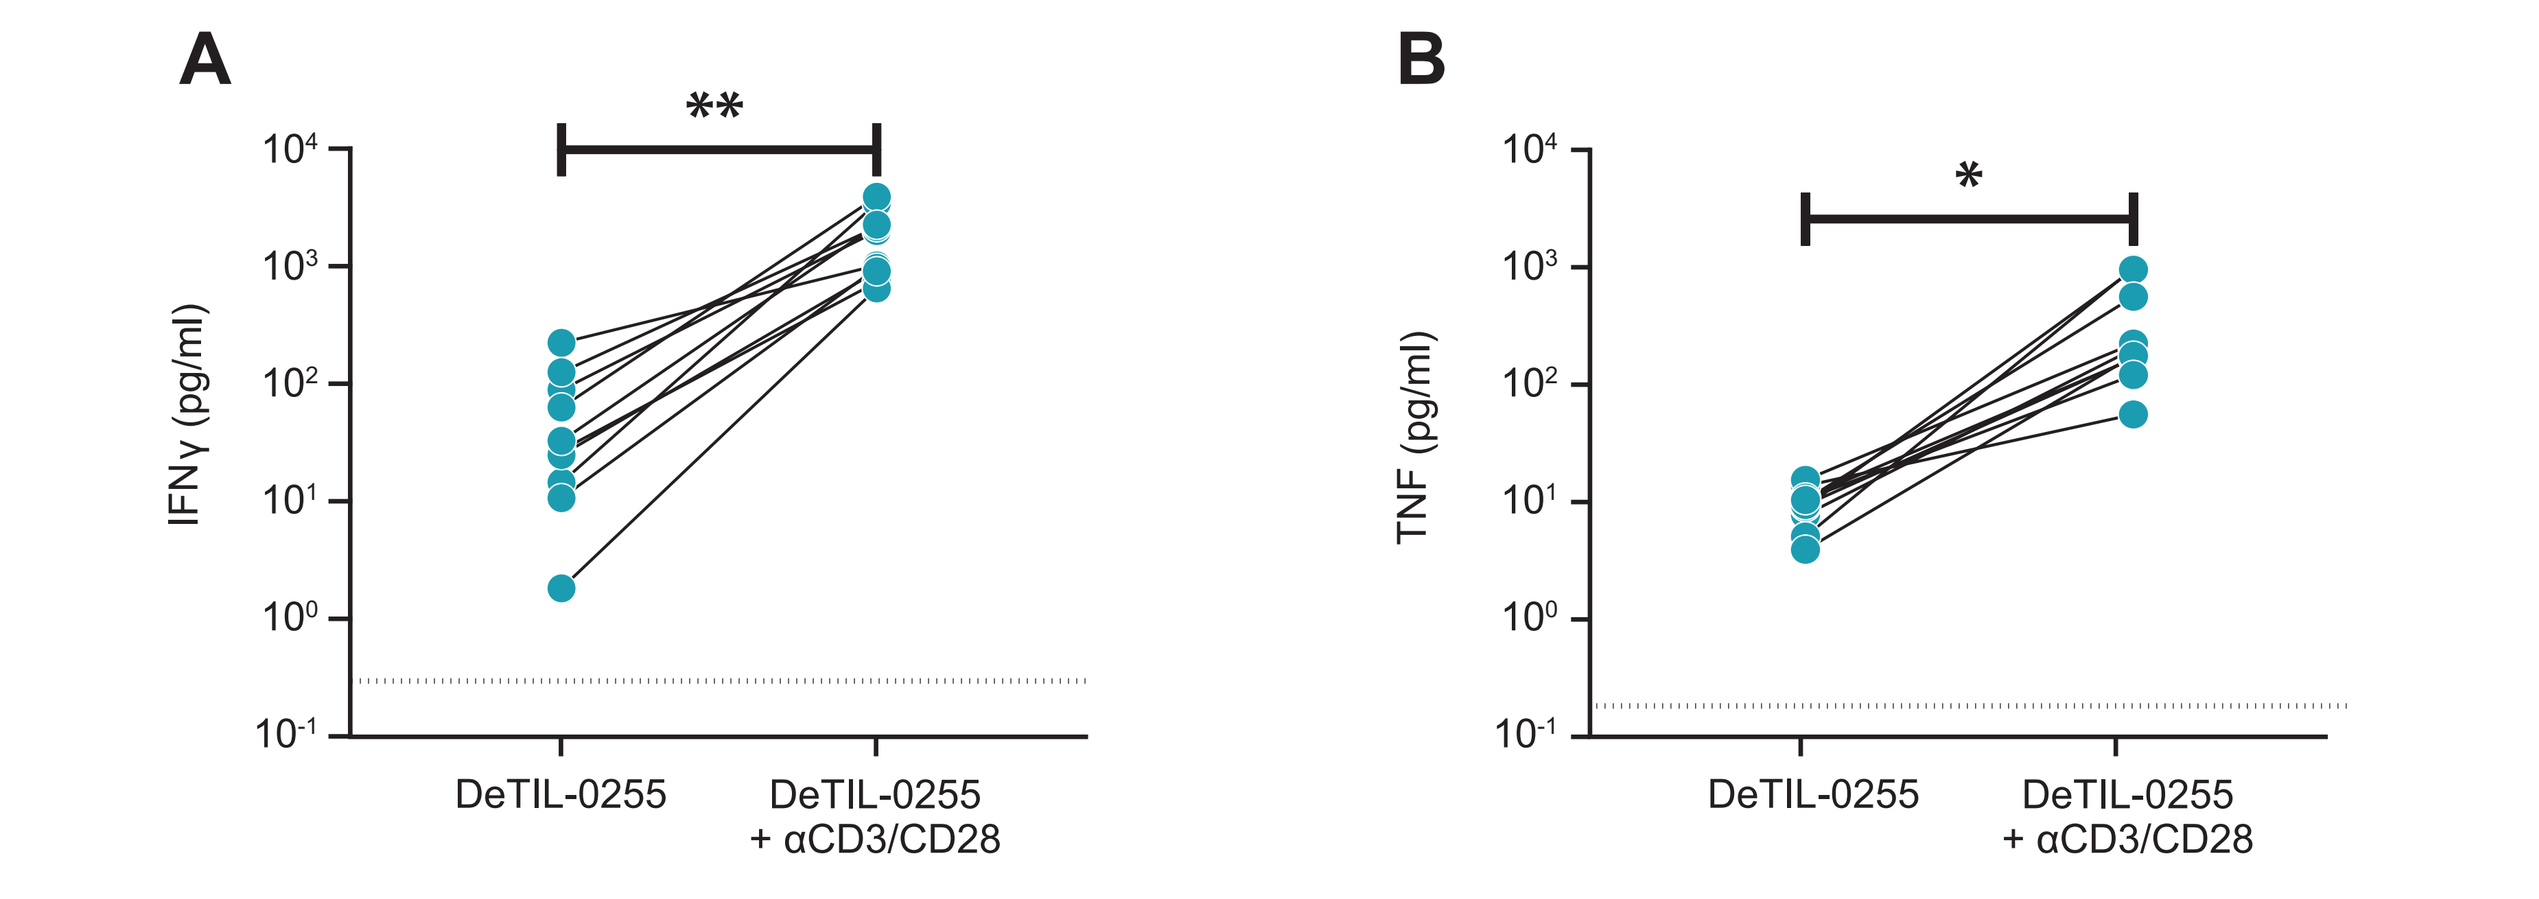

A
**
IFNγ (pg/ml)
DeTIL-0255
DeTIL-0255 + αCD3/CD28
B
*
TNF (pg/ml)
DeTIL-0255
DeTIL-0255 + αCD3/CD28

(A) Interferon-γ (IFNγ) or **(B)** tumor necrosis factor (TNF) secretion following stimulation with αCD3/CD28 antibody complexes (ImmunoCult™) for 48 hours as assessed by multi-analyte enzyme-linked immunosorbent assay (ELISA). Lower limit of quantification indicated by dotted lines. Paired t test: * P < 0.5, ** P < 0.001.

References

1. Zhang L, et al. NEJM. 2003;348:203-13.
2. Sato E, et al. Proc Natl Acad Sci USA 2005;102:18538-43.
3. Anadon CM, et al. Cancer Cell. 2022;40:545-57.e13.
4. Matulonis UA, et al. Ann Oncol. 2019;30:1080-7.
5. Zamarin D, et al. J Clin Oncol. 2020;38:1814-23.
6. Duraiswamy J, et al. Cancer Cell. 2021;39:1623-42.e20.
7. Banta KL, et al. Immunity. 2022;55:512-26.e9.
8. Goff SL, et al. J Clin Oncol. 2016;34:2389-97.
9. Stevanović S, et al. Clin Cancer Res. 2019;25:1486-93.
10. Creelan BC, et al. Nat Med. 2021;27:1410-8.
11. Zacharakis N, et al. J Clin Oncol. 2022;40:1741-54.
12. Pedersen M, et al. Oncoimmunology. 2018;7:e1502905.
13. Kverneland AH, et al. Oncotarget. 2020;11:2092-105.
14. Chiang YJ, et al. Nature. 2000;403:216-20.
15. Whelan S, et al. J Immunother Cancer. 2021;9 (Suppl 2):A107:abstr 98.
16. Gallotta M, et al. Cancer Res. 2022; 82 (Suppl 12):abstr 573.
17. Van Gassen S, et al. Cytometry A. 2015;87:636-45.

Conclusions

- Conventional TIL therapy in ovarian cancer has thus far shown limited clinical responses, with cell products (median 39-day manufacturing time) showing a differentiated and exhausted phenotype.[12,13]
- Here we demonstrate universal expansion of ovarian cancer DeTIL-0255 even with minimal (5–12) input tumor fragments in a 22-day manufacturing process.
- Ovarian cancer DeTIL-0255 are comprised of a balance of central memory and effector memory CD4 and CD8 TIL with reduced expression of checkpoint receptors and evidence of functional potential.
- Ovarian cancer DeTIL-0255 also show high expression of the co-stimulatory receptor CD226, previously shown to be predictive of immunotherapy response in multiple clinical studies.
- Ovarian cancer DeTIL-0255 demonstrates a favorable phenotype amenable for adoptive cell therapy.
- A Phase 1 clinical study of DeTIL-0255 in women with recurrent/platinum resistant ovarian cancer, endometrial cancer, and cervical cancer is ongoing (NCT05107739).

Acknowledgements

Research was sponsored by Nurix Therapeutics, Inc. Medical writing was provided by Pranav Murthy of Nurix Therapeutics, Inc. Editorial support was provided by Miller Medical Communications, which was funded by Nurix Therapeutics, Inc.

Other DeTIL-0255 posters presented at SITC 2022

10 November 2022: #671 Girda E, et al. A phase 1 adoptive cell therapy using drug-enhanced, tumor-infiltrating lymphocytes, DeTIL-0255, in adults with advanced malignancies

11 November 2022: #254 Liang X, et al. The CBL-B inhibitor, NX-0255, enhances human drug enhanced tumor infiltrating lymphocyte (DeTIL) expansion and T cell function in full-scale runs

11 November 2022: #331 Gallotta M, et al. A novel small molecule inhibitor of CBL-B shows potent antitumor activity in combination with Pmel-1 adoptive transfer in an aggressive mouse melanoma model

nurix

Presented at the SITC 37th Annual Meeting, November 10–12, 2022, Boston, MA, USA