Initial Clinical Characterization of Novel Proximal Biomarkers for NX-1607, a First-in-Class Oral CBL-B Inhibitor, in Patients with Advanced Malignancies

Sarah Whelan,[1] Christopher Karim,[1] Jordan Ye,[1] Timothy Ingallinera,[1] Ganesh Cherala,[1] Katherine Jameson,[1] Anja Williams,[2] Adam Sharp,[3] Matthew G. Krebs,[4] Simon Pacey,[5] Sarah Blagden,[6] Ruth Plummer,[7] Daniel Hochhauser,[8] Jeff Evans,[9] Johann de Bono,[3] and Janine Powers[1]

[1]Nurix Therapeutics, Inc., San Francisco, CA, USA; [2]Sarah Cannon Research Institute (SCRI), London, UK; [3]Institute of Cancer Research, Royal Marsden NHS Foundation Trust, London, UK; [4]The Christie NHS Foundation Trust, Manchester, UK; [5]University of Cambridge & Cambridge University Hospitals NHS Foundation Trust, Cambridge, UK; [6]University of Oxford, Oxford, UK; [7]Newcastle University Centre for Cancer, Newcastle, UK; [8]University College Hospital, London, UK; [9]Institute of Cancer Sciences, CR-UK Beatson Institute, Glasgow, UK

777

NX-1607: Proposed mechanism of action

- CBL-B E3 ligase is a master orchestrator of the immune response:
 - – CBL-B knockout mice show robust T and NK cell mediated anti-tumor immunity
- Our CBL-B inhibitor NX-1607 demonstrates an exciting profile:
 1. Enhanced IL-2 and IFN-γ secretion in T cells
 2. Enhanced T cell function and T cell memory generation
 3. Enhanced NK and dendritic cell function
 4. Single agent and combination activity in multiple tumor models

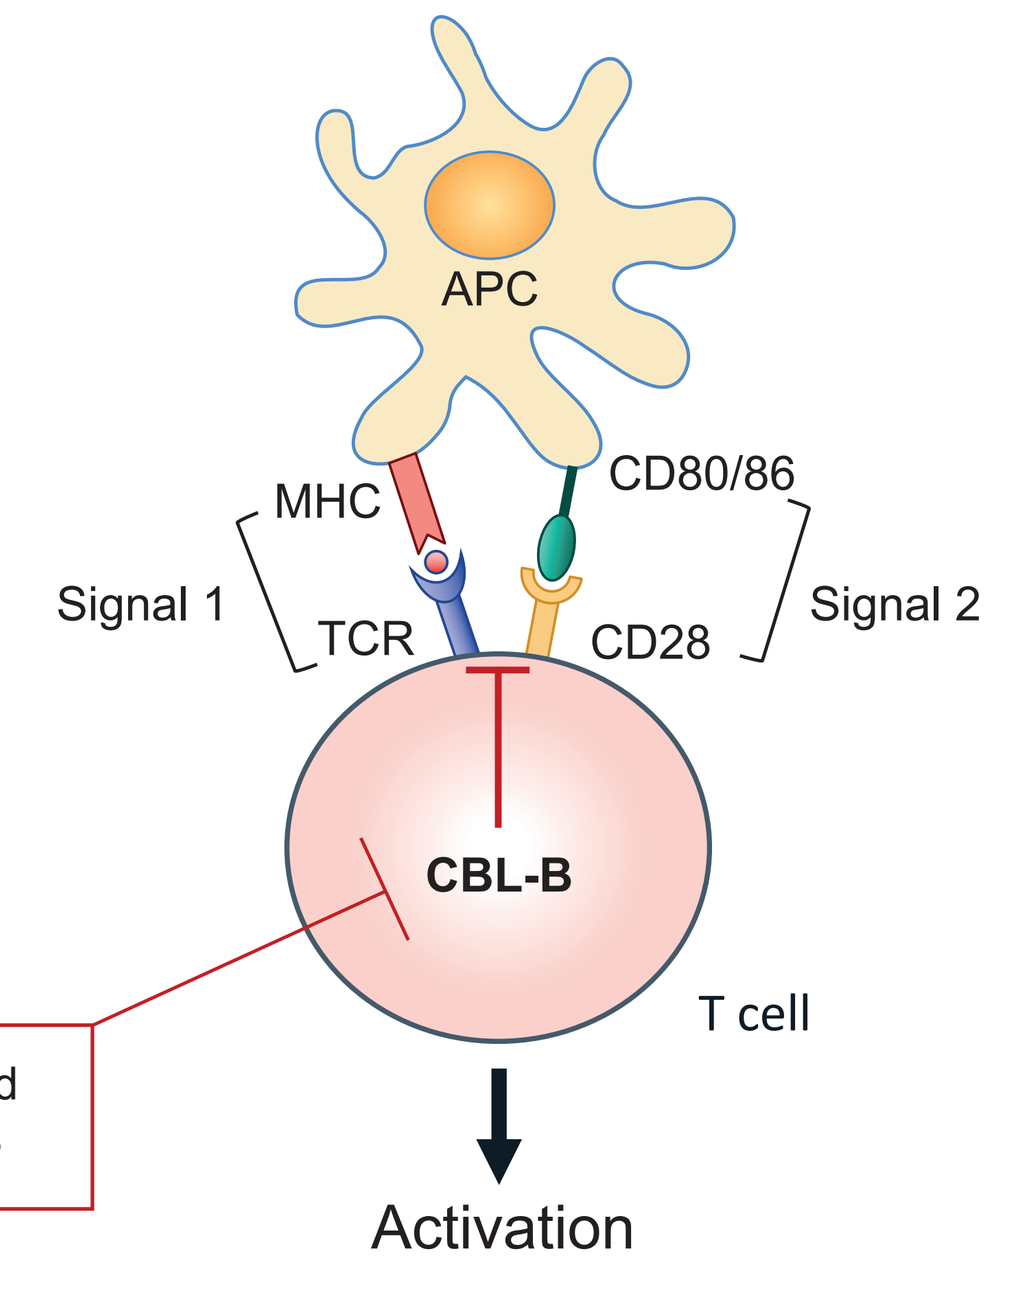
APC
MHC
CD80/86
Signal 1
Signal 2
TCR
CD28
CBL-B
T cell
Activation

NX-1607 is an oral, potent, reversible, and specific small-molecule inhibitor of CBL-B

Methods

Biomarker identification and validation

- Agnostic screening campaigns were performed to identify multiple proprietary proximal biomarkers of CBL-B inhibition in activated T cells.
- Validation of these biomarkers in non-human primate (NHP) and mouse *in vivo* models, coupled with allometric scaling of PK profiles, were used to identify the predicted therapeutic range of NX-1607 and inform clinical dose selection.
- Dose projection modeling using non-clinical data was used to determine a minimal anticipated biological effect level (MABEL) predictive of activating proximal biomarkers following oral (PO) administration of NX-1607.

First-in-human trial: NX-1607-101

- NX-1607-101 is a first-in-human Phase 1 trial evaluating NX-1607 in patients with relapsed or refractory cancers (**Figure 1**).
- During dose escalation, NX-1607 is given orally once daily at 4 ascending dose levels (5, 15, 25 and 50 mg) using an accelerated modified Fibonacci design that transitions to a standard 3 + 3 design based on protocol-specific criteria.
- Efficacy, safety, PK and PD data are currently being evaluated.

Figure 1. NX-1607-101: Phase 1 first-in-human clinical trial design

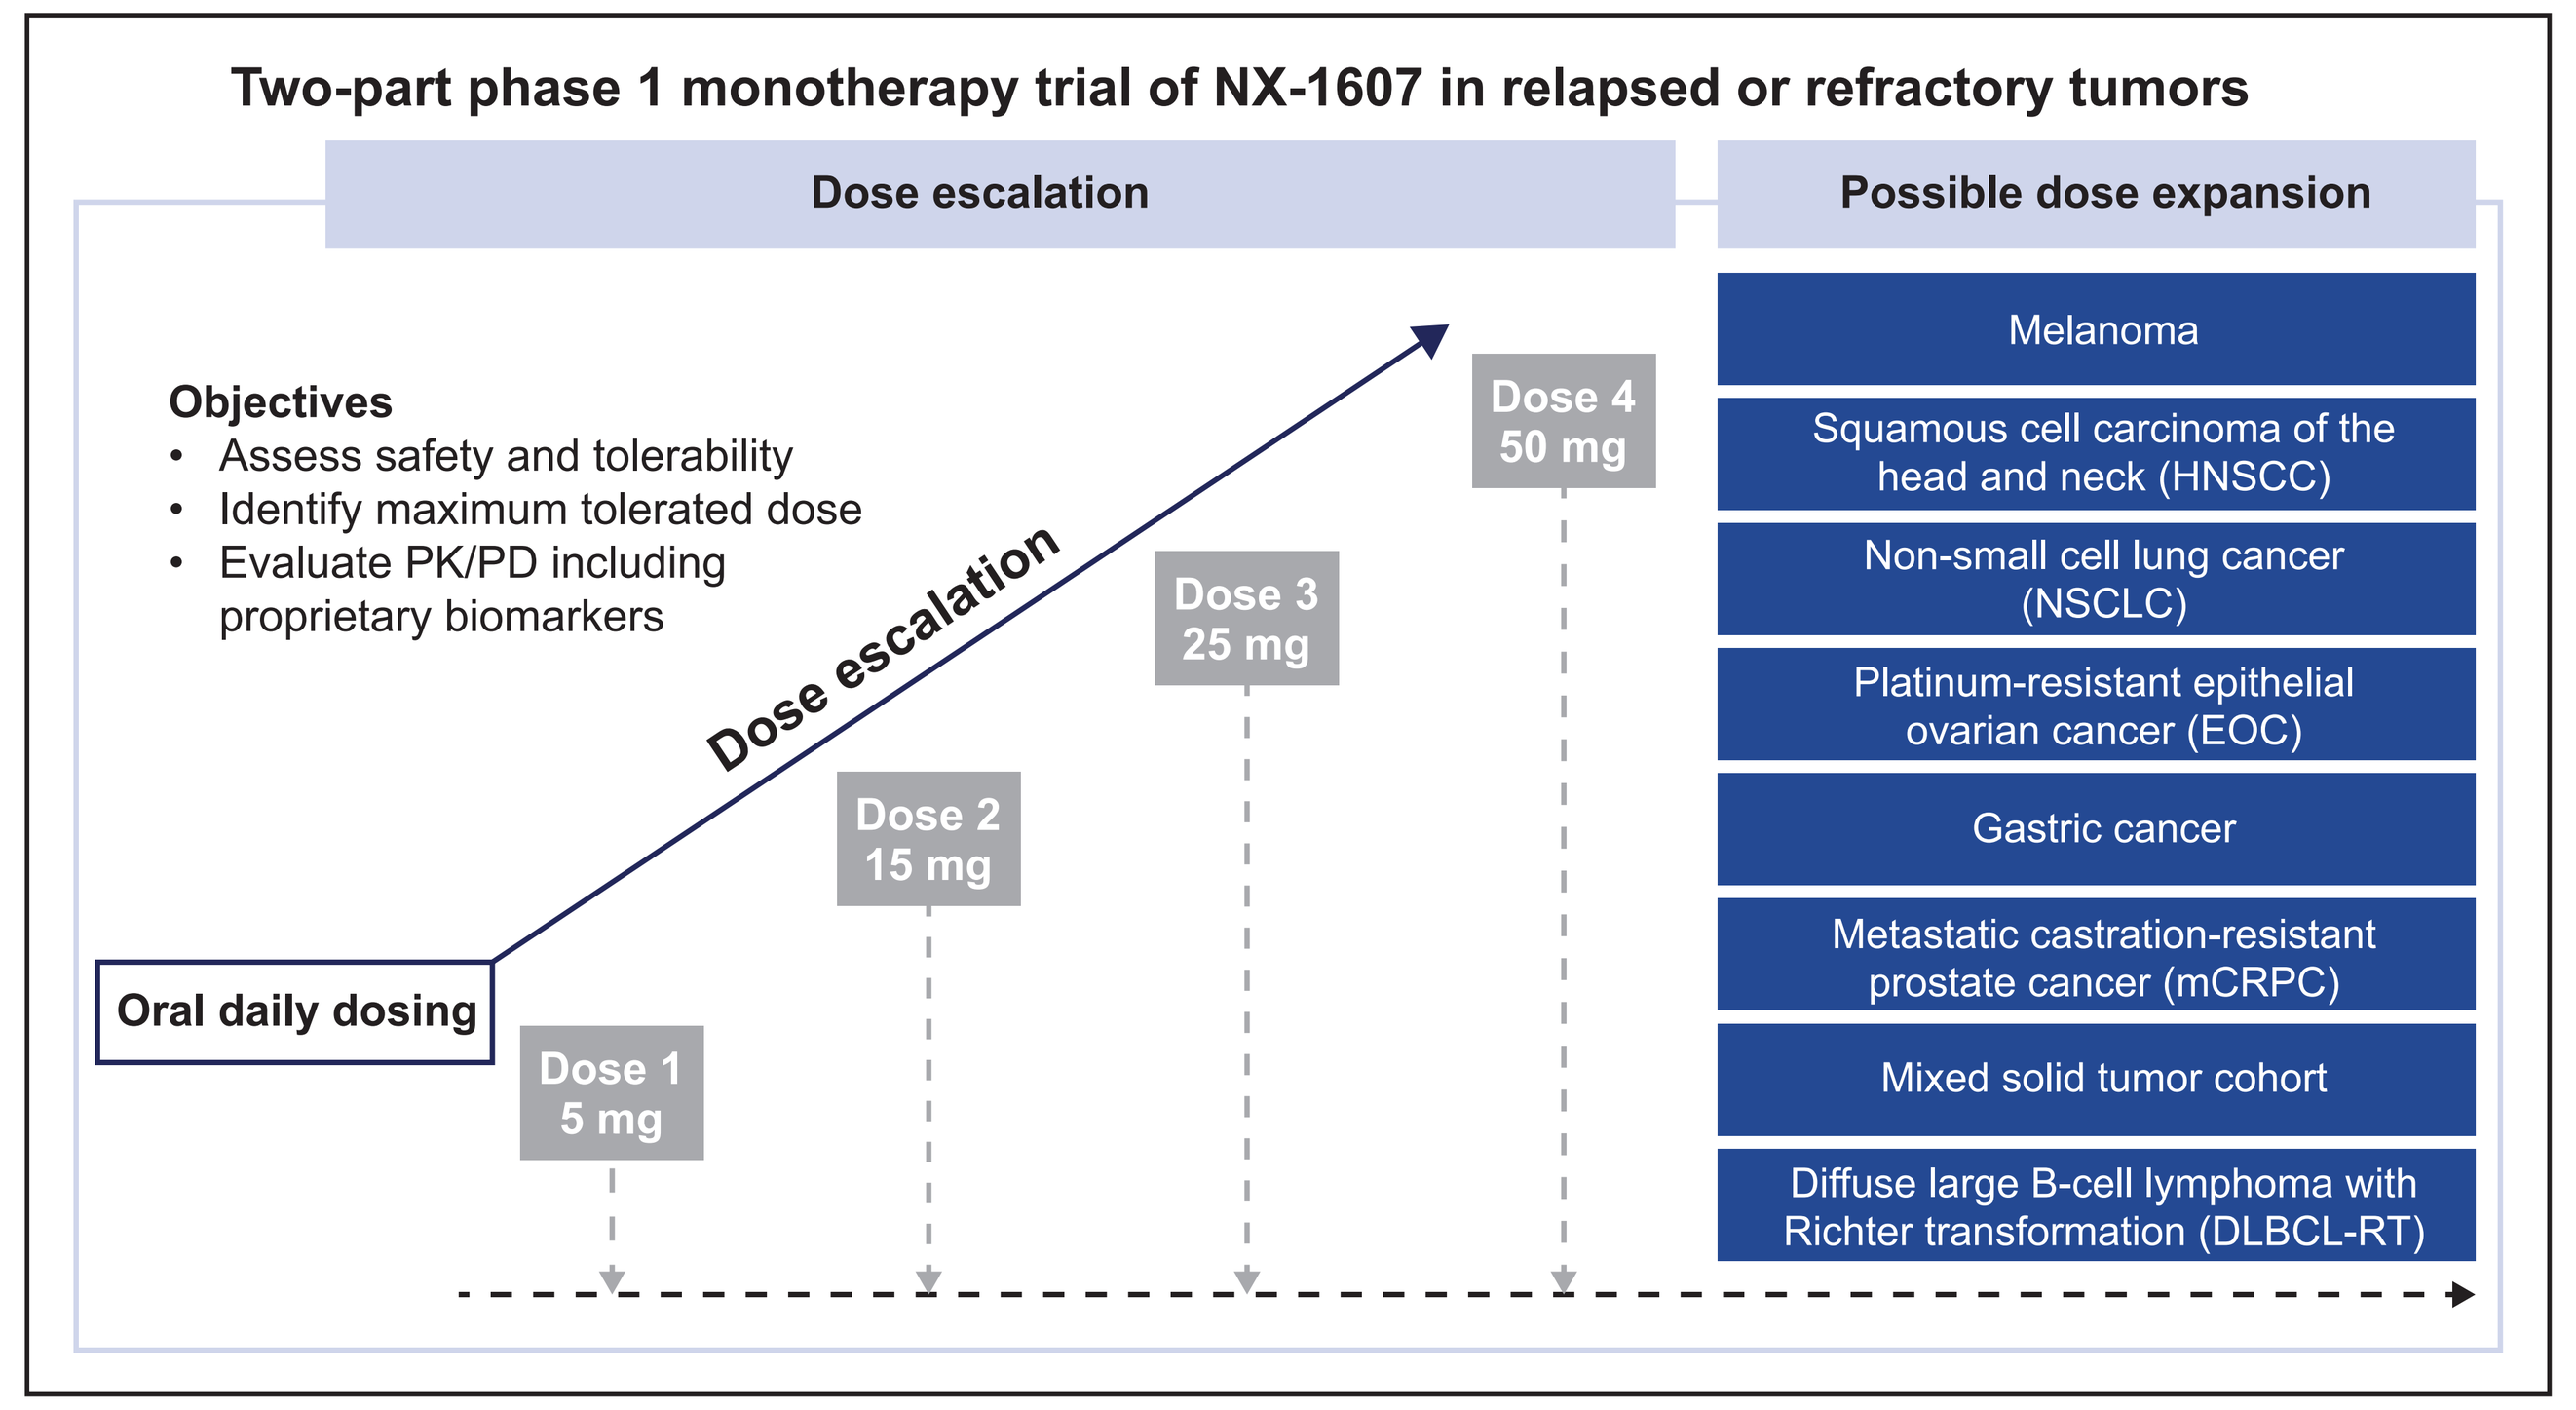
Two-part phase 1 monotherapy trial of NX-1607 in relapsed or refractory tumors
Dose escalation
Possible dose expansion
Objectives
Assess safety and tolerability
Identify maximum tolerated dose
Evaluate PK/PD including proprietary biomarkers
Dose escalation
Oral daily dosing
Dose 1 5 mg
Dose 2 15 mg
Dose 3 25 mg
Dose 4 50 mg
Melanoma
Squamous cell carcinoma of the head and neck (HNSCC)
Non-small cell lung cancer (NSCLC)
Platinum-resistant epithelial ovarian cancer (EOC)
Gastric cancer
Metastatic castration-resistant prostate cancer (mCRPC)
Mixed solid tumor cohort
Diffuse large B-cell lymphoma with Richter transformation (DLBCL-RT)

Results

Biomarker identification and validation

- Phosphorylated hematopoietic lineage cell-specific protein 1 (pHS1), phospholipase C gamma 2 (pPLCγ2), and zeta chain of T cell receptor-associated protein kinase 70 (pZAP70), regulators of T-cell receptor (TCR) signaling, were identified as biologically relevant proximal biomarkers for monitoring pharmacologic inhibition of CBL-B in whole blood (**Figure 2**).
- Validation of proximal biomarkers *in vivo* demonstrated target engagement with dose-dependent increases of phosphorylated biomarkers in CD8+ T cells in both NHP (**Figure 3**) and mice dosed orally with NX-1607 (**Figure 4**):
 - – Dose-dependent increases of CBL-B proximal biomarkers observed in NHP dosed orally with NX-1607.
 - – *In vivo* efficacy observed with NX-1607 at doses of 10 to 60 mg/kg in mice corresponded to >20% pHS1+/CD8+ T cells, and is consistent with dose-projection model.
- Dose-responsive biomarker activity was evident with NX-1607 in human whole blood (**Figure 5A**). A human dose-projection model suggests that minimum anticipated biological effective level (MABEL) of NX-1607 will be reached starting at 5 mg once daily (i.e. dose level 1 in NX-1607-101) (**Figure 5B**).

Figure 2. Overview of UbiScan® technology, identification of direct CBL-B substrates within the TCR signaling cascade and proximal biomarkers of CBL inhibition (Phospho-Screen)

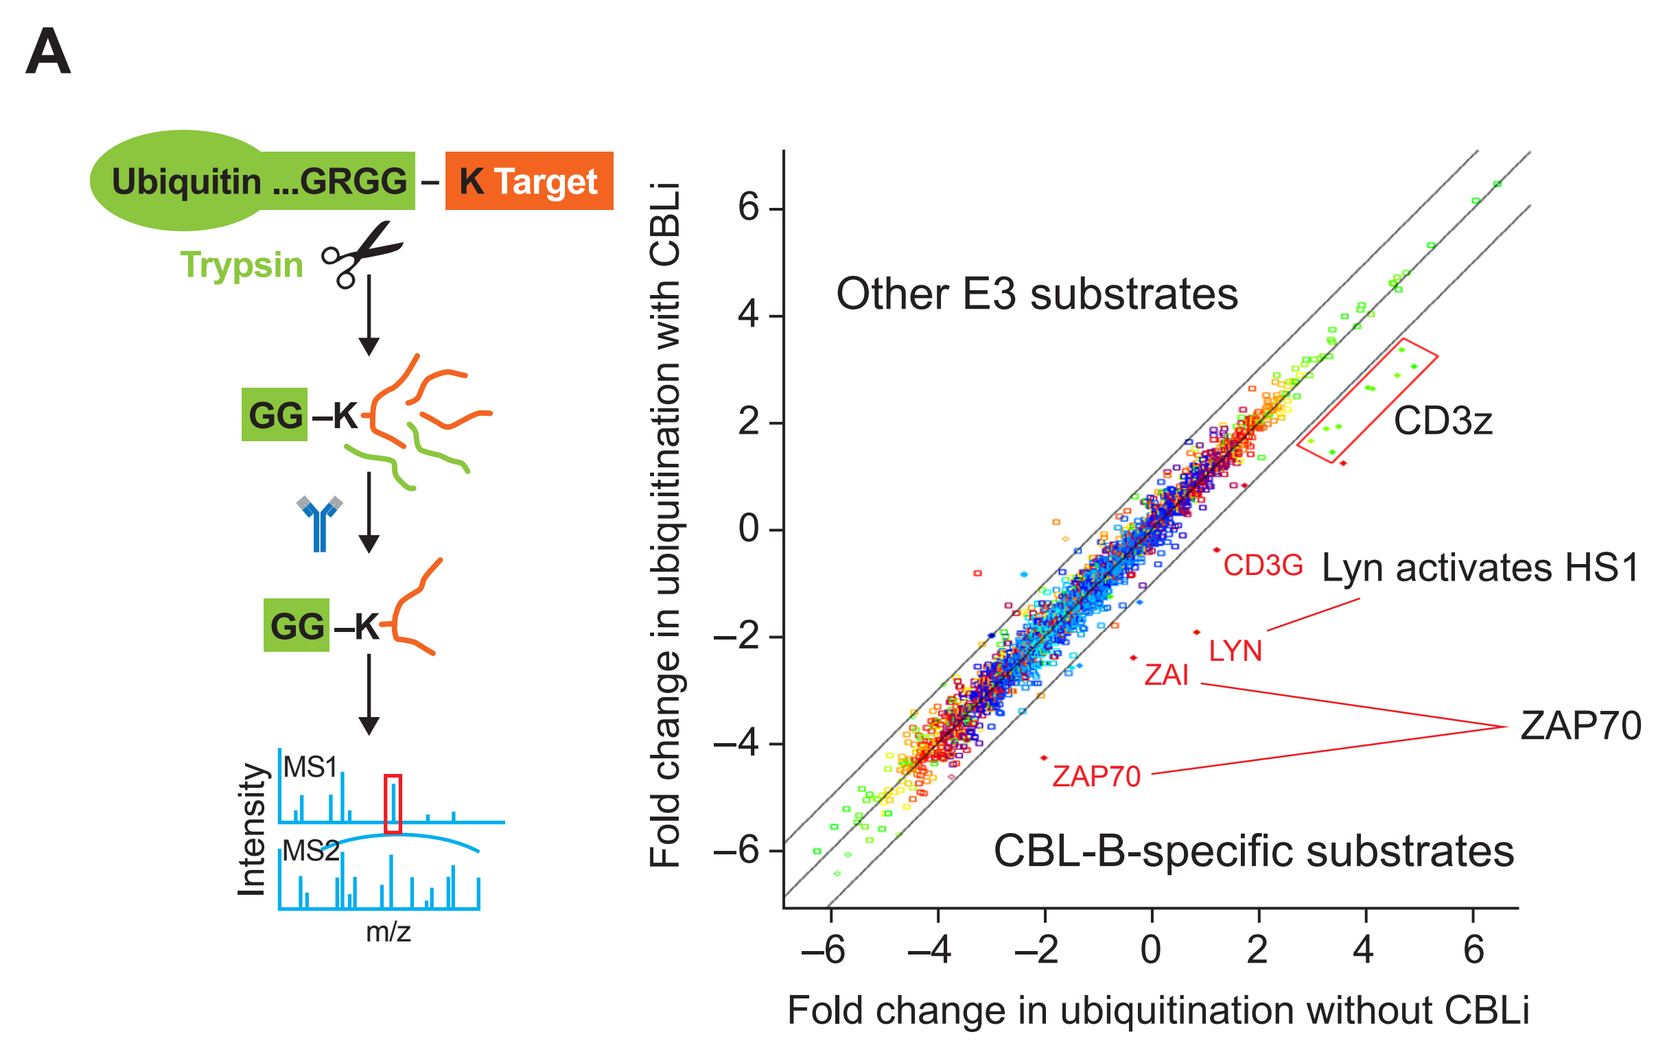
A
Ubiquitin ...GRGG – K Target
Trypsin
GG –K
Intensity
MS1
MS2
m/z
Other E3 substrates
CD3z
CD3G Lyn activates HS1
LYN
ZAI
ZAP70
CBL-B-specific substrates
Fold change in ubiquitination with CBLi
Fold change in ubiquitination without CBLi

- Anti-di-glycine (K-ϵ-GG) antibody measures remnants left on ubiquitinated lysine residues after trypsin digestion
- Decreased signal represents direct substrates ubiquitinated by CBL-B ligase activity
- Inhibiting CBL-B decreases ubiquitination of important TCR signaling molecules

To analyze the CBL-B ubiquitinome, we applied UbiScan, a technique that uses a ubiquitin remnant motif (K-ϵ-GG) antibody–bead conjugate to isolate ubiquitinated peptides. Digestion of proteins with trypsin during mass spectrometry sample preparation cleaves ubiquitin at the C-terminus, leaving a Gly-Gly residue (K-ϵ-GG) that is still attached to a lysine on the target protein, thus providing evidence of a ubiquitinated protein. T cells were incubated with 10uM CBL-B inhibitors and stimulated with anti-CD3. Direct substrates ubiquinated by CBL-B ligase activity upon stimulation were decreased.

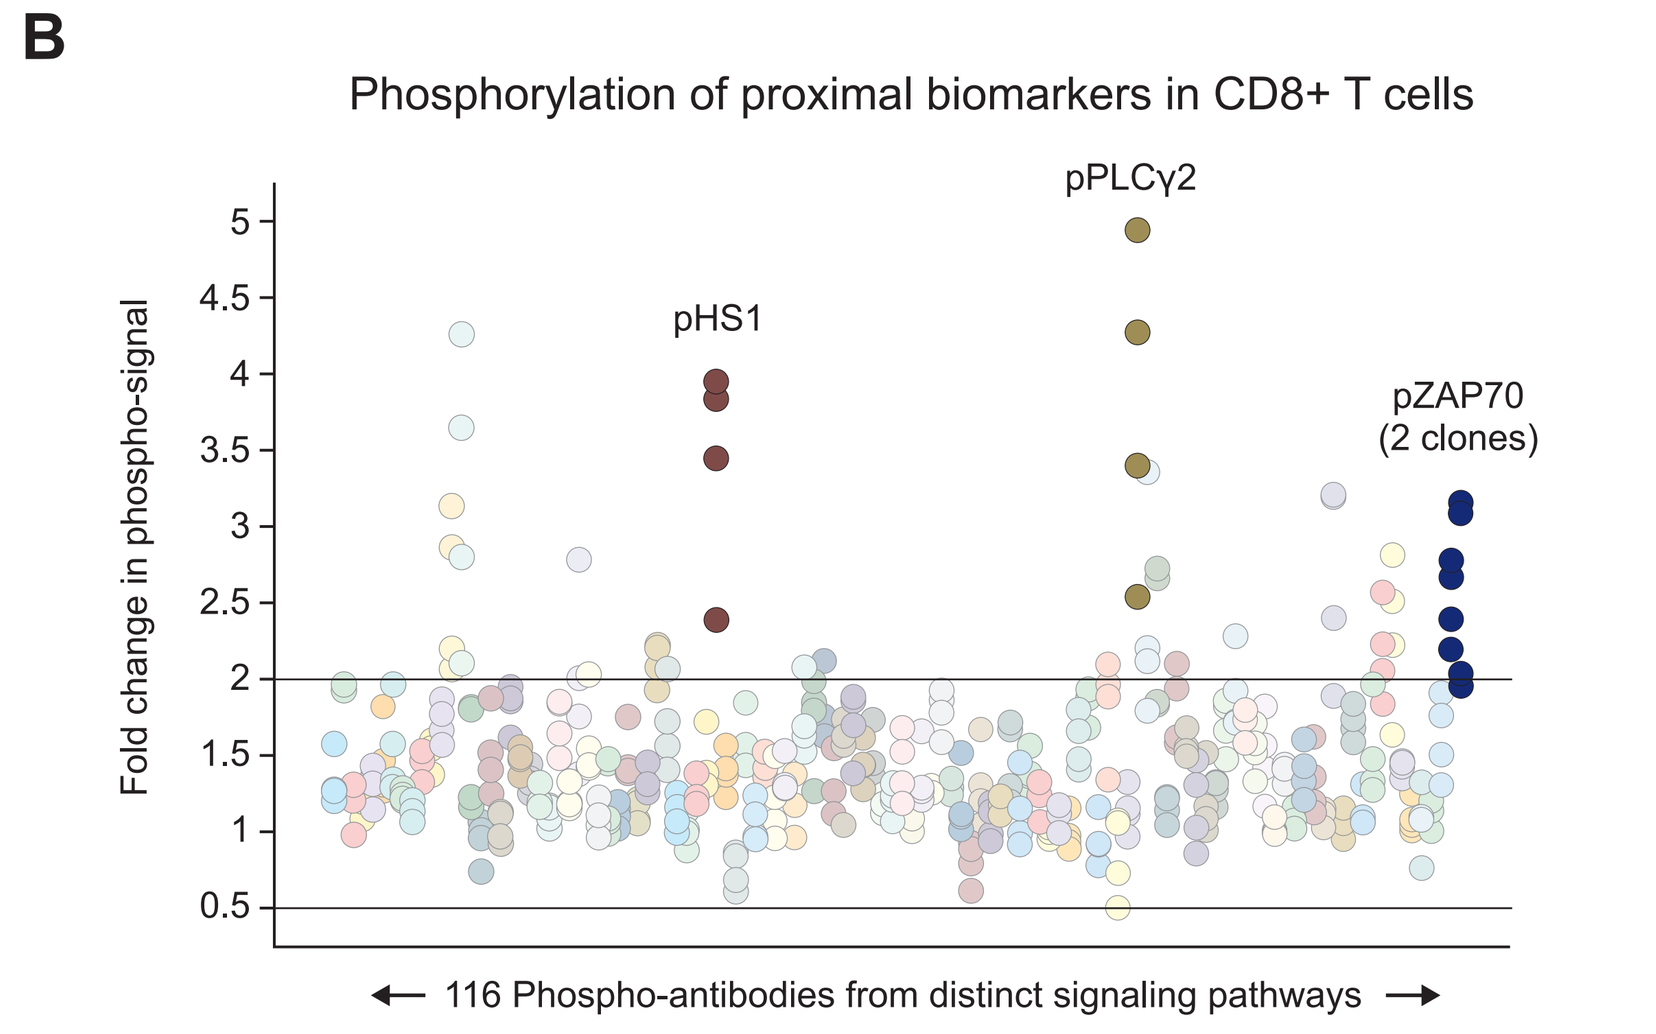
B
Phosphorylation of proximal biomarkers in CD8+ T cells
pHS1
pPLCγ2
pZAP70 (2 clones)
Fold change in phospho-signal
116 Phospho-antibodies from distinct signaling pathways

Upon CBL-B inhibition, stimulation of TCR results in a substantial increase in phosphorylation of:

- pHS1
 A substrate for LYN and essential actin-regulatory adaptor protein at the immune synapse, via VAV1
- pZAP70
 Key organizer of downstream TCR signaling.
- pPLCγ2
 Expressed in both B and T cells

 Associates with LAT and SLP-76 and becomes phosphorylated upon TCR stimulation[2]

Human PBMCs were treated with CBL inhibitor ± anti-CD3/CD28. Cells were stained with a panel of phospho-specific antibodies for proteins downstream of the TCR signaling pathway and expression levels were assessed by flow cytometry. Three proximal biomarkers were prioritized (highlighted) based on their robustness and stability in multiple donors.

Figure 3. Single dose NX-1607 exposure in NHP and induction of phosphorylated proximal biomarkers of CBL-B inhibition upon stimulation in vivo in cynomolgus circulating CD8+ T cells

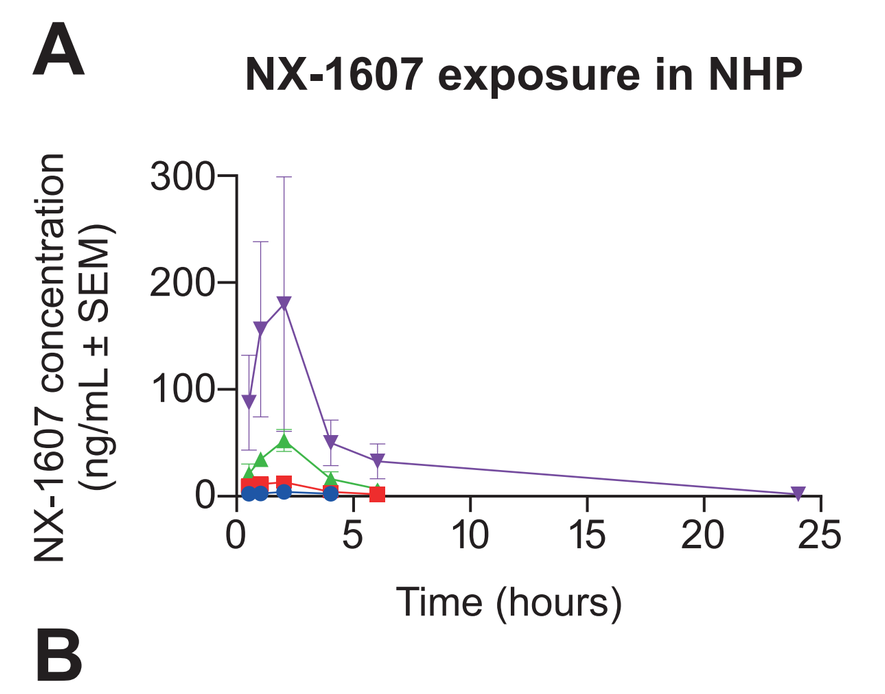
A
NX-1607 exposure in NHP
NX-1607 concentration (ng/mL ± SEM)
Time (hours)

- Proximal biomarker signals were validated in vivo, and confirmed dose responsive target engagement of NX-1607 after a single oral dose in Non-Human Primates (NHP)

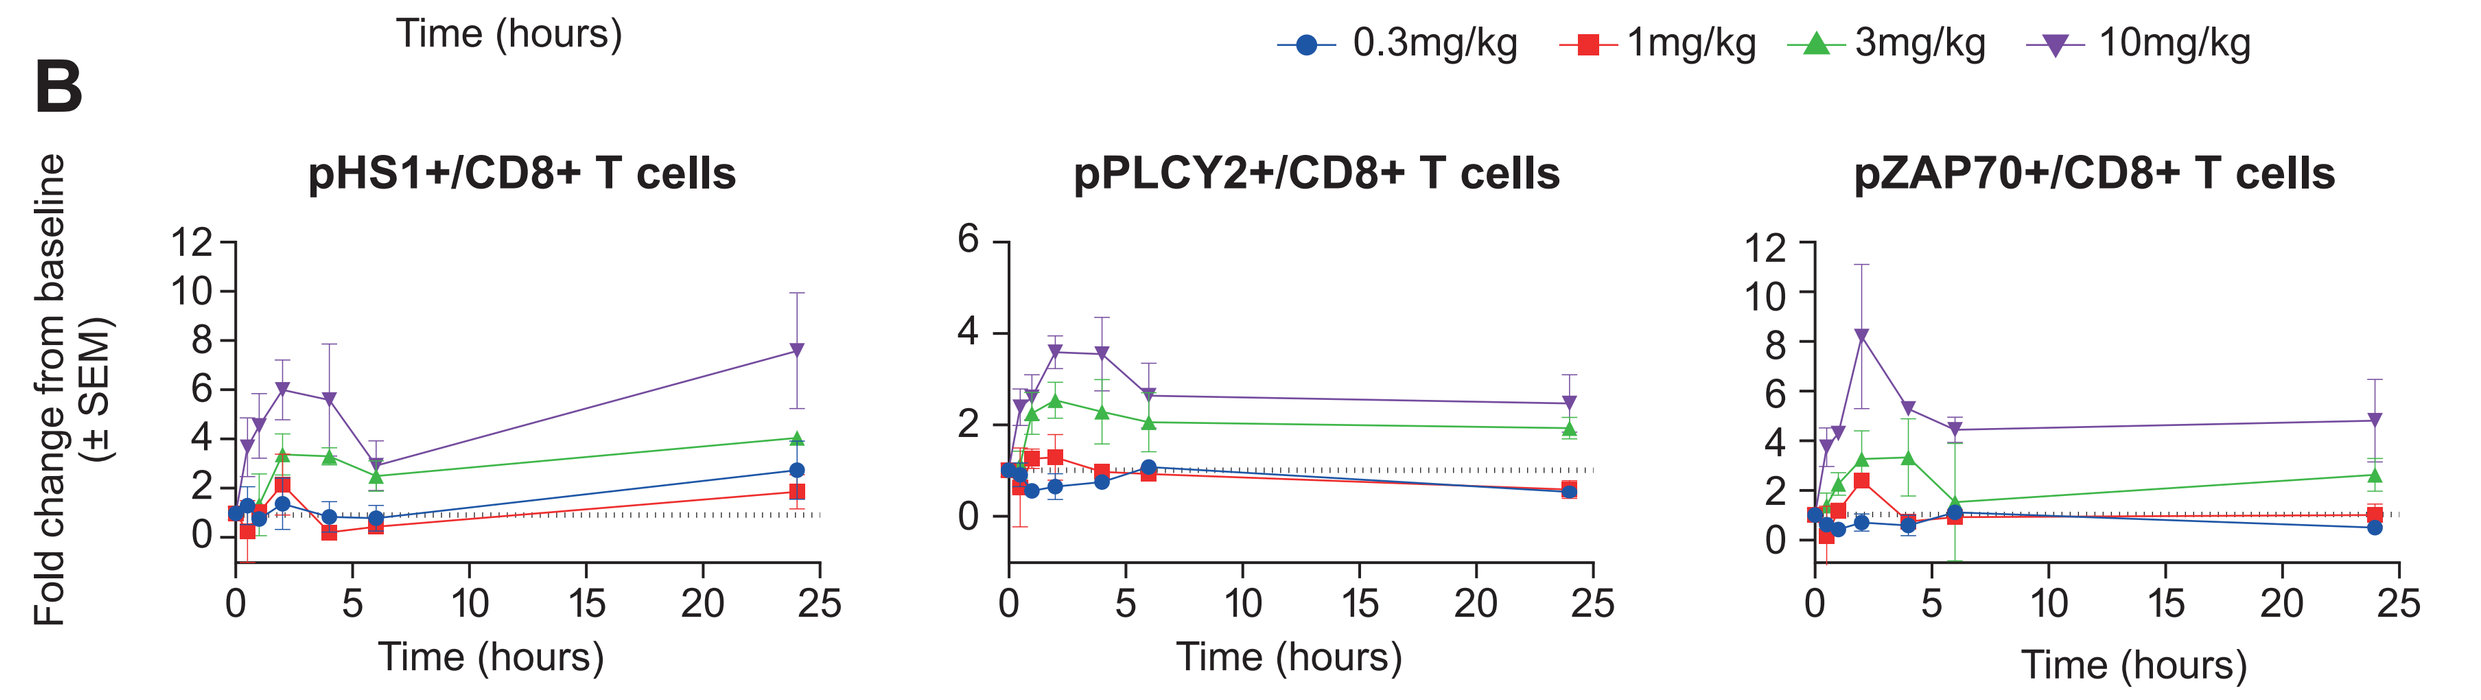
0.3mg/kg
1mg/kg
3mg/kg
10mg/kg
B
pHS1+/CD8+ T cells
pPLCY2+/CD8+ T cells
pZAP70+/CD8+ T cells
Fold change from baseline (± SEM)
Time (hours)

Cynomolgus monkeys were orally administered either 0.3, 1, 3 or 10 mg/kg NX-1607 and whole blood was collected to assess both PK & PD.
Figure 3A: Following oral administration of NX-1607 to NHP, NX-1607 concentrations were measured in plasma, predose through 24 hours post dose for 24hrs.
Figure 3B: Whole blood was collected into 2 types of TruCulture tubes ("Null" for aliquot 1; "CD3/CD28" for aliquot 2) at each indicated timepoints pre and post dosing (n=3/group). % Phosphorylated HS1, PLCY2 & ZAP70 were quantitated in T cells (CD45+/CD8+) using flow cytometry. Graph represents the mean (± SEM) fold change of phosphorylated HS1, PLCY2 & ZAP70 at each time point after correcting for background in unstimulated cells and normalizing to pre-dose (T=0) phosphorylated HS1, PLCY2 & ZAP70 levels.

Figure 4. Dose-dependent increases of percentage pHS1+/CD8+ T cells correlates with anti-tumor effects of NX-1607 in mice

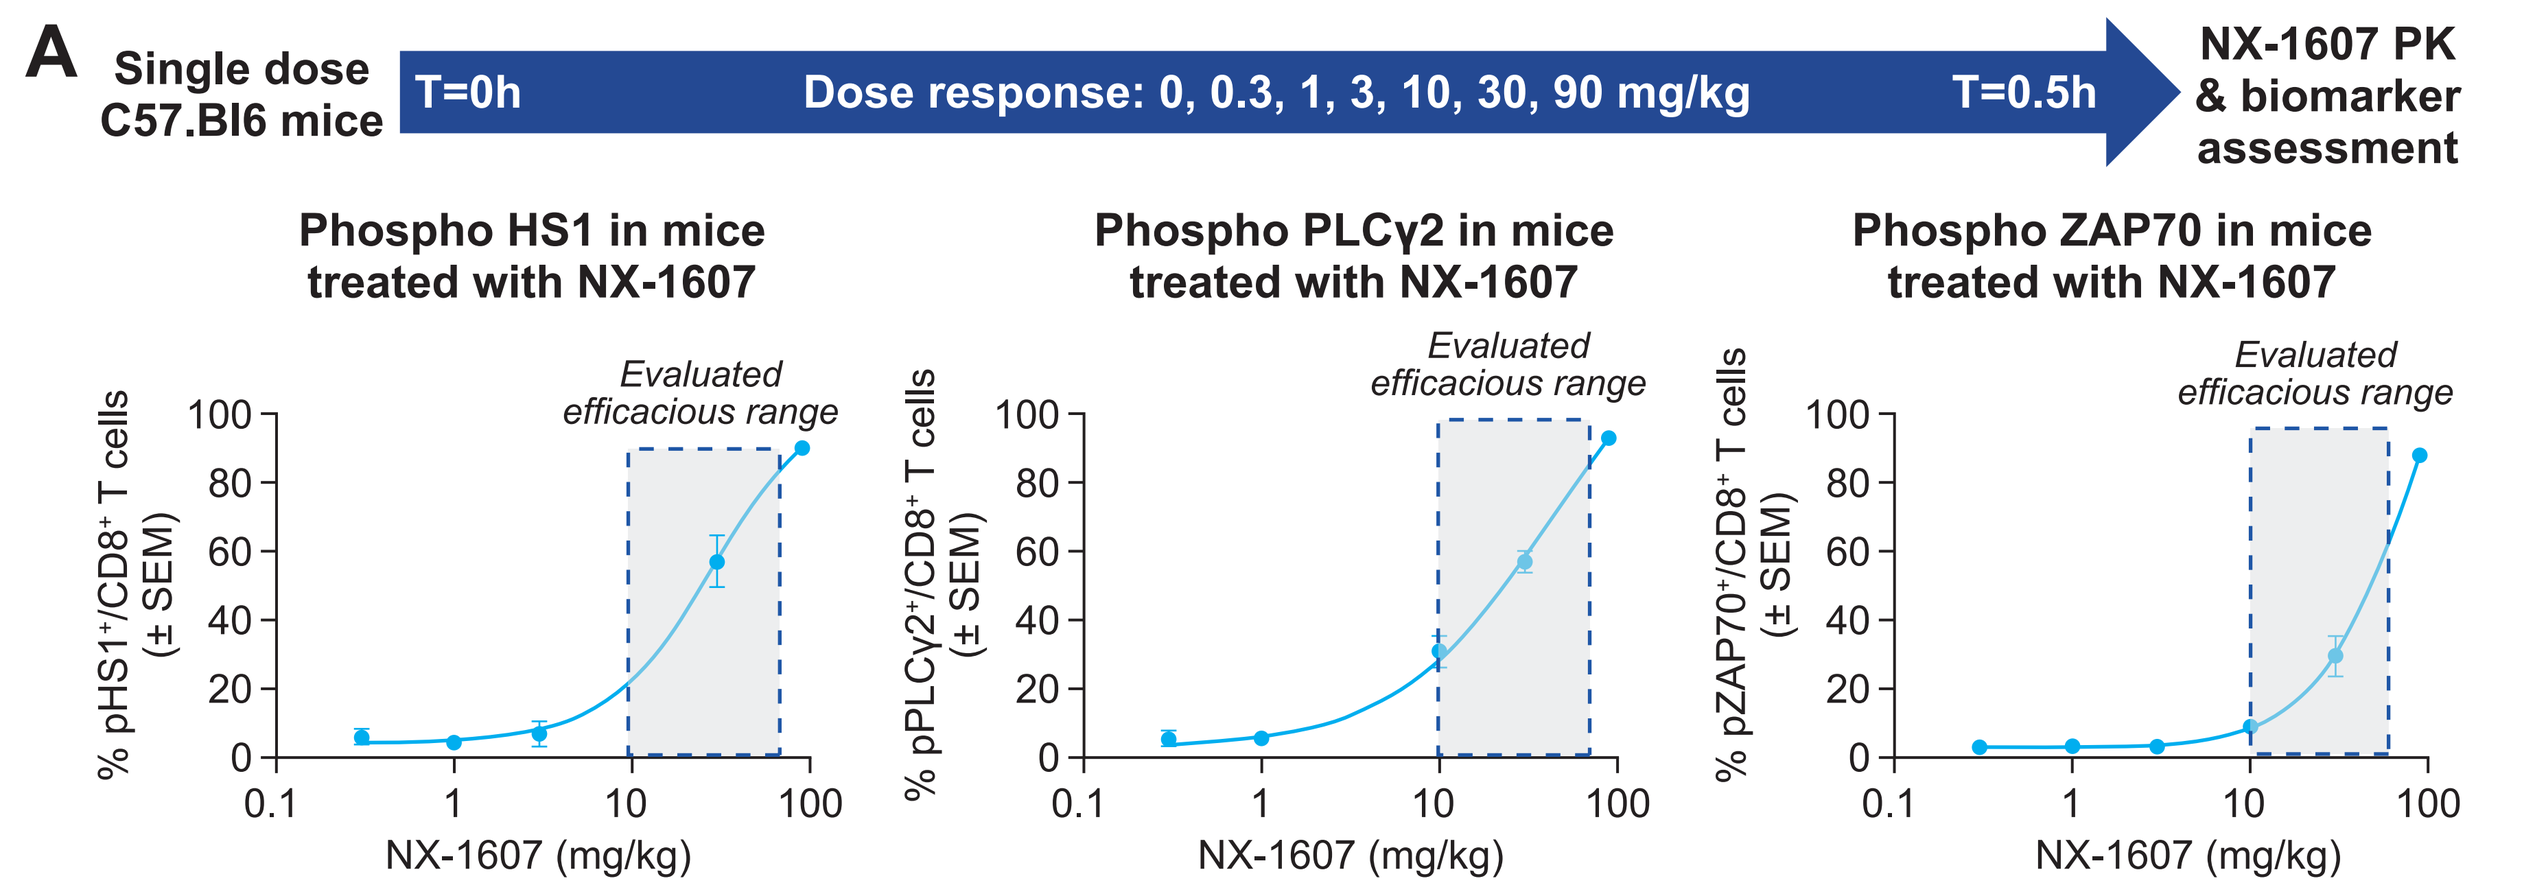
A
Single dose C57.Bl6 mice
T=0h
Dose response: 0, 0.3, 1, 3, 10, 30, 90 mg/kg
T=0.5h
NX-1607 PK & biomarker assessment
Phospho HS1 in mice treated with NX-1607
Phospho PLCγ2 in mice treated with NX-1607
Phospho ZAP70 in mice treated with NX-1607
Evaluated efficacious range
NX-1607 (mg/kg)

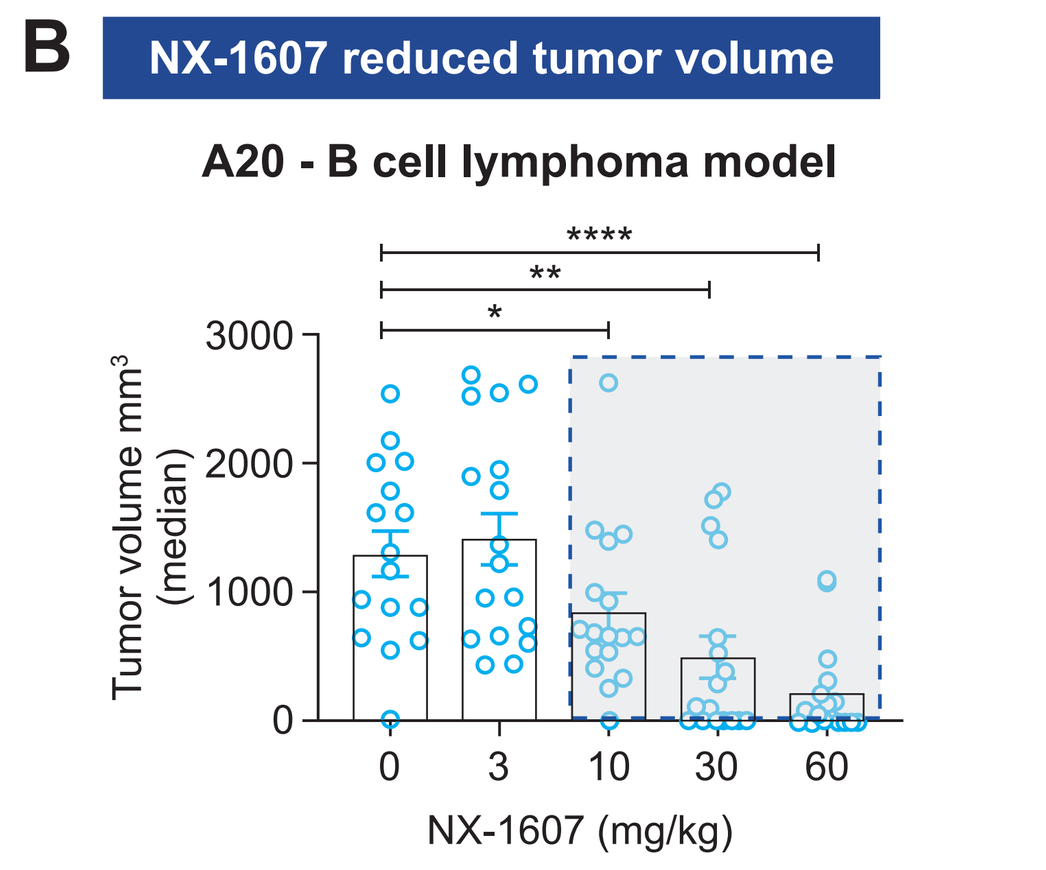
B
NX-1607 reduced tumor volume
A20 - B cell lymphoma model
Tumor volume mm³ (median)
NX-1607 (mg/kg)

Figure 4A: Whole blood from groups of mice (n=4 per group) administered a single oral dose of either vehicle (0.5% MC/0.2% Polysorbate 80 in deionized water) or increasing concentrations of NX-1607 at 0.3, 1, 3, 10, 30, or 90 mg/kg were collected in sodium heparin anti-coagulant. NX-1607 or vehicle treated blood was incubated with 2 μg/mL anti-CD3 & anti-CD28 for 5 mins and subsequently stimulated with crosslinker for 60 minutes. % Phosphorylated HS1, PLCY2 and ZAP70 was quantitated in T cells (CD8+) using flow cytometry.

Figure 4B: Mice bearing A20 tumors established on their right flanks were treated from Day 14 to Day 25 post tumor implant, by daily oral administration of either: vehicle (0.5% methylcellulose, 0.2% Polysorbate 80 in deionized water + 1 eq HCl) or NX-1607 at either 3, 10, 30 or 60 mg/kg. Beginning Day 14, tumor volumes were measured twice weekly and individual mean tumor volumes taken on Day 28 are shown. Statistical significance of differences in mean tumor volumes between treated groups and vehicle control group on Day 28 was evaluated using two tailed mann whitney for each arm compared to control. Data are mean ± s.e.m. ns, P > 0.05. **P ≤ 0.005, ***P ≤ 0.0005.

First-in-human trial

- As of June 16, 2022, 10 patients have enrolled at 4 ascending oral dose levels of NX-1607 (5, 15, 25 and 50 mg once daily):
 - – Preliminary PK data suggest a half-life of 4 to 10 hours and dose-proportional exposures (**Figure 6**).
 - – Dose-proportional increases of pHS1-expressing T cells were observed in Cycle 1 (**Figure 7A**).
 - – Together these data are consistent with preclinical human dose projections and are similar to pHS1 levels associated with anti-tumor activity in mouse models (**Figure 7B**).

Figure 5. Dose-response biomarker activity in human whole blood and human dose projection model based on PK/PD/efficacy and allometric scaling

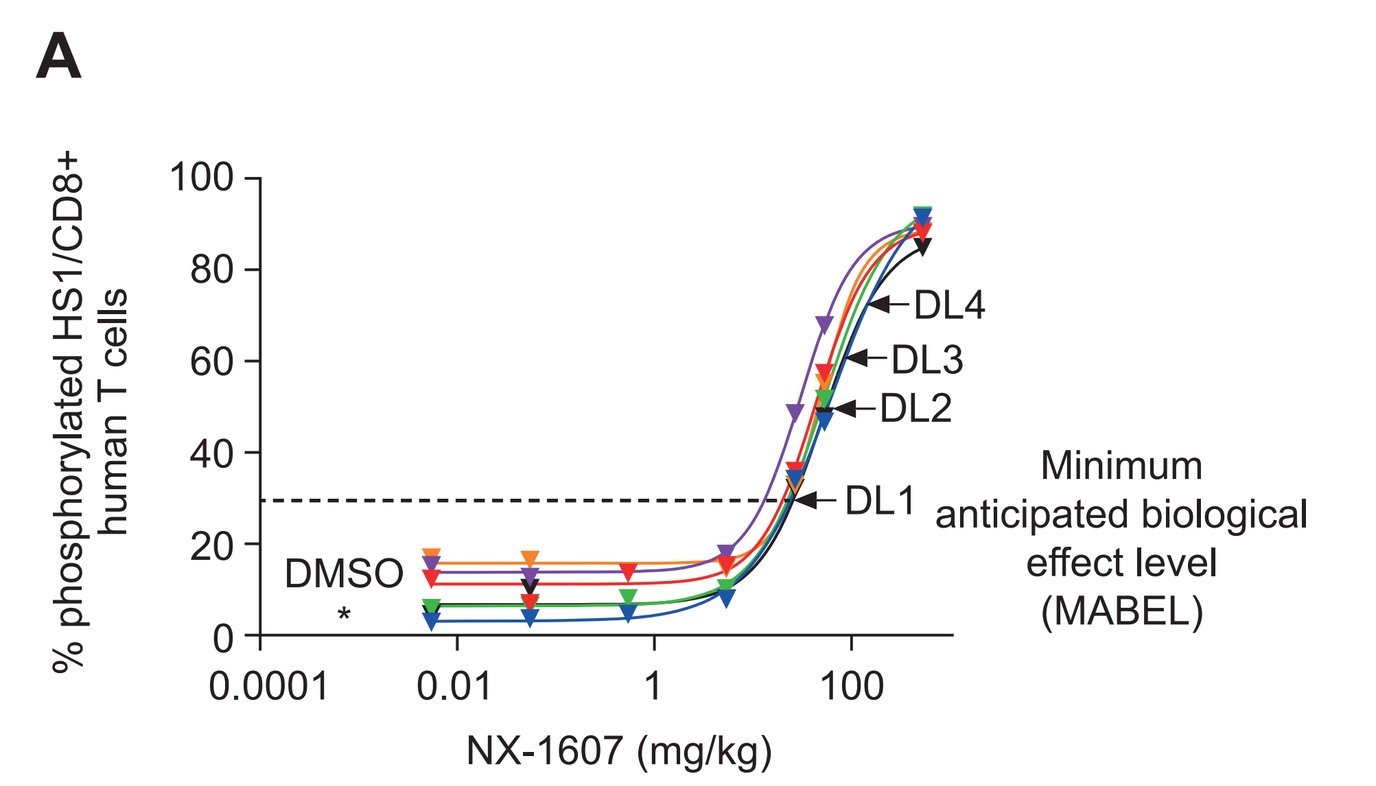
A
% phosphorylated HS1/CD8+ human T cells
DMSO
DL4
DL3
DL2
DL1
Minimum anticipated biological effect level (MABEL)
NX-1607 (mg/kg)

B

Proposed dose level[a]	NX-1607 dose (mg)	Estimated % HS1+/CD8+ T cells
-1	2.5	22.2
1[b]	5	30.0
2	15	49.7
3	25	60.6
4	50	74.0

[a]Dose levels in NX-1607-101.
[b]Minimum anticipated biological effect level (MABEL).

Figure 5A: Whole blood from six human donors collected in sodium heparin anti-coagulant was treated with increasing concentrations of NX-1607 for 2 hrs at 37˚C. Treated blood was incubated with 0.2 ug/mL anti-CD3 & 0.3ug/mL anti-CD28 for 5 mins and subsequently stimulated with crosslinker for 60 minutes. Percentage phosphorylated HS1 was quantitated in T cells (CD8+) using flow cytometry. Graph represents the mean (± SEM) percentage phosphorylated HS1 at each concentration.
Figure 5B: The predicted percentage phosphorylated HS1 at indicated doses was summarized.

Figure 6. NX-1607-101: Interim PK results suggest linear PK (Cycle 1 Day 1)

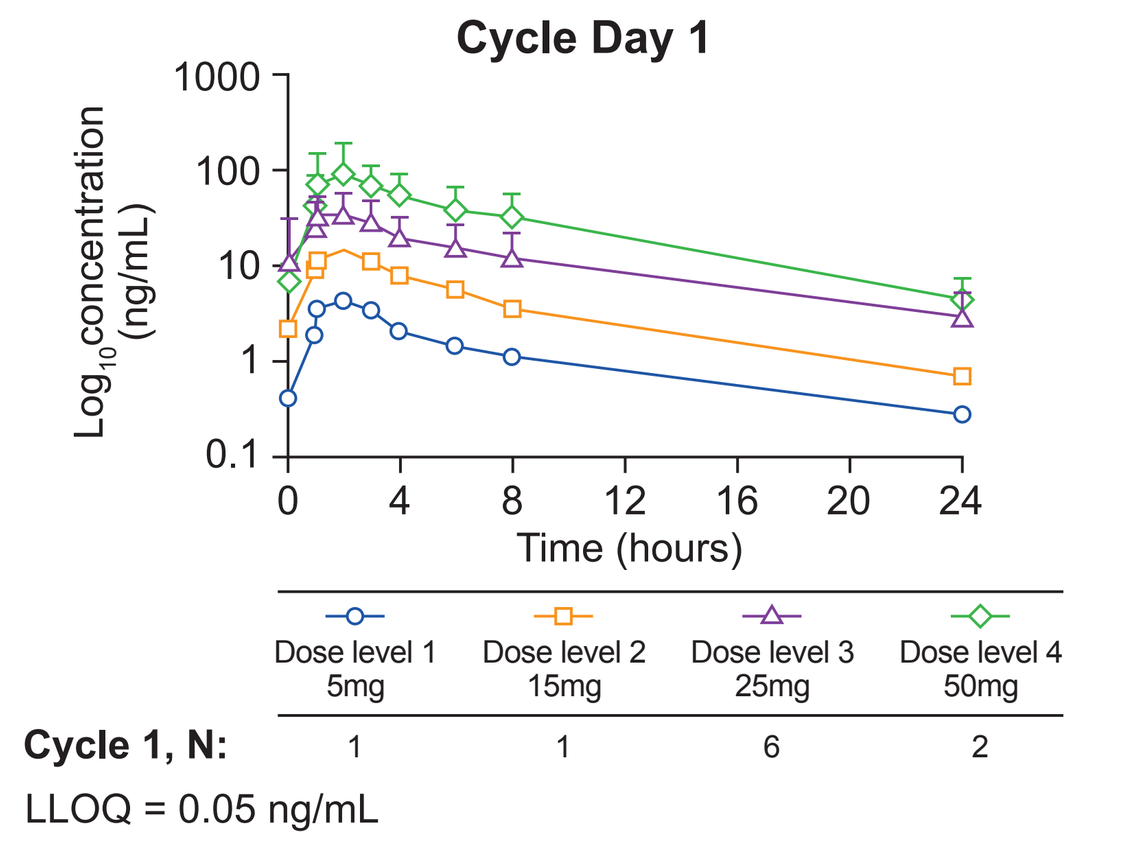
Cycle Day 1
Log10concentration (ng/mL)
Time (hours)
Dose level 1 5mg
Dose level 2 15mg
Dose level 3 25mg
Dose level 4 50mg
Cycle 1, N:
1
1
6
2
LLOQ = 0.05 ng/mL

Dose	C_{max} (ng/mL)	AUC_{last} (ng*h/mL)	T_{max} (h)	$T_{1/2}$ (h)
5 mg (n=1)	4.35	26.2	2.0	7.72
15 mg (n=1)	13.6	86.8	2.0	6.29
25 mg (n=6)	30.1 (109)	201 (37.9)	1.5 (1.0–3.0)	6.82 (27.5)
50 mg (n=2)	79.2 (134)	502 (113)	2.5 (2.0–3.0)	5.88 (7.7)

C_{max}, AUC_{last} are presented as geometric mean (geometric %CV); T_{max} is presented as median (range); $T_{1/2}$ is presented as mean (%CV).

Figure 7. NX-1607-101: Interim PD results show dose-proportionate increases of proximal biomarker pHS1, which correspond with levels associated with potent anti-tumor activity in mouse models

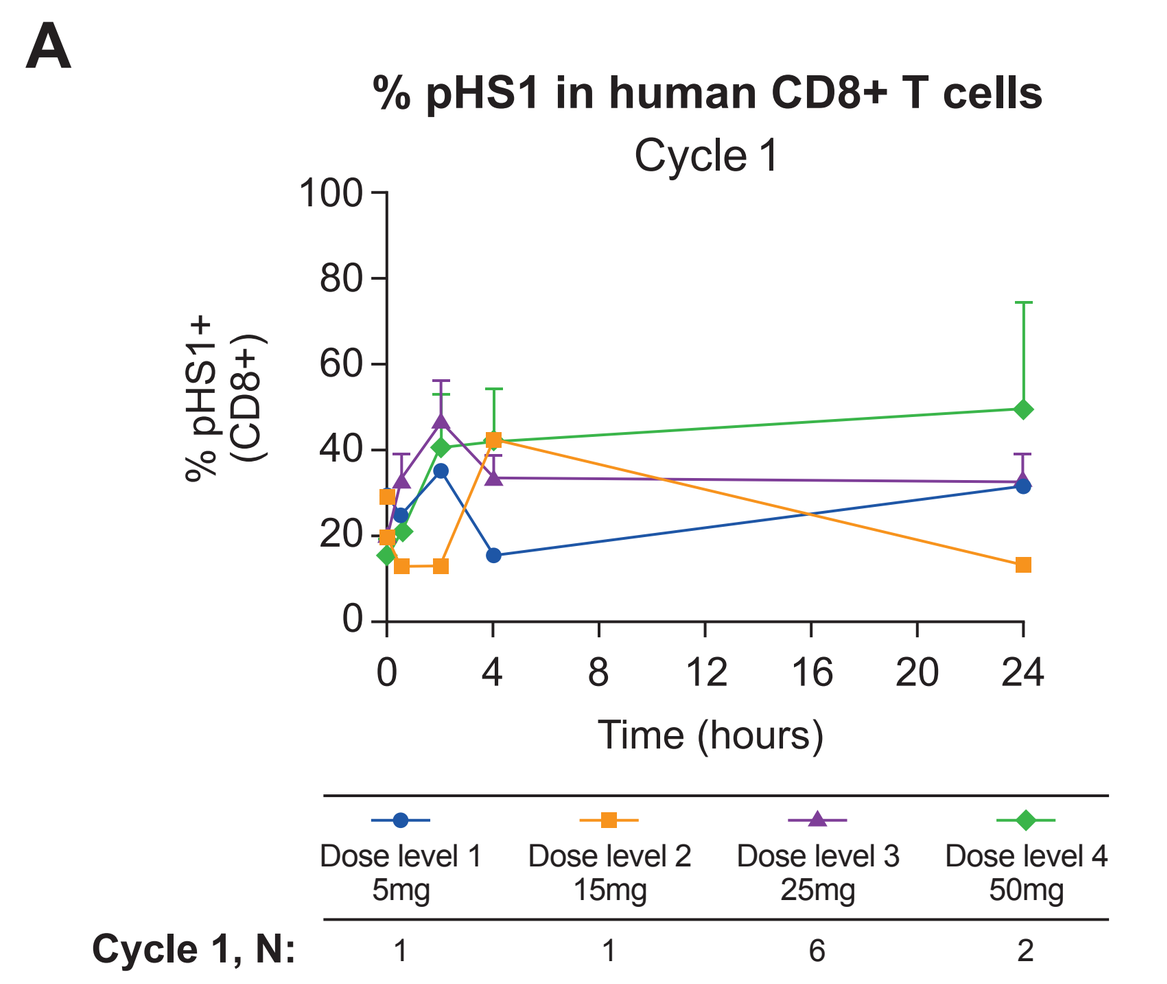
A
% pHS1 in human CD8+ T cells
Cycle 1
% pHS1+ (CD8+)
Time (hours)
Dose level 1 5mg
Dose level 2 15mg
Dose level 3 25mg
Dose level 4 50mg
Cycle 1, N:
1
1
6
2

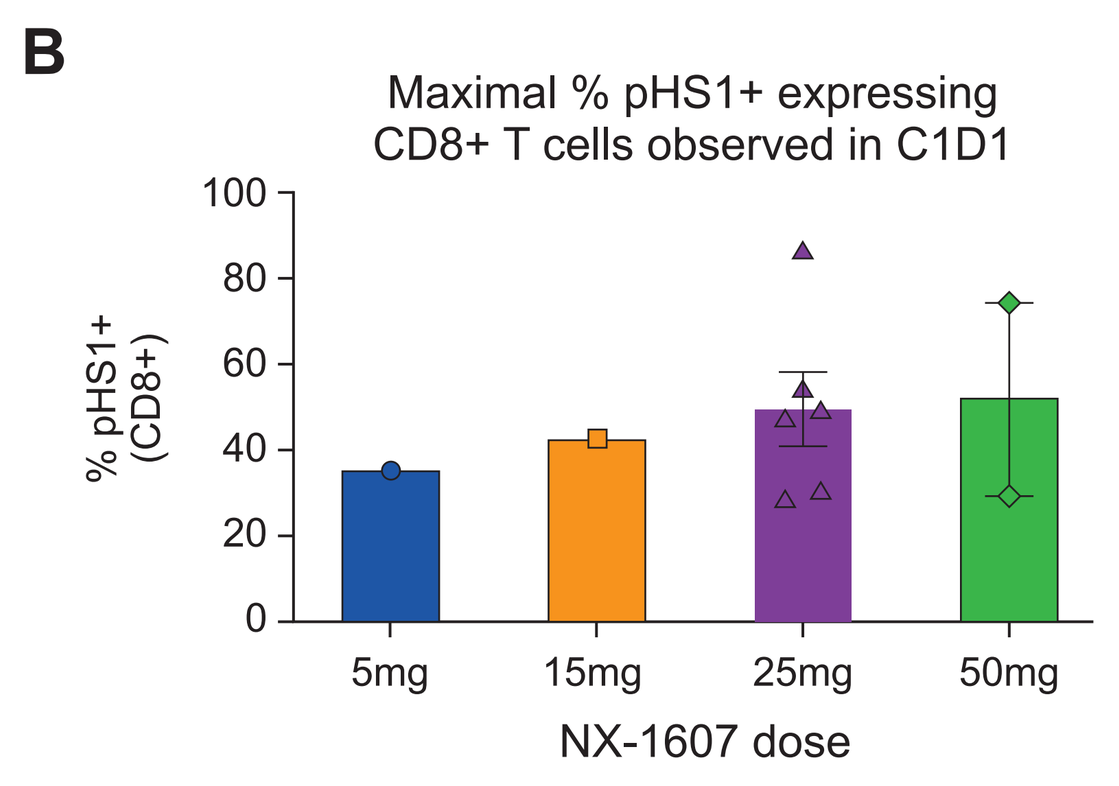
B
Maximal % pHS1+ expressing CD8+ T cells observed in C1D1
% pHS1+ (CD8+)
5mg
15mg
25mg
50mg
NX-1607 dose

- Patient cohorts dosed with NX-1607 once daily: 5, 15, 25 and 50 mg.
- Blood sampled for exposure and PD longitudinally:
 - – Cycle 1 day 1: pre, 0.5h, 2h, 4h, 24h trough.

Figure 7A: Pharmacodynamic profiles on Cycle 1 Day 1 in patients treated with 5 to 50 mg of NX-1607 once daily. Whole blood was collected into 2 types of TruCulture tubes (Tubes containing no stimulant, "Null" for aliquot 1; and tubes containing anti-CD3/CD28, "CD3/CD28" for aliquot 2) at each indicated timepoints pre- and post-dosing on scheduled visits (excluding end of treatment) in cycle 1. Percentage phosphorylated HS1 upon stimulation was quantitated in T cells (CD45+/CD8+) using flow cytometry. Graphs represent the mean (± SEM) percentage phosphorylated HS1 at each time point. Maximal change in phosphorylated HS1+ CD8+ T cells observed was graphed per dose treatment group. Baseline signal observed at approximately 20%. Graphs represent the mean (± SEM) percentage phosphorylated HS1 at each dose level.

Figure 7B: Percentage phosphorylated HS1 upon stimulation was quantitated in T cells (CD45+/CD8+) using flow cytometry. Graphs represent the mean (± SEM) percentage phosphorylated HS1 at each time point. Maximal change in phosphorylated HS1+ CD8+ T cells observed was graphed per dose treatment group. Baseline signal observed at approximately 20%. Graphs represent the mean (± SEM) percentage phosphorylated HS1 at each dose level.

Conclusions

- NX-1607 is a potent, reversible, and selective CBL-B inhibitor optimized for *in vivo* T cell activation.
- Multiple proprietary proximal biomarkers of CBL-B inhibition were identified in activated T cells.
- Validation of biomarkers in NHP and mouse *in vivo* models dosed orally with NX-1607 demonstrated target engagement with dose-dependent increases of phosphorylated biomarkers in CD8+ T cells.
- Phosphorylated HS1, a regulator of TCR signaling, was identified as the most robust and reproducible biologically relevant proximal biomarker for monitoring pharmacologic inhibition of CBL-B in whole blood.
- Percentage pHS1 coupled with allometric scaling of PK profiles were used to inform clinical dose selection and to characterize the activity of NX-1607 in a first-in-human clinical trial (NCT05107674):
 - – 10 patients have enrolled on study as of the data cut-off date (16 June 2022) and received NX-1607 at 4 ascending oral dose levels (5, 15, 25 and 50 mg once daily).
 - – Dose-dependent increases of pHS1-expressing T cells were observed in Cycle 1.
 - – Preliminary PK data suggest dose-proportional exposures.
- PK and PD data from non-clinical and clinical studies of NX-1607 have shown remarkable concordance, and biomarker levels observed in the clinic correspond with levels associated with potent anti-tumor activity in mouse models, suggesting that doses of NX-1607 being tested in the first-in-human trial are within target therapeutic range.

References

1. Rountree R, et al. Cancer Res 2021;81(13_Suppl): Poster #1595.
2. Fu G, et al. J Immunol 2012;189:2326–32.

Glossary

AUC_{last}, area under the time-concentration curve to last measurable concentration; CBL-B, Casitas B-lineage lymphoma proto-oncogene B; CBL-Bi, CBL-B inhibition; CD8, cluster of differentiation 8; C_{max}, maximum plasma concentration; LAT, linker for activated T cells; LLOQ, lower limit of quantification; MABEL, minimum anticipated biological effective level; PBMCs, peripheral blood mononuclear cells; PD, pharmacodynamics; PK, pharmacokinetics; NHP, non-human primate; pHS1, phosphorylated hematopoietic lineage cell-specific protein 1; pPLCγ2, phosphorylated phospholipase C gamma 2; pZAP70, phosphorylated zeta chain of T cell receptor-associated protein kinase 70; SLP76, SH2-domain-containing leukocyte protein (76 kDa); $T_{½}$, elimination half-life; TCR, T-cell receptor; T_{max}, time to C_{max}.

Acknowledgements

NX-1607-101 was funded by Nurix Therapeutics, Inc., and supported by the NIHR Cambridge Clinical Research Facility and Cambridge Experimental Cancer Medicine Centre. The Cambridge Human Research Tissue Bank is supported by the NIHR Cambridge Biomedical Research Centre. Nurix Therapeutics, Inc. also funded the provision of editorial support provided by Miller Medical Communications.

Other NX-1607 posters presented at SITC 2022

10 November 2022
#331 Gallotta M, et al. A novel small molecule inhibitor of CBL-B shows potent antitumor activity in combination with Pmel-1 adoptive cell transfer in an aggressive mouse melanoma model

11 November 2022
#824 Gallotta M, et al. NX-1607: a small molecule inhibitor of the CBL-B E3 ubiquitin ligase, promotes T and NK cell activation and enhances NK-mediated ADCC in a mouse lymphoma tumor model

Presented at the Society for Immunotherapy of Cancer (SITC) 37th Annual Meeting, Nov 8–12, 2022, in Boston, MA, USA.