UNIVERSITY OF MIAMI
MILLER SCHOOL of MEDICINE

Drug-resistance mutations in BTK occur in distinct enzymatic classes and are overcome by BTK degradation

Skye Montoya[1], Jessie Bourcier[2], Mark Noviski[3], Hao Lu[3], Meghan C. Thompson[4], Jacob Jahn[1], Anya K. Sondhi[1], Stefan Gajewski[3], Ying Siow (May) Tan[3], Stephanie Yung[3], Aleksandra Urban[2], Eric Wang[2], Xiaoli Mi[2], Hugo Bousquet[3], Nivetha Brathaban[3], Brandon Bravo[3], Melissa Gessner[3], Cristiana Guiducci[3], James N. Iuliano[3], Tim Kane[3], Ratul Mukerji[3], Janine Powers[3], Mateo Sanchez Garcia de los Rios[3], Jordan Ye[3], Carla Barrientos Risso[1], Daniel Tsai[1], Gabriel Pardo[1], Ryan Q. Notti[2], Alex Pardo[1], Maurizio Affer[1], Vindhya Nawaratne[1], Tulasigeri Totiger[1], Camila Pena-Velasquez[2], Joanna M. Rhodes[2], Andrew D. Zelenetz[2], Alvaro Alencar[1], Lindsey E. Roeker[2], Adam Linley[2], Rajesh Kumar Soni[2], Sigrid S. Skånland[2], Robert J. Brown[3], Anthony R. Mato[2], Gwenn M. Hansen[3], Omar Abdel-Wahab[2], and Justin Taylor[1]

Affiliations: 1. Sylvester Comprehensive Cancer Center at the University of Miami , Miami, FL, USA 2. Memorial Sloan Kettering Cancer Center, New York, NY, USA 3. Nurix Therapeutics, San Francisco, CA, USA

Background

Bruton's tyrosine kinase (BTK) inhibitors have transformed the therapeutic landscape for patients with chronic lymphocytic leukemia (CLL) and Non-Hodgkin lymphomas (NHL), due to the critical role of BTK in the proliferation and survival of B-cell malignancies.

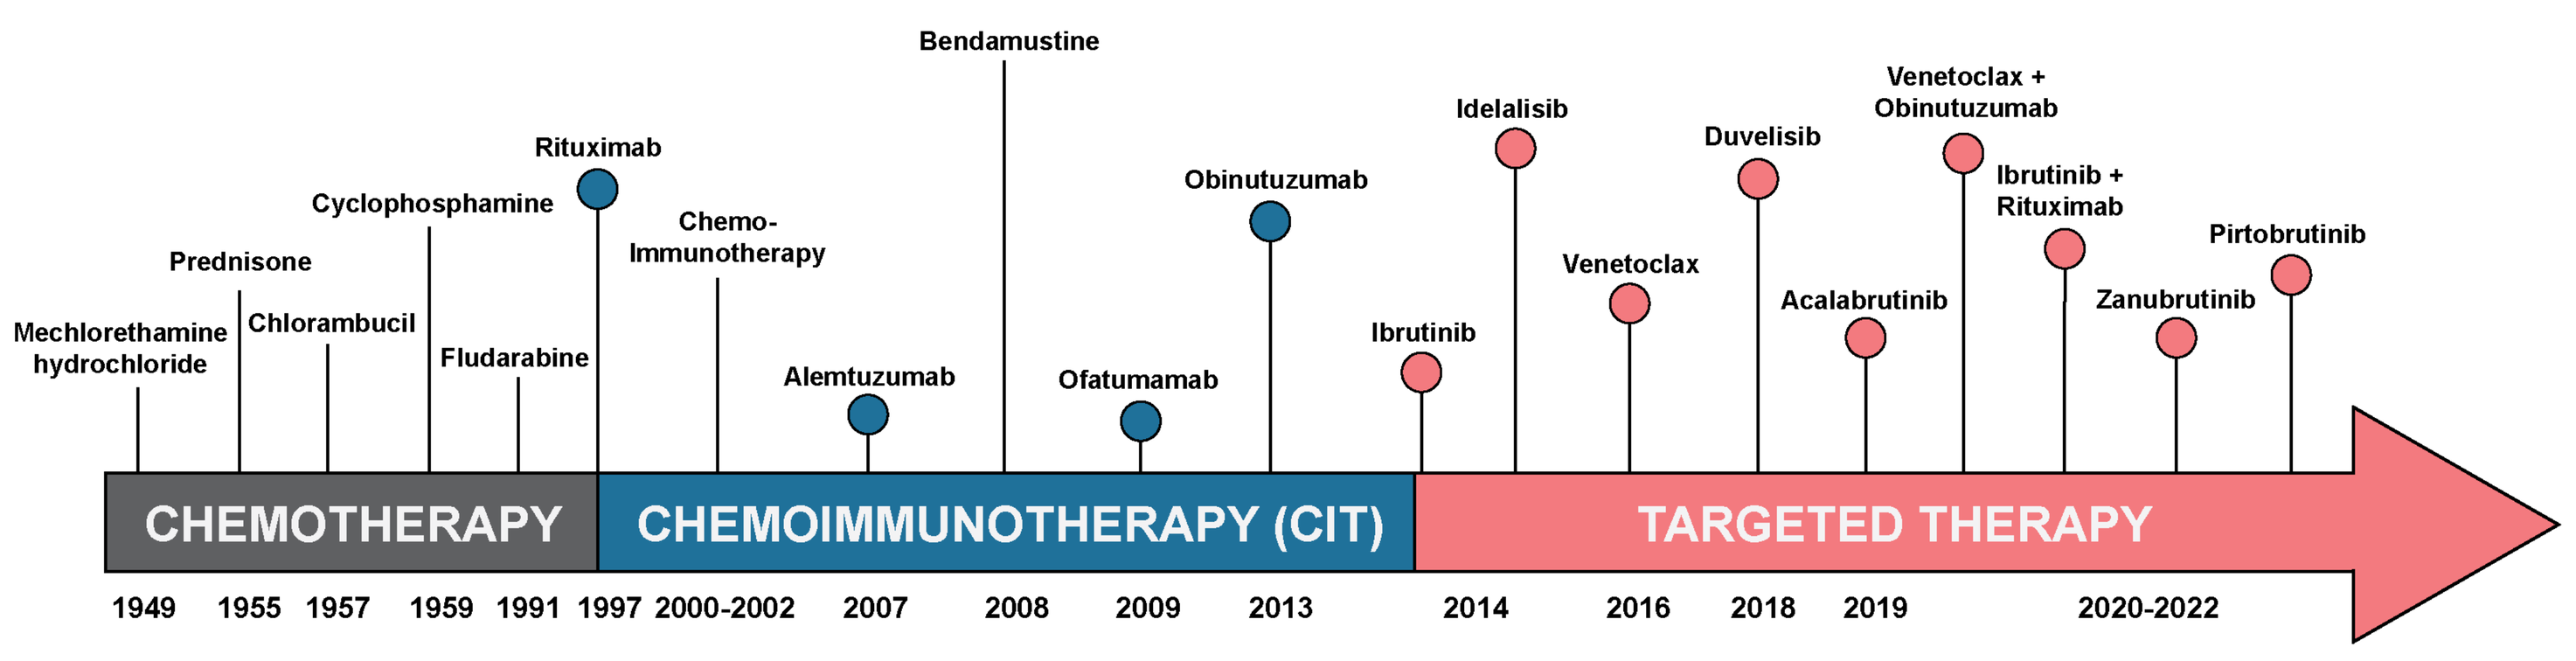

Mechlorethamine hydrochloride
Prednisone
Chlorambucil
Cyclophosphamine
Fludarabine
Rituximab
Chemo-Immunotherapy
Bendamustine
Alemtuzumab
Ofatumamab
Obinutuzumab
Ibrutinib
Idelalisib
Venetoclax
Duvelisib
Acalabrutinib
Venetoclax + Obinutuzumab
Ibrutinib + Rituximab
Zanubrutinib
Pirtobrutinib
CHEMOTHERAPY
CHEMOIMMUNOTHERAPY (CIT)
TARGETED THERAPY
1949 1955 1957 1959 1991 1997 2000-2002 2007 2008 2009 2013 2014 2016 2018 2019 2020-2022

Increasing use of covalent Bruton tyrosine kinase (BTK) inhibitors ibrutinib, acalabrutinib, and zanubrutinib as well as noncovalent inhibitors nemtabrutinib and pirtobrutinib, have elucidated a series of acquired BTK mutations, some of which can confer cross-resistance to other BTK inhibitors in patients with B-cell malignancies.

BTK Mutations in CLL patients confer resistance to non-covalent BTK inhibitors

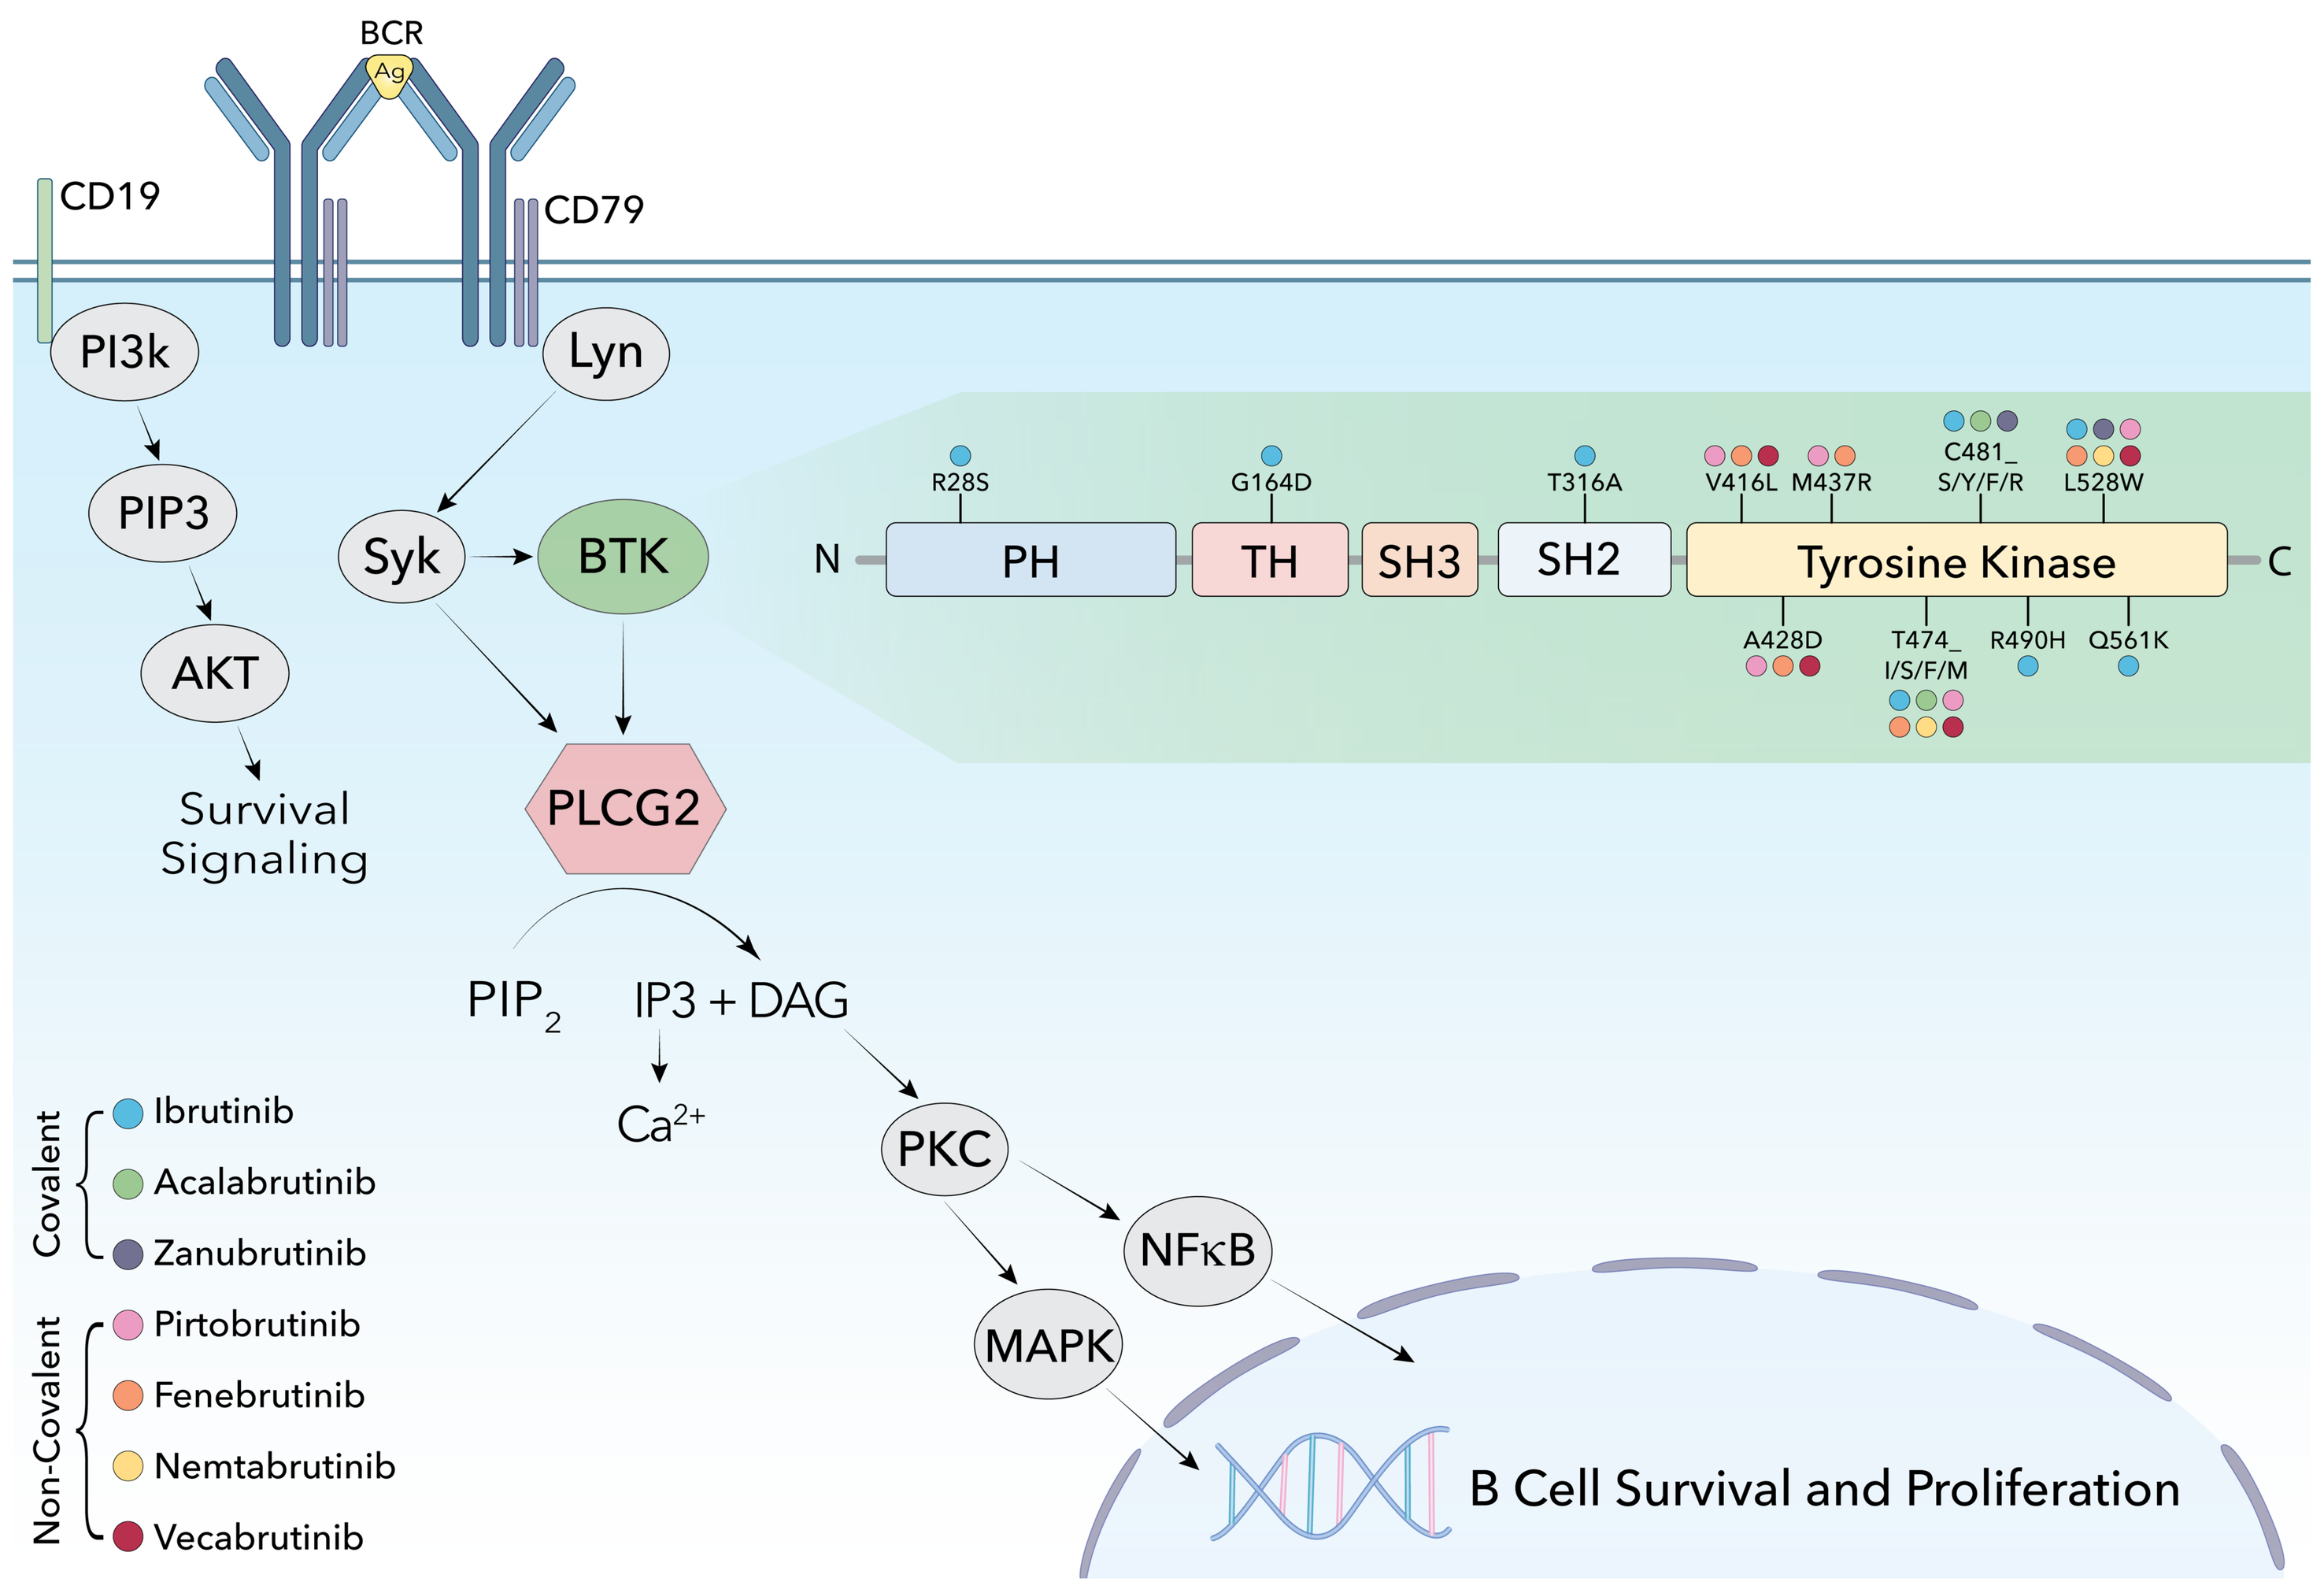

BCR
Ag
CD19
CD79
PI3k
Lyn
PIP3
Syk
BTK
AKT
Survival Signaling
PLCG2
PIP2
IP3 + DAG
Ca2+
PKC
NFκB
MAPK
B Cell Survival and Proliferation
N
PH
TH
SH3
SH2
Tyrosine Kinase
C
R28S
G164D
T316A
V416L
M437R
C481_ S/Y/F/R
L528W
A428D
T474_ I/S/F/M
R490H
Q561K
Covalent
Ibrutinib
Acalabrutinib
Zanubrutinib
Non-Covalent
Pirtobrutinib
Fenebrutinib
Nemtabrutinib
Vecabrutinib

BTK mutants lack enzymatic function but sustain downstream BCR signaling

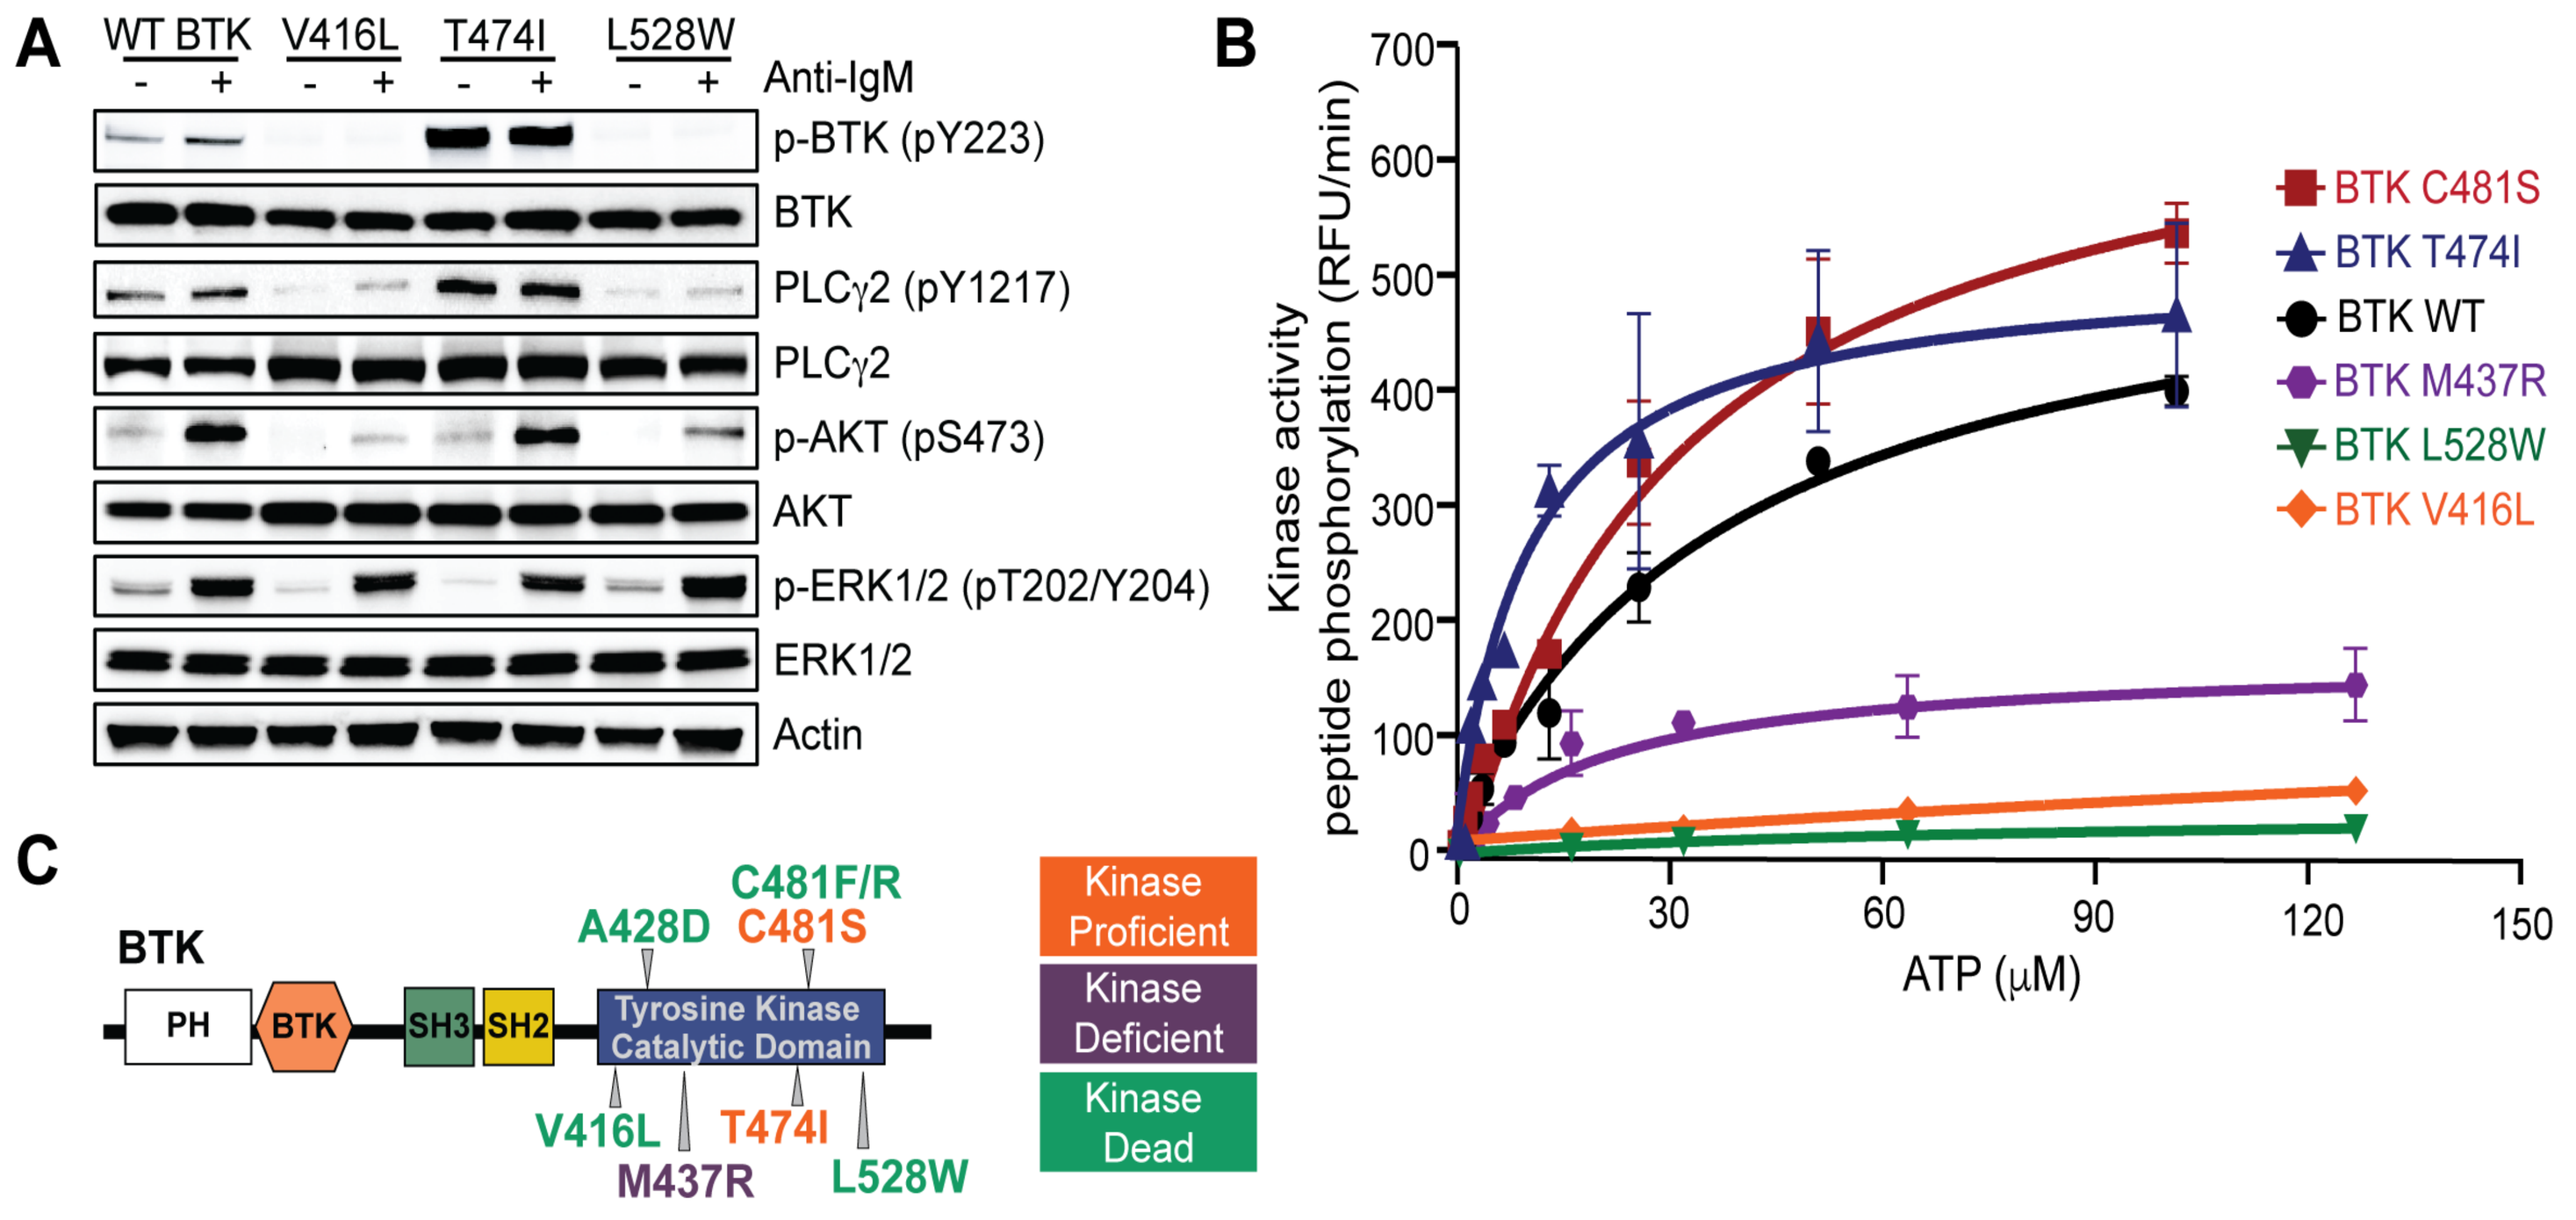

A
WT BTK
V416L
T474I
L528W
Anti-IgM
p-BTK (pY223)
BTK
PLCγ2 (pY1217)
PLCγ2
p-AKT (pS473)
AKT
p-ERK1/2 (pT202/Y204)
ERK1/2
Actin
B
Kinase activity peptide phosphorylation (RFU/min)
ATP (μM)
BTK C481S
BTK T474I
BTK WT
BTK M437R
BTK L528W
BTK V416L
C
BTK
PH
BTK
SH3
SH2
Tyrosine Kinase Catalytic Domain
A428D
C481F/R
C481S
V416L
M437R
T474I
L528W
Kinase Proficient
Kinase Deficient
Kinase Dead

Methodology

BTK Mutant Signaling Studies: To define the signaling mechanisms of kinase dead BTK mutants, we generated CRISPR-CAS9 knockin mutant cells and utilize several orthogonal proteomic approaches in BTK-dependent human lymphoma B cells expressing WT or mutant (C481S, V416L, T474I and L528W) BTK. We performed global phosphoproteomics, kinobead assays, BTK immunoprecipitation mass spectrometry studies, and 2D differential gel electrophoresis to unbiasedly elucidate a novel scaffolding function of BTK.

BTK Degrader Studies: Given that mutations conferring resistance to BTK inhibitors lack enzymatic activity, we evaluated means to eliminate, rather than enzymatically inhibit, mutant BTK proteins. Nurix generated NX-2127, a heterobifunctional degrader molecule which brings BTK into close proximity with cereblon, leading to ubiquitylation and proteasomal degradation of BTK (as demonstrated by biophysical and structural data).

Results

Kinase Dead BTK mutants sustain downstream BCR signaling utilizing novel signaling interactions

Figure 1

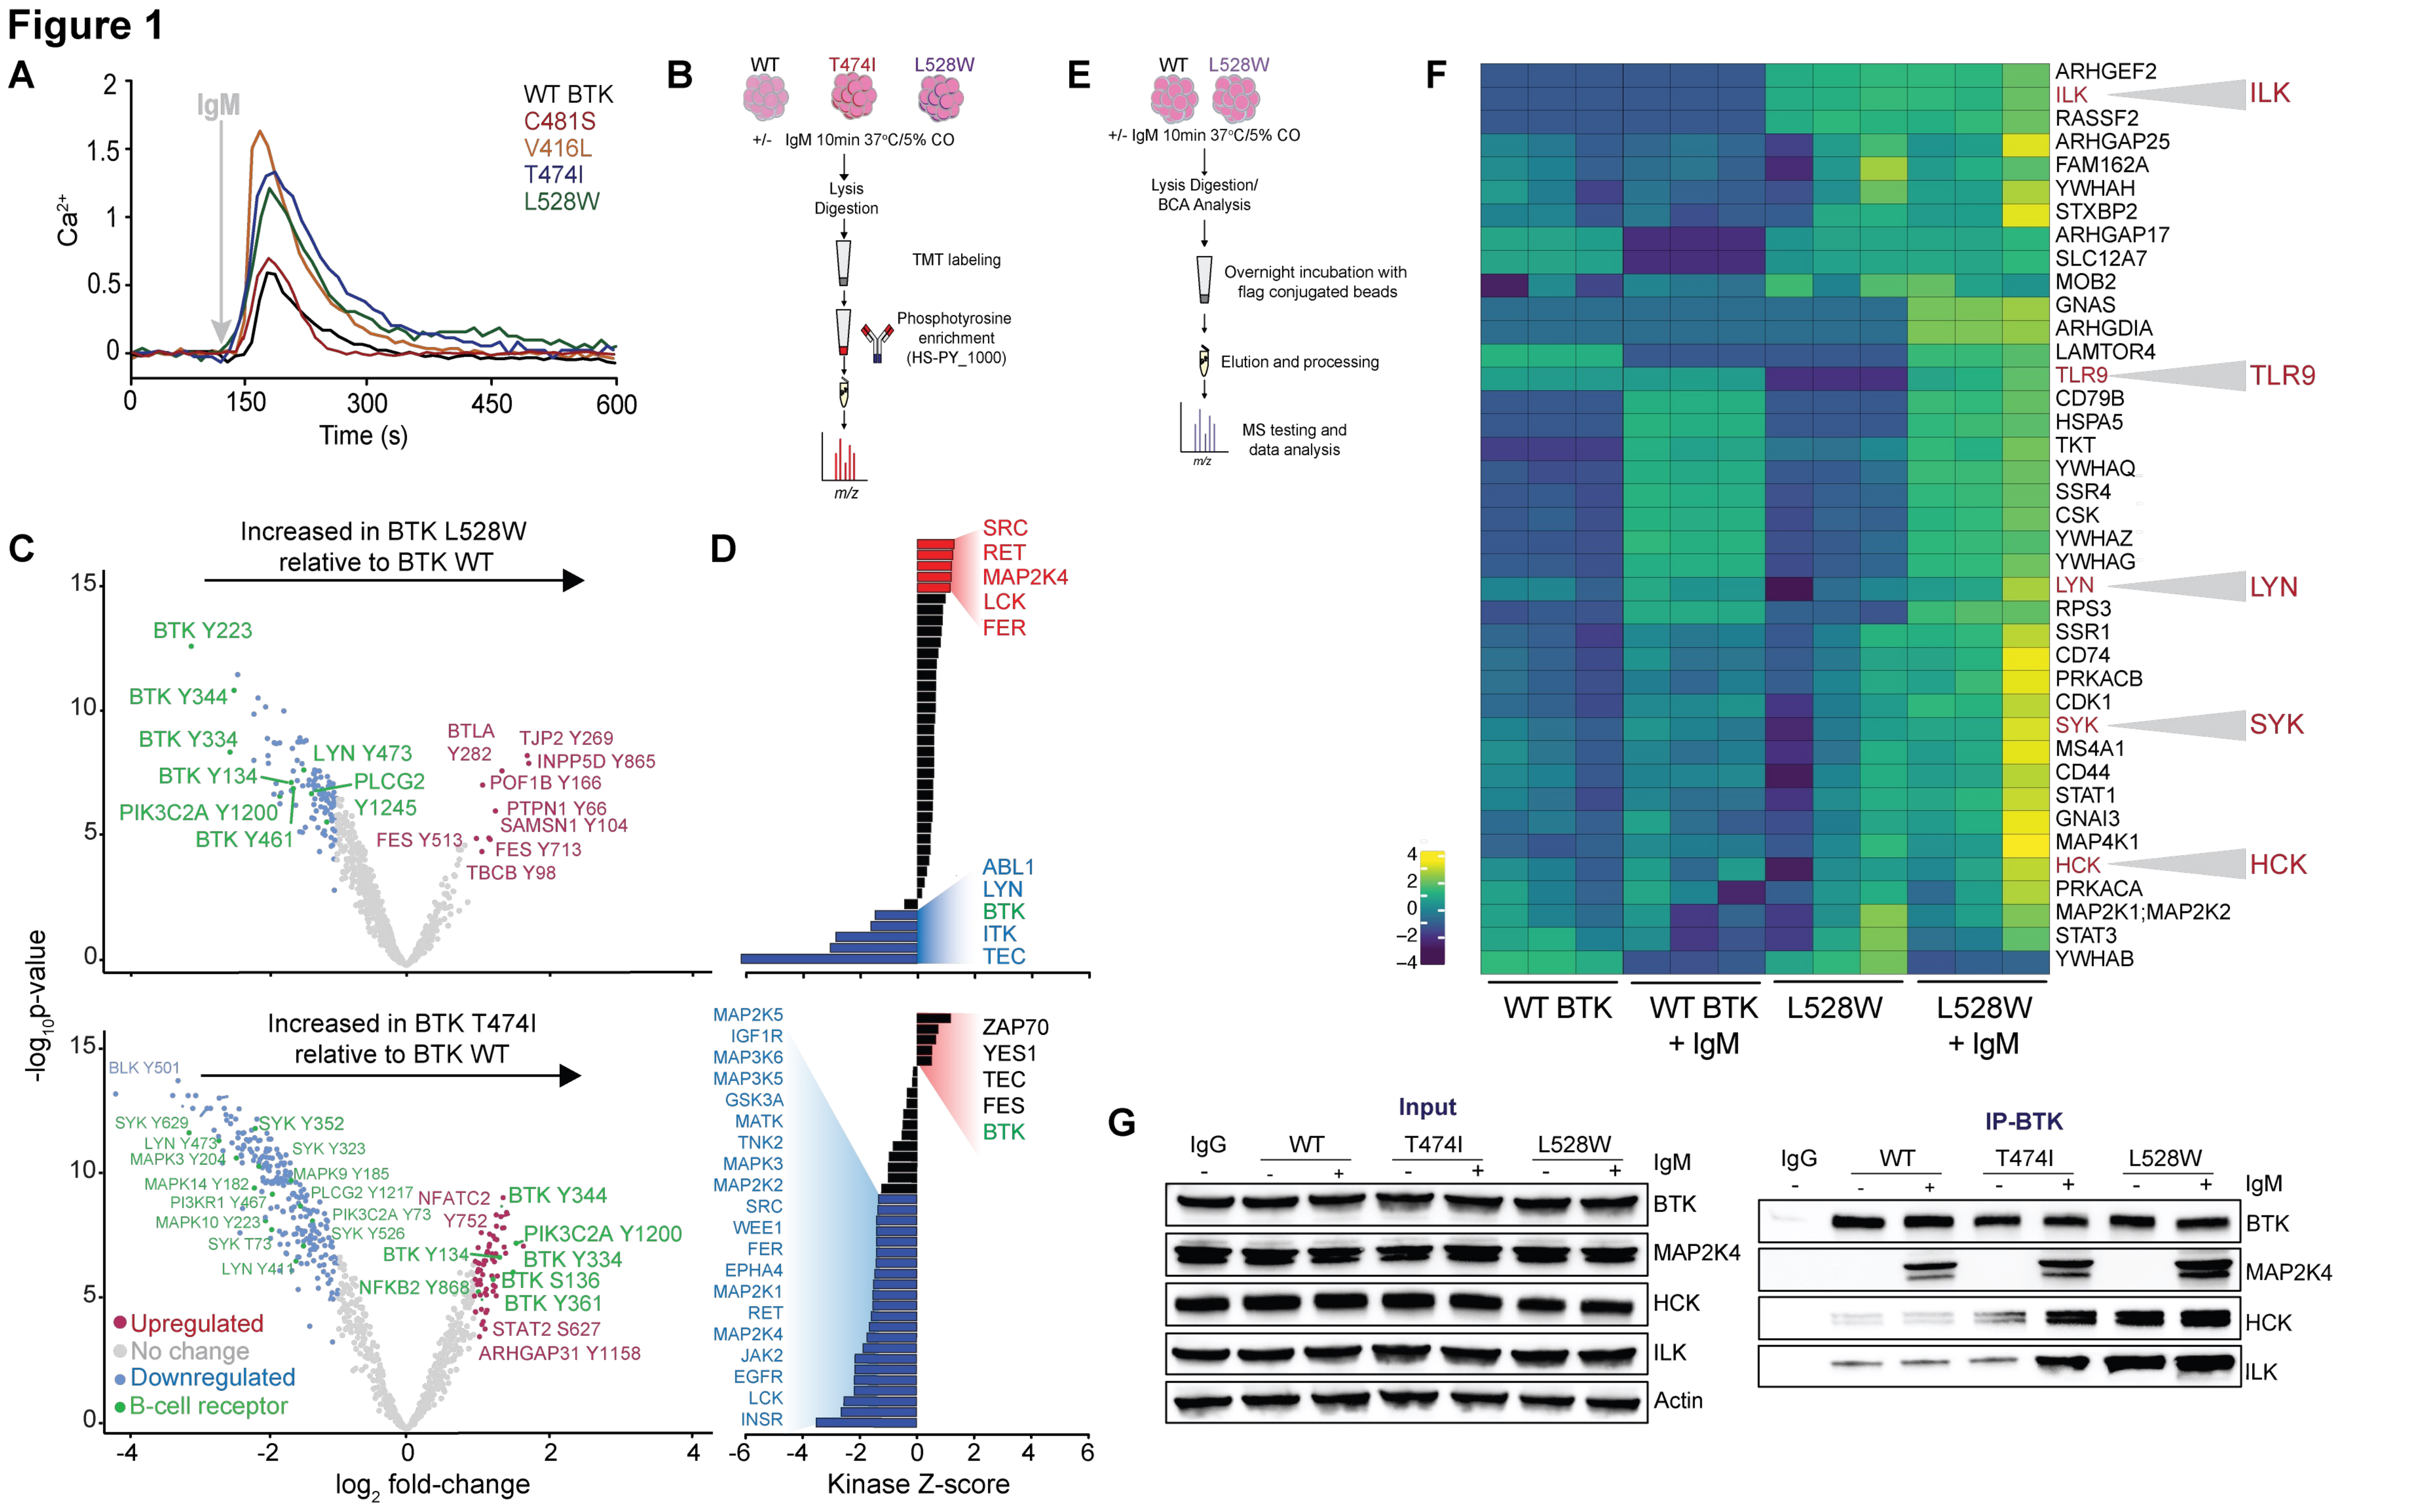

A
IgM
WT BTK
C481S
V416L
T474I
L528W
Ca2+
Time (s)
B
WT
T474I
L528W
+/- IgM 10min 37°C/5% CO
Lysis Digestion
TMT labeling
Phosphotyrosine enrichment (HS-PY_1000)
m/z
E
WT
L528W
+/- IgM 10min 37°C/5% CO
Lysis Digestion/ BCA Analysis
Overnight incubation with flag conjugated beads
Elution and processing
MS testing and data analysis
F
ARHGEF2
ILK
RASSF2
ARHGAP25
FAM162A
YWHAH
STXBP2
ARHGAP17
SLC12A7
MOB2
GNAS
ARHGDIA
LAMTOR4
TLR9
CD79B
HSPA5
TKT
YWHAQ
SSR4
CSK
YWHAZ
YWHAG
LYN
RPS3
SSR1
CD74
PRKACB
CDK1
SYK
MS4A1
CD44
STAT1
GNAI3
MAP4K1
HCK
PRKACA
MAP2K1;MAP2K2
STAT3
YWHAB
WT BTK
WT BTK + IgM
L528W
L528W + IgM
C
Increased in BTK L528W relative to BTK WT
BTK Y223
BTK Y344
BTK Y334
BTK Y134
LYN Y473
PLCG2 Y1245
PIK3C2A Y1200
BTK Y461
BTLA Y282
TJP2 Y269
INPP5D Y865
POF1B Y166
PTPN1 Y66
SAMSN1 Y104
FES Y513
FES Y713
TBCB Y98
Increased in BTK T474I relative to BTK WT
BLK Y501
SYK Y629
LYN Y473
MAPK3 Y204
SYK Y352
SYK Y323
MAPK9 Y185
MAPK14 Y182
PI3KR1 Y467
PLCG2 Y1217
MAPK10 Y223
PIK3C2A Y73
SYK Y526
SYK T73
LYN Y411
NFATC2 Y752
BTK Y344
PIK3C2A Y1200
BTK Y334
BTK Y134
BTK S136
BTK Y361
NFKB2 Y868
STAT2 S627
ARHGAP31 Y1158
Upregulated
No change
Downregulated
B-cell receptor
-log10p-value
log2 fold-change
D
SRC
RET
MAP2K4
LCK
FER
ABL1
LYN
BTK
ITK
TEC
MAP2K5
IGF1R
MAP3K6
MAP3K5
GSK3A
MATK
TNK2
MAPK3
MAP2K2
SRC
WEE1
FER
EPHA4
MAP2K1
RET
MAP2K4
JAK2
EGFR
LCK
INSR
ZAP70
YES1
TEC
FES
BTK
Kinase Z-score
G
Input
IP-BTK
IgG
WT
T474I
L528W
IgM
BTK
MAP2K4
HCK
ILK
Actin

Figure 1: Kinase dead forms of BTK have sustained B-cell receptor (BCR) signaling through enhanced interactions with additional signaling proteins downstream of BCR. **(A)** IgM-mediated intracellular calcium release in BTK mutants. **(B)** Phosphoproteomics design schematic with differentially phosphorylated peptides of kinase active BTK mutant, T474I, and kinase dead BTK mutant, L528W **(C)** with predicted alterations in protein kinase mediated signaling **(D)**. BTK co-immunoprecipitation mass spectrometry experiment **(E)** revealed enhanced physical interactions of kinase dead BTK with several protein kinases malignant B cells **(F)**. Interactions with HCK and ILK were further validated by western blot analysis **(G)**.

BTK degrader NX-2127 overcomes non-covalent and covalent BTK inhibitor resistance

Figure 2

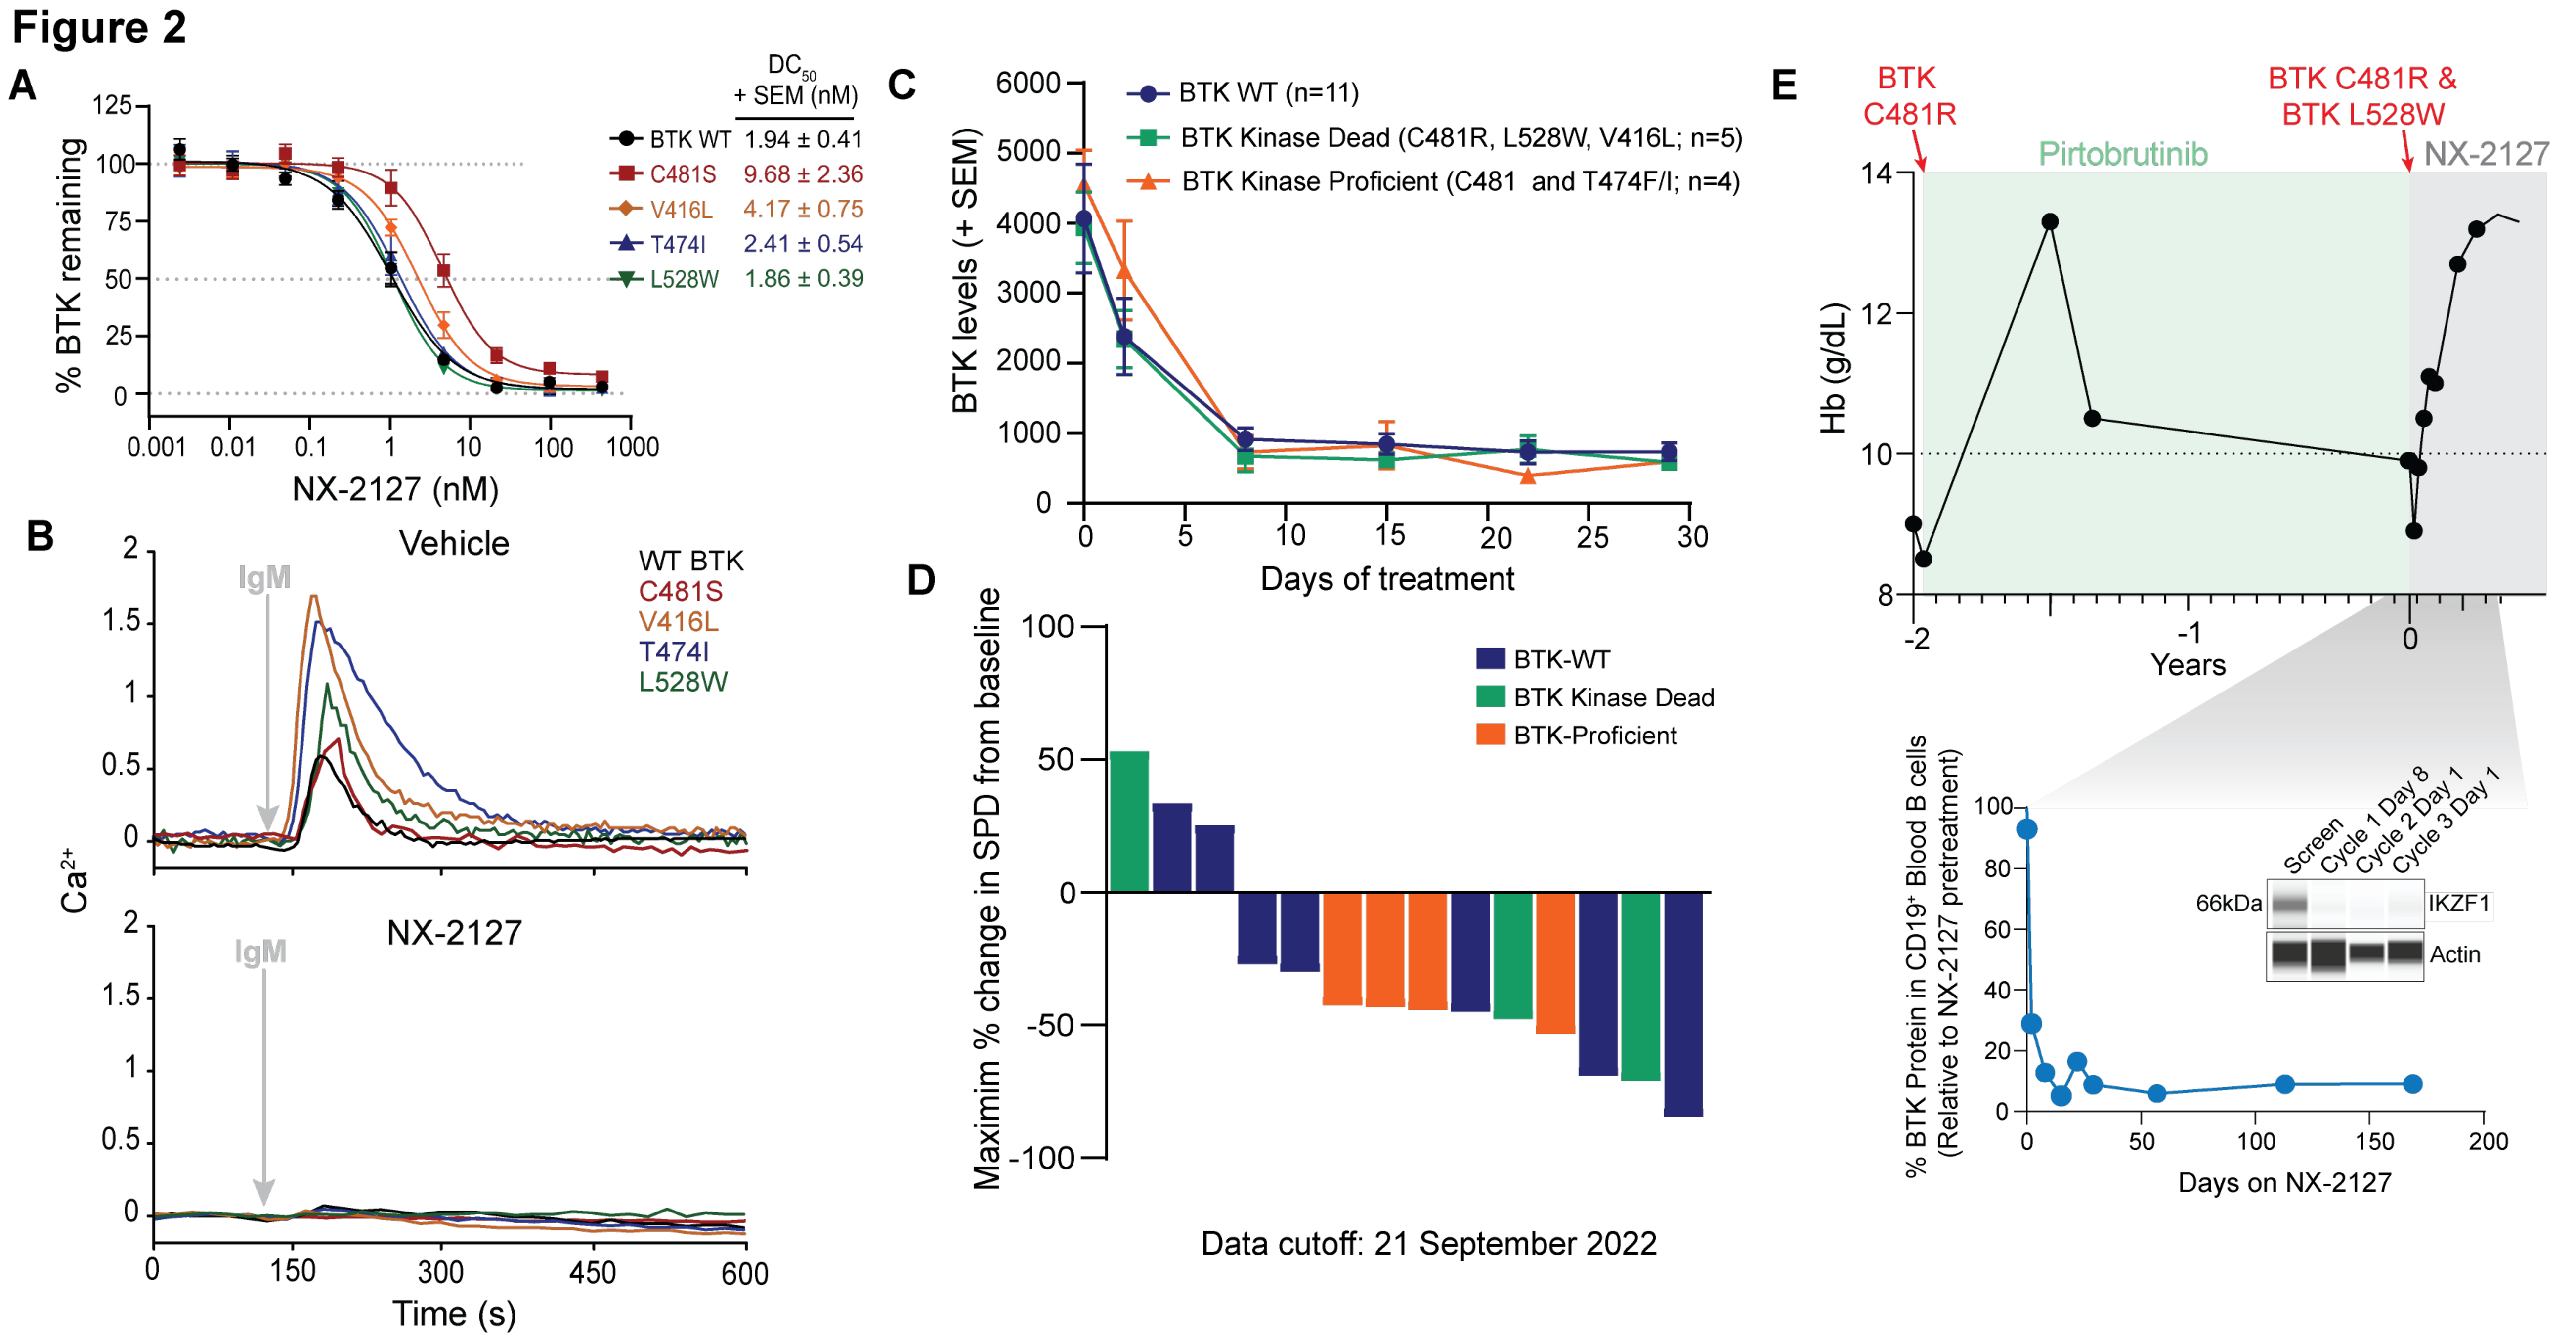

A
DC50 + SEM (nM)
BTK WT 1.94 ± 0.41
C481S 9.68 ± 2.36
V416L 4.17 ± 0.75
T474I 2.41 ± 0.54
L528W 1.86 ± 0.39
% BTK remaining
NX-2127 (nM)
B
Vehicle
NX-2127
IgM
WT BTK
C481S
V416L
T474I
L528W
Ca2+
Time (s)
C
BTK WT (n=11)
BTK Kinase Dead (C481R, L528W, V416L; n=5)
BTK Kinase Proficient (C481 and T474F/I; n=4)
BTK levels (+ SEM)
Days of treatment
D
BTK-WT
BTK Kinase Dead
BTK-Proficient
Maximim % change in SPD from baseline
Data cutoff: 21 September 2022
E
BTK C481R
Pirtobrutinib
BTK C481R & BTK L528W
NX-2127
Hb (g/dL)
Years
% BTK Protein in CD19+ Blood B cells (Relative to NX-2127 pretreatment)
Days on NX-2127
Screen
Cycle 1 Day 8
Cycle 2 Day 1
Cycle 3 Day 1
66kDa
IKZF1
Actin

NX-2127 degrades BTK drug resistant mutant proteins and abrogates BCR signaling. **(A)** Dose curves to determine degradation concentration (DC_{50}) of BTK for WT and mutant BTK treated with NX-2127 for 24 hours. **(B)** IgM-mediated intra-cellular calcium release in BTK mutants treated with either vehicle (DMSO) or NX-2127. **(C-D)** Patient data from Phase 1b dose expansion trial of NX-2127 with one exemplary patient highlighted in **(E)**.

Conclusion

These data reveal a distinct oncogenic scaffolding function of kinase dead BTK which confers resistance across FDA-approved BTK inhibitors. Importantly, regardless of enzymatic group, the BTK mutants evaluated in this study are susceptible to BTK degradation both preclinically and in patients currently being treated in a first-in-human Phase 1b dose expansion trial of NX-2127 (clinicaltrials.gov NCT04830137).

Acknowledgements

Skye Montoya is supported by the Ruth L. Kirschstein National Research Service Award for Individual Predoctoral Fellows (F31) under the award number 1F31CA275378-01. JT is supported by the Doris Duke Charitable Foundation and the Sylvester Comprehensive Cancer Center.

NIH NATIONAL CANCER INSTITUTE Center for Cancer Research

DORIS DUKE CHARITABLE FOUNDATION