Robust Bruton's tyrosine kinase (BTK) degradation with NX-5948, an oral BTK degrader, in a first-in-human phase 1a trial in patients with relapsed/refractory B cell malignancies

[1]Kim Linton, [2]Francesco Forconi, [3]David Lewis, [4]John Riches, [5]Dima El-Sharkawi, [6]Mary Gleeson, [7]Sarah G. Injac, [7]Srinand Nandakumar, [7]May Tan, [7]Ganesh Cherala, [8]Graham P. Collins

[1]Manchester Cancer Research Centre, Division of Cancer Sciences, Manchester, UK; [2]University Hospital Southampton NHS Trust, Southampton, UK; [3]Derriford Hospital, Plymouth, UK; [4]Barts Cancer Institute, Queen Mary University of London, UK; [5]Royal Marsden NHS Foundation Trust, Sutton, UK; [6]Sarah Cannon Research Institute, Guy's and St Thomas' NHS Foundation Trust, London, UK; [7]Nurix Therapeutics, Inc., San Francisco, CA, USA; [8]Oxford University Hospitals NHS Foundation Trust, Oxford, UK

Background

- Bruton's tyrosine kinase (BTK) is a key component of the B cell receptor signaling pathway; chronic activation of BTK-mediated B cell receptor signaling is a hallmark of many B cell malignancies.
- BTK inhibition has been validated as an effective therapeutic strategy in patients with B cell malignancies; however, emerging patterns of resistance and intolerance limit the utility of covalent and/or non-covalent BTK inhibitors in later lines of treatment.
- Novel therapeutics that can overcome emerging resistance mutations may represent an alternative treatment option for patients who have developed resistance to BTK inhibitors or in B cell indications where treatment with BTK inhibitors has been less effective.[1]
- NX-5948 is a novel, orally administered, small molecule that induces BTK degradation via recruitment of the cereblon E3 ubiquitin ligase complex, without inducing degradation of other cereblon neo-substrates (Figure 1):
 - NX-5948 induces sub-nanomolar potency degradation of both wild-type and known mutant forms of BTK *in vitro*.[2]
 - Orally administered NX-5948 results in BTK degradation in circulating and splenic B cells, and exhibits potent tumor growth inhibition in TMD8 xenograft models that contain either wild-type BTK or BTKi-resistant mutations.[3]
 - NX-5948 can cross the blood-brain barrier and degrade BTK intracranially, translating to preclinical efficacy in mouse brain lymphoma disease models.[3]

Figure 1. NX-5948: Mechanism of action

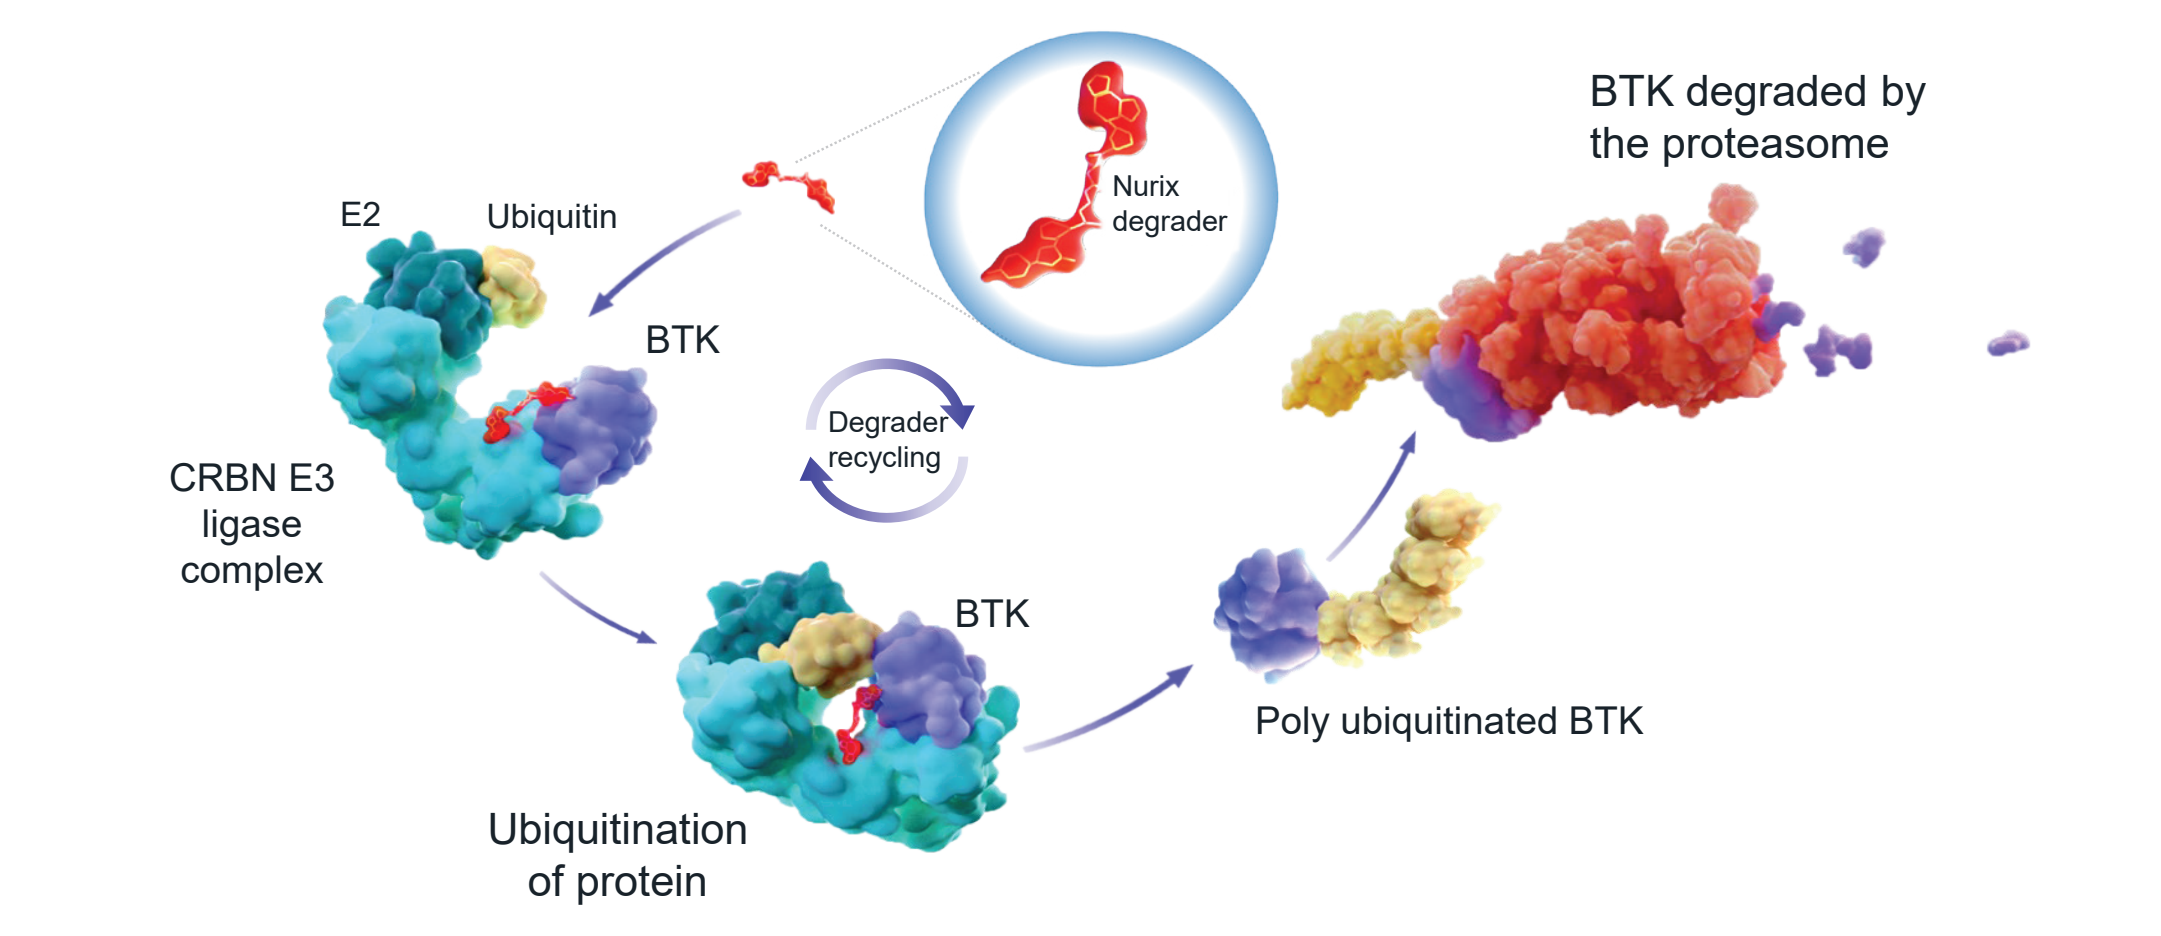
Nurix
degrader
BTK degraded by
the proteasome
E2
Ubiquitin
BTK
Degrader
recycling
CRBN E3
ligase
complex
BTK
Poly ubiquitinated BTK
Ubiquitination
of protein

Methods

- NX-5948-301 is a first-in-human, dose-escalation (Phase 1a) and cohort-expansion (Phase 1b) study designed to evaluate the safety, tolerability, and preliminary efficacy of NX-5948 in adult patients with relapsed and refractory B cell malignancies (Figure 2):
 - Phase 1a (dose escalation) will utilize a standard 3+3 dose-escalation design of 3 patients per dose level (starting dose 50 mg qd), in the absence of dose-limiting toxicities (DLTs).
 - Phase 1b (dose expansion) will include up to 4 expansion cohorts. The recommended Phase 1b dose will be determined following assessment of pharmacokinetics (PK)/pharmacodynamics (PD), safety, and anti-tumor activity in Phase 1a.
- Eligible tumor types include chronic lymphocytic leukemia (CLL), small lymphocytic lymphoma (SLL), diffuse large B-cell lymphoma (DLBCL), follicular lymphoma (FL), mantle cell lymphoma (MCL), marginal zone lymphoma (MZL), Waldenström macroglobulinemia (WM), including those with secondary central nervous system (CNS) involvement in any disease indication listed or primary CNS lymphoma (PCNSL).
- The primary objectives are to evaluate safety, determine the maximum tolerated dose and/or recommended Phase 1b dose (Phase 1a), and to evaluate the anti-tumor activity of NX-5948 in expansion cohorts (Phase 1b; Figure 3).

Figure 2. NX-5948: Study design

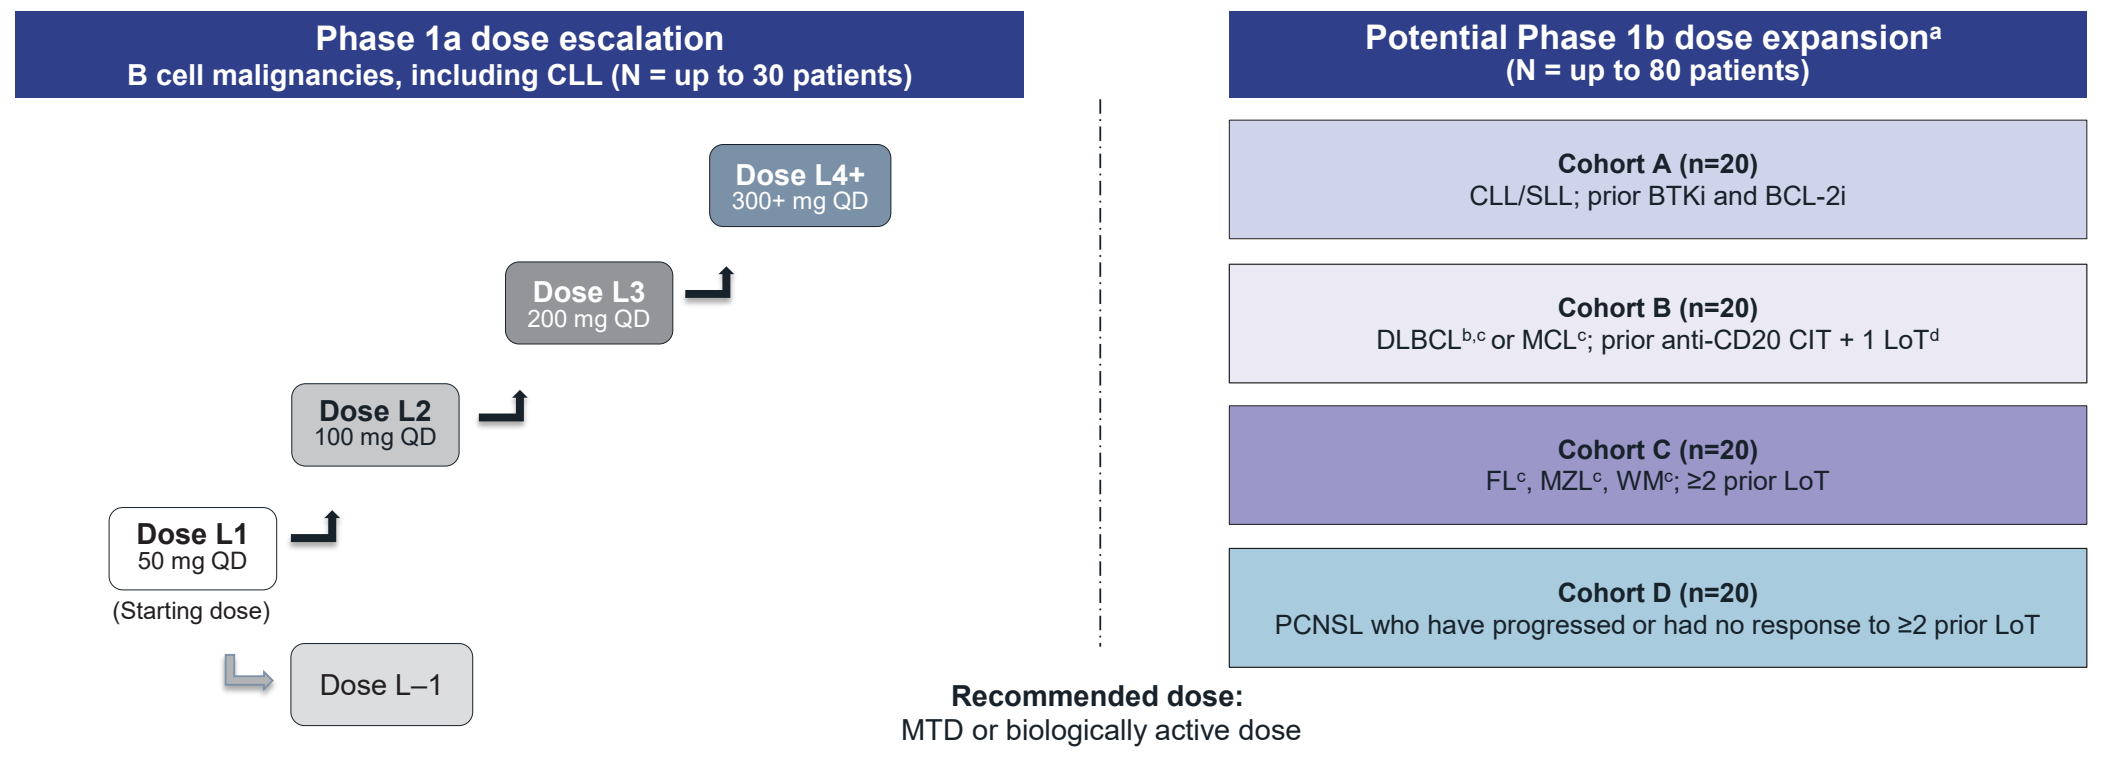
Phase 1a dose escalation
B cell malignancies, including CLL (N = up to 30 patients)
Dose L4+
300+ mg QD
Dose L3
200 mg QD
Dose L2
100 mg QD
Dose L1
50 mg QD
(Starting dose)
Dose L–1
Potential Phase 1b dose expansion[a]
(N = up to 80 patients)
Cohort A (n=20)
CLL/SLL; prior BTKi and BCL-2i
Cohort B (n=20)
DLBCL[b,c] or MCL[c]; prior anti-CD20 CIT + 1 LoT[d]
Cohort C (n=20)
FL[c], MZL[c], WM[c]; ≥2 prior LoT
Cohort D (n=20)
PCNSL who have progressed or had no response to ≥2 prior LoT
Recommended dose:
MTD or biologically active dose

[a]Potential dose-expansion cohorts are expected to open in the second half of 2023; [b]Subtypes include: transformed indolent lymphoma (e.g., grade 3b/transformed FL), Richter-transformed DLBCL, high-grade B-cell lymphoma with MYC and BCL-2 and/or BCL-6 rearrangements, high-grade B-cell lymphomas NOS; [c]Includes patients with secondary CNS involvement; [d]Additional lines of therapy include anthracycline for non-GCB DLBCL and BTKi for MCL; *Abbreviations:* BCL-2i, B-cell lymphoma-2 inhibitor; BTKi, Bruton's tyrosine kinase inhibitor; CIT, chemo-immunotherapy; CLL, chronic lymphocytic leukemia; CNS, central nervous system; DLBCL, diffuse large B-cell lymphoma; EMZL, extranodal marginal zone lymphoma; FL, follicular lymphoma; GCB, germinal center B cell; L, level; MCL, mantle cell lymphoma; LoT, line of therapy; MALT, mucosa-associated lymphoid tissue; MTD, maximum tolerated dose; MZL, marginal zone lymphoma; NMZL, nodal marginal zone lymphoma; NOS, not otherwise specified; PCNSL, primary central nervous system lymphoma; SLL, small lymphocytic lymphoma; SMZL, splenic marginal zone lymphoma; WM, Waldenströms macroglobulinemia.

Figure 3. Key study objectives and endpoints

Primary objectives

1a
- Evaluate safety and tolerability
- Evaluate MTD and/or recommended Ph1b dose

1b
- Evaluate anti-tumor activity
- Evaluate safety and tolerability

Primary endpoints

- Incidence of DLTs, TEAEs, and deaths
- Change in baseline safety parameters

- ORR per disease specific response criteria (e.g., iwCLL)
- Incidence of TEAEs, deaths, and changes in safety parameters

Secondary objectives

1a
- Characterization of PK and PD
- Assess preliminary anti-tumor activity

1b
- Evaluate anti-tumor activity
- Characterization of PK and PD

Secondary endpoints

- Plasma PK parameters, changes in BTK levels in B cells
- ORR, CR, time to first response, DOR, PFS, TTNT

- CR, time to first response, DOR, PFS, TTNT
- Plasma and CNS (patients with CNS disease) PK parameters

Exploratory objectives

- OS, metabolism, BTK inhibition, PK/PD, mechanisms of response/resistance, impact on cognitive function

Exploratory endpoints

- OS, NX-5948 metabolites, biomarkers of the BTK signaling and resistance pathways and target engagement, MMSE

Abbreviations: BTK, Bruton's tyrosine kinase; CNS, central nervous system; CR, complete response; DLT, dose limiting toxicities; DOR, duration of response; MMSE, mini-mental state examination; MTD, max tolerated dose; ORR, overall response rate; OS, overall survival; PD, pharmacodynamics; PFS, progression free survival; Ph1b, phase 1b; PK, pharmacokinetics; TEAEs, treatment emergent adverse events; TTNT, time to next treatment

Results

- As of December 1, 2022, seven patients have been enrolled in Phase 1a and received NX-5948 at 50 mg (n=4) or 100 mg (n=3).

Table 1. Baseline demographics/disease characteristics

Characteristics	Overall population (N=7)
Median age, years (range)	59.0 (46.0–79.0)
Male, n (%)	4 (57.1)
Female, n (%)	3 (42.9)
White, n (%)	7 (100)
ECOG performance status, n (%)	
0	3 (42.9)
1	4 (57.1)
Primary diagnosis, n (%)	
DLBCL	2 (28.6)
MCL	2 (28.6)
MZL	2 (28.6)
FL	1 (14.2)
Median time since initial diagnosis, years (range)	7.6 (2.9–23.5)
Median number of lines of prior therapy (range)	4 (3–10)
Previous targeted treatments, n (%)	
BTK inhibitors	3 (42.9)
CAR-T	3 (42.9)
Bispecific antibody	3 (42.9)

Data cutoff: December 1, 2022

Figure 4. NX-5948 pharmacokinetic profiles

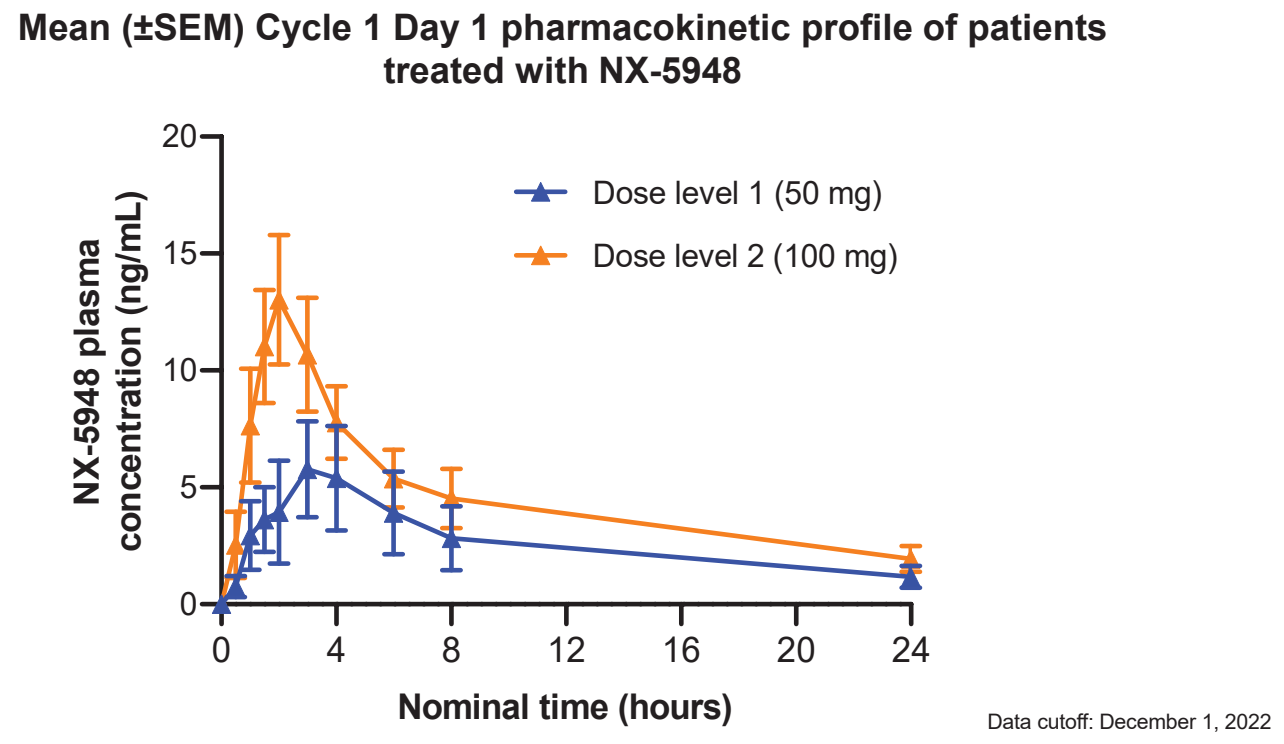
Mean (±SEM) Cycle 1 Day 1 pharmacokinetic profile of patients treated with NX-5948
NX-5948 plasma concentration (ng/mL)
20
15
10
5
0
0
4
8
12
16
20
24
Nominal time (hours)
Dose level 1 (50 mg)
Dose level 2 (100 mg)

Data cutoff: December 1, 2022

- Preliminary data suggest that NX-5948 exhibits dose-proportional pharmacokinetics (Figure 4; Table 2):
 - The half life of ~12.6 hours supports daily dosing.
 - The T_{max} of 2–3 hours suggests fast absorption.
 - Exposures (both AUC and C_{max}) increase with dose.
- NX-5948 resulted in rapid, robust and sustained BTK degradation in all patients dosed, regardless of their absolute BTK starting level, tumor type, or dose level of NX-5948 (Figure 5).

Table 2. NX-5948 pharmacokinetics

Dose	Cycle 1, Day 1 C_{max} (ng/mL)	$AUC_{0\text{–}last}$ (h x ng/mL)	T_{max} (hours)	$t_{1/2}$* (hours)
50 mg n=4	4.52 (102)	42.1 (152)	3.0	12.8 (19.9)
100 mg n=3	12.3 (45.3)	99.6 (50.2)	2.0	12.4 (9.39)

C_{max} and $AUC_{0\text{-last}}$ are presented as geometric mean (geometric %CV); T_{max} is presented as median; $t_{1/2}$ is presented as mean (%CV); *AUC extrapolation >20%

Data cutoff: December 1, 2022

Figure 5. NX-5948 pharmacodynamics

- NX-5948 induced sustained BTK degradation of 89±4% at Cycle 2 Day 1 across dose levels

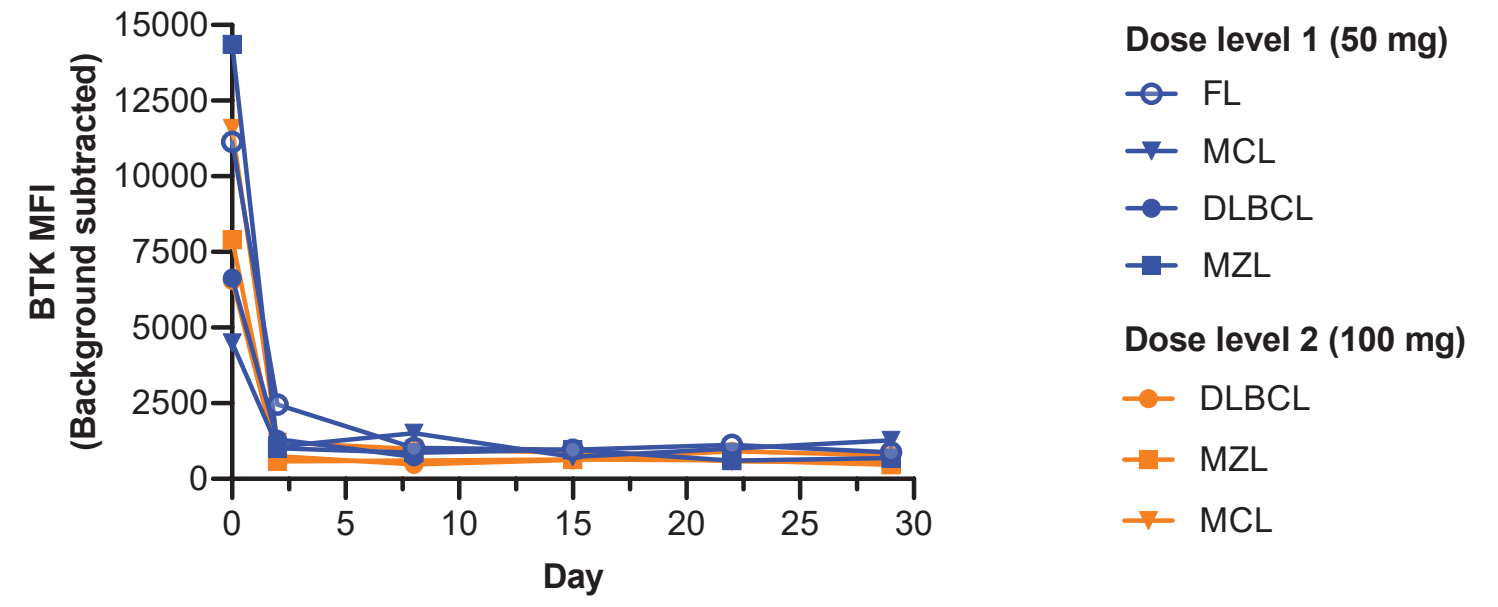
BTK MFI (Background subtracted)
15000
12500
10000
7500
5000
2500
0
0
5
10
15
20
25
30
Day
Dose level 1 (50 mg)
FL
MCL
DLBCL
MZL
Dose level 2 (100 mg)
DLBCL
MZL
MCL

FL (follicular lymphoma), DLBCL (diffuse large B cell lymphoma), MCL (mantle cell lymphoma), MZL (marginal zone lymphoma)

Data cutoff: December 1, 2022

Summary/conclusion

- Preliminary findings suggest that NX-5948 exhibits dose-proportional pharmacokinetics.
- Exposure to NX-5948 results in rapid, robust and sustained BTK degradation, supporting daily dosing.
- Additional indications, including CLL, and additional dosing levels are currently being explored. Potential dose-expansion cohorts are expected to open in the second half of 2023.
- The study continues to enroll patients and further data will be shared at an upcoming meeting.

References/acknowledgements

1. Wang E. et al. N Engl J Med 2022;386;735–43.
2. Noviski M. et al. AACR 2023. Abstract presentation number 2850.
3. Robbins DW. et al. ASH 2021. Poster number 2251.
4. Hallek M, et al. Blood 2018;131:2745–60.
5. Owen RG, et al. Br J Haematol 2013;160:171–6.
6. Cheson BD, et al. J Clin Oncol 2014;32:3059–68.
7. Abrey LE, et al. J Clin Oncol 2005;23:5034–43.

- NX-5948-301 is sponsored by Nurix Therapeutics, Inc.
- Nurix Therapeutics, Inc. also funded the medical writing/layout support for this poster, which was provided by Miller Medical Communications.

nurix

Presented at the 17th ICML meeting, June 13–17, 2023, Lugano, Switzerland.

Study contact: nx5948301@nurixtx.com