BTK Degradation as a Novel Therapeutic Strategy in Relapsed CNS Lymphoma: Preclinical Proof of Concept Studies in Intracranial Patient-Derived Model

Jun Ma[2], Ming Lu[1], Lingjing Chen[1], Huimin Geng[1], Michael Randall[1], Aishwarya Kumar[2], Mark Noviski[2], Ryan Rountree[2], Cristiana Guiducci[2], James L. Rubenstein[1]

[1]Department of Medicine, University of California, San Francisco, San Francisco, CA. [2]Nurix Therapeutics, San Francisco, CA.

University of California San Francisco

INTRODUCTION

There is substantial need for more effective therapeutic strategies for relapsed, refractory primary and secondary CNS lymphomas. Bruton's tyrosine kinase (BTK) is a key driver of NF-kB activation and an important target in primary CNS lymphoma, largely dependent on B-cell receptor (BCR) signaling. Limitations of current covalent and non-covalent BTK inhibitors include the susceptibility to mutational escape as a basis for resistance.

Recent evidence also suggests a potential scaffolding function of BTK, active in the setting of catalytically inactive BTK kinase mutations and not antagonized by kinase inhibitors, that permits downstream growth pathway activation.

BTK degradation represents a unique therapeutic strategy. NX-5948 is a novel oral bifunctional degrader that induces BTK degradation via recruitment of the cereblon E3 ligase complex. NX-5948 is being evaluated in a Phase 1 trial as treatment for patients with B cell malignancies who have progressed despite prior treatment with covalent and/or non-covalent BTK inhibitors or in B cell indications where treatment with BTK inhibitors has been less effective. NX-5948 induces sub-nanomolar potency degradation of both wild-type and mutant forms of BTK *in vitro*, demonstrating rapid *in vivo* degradation in mouse and non-human primate B cells within four hours of oral administration. In mice, NX-5948 inhibits growth of subcutaneously implanted ABC-type DLBCL, TMD8 tumors, harboring BTK wild type or ibrutinib resistant C481S mutation. In addition, NX-5948 crosses an intact blood-brain barrier in mice and promotes BTK degradation in microglia and brain-resident lymphoma cells.

Here we evaluated the pharmacodynamic properties of NX-5948 in an intracranial model of CNS lymphoma using patient-derived SC1 cells. SC1 cells are derived from a patient with highly refractory CD79b and EVT6-mutant large B-cell secondary CNS lymphoma, resistant to R-CHOP, high-dose methotrexate/rituximab, etoposide, Ara-C and irradiation. Upon intracranial implantation, SC1 cells grow aggressively and exhibit diffuse bi-hemispheric brain infiltration in RAG-/- mice, with localization along brain microvasculature as demonstrated by fluorescein labeled *lysopersicon esclutentum* staining. Oral administration of NX-5948 for three days at 90 mg/kg in mice bearing established intracranial SC1 tumors yielded 98% degradation of BTK in SC1 lymphoma cells isolated six hours after NX-5948 dosing, as quantified by immunoblot analysis. Daily treatment with NX-5948 was associated with marked prolongation of survival in mice with intracranial SC1 CNS lymphoma compared to control, p= 5.6×10^{-5} and to mice treated with daily ibrutinib p= 8.6×10^{-5} (N=6 mice/cohort). Survival prolongation was further evident following cessation of dosing at 100 days. Taken together, these preclinical results support, in part, the rationale for a phase I study of NX-5948 in relapsed primary CNS lymphoma (NCT05131022).

MECHANISM OF ACTION

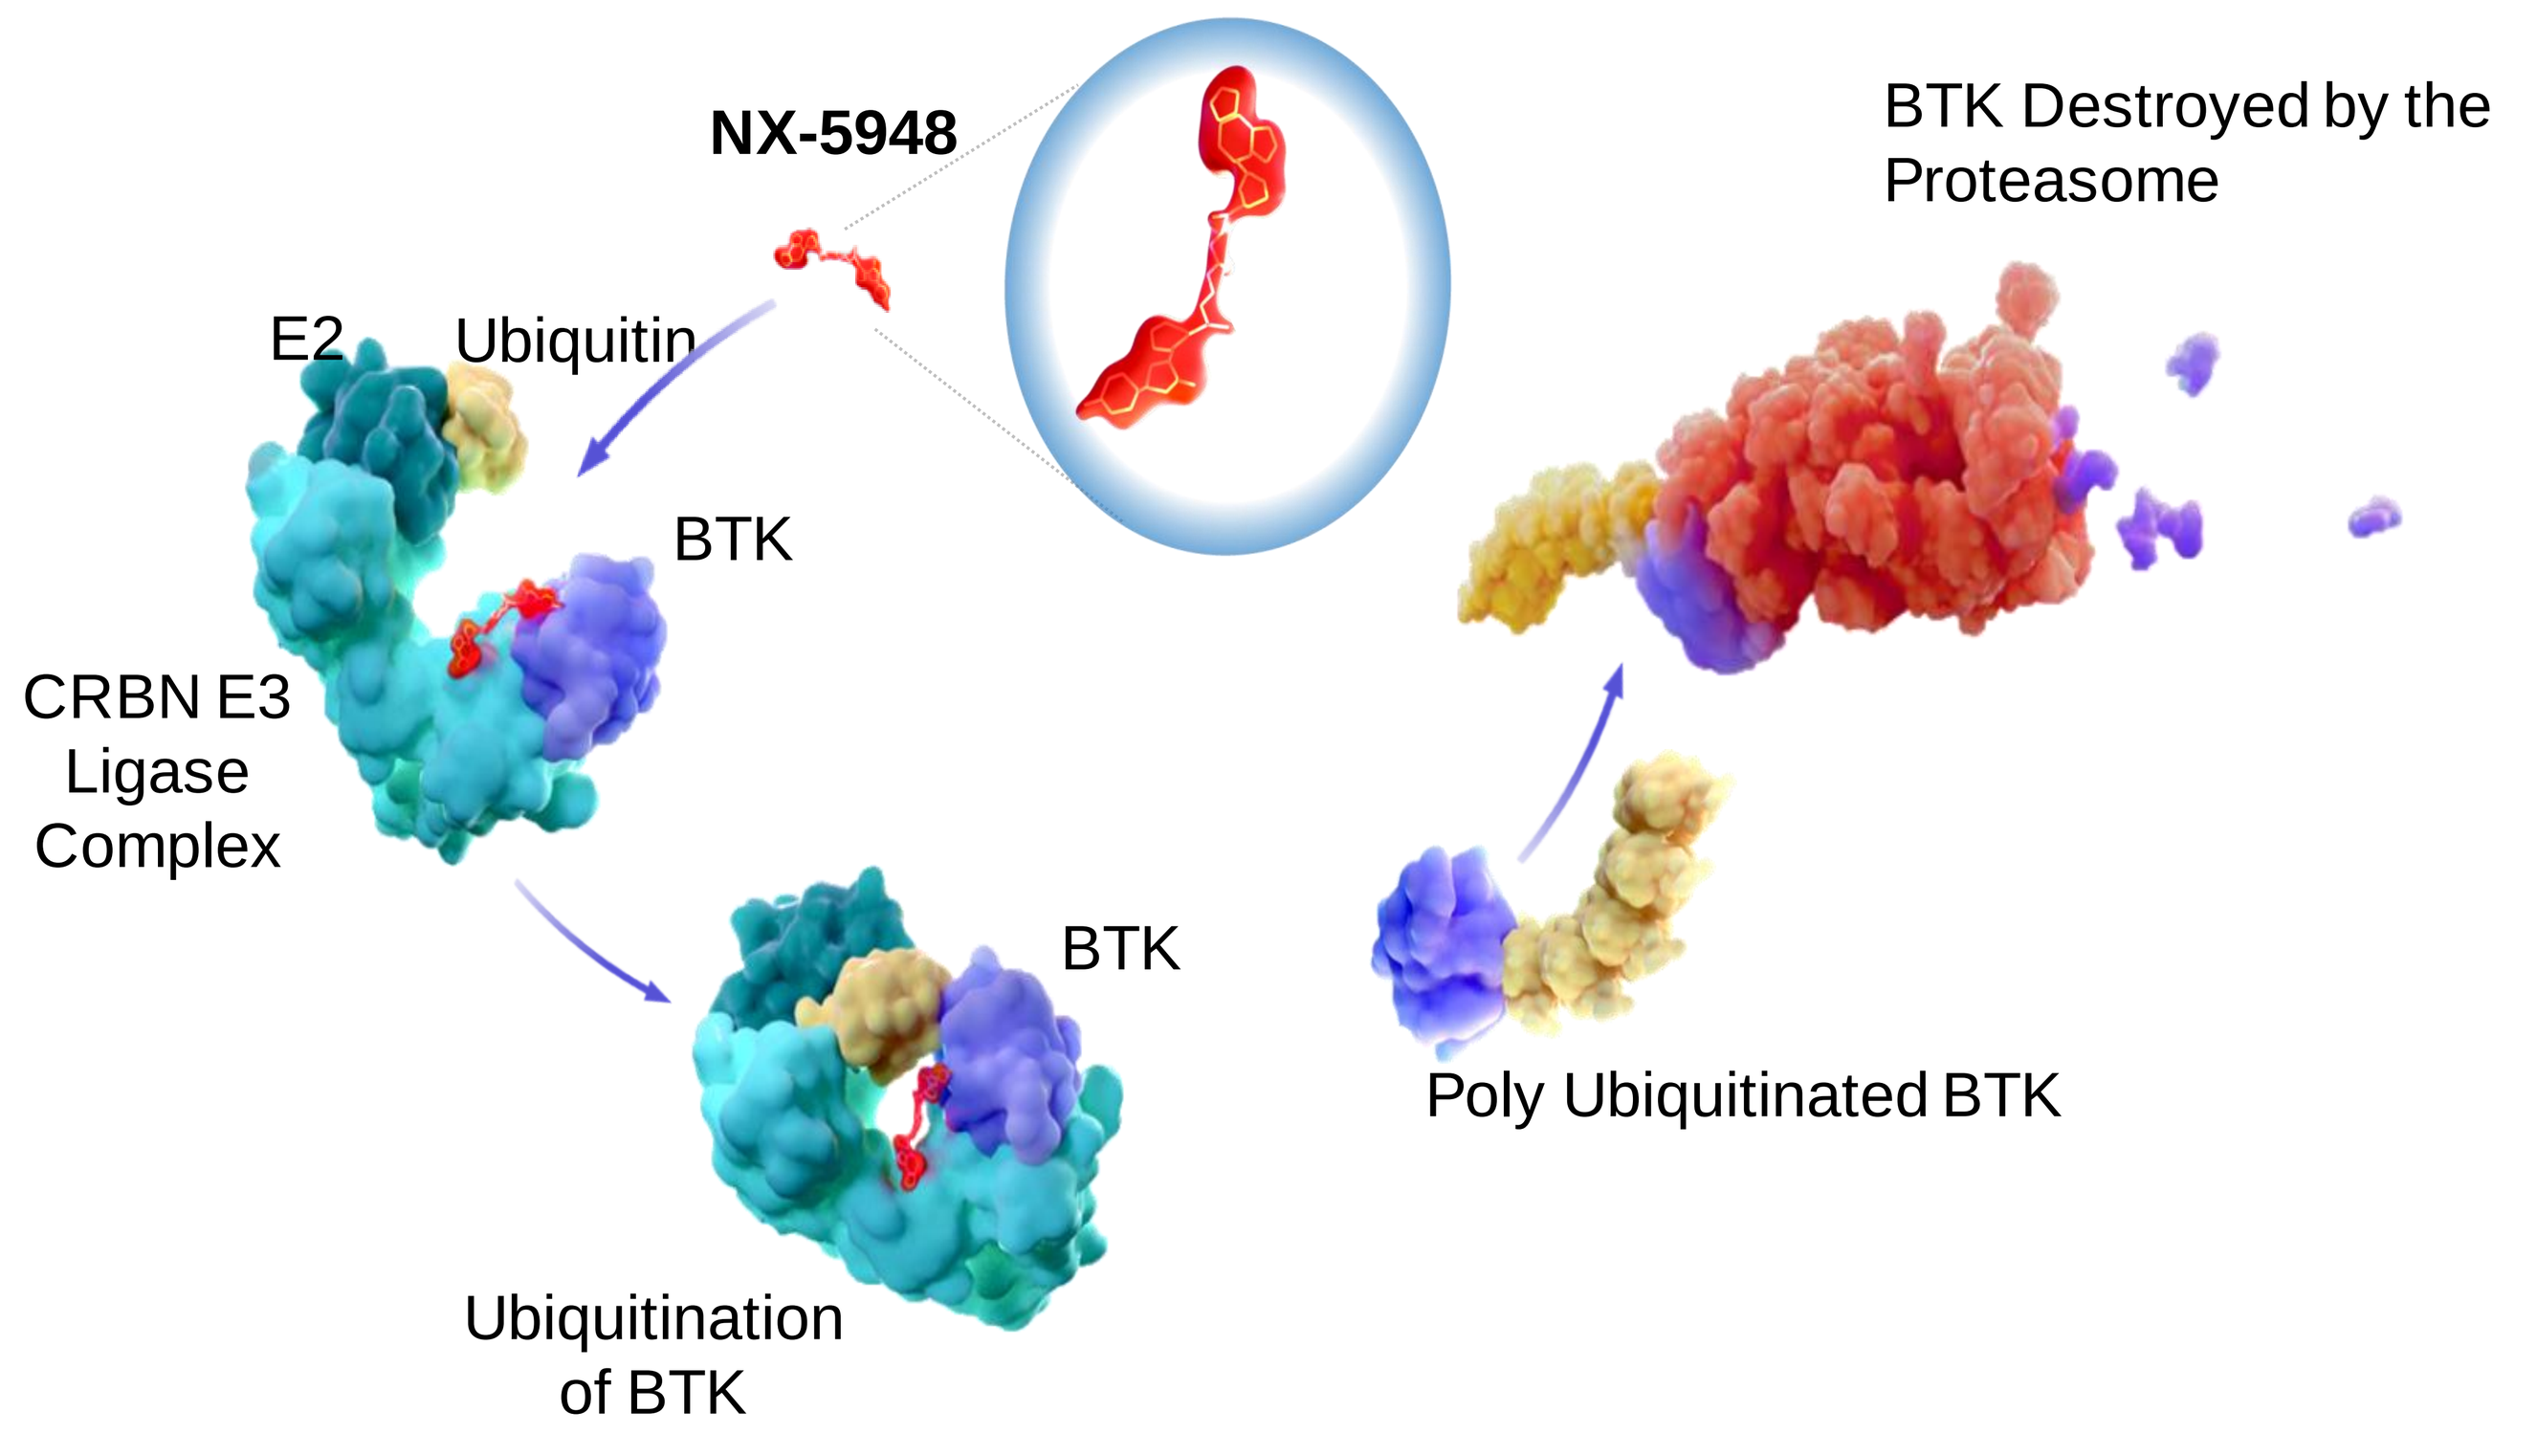

NX-5948
E2
Ubiquitin
BTK
CRBN E3 Ligase Complex
BTK
Ubiquitination of BTK
BTK Destroyed by the Proteasome
Poly Ubiquitinated BTK

- NX-5948 is an orally bioavailable bifunctional molecule that induces BTK degradation via recruitment of the cereblon E3 ligase complex, which ubiquitylates BTK and targets BTK for proteasomal degradation

RESULTS

NX-5948 is a Potent and Rapid BTK Degrader

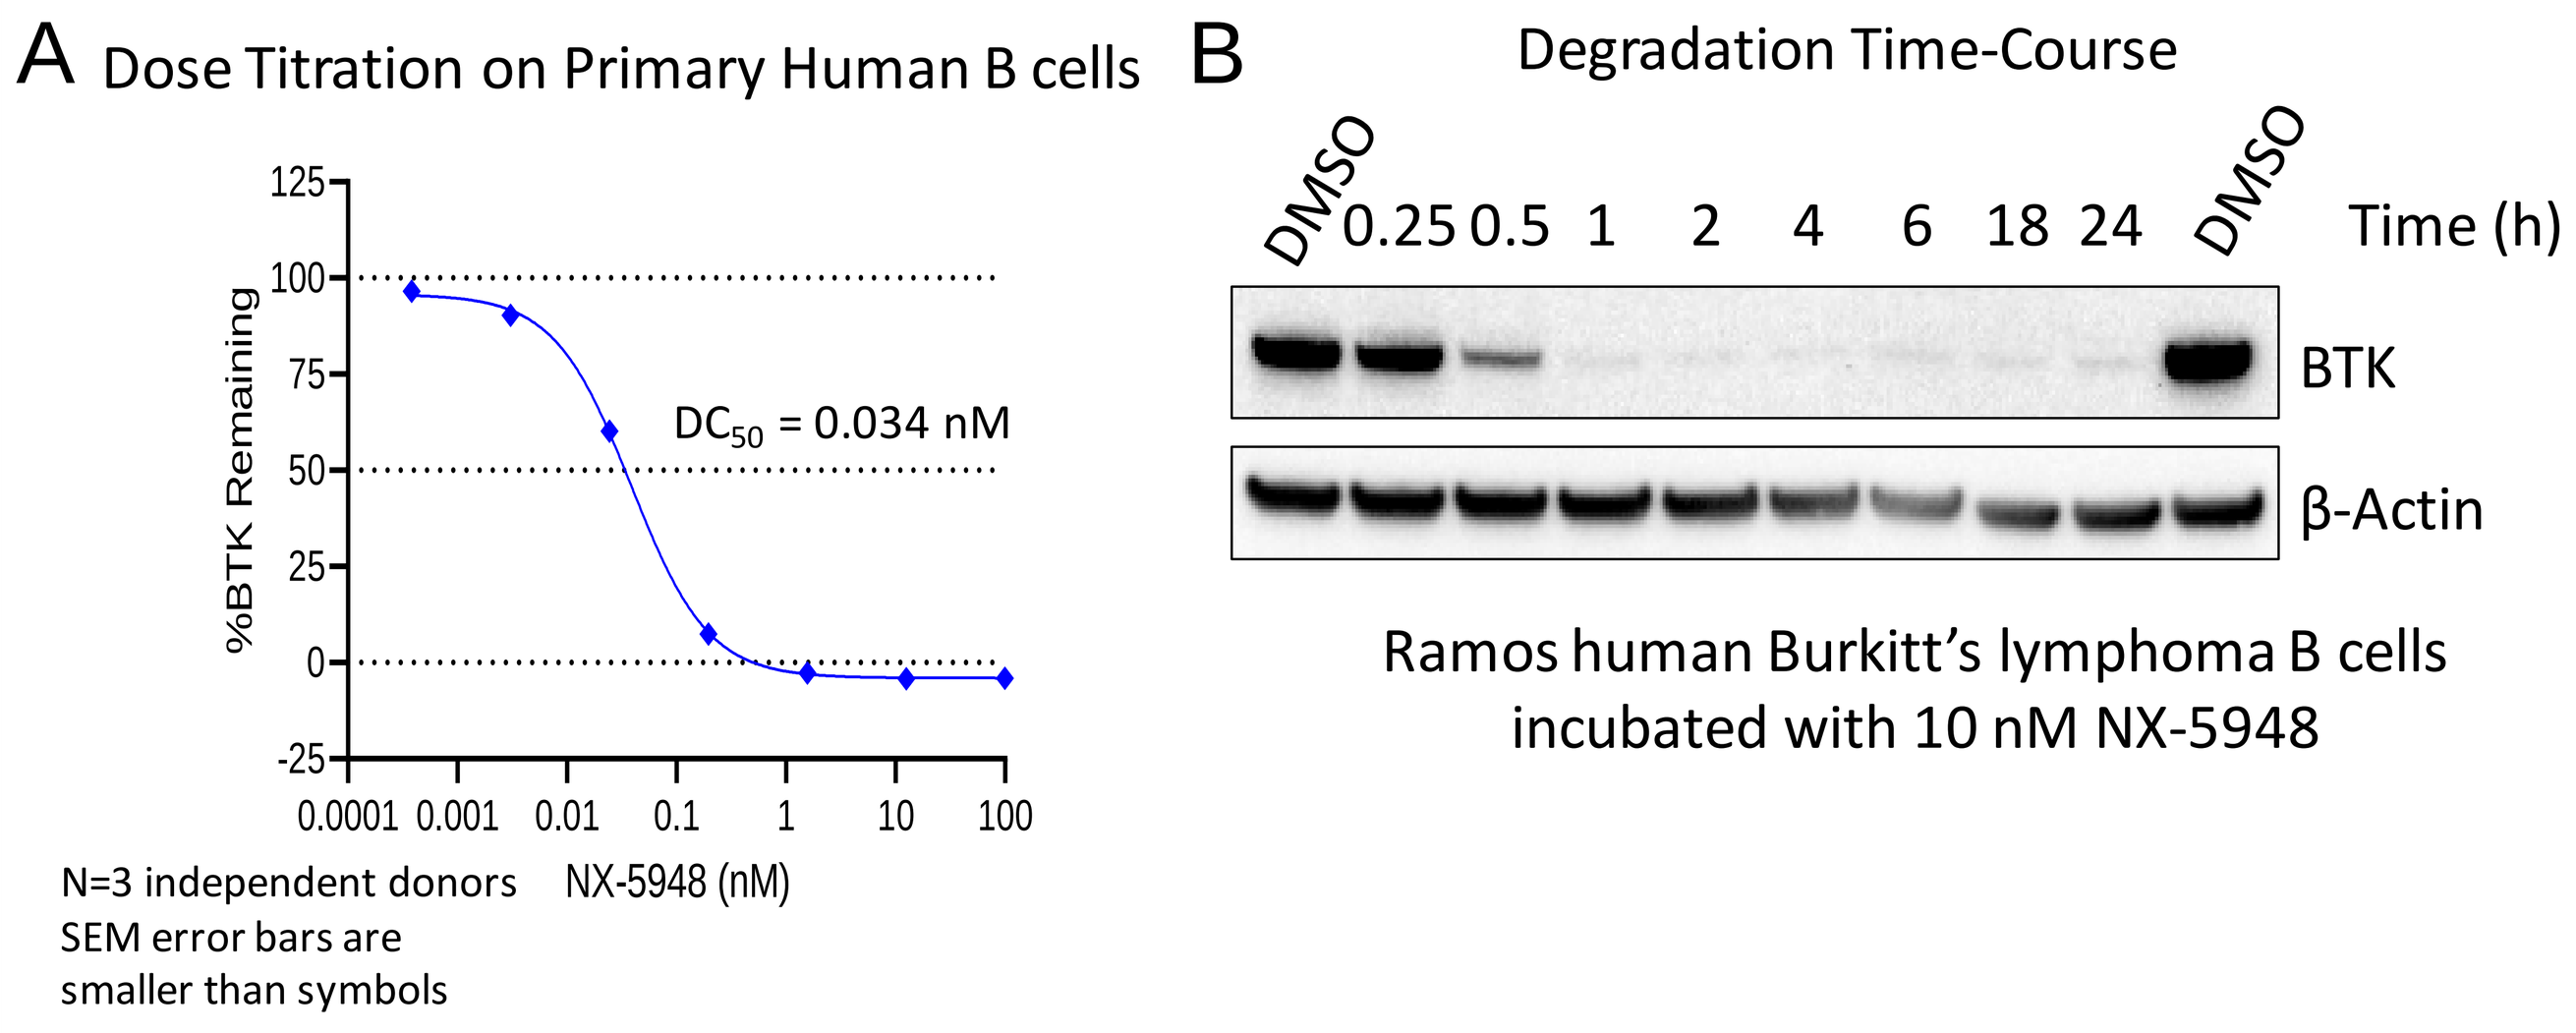

A Dose Titration on Primary Human B cells
%BTK Remaining
DC50 = 0.034 nM
NX-5948 (nM)
N=3 independent donors
SEM error bars are smaller than symbols
B Degradation Time-Course
DMSO 0.25 0.5 1 2 4 6 18 24 DMSO Time (h)
BTK
β-Actin
Ramos human Burkitt's lymphoma B cells incubated with 10 nM NX-5948

Figure 1. NX-5948 degraded BTK with subnanomolar potency (A) and resulted in near-complete BTK degradation after 1 hour (B). (A) Flow cytometric analysis of intracellular BTK levels in primary human B cells incubated with a dose titration of NX-5948 (DC_{50} is the NX-5948 concentration leading to 50% BTK degradation). (B) Western blot analysis of BTK in the Ramos Burkitt's lymphoma cell line following a time-course of incubation with 10 nM NX-5948.

Daily Oral Administration of NX-5948 Drives Dose-Dependent BTK Degradation and Tumor Growth Inhibition in Mice with Subcutaneous TMD8 DLBCL Tumors

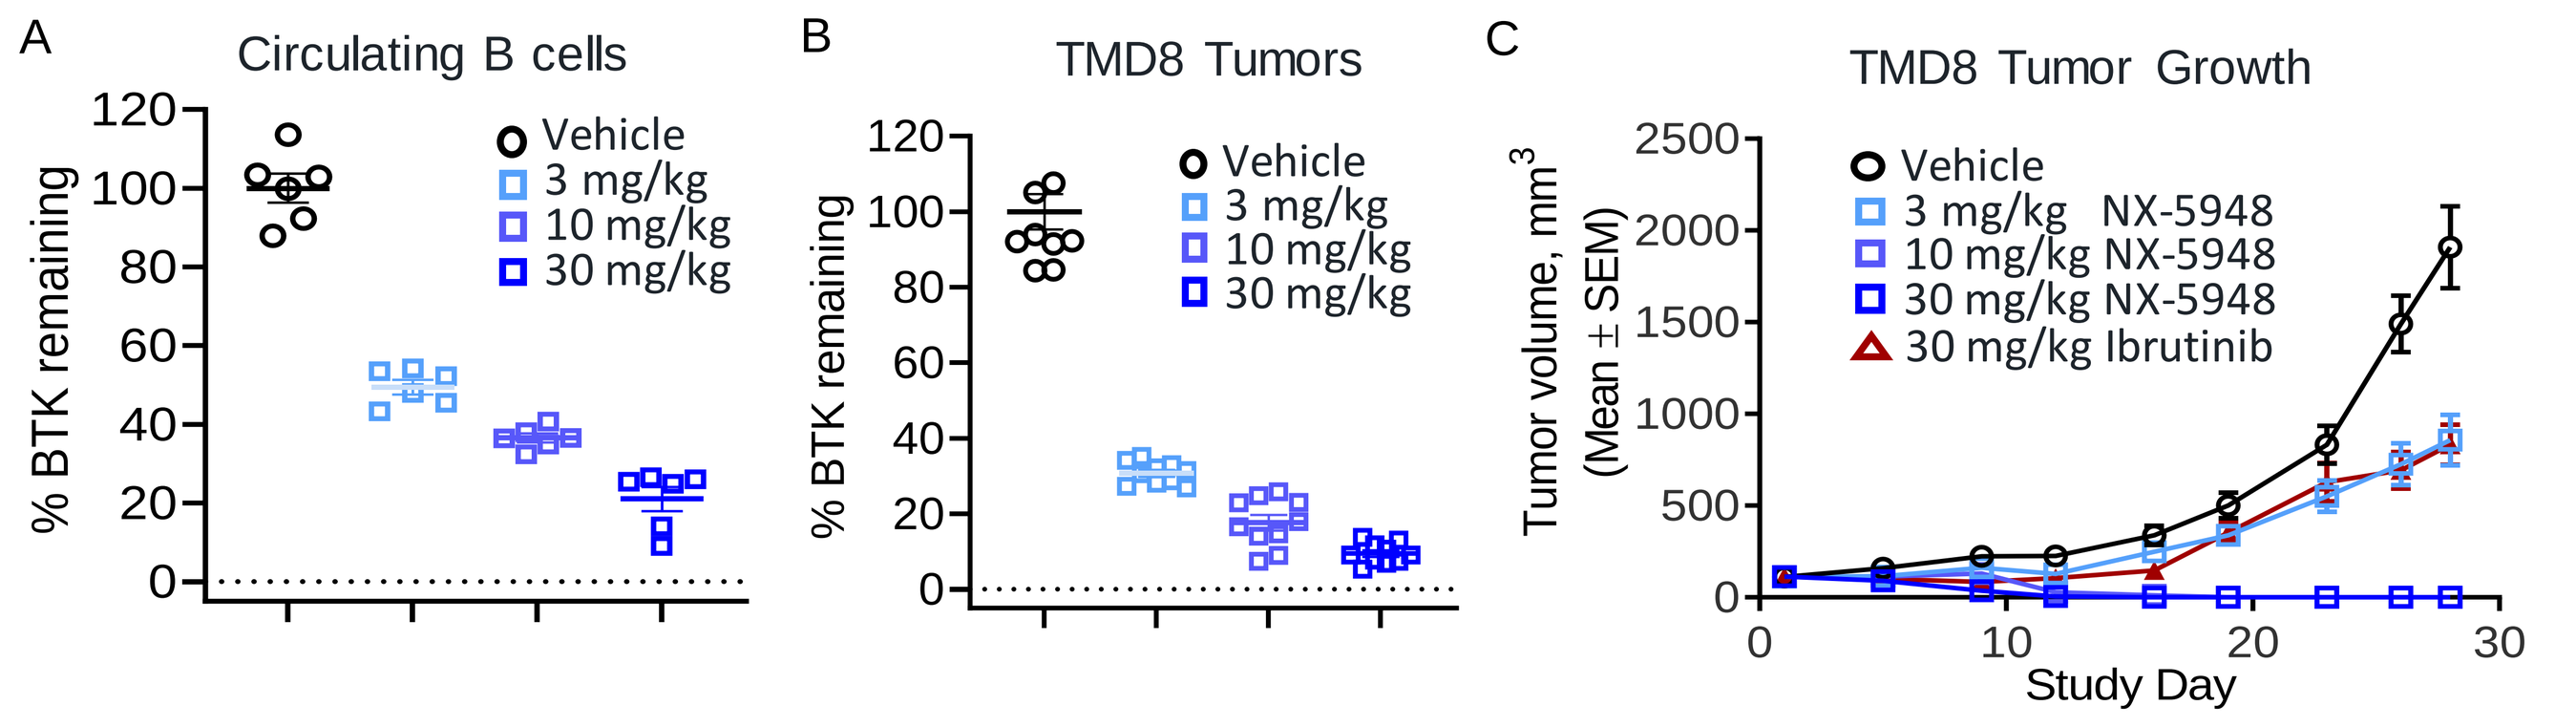

A Circulating B cells
% BTK remaining
Vehicle
3 mg/kg
10 mg/kg
30 mg/kg
B TMD8 Tumors
% BTK remaining
C TMD8 Tumor Growth
Tumor volume, mm3 (Mean ± SEM)
Vehicle
3 mg/kg NX-5948
10 mg/kg NX-5948
30 mg/kg NX-5948
30 mg/kg Ibrutinib
Study Day

Treatment	Oral gavage dose (mg/kg)	% BTK degradation in circulating B cells	% BTK degradation in TMD8 tumor tissue	% TGI vs Vehicle (Day 26)	*P* value vs Vehicle
Vehicle	0	0.0±3.7	0.0±4.7	N/A	N/A
NX-5948	3	50.5±1.9	69.2±0.9	54%	0.0025
	10	63.5±1.1	82.4±2.1	100%	<0.0001
	30	79.0±3.1	90.5±0.5	100%	<0.0001
Ibrutinib	30	N/A	N/A	57%	0.0015

N/A: Not applicable; TGI: tumor growth inhibition.
P values determined on tumor volume by mixed-effect analysis with Dunnett's multiple comparisons test

Figure 2. Daily, oral NX-5948 administration resulted in dose-dependent BTK degradation (A-B) and superior anti-tumor activity compared to ibrutinib (C). (A) Wild-type CD1 mice were administered either vehicle control or NX-5948 at 3, 10, or 30 mg/kg for 5 days, and BTK levels were determined by flow cytometry in circulating B cells from tail bleeds collected 24 hours after the fifth dose. (B-C) 1 x 10E7 TMD8 ABC-type DLBCL cells were implanted subcutaneously into the flank of CB.17 SCID mice, and mice were treated with daily oral administration of either vehicle, NX-5948, or ibrutinib starting when average tumor volume was approximately 100 mm^3. (B) 24 hours after five days of daily dosing, BTK levels were determined in TMD8 tumor tissue by western blot analysis. (C) NX-5948 treatment resulted in dose-dependent anti-tumor activity with 100% tumor growth inhibition (TGI) when administered daily at 10 or 30 mg/kg. In contrast, daily oral administration of ibrutinib at 30 mg/kg resulted in 57% TGI.

Daily Oral Administration of NX-5948 to Mice with Intracranial TMD8 DLBCL Tumors Degrades BTK in Brain-Resident Cells and Prolongs Survival

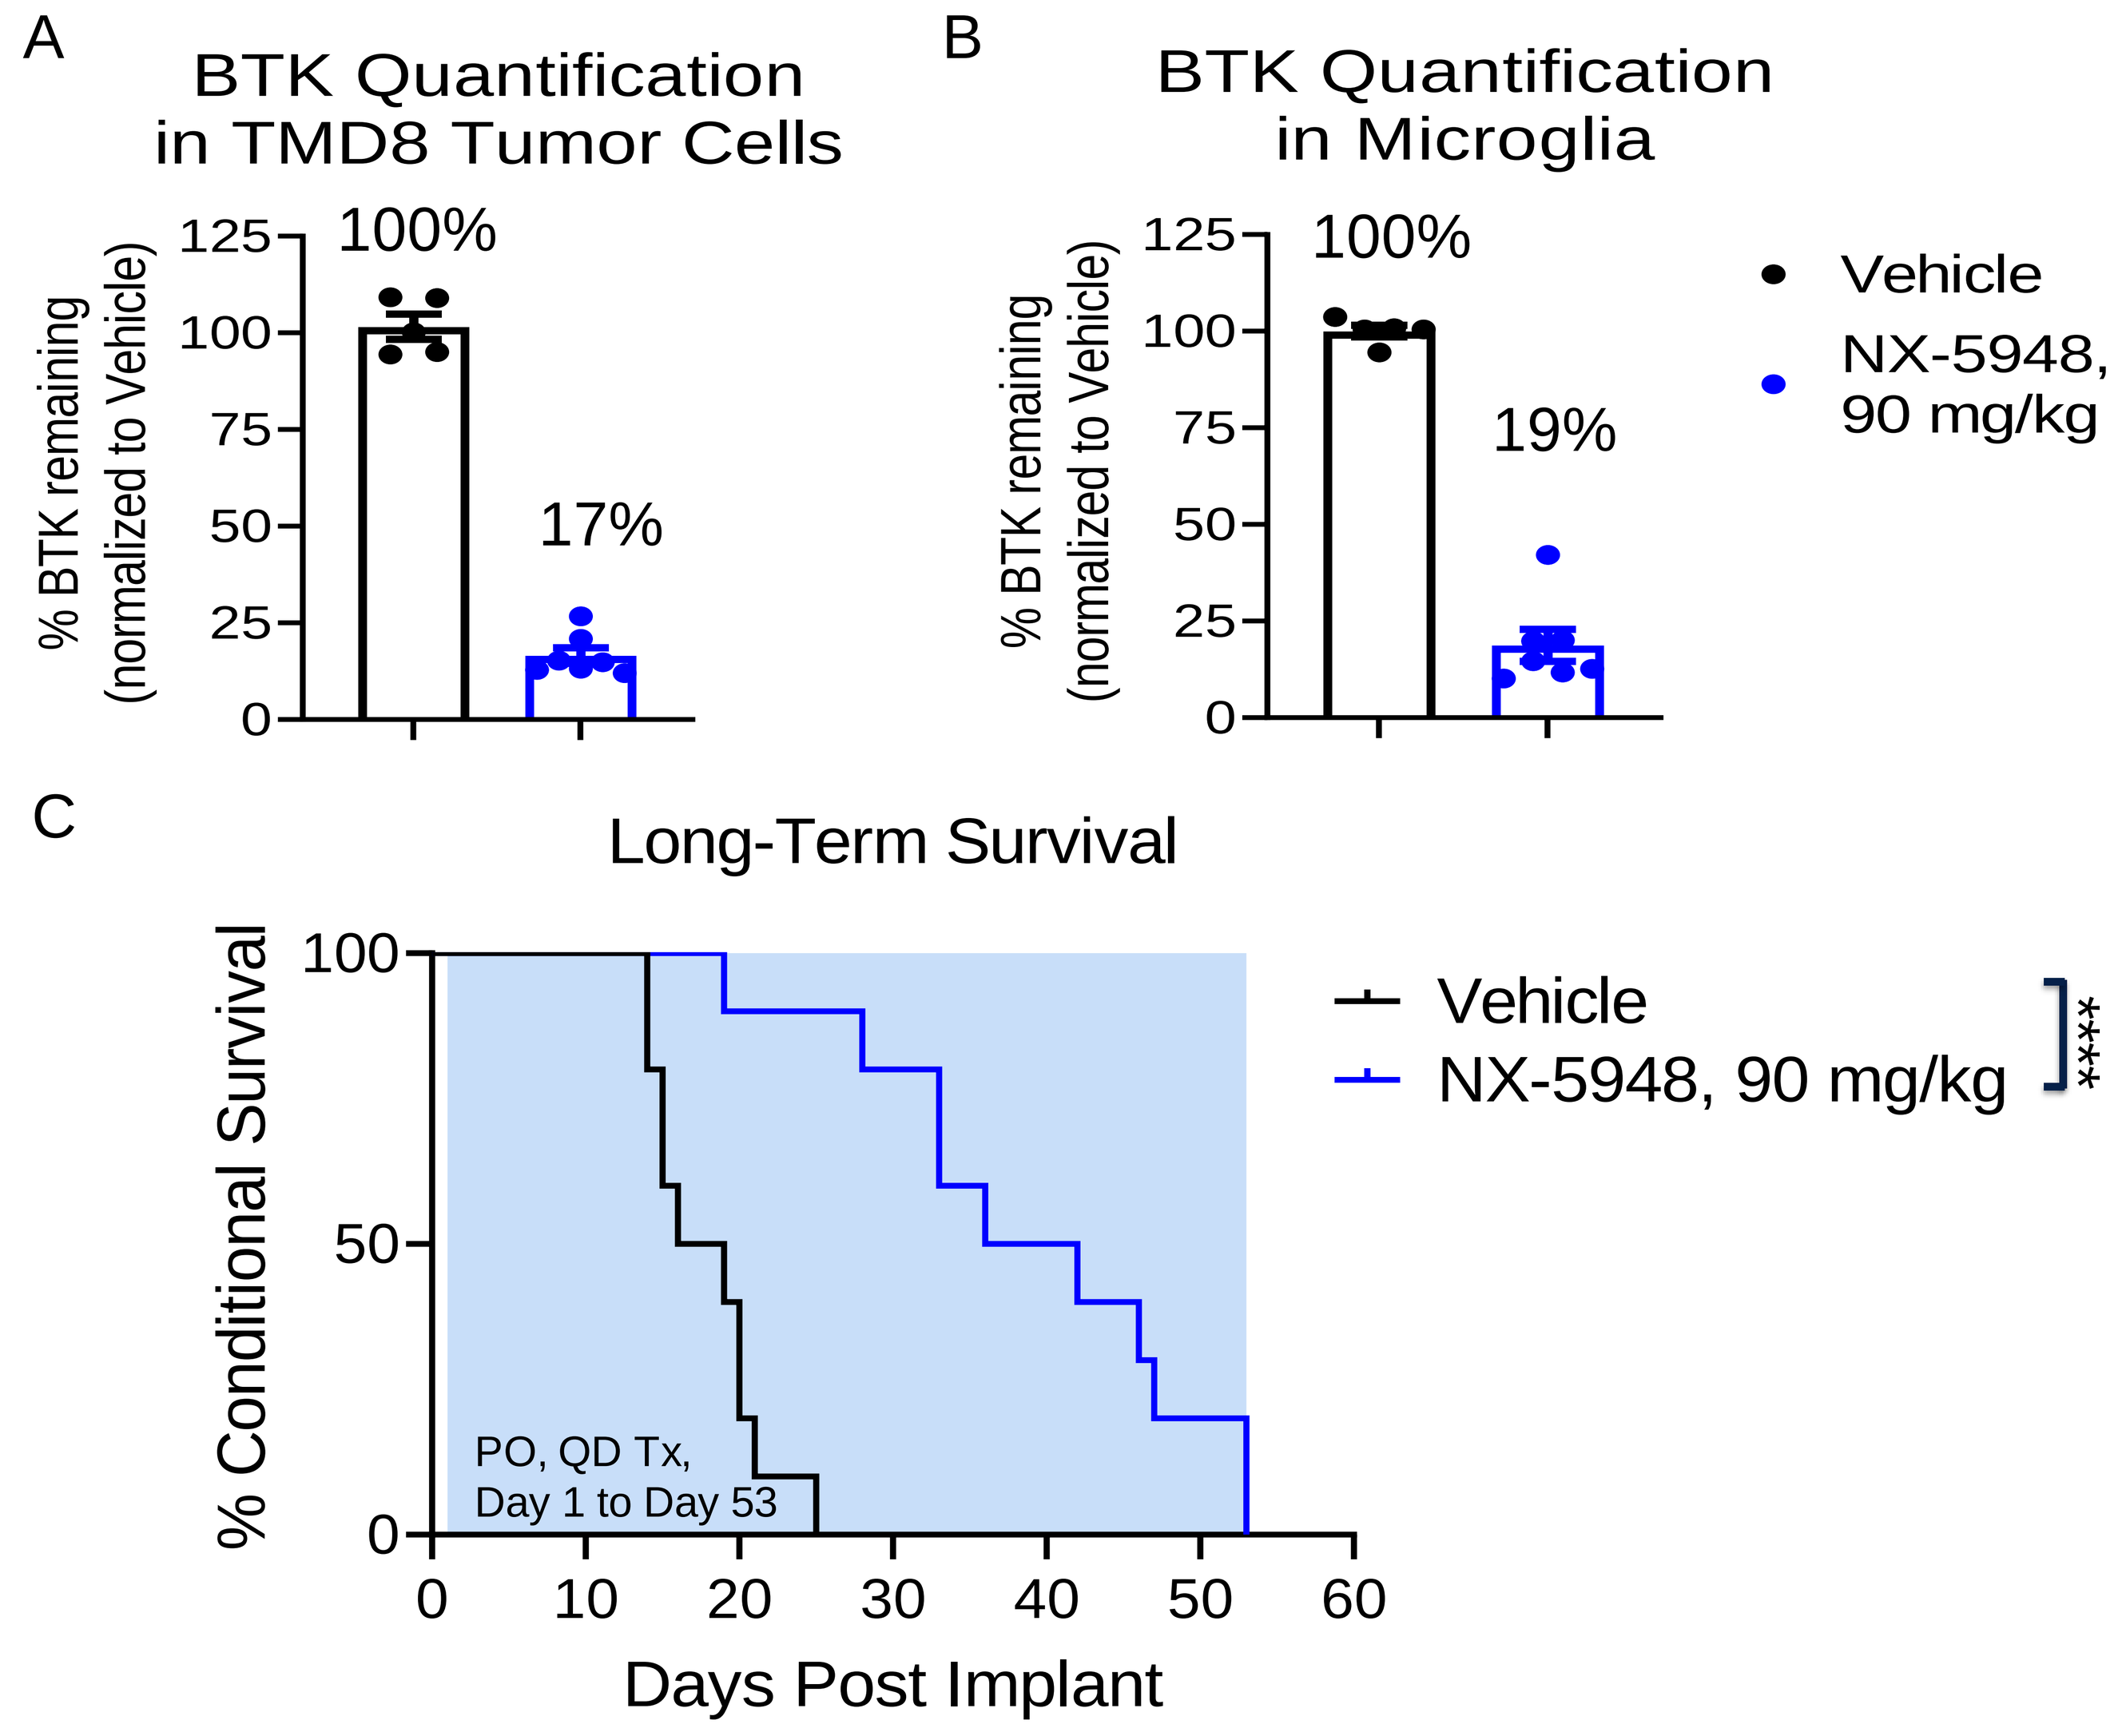

A BTK Quantification in TMD8 Tumor Cells
% BTK remaining (normalized to Vehicle)
100%
17%
B BTK Quantification in Microglia
100%
19%
Vehicle
NX-5948, 90 mg/kg
C Long-Term Survival
% Conditional Survival
PO, QD Tx, Day 1 to Day 53
Days Post Implant
Vehicle
NX-5948, 90 mg/kg

Figure 3. 5 x 10E5 TMD8 ABC-type DLBCL cells were implanted by intracranial injection on Day 0, and NX-5948 or vehicle control was orally administered daily at 90 mg/kg beginning on Day 1. (A) 24 h after the 11th daily dose, brain tissue was harvested from a subset of mice, single-cell suspensions were prepared, and BTK levels were determined by intracellular flow cytometry in (A) TMD8 tumor cells or (B) microglia. Daily oral NX-5948 treatment robustly degraded BTK in both cell types. (C) NX-5948 treatment significantly prolonged median overall survival to 39 days compared to 17.5 days in the vehicle control group (**** p<0.0001, Log-rank test).

The SC1 Murine DLBCL PDX Model Retains Characteristics of CNS Lymphoma

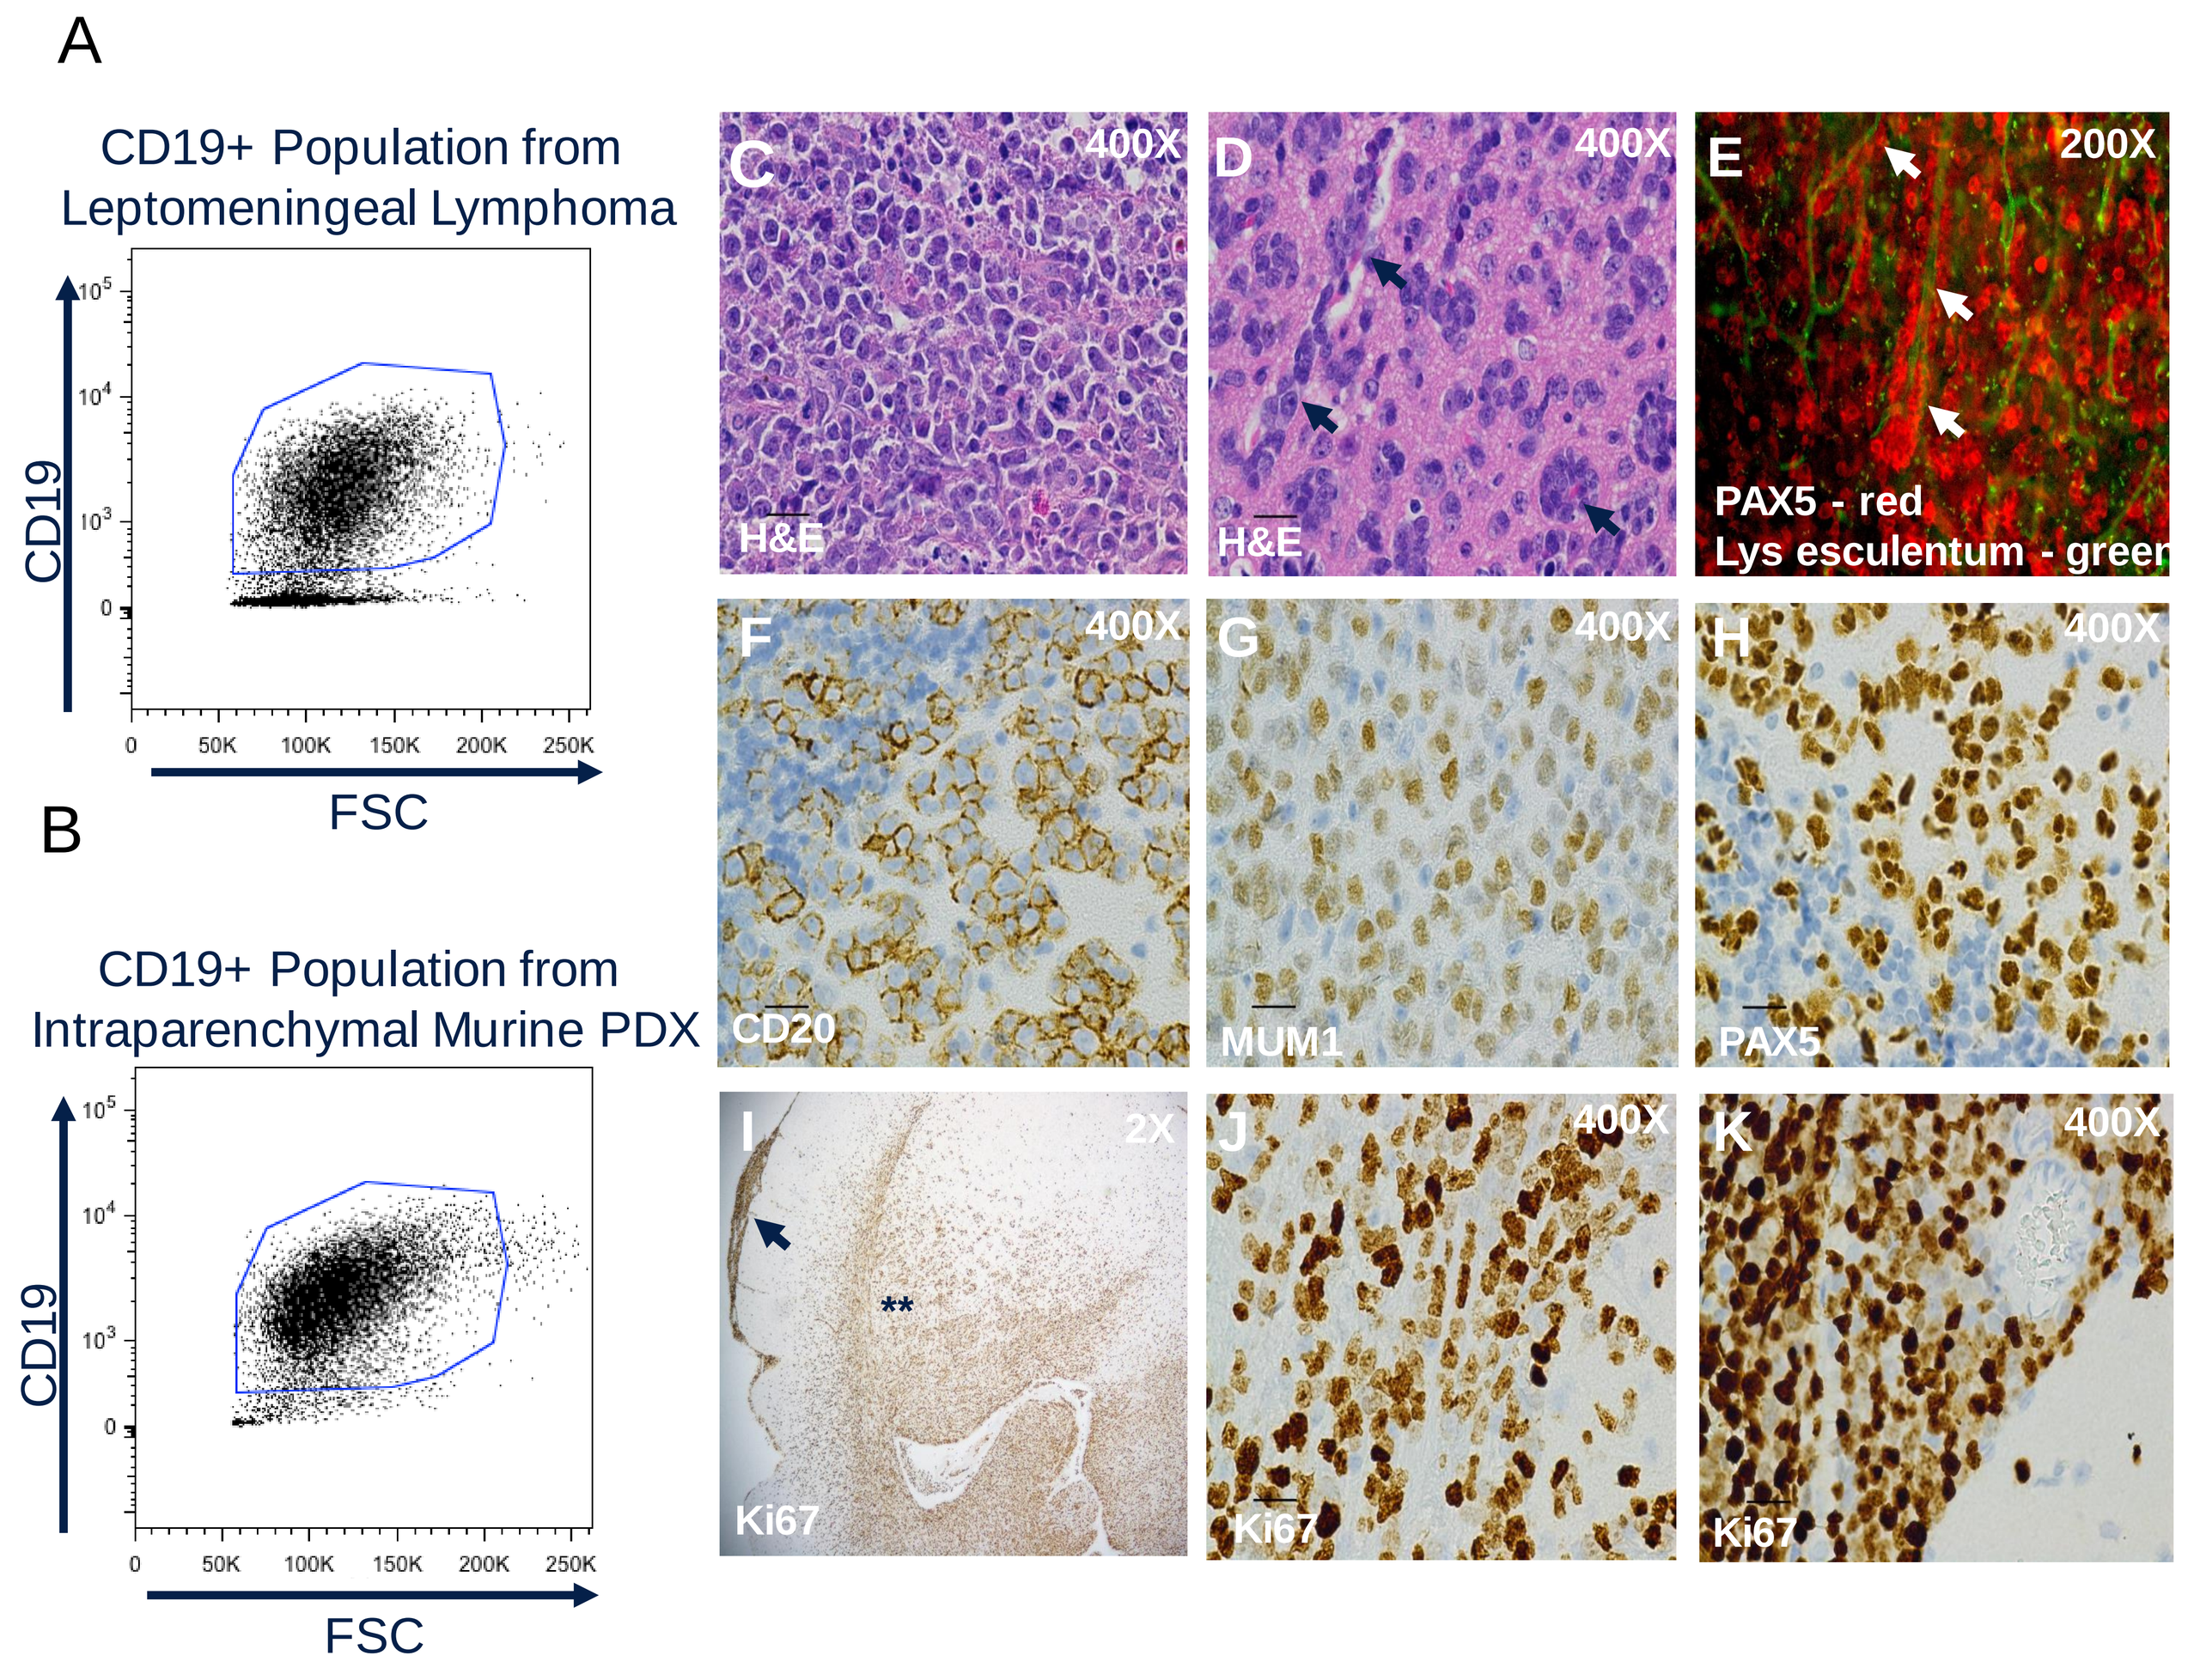

A CD19+ Population from Leptomeningeal Lymphoma
CD19
FSC
B CD19+ Population from Intraparenchymal Murine PDX
CD19
FSC
C 400X H&E
D 400X H&E
E 200X PAX5 - red Lys esculentum - green
F 400X CD20
G 400X MUM1
H 400X PAX5
I 2X Ki67 **
J 400X Ki67
K 400X Ki67

Figure 4. (A) CD19+ secondary leptomeningeal lymphoma cells isolated from cerebrospinal fluid (CSF) of a patient diagnosed with CD79b mutant DLBCL involving skin. (B) Intraparenchymal cells isolated from the corresponding SC1 murine PDX model share similar characteristics as measured by CD19 levels and FSC using flow cytometry. (C) Photomicrograph characterizing DLBCL diagnostic specimen of original skin biopsy. (D-H) Photomicrographs show PDX SC1 cells invading brain parenchyma in RAG -/- mice and evidence of vascularization (arrows in D and E). C) Immunofluorescence analysis of PDX model with SC1 cells positive for PAX5 (red) and tumor vasculature labeled with lysoperisicum esculentum (green). (F) CD20, (G) MUM1, and (H) PAX5 staining of PDX SC1 cells invading brain parenchyma. (I) Ki67 staining of a cross section of mouse brain with implanted PDX SC1 cells. SC1 cells spread to meninges (arrow in I, K) and across white matter in a periventricular distribution (** in I and J). (J) 400X magnification of brain parenchyma (** in I). (K) 400X magnification of meninges (arrow in I).

Daily Oral Administration of NX-5948 to Mice Implanted with SC1 DLBCL PDX Cells in Parenchyma Drives potent BTK Degradation and Prolongs Survival

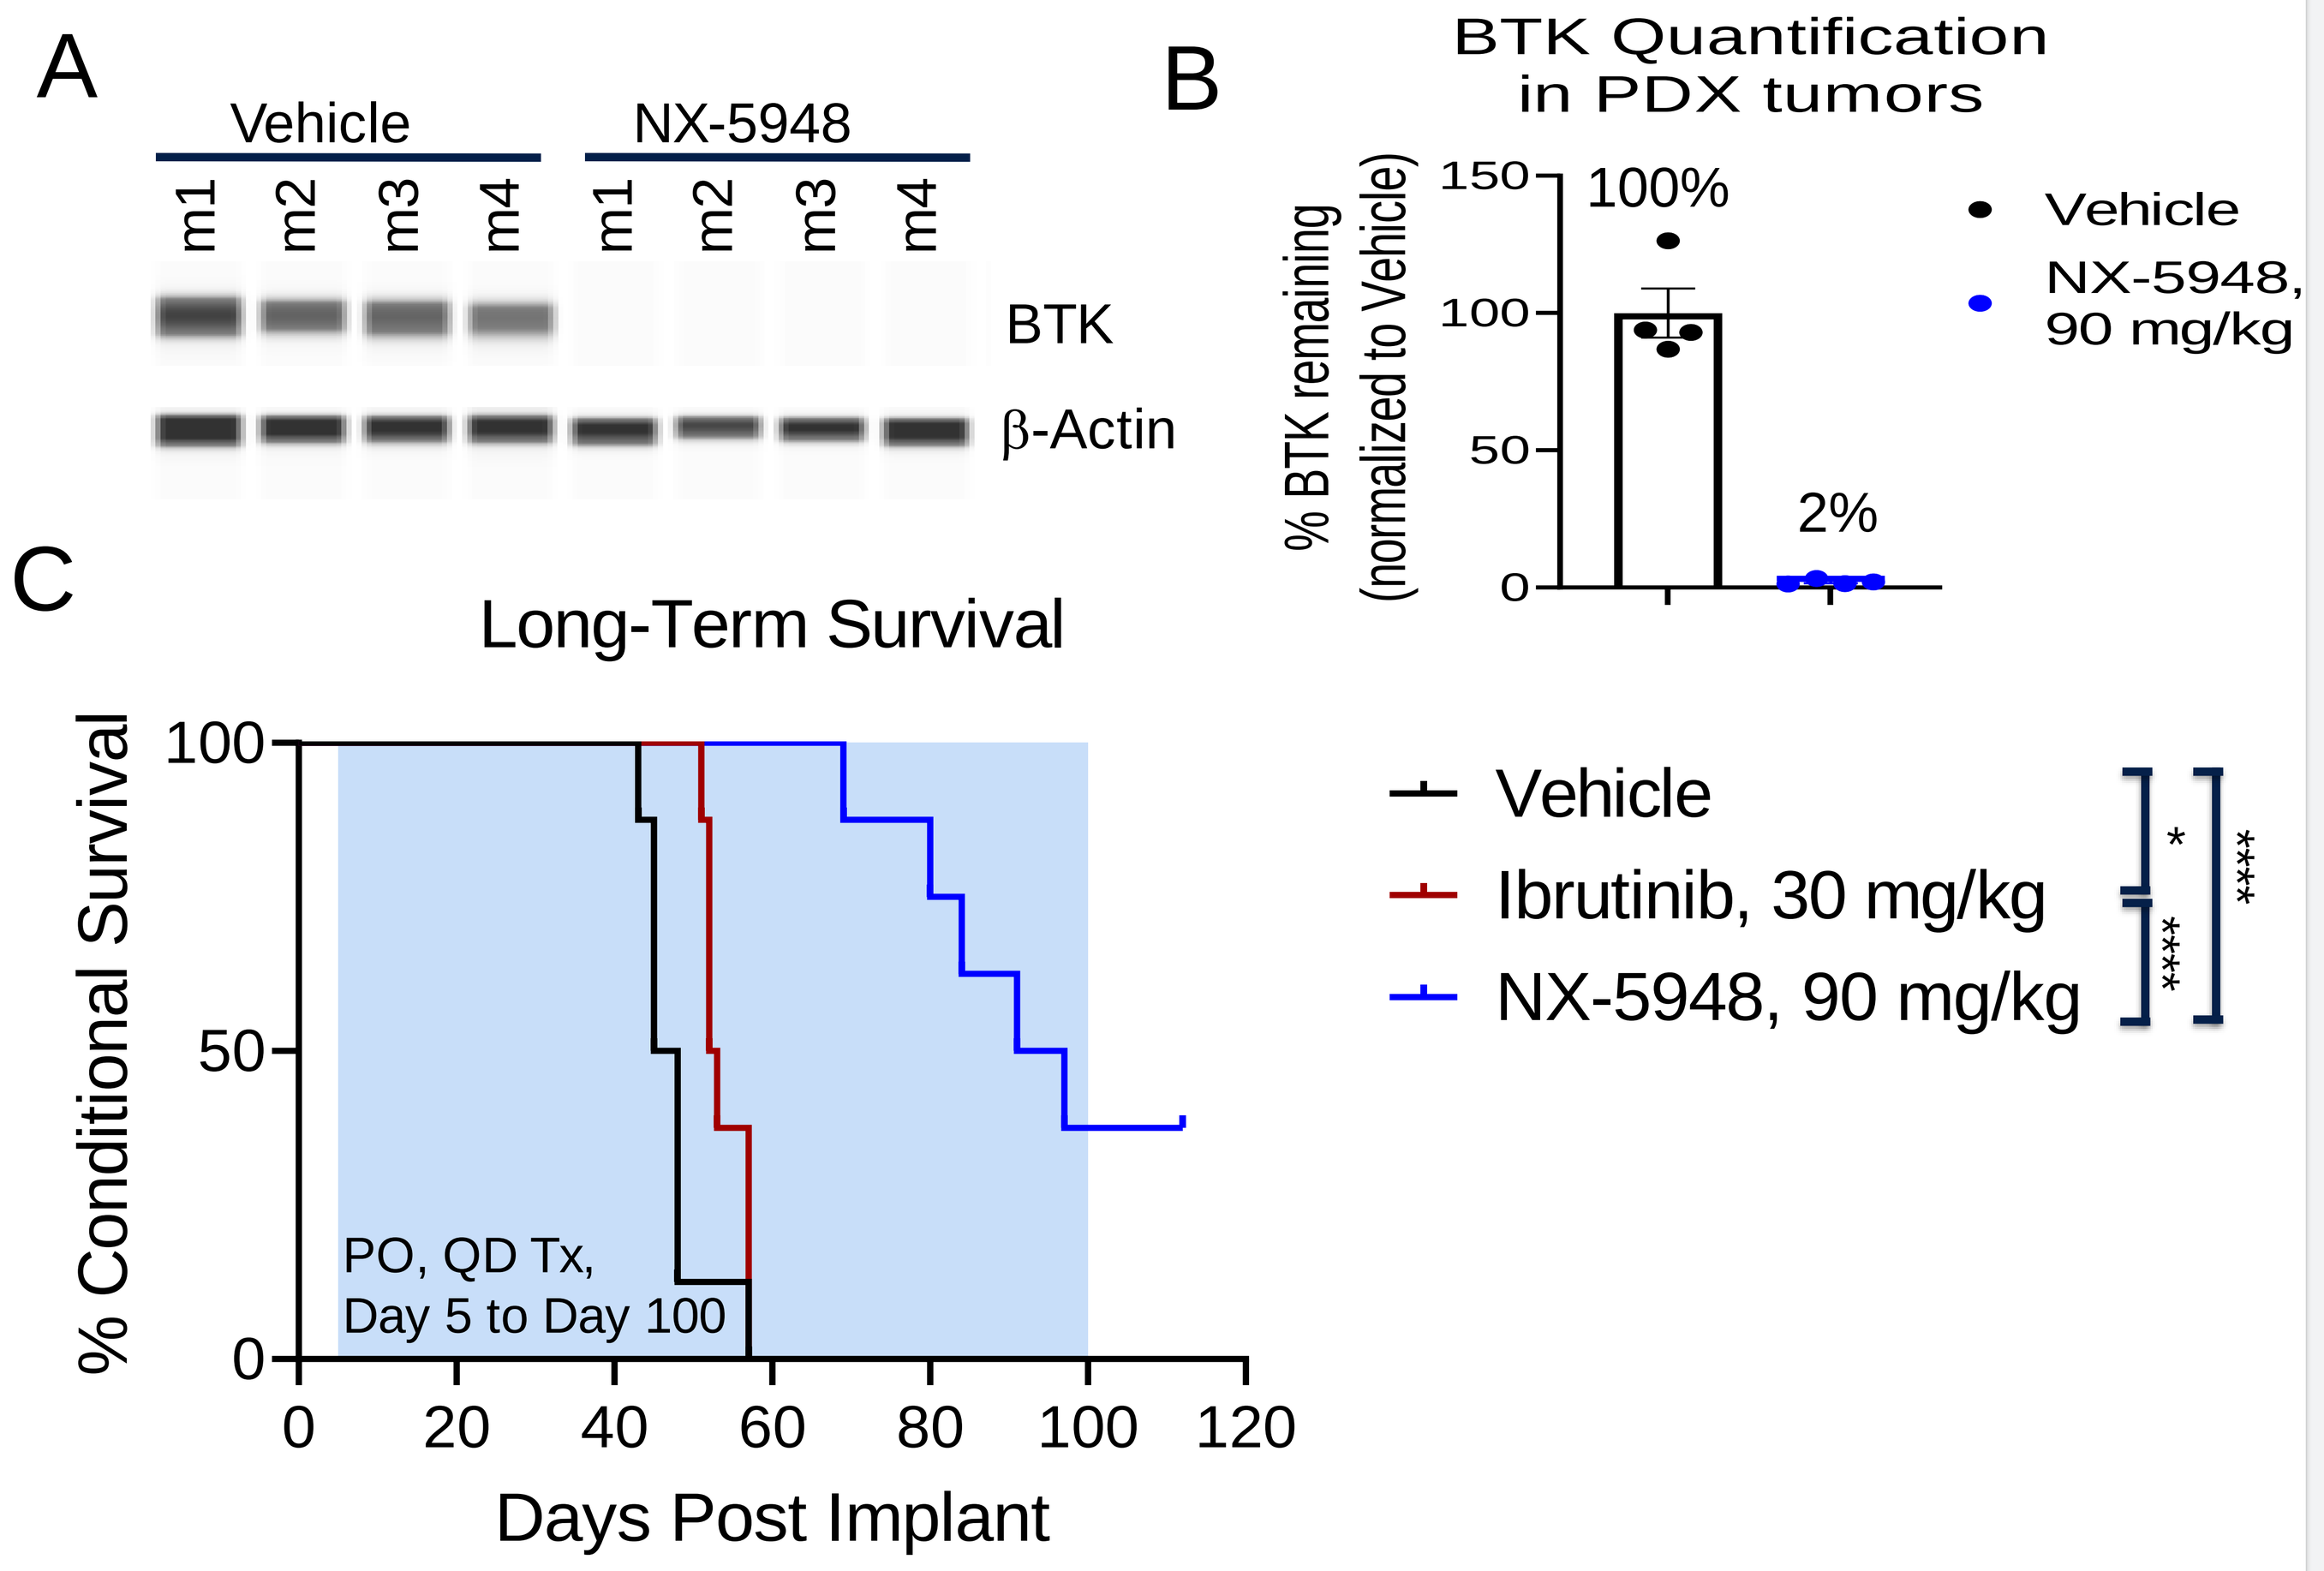

A
Vehicle m1 m2 m3 m4
NX-5948 m1 m2 m3 m4
BTK
β-Actin
B BTK Quantification in PDX tumors
% BTK remaining (normalized to Vehicle)
100%
2%
Vehicle
NX-5948, 90 mg/kg
C Long-Term Survival
% Conditional Survival
PO, QD Tx, Day 5 to Day 100
Days Post Implant
Vehicle
Ibrutinib, 30 mg/kg
NX-5948, 90 mg/kg
*

Figure 5. Daily, oral NX-5948 administration resulted in 98% BTK degradation (A-B), superior anti-tumor activity compared to ibrutinib, and prolonged protection after discontinued treatment (C). (A-B) RAG-/- mice with intraparenchymal implanted SC1 cells were administered either vehicle control or NX-5948 for 3 days and BTK levels were determined 6 hours after the final dose by western blot analysis of human CD19+ cells isolated from mouse brains. (C) PDX mice were treated with daily oral administration of either vehicle, NX-5948, or ibrutinib starting on day 5 post intraparenchymal implantation. Survival was followed for 112 days with NX-5948 treatment discontinued on day 100.

CONCLUSIONS

- NX-5948 is an orally bioavailable bifunctional degrader currently in a Phase 1 clinical trial for treatment of patients with B cell malignancies (NCT05131022).
- Daily oral administration of NX-5948 to mice bearing subcutaneous or intracranial TMD8 ABC-type DLBLC tumors crosses the blood-brain barrier, degrades BTK in tumor cells, provides robust anti-tumor activity, and significantly extends survival.
- The SC1 murine PDX model of secondary CD79b mutant CNS DLBCL expresses markers of ABC DLBCL with evidence of vascular tropism and retains the brain-invasive characteristics of CNS lymphoma.
- Oral administration of NX-5948 once daily to mice bearing intraparenchymal SC1 PDX cells potently degrades BTK in tumor cells and significantly extends survival compared to ibrutinib.
- These preclinical results support the rationale for a Phase 1 study of NX-5948 in relapsed primary CNS lymphoma.

REFERENCES

Evolving Treatments for Primary Central Nervous System Lymphoma. Ferreri AJM, Holdhoff M, Nayak L, Rubenstein JL. American Society for Clinical Oncology Education Book 2019 39:454-466

Protein Degraders Enter the Clinic – a New Approach to Cancer Therapy. Chirnomas, D., Hornberger, K.R., Crews, C.M. Nature Reviews Clinical Oncology Volume 20 265-278 (2023)

Biology of CNS lymphoma and the potential of novel agents. Rubenstein JL. Hematology Am Soc Hematol Educ Program. 2017 (1):556-564.

Pathologic correlates of primary central nervous system lymphoma defined in an orthotopic xenograft model. Kadoch C, Dinca EB, Voicu R, Chen L, Nguyen D, Parikh S, Karrim J, Shuman MA, Lowell CA, Treseler PA, James CD, Rubenstein JL. Clin Cancer Res. 2009;15(6):1989-97.

Use of Labeled Tomato Lectin for Imaging Vasculature Structures. Robertson, R.T., Levine, S.T., Haynes, S.M., Gutierrez, P., Baratta, J.L., Tan, Z., Longmuir, K.J. Histochemistry and Cell Biology 2015 143 (2): 225-34

ACKNOWLEDGEMENTS

Byron Hann, M.D., Ph.D., Veronica Steri, Ph.D. Juan Antonio Camara Serrano, M.D., Ph.D., Paul Phojanakong, Fernando Salangsang, Cigall Kadoch, Ph.D., Hua-Xin Gao, Ph.D.